Strategies for multi-gene editing and reduction of translocations with CRISPR-Cpf1 in T cells for the development of improved cell therapies

P43

J.A. Zuris[1], A. Bothmer[1], R. Viswanathan[1], H.S. Abdulkerim[1], K. Gareau[1], S.M. Winston[1], S.N. Scott[1], J.W. Fang[1], M.S. Chin[1], J.A. DaSilva[1], T. Wang[1], G.M. Gotta[1], C.M. Borges[1], F. Harbinski[1], E. Marco[1], C.J. Wilson[1], G.G. Welstead[1], V.E. Myer[1], C.A. Fernandez[1]

Editas Medicine, Inc. | 11 Hurley Street, Cambridge, MA 02141

Introduction

Gene editing using RNA-guided nuclease technology has gained widespread attention for its potential application to current cell therapies. The CRISPR-Cpf1 system (also known as Cas12a) is complementary to Cas9 with several distinct differences. Cpf1 uses a single ~40 nucleotide crRNA and can target T- and C- rich PAMs with the WT and engineered PAM variants. The expanded targeting space, when compared to the purine rich PAMs of Cas9, makes it an attractive addition to enable broader targeting opportunities. Unlike SpCas9, Cpf1 makes a staggered cut in the DNA leaving behind a 4-5 nucleotide 5'-overhang, which could result in different editing outcomes. We targeted *TRAC*, *B2*M, and *CIITA* in primary human CD4+ and CD8+ T cells with Cpf1 and its engineered RR and RVR PAM variants with these different enzymes delivered as ribonucleoproteins (RNPs). We were able to consistently achieve robust multiplexed (>80% triple KO) gene disruption. To examine the genomic consequences of multiplexed editing, we applied a set of detection technologies including targeted and genome-wide methods to quantitate editing efficiency and genomic rearrangements within and between target loci.

Methods

- Used Cpf1 or SpCas9 to KO single or multiple combinations of *TRAC*, *B2*M, and *CIITA* in a single delivery of RNP to CD4+ and/or CD8+ T cells as measured by flow cytometry and NGS
- Conducted specificity studies on top RNPs using GUIDE-seq, Digenome-seq, and *in silico* analyses followed by NGS
- Applied a set of translocation detection technologies including targeted and genome-wide methods to quantitate editing efficiency and genomic rearrangements within and between target loci
- Developed strategies to reduce translocation frequencies that were observed in a multi-editing context

Results

Multiple Gene Edits Allow for the Generation of an "Off-the-shelf" Allogeneic T Cell Product

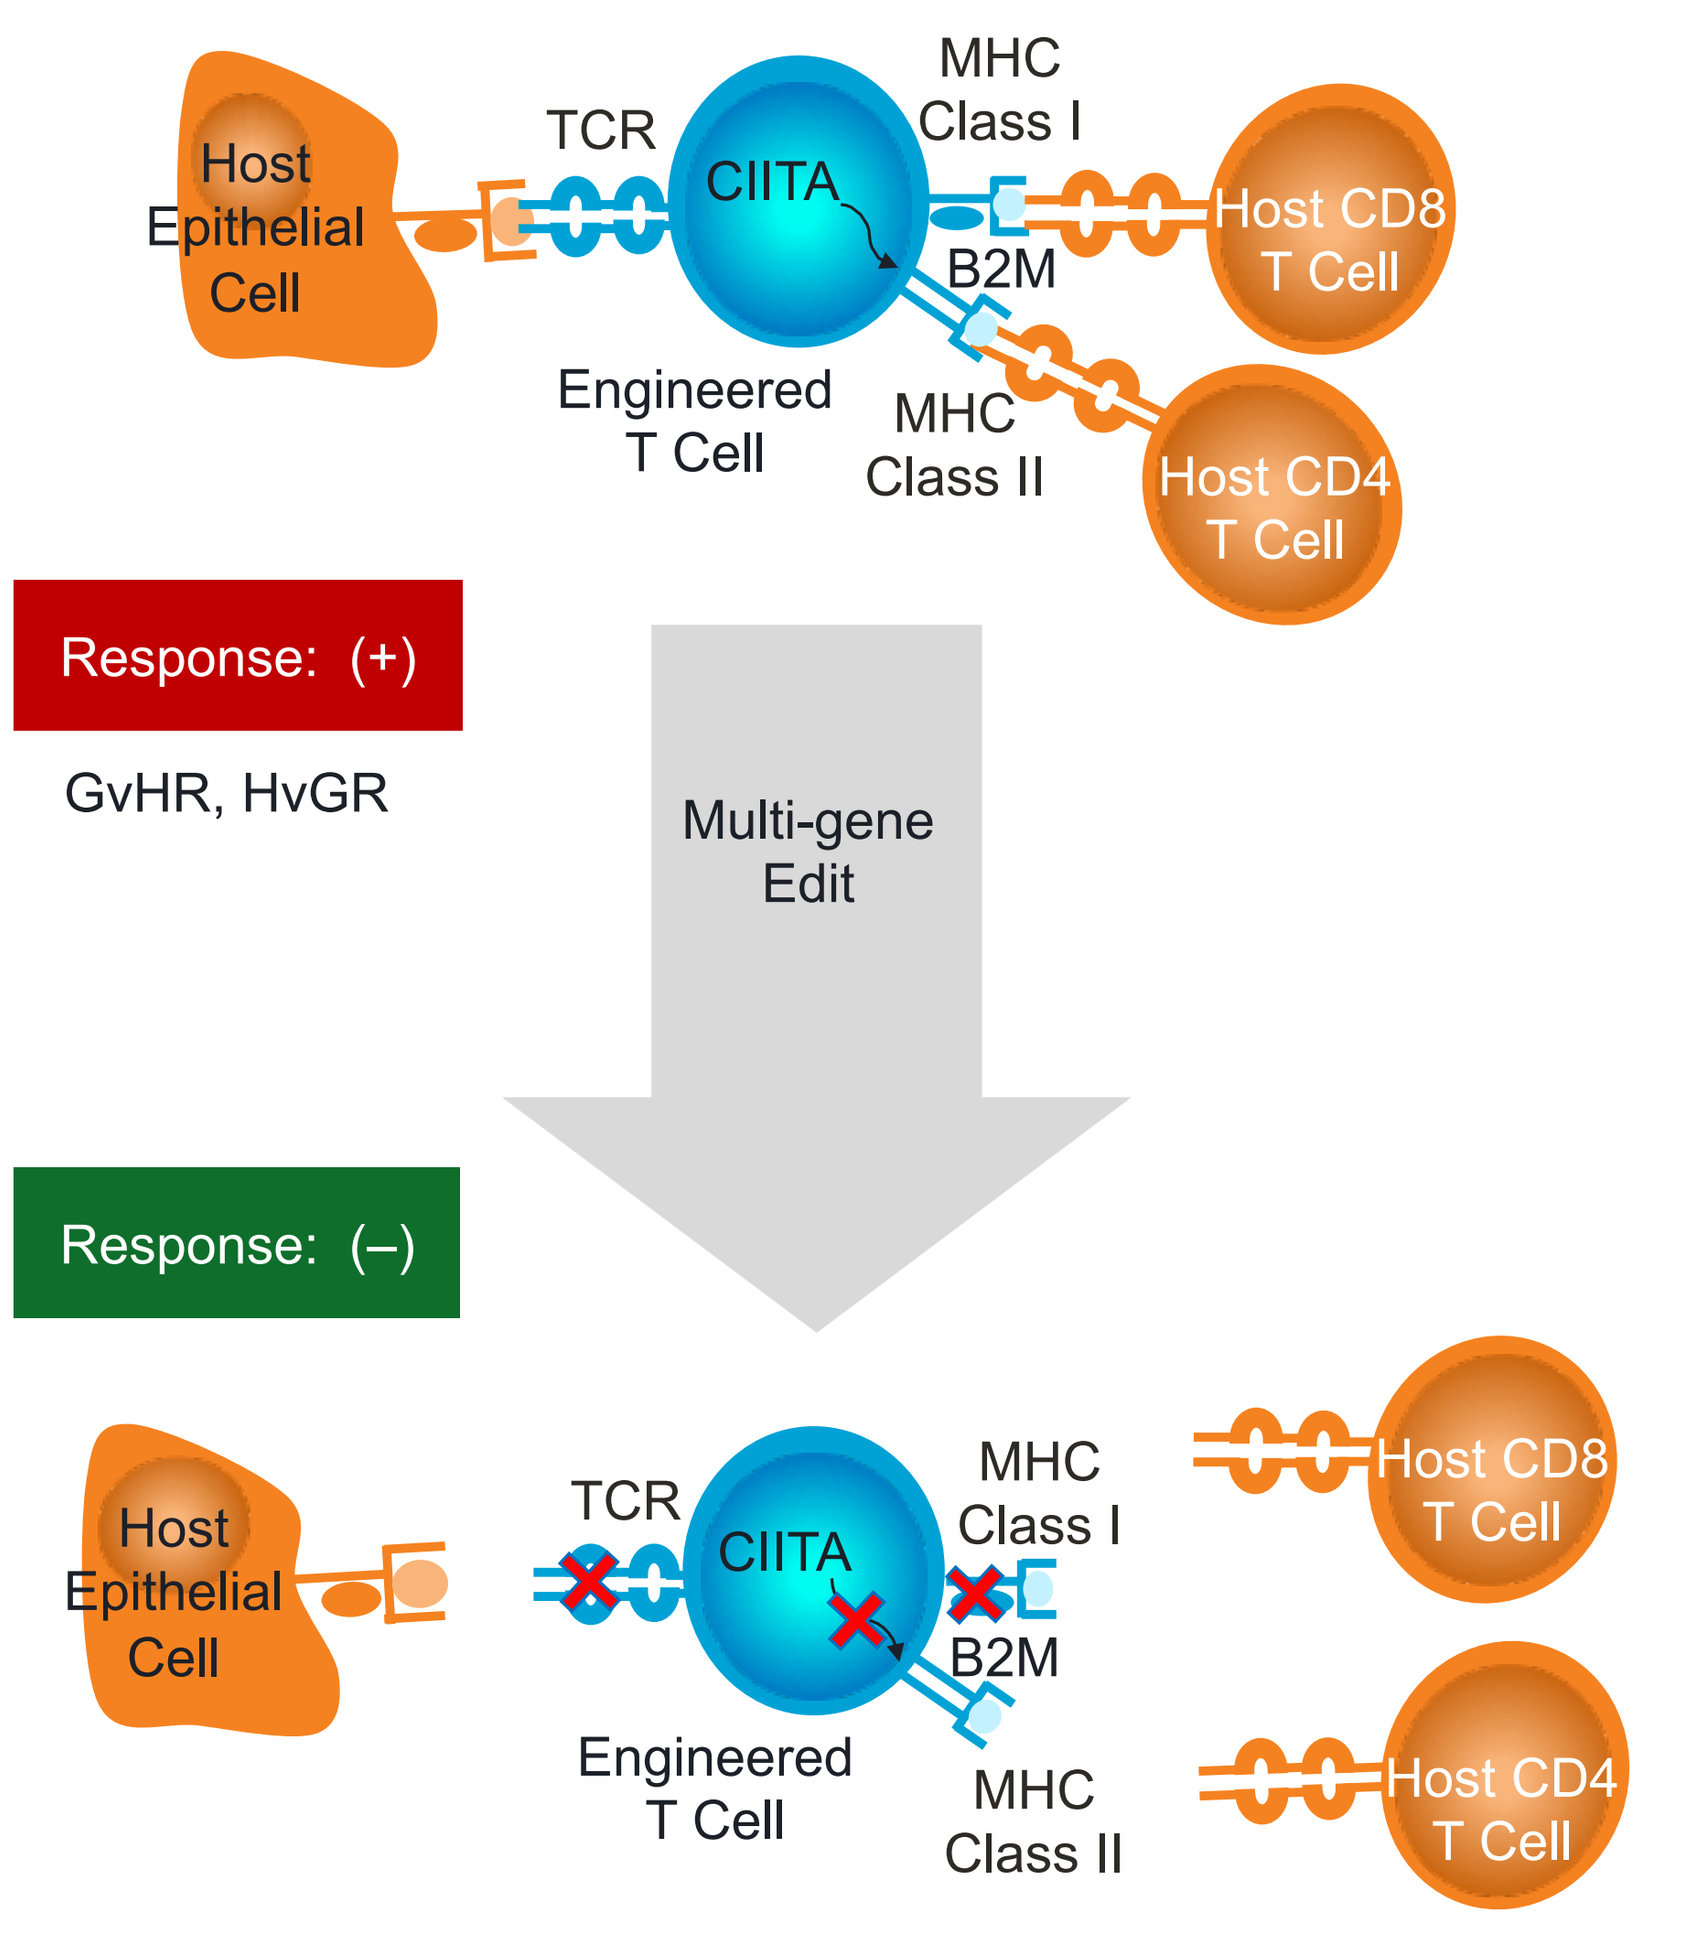
Host Epithelial Cell
TCR
CIITA
MHC Class I
B2M
Host CD8 T Cell
Engineered T Cell
MHC Class II
Host CD4 T Cell
Response: (+)
GvHR, HvGR
Multi-gene Edit
Response: (–)

Cpf1 Offers Several Potentially Advantageous Features for the Genome Editing Tool Kit

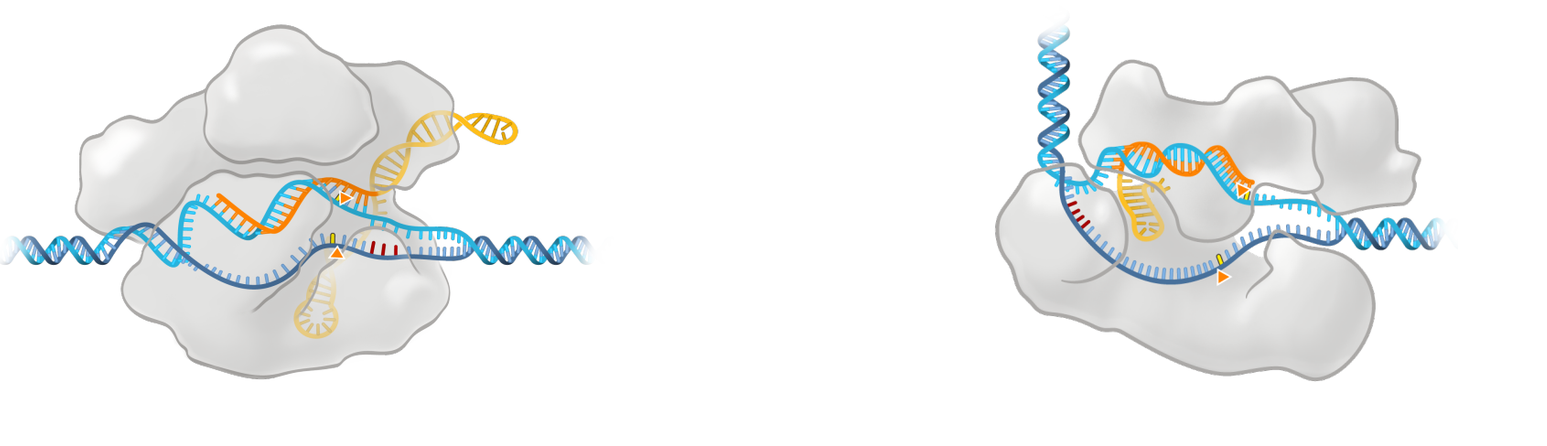
Cas9
Cpf1 (Cas12a)

Cas9	Cpf1 (Cas12a)
Targets G-rich PAMs	Targets T- and C-rich PAMs
Separate crRNA and trRNA that can be linked (~100 nt)	Naturally occurring ~40 nt single guide RNA
Predominantly blunt DNA cut or 1 nt overhang	5' staggered DNA cut with 4 nt overhangs

Variant	PAM	Frequency (bp)
SpCas9	NGG	1 in 8
SaCas9	NNGRRT	1 in 32
SaCas9 KKH	NNNRRT	1 in 8
AsCpf1	TTTV	1 in 43
AsCpf1 RR	TYCV/CCCC	1 in 18
AsCpf1 RVR	TATV	1 in 43

Single KO Efficiency Comparable to SpCas9 with a Cpf1 RNP for Each Allo Target

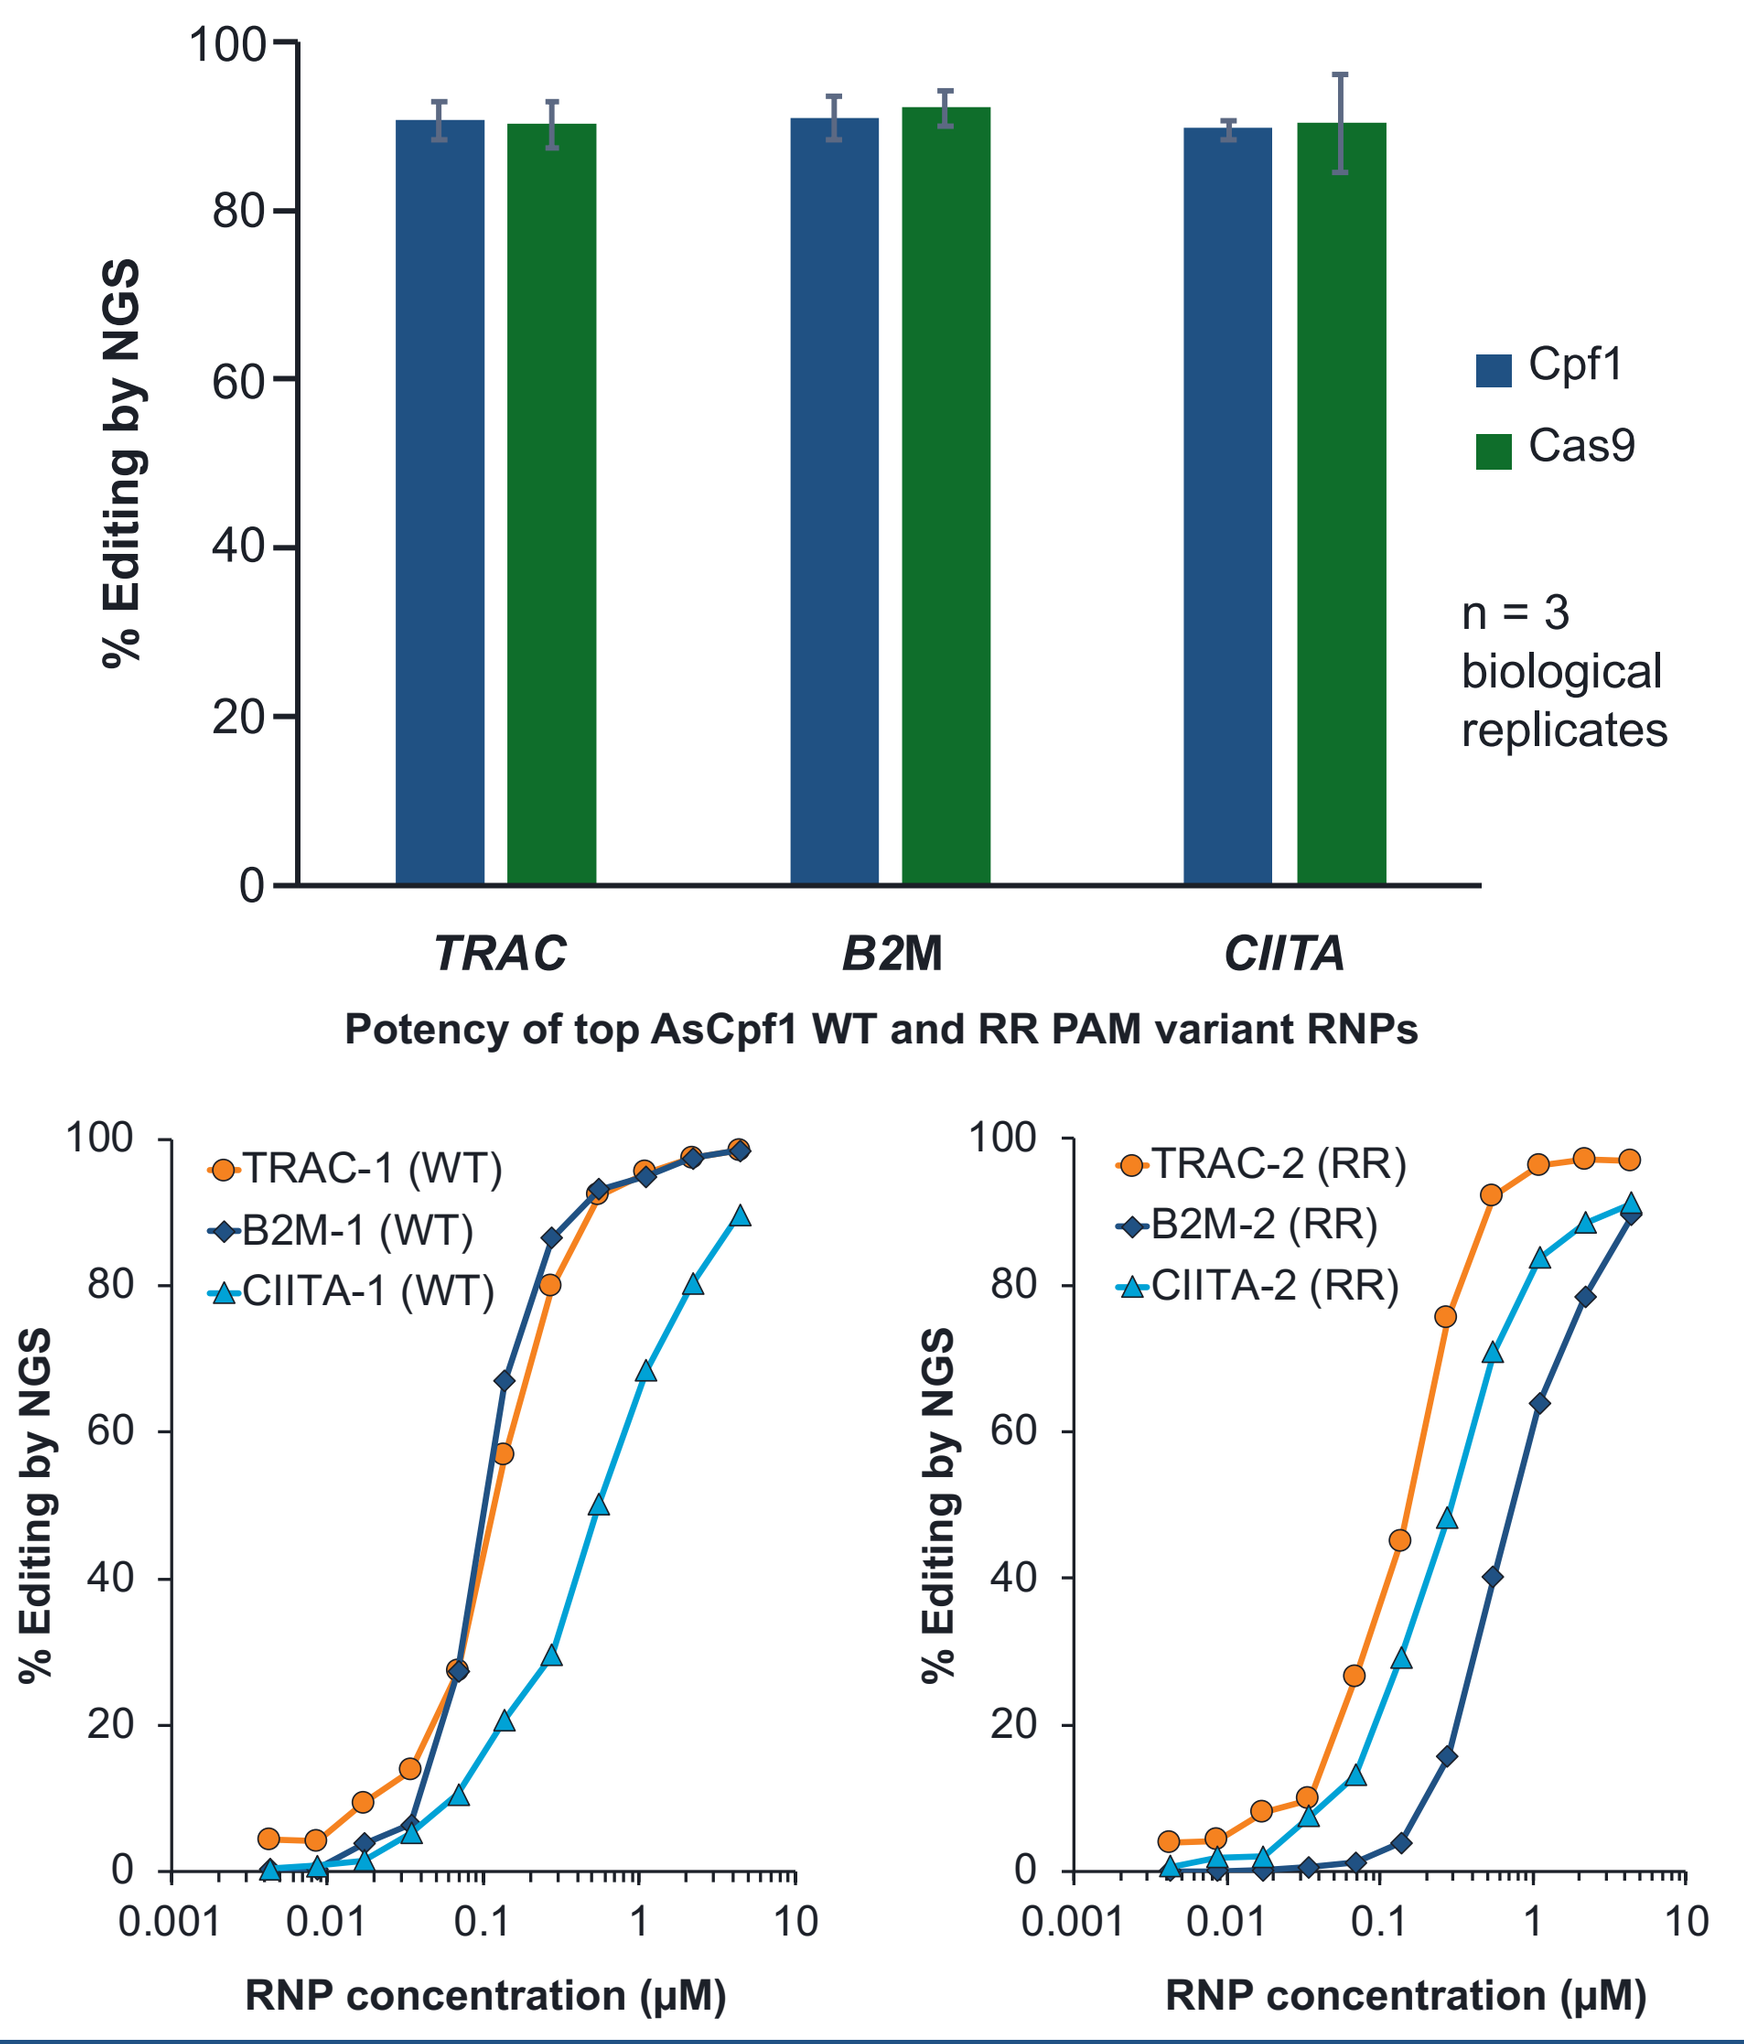
% Editing by NGS
TRAC
B2M
CIITA
Cpf1
Cas9
n = 3 biological replicates
Potency of top AsCpf1 WT and RR PAM variant RNPs
TRAC-1 (WT)
B2M-1 (WT)
CIITA-1 (WT)
TRAC-2 (RR)
B2M-2 (RR)
CIITA-2 (RR)
RNP concentration (µM)

Top Cpf1 RNPs Appear to be Specific by Multiple Orthogonal Specificity Assays

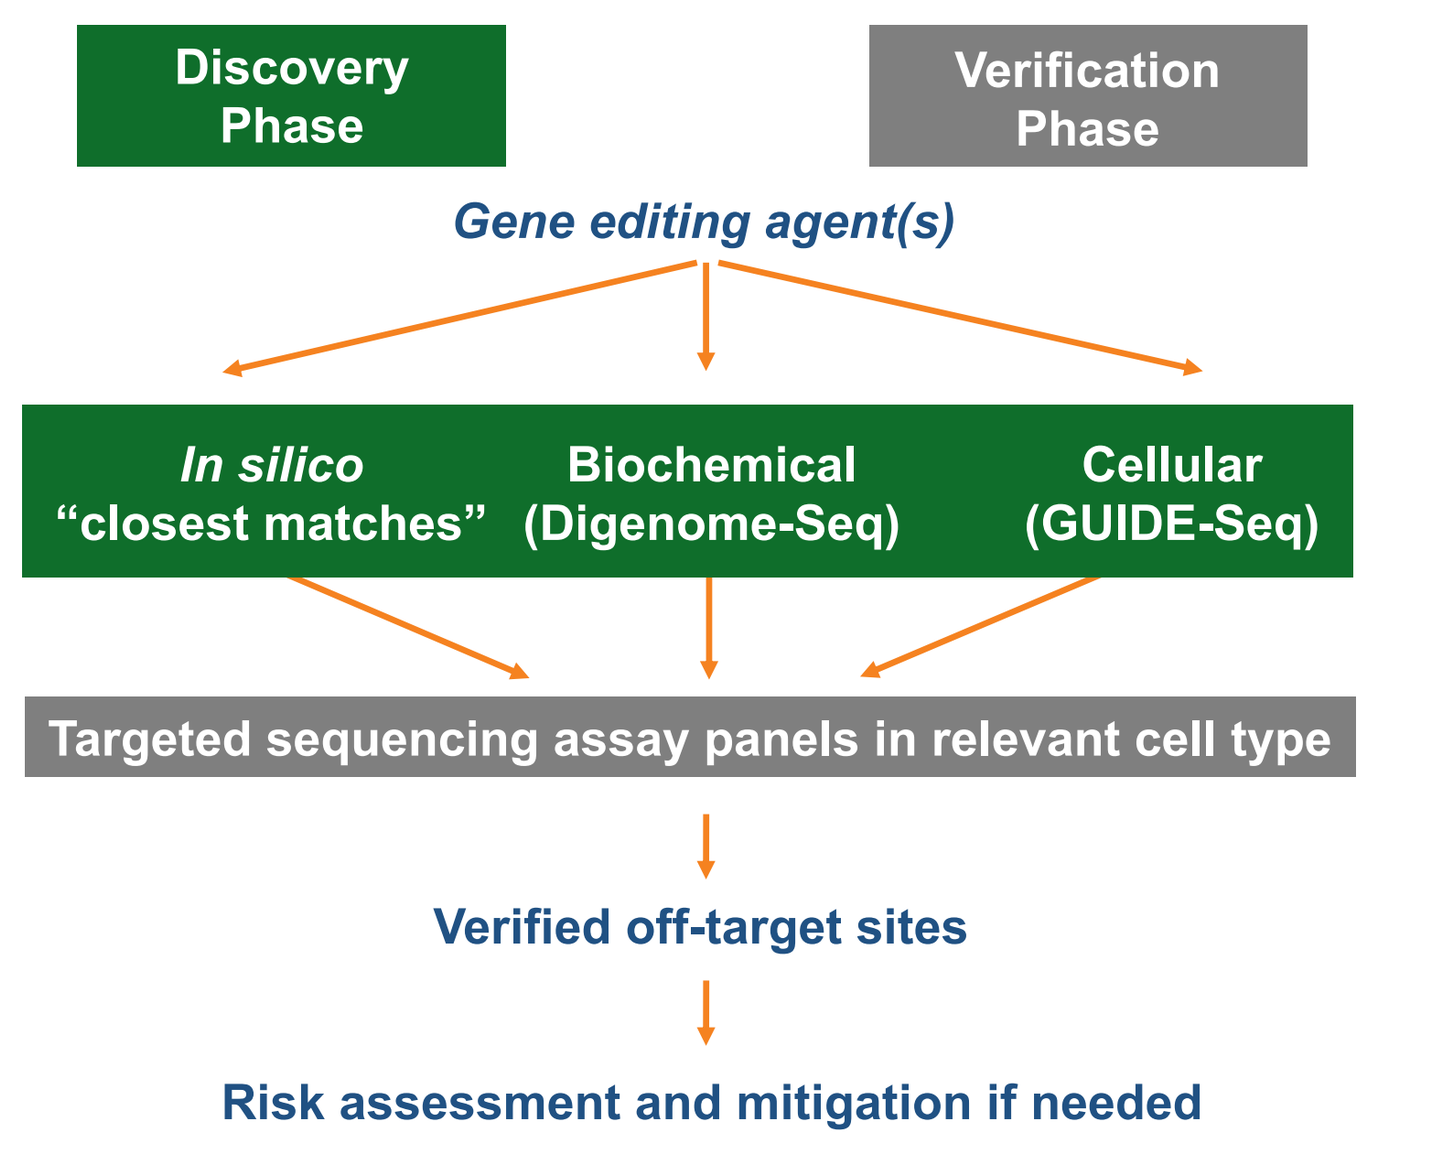
Discovery Phase
Verification Phase
Gene editing agent(s)
In silico "closest matches"
Biochemical (Digenome-Seq)
Cellular (GUIDE-Seq)
Targeted sequencing assay panels in relevant cell type
Verified off-target sites
Risk assessment and mitigation if needed

Guide (variant)	% Editing in T cells	*In silico* (off-by 1, 2, 3)	GUIDE-seq off-targets	Digenome-seq off-targets	Amp-seq verified off-targets
B2M-1 (WT)	98.3	0, 1, 23	0	6	0
B2M-2 (RR)	90.0	0, 0, 13	0	0	0
TRAC-1 (WT)	94.5	0, 2, 14	0	0	0
TRAC-2 (RR)	97.8	0, 2, 18	0	9	0
CIITA-1 (WT)	89.7	0, 1, 10	0	14	0
CIITA-2 (RR)	91.3	0, 2, 21	0	2	0
Control (SpCas9)	93.0	1, 4, 39	12	98	2

Efficient Multi-Gene KO in T cells with Cpf1 When Using a Single Delivery of Three RNPs

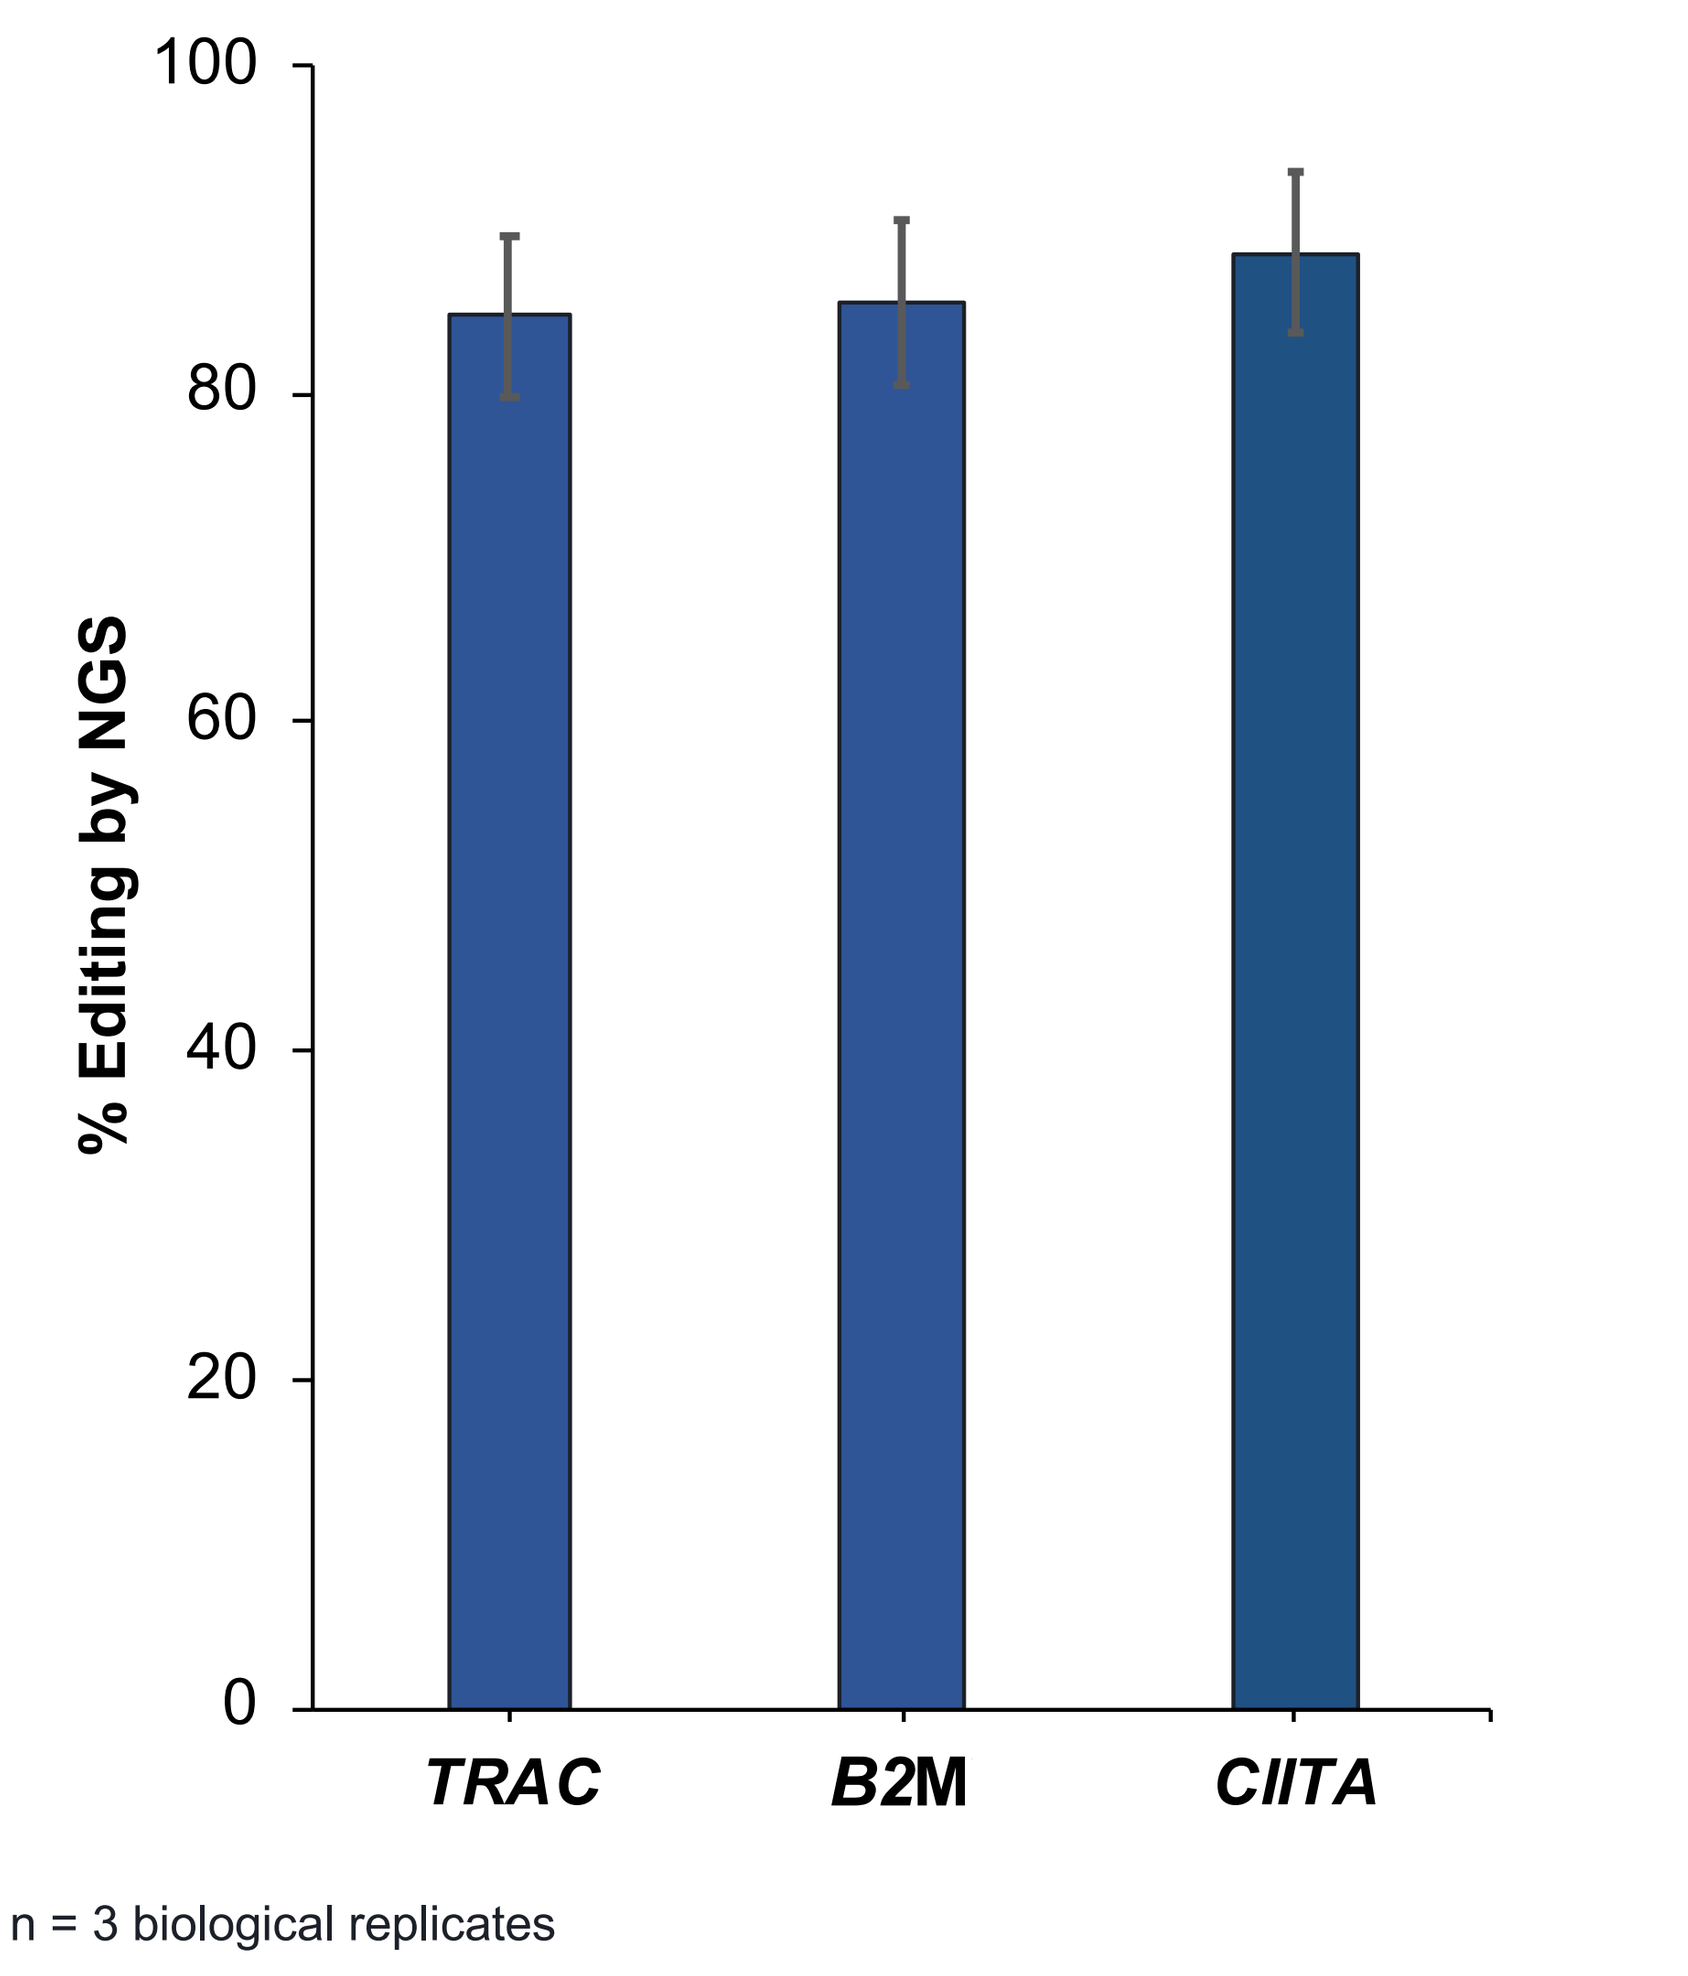
% Editing by NGS
TRAC
B2M
CIITA

n = 3 biological replicates

Translocation Detection in the Context of Multiplexing with CRISPR Nucleases

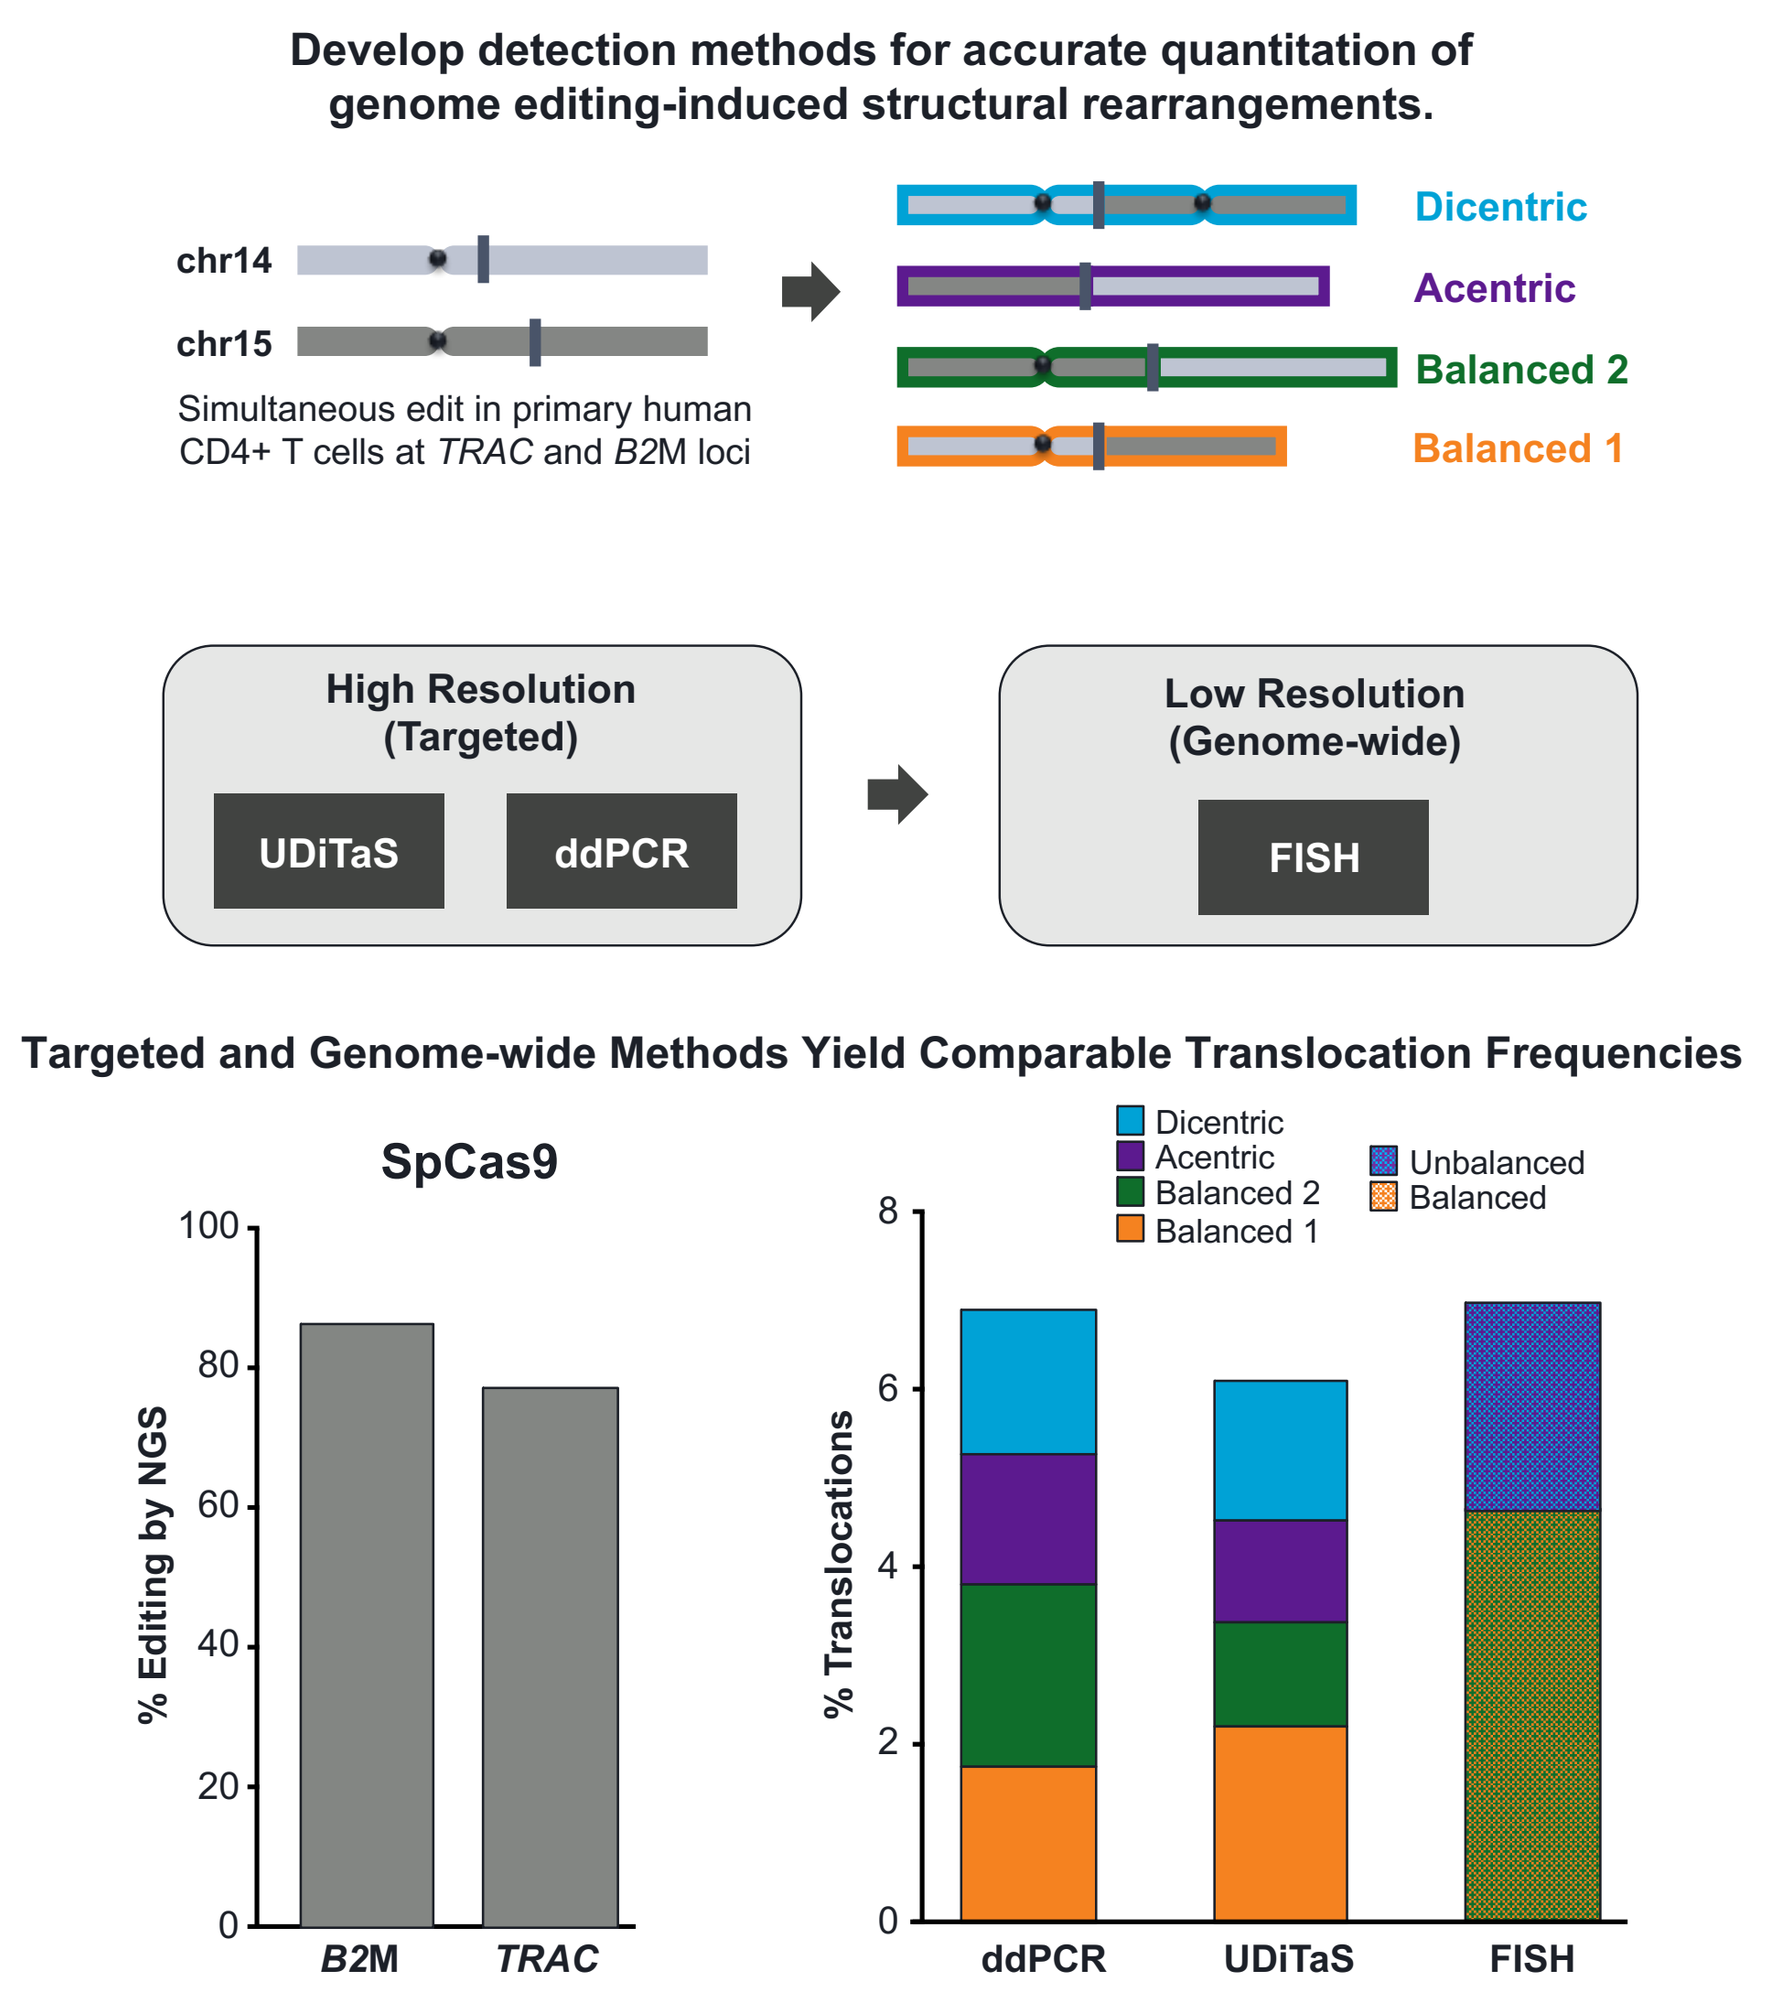
Develop detection methods for accurate quantitation of genome editing-induced structural rearrangements.
chr14
chr15
Simultaneous edit in primary human CD4+ T cells at TRAC and B2M loci
Dicentric
Acentric
Balanced 2
Balanced 1
High Resolution (Targeted)
UDiTaS
ddPCR
Low Resolution (Genome-wide)
FISH
Targeted and Genome-wide Methods Yield Comparable Translocation Frequencies
SpCas9
% Editing by NGS
% Translocations
Unbalanced
Balanced

Translocation Rates Follow Editing Rates for Both On-Target and Off-Target Edits

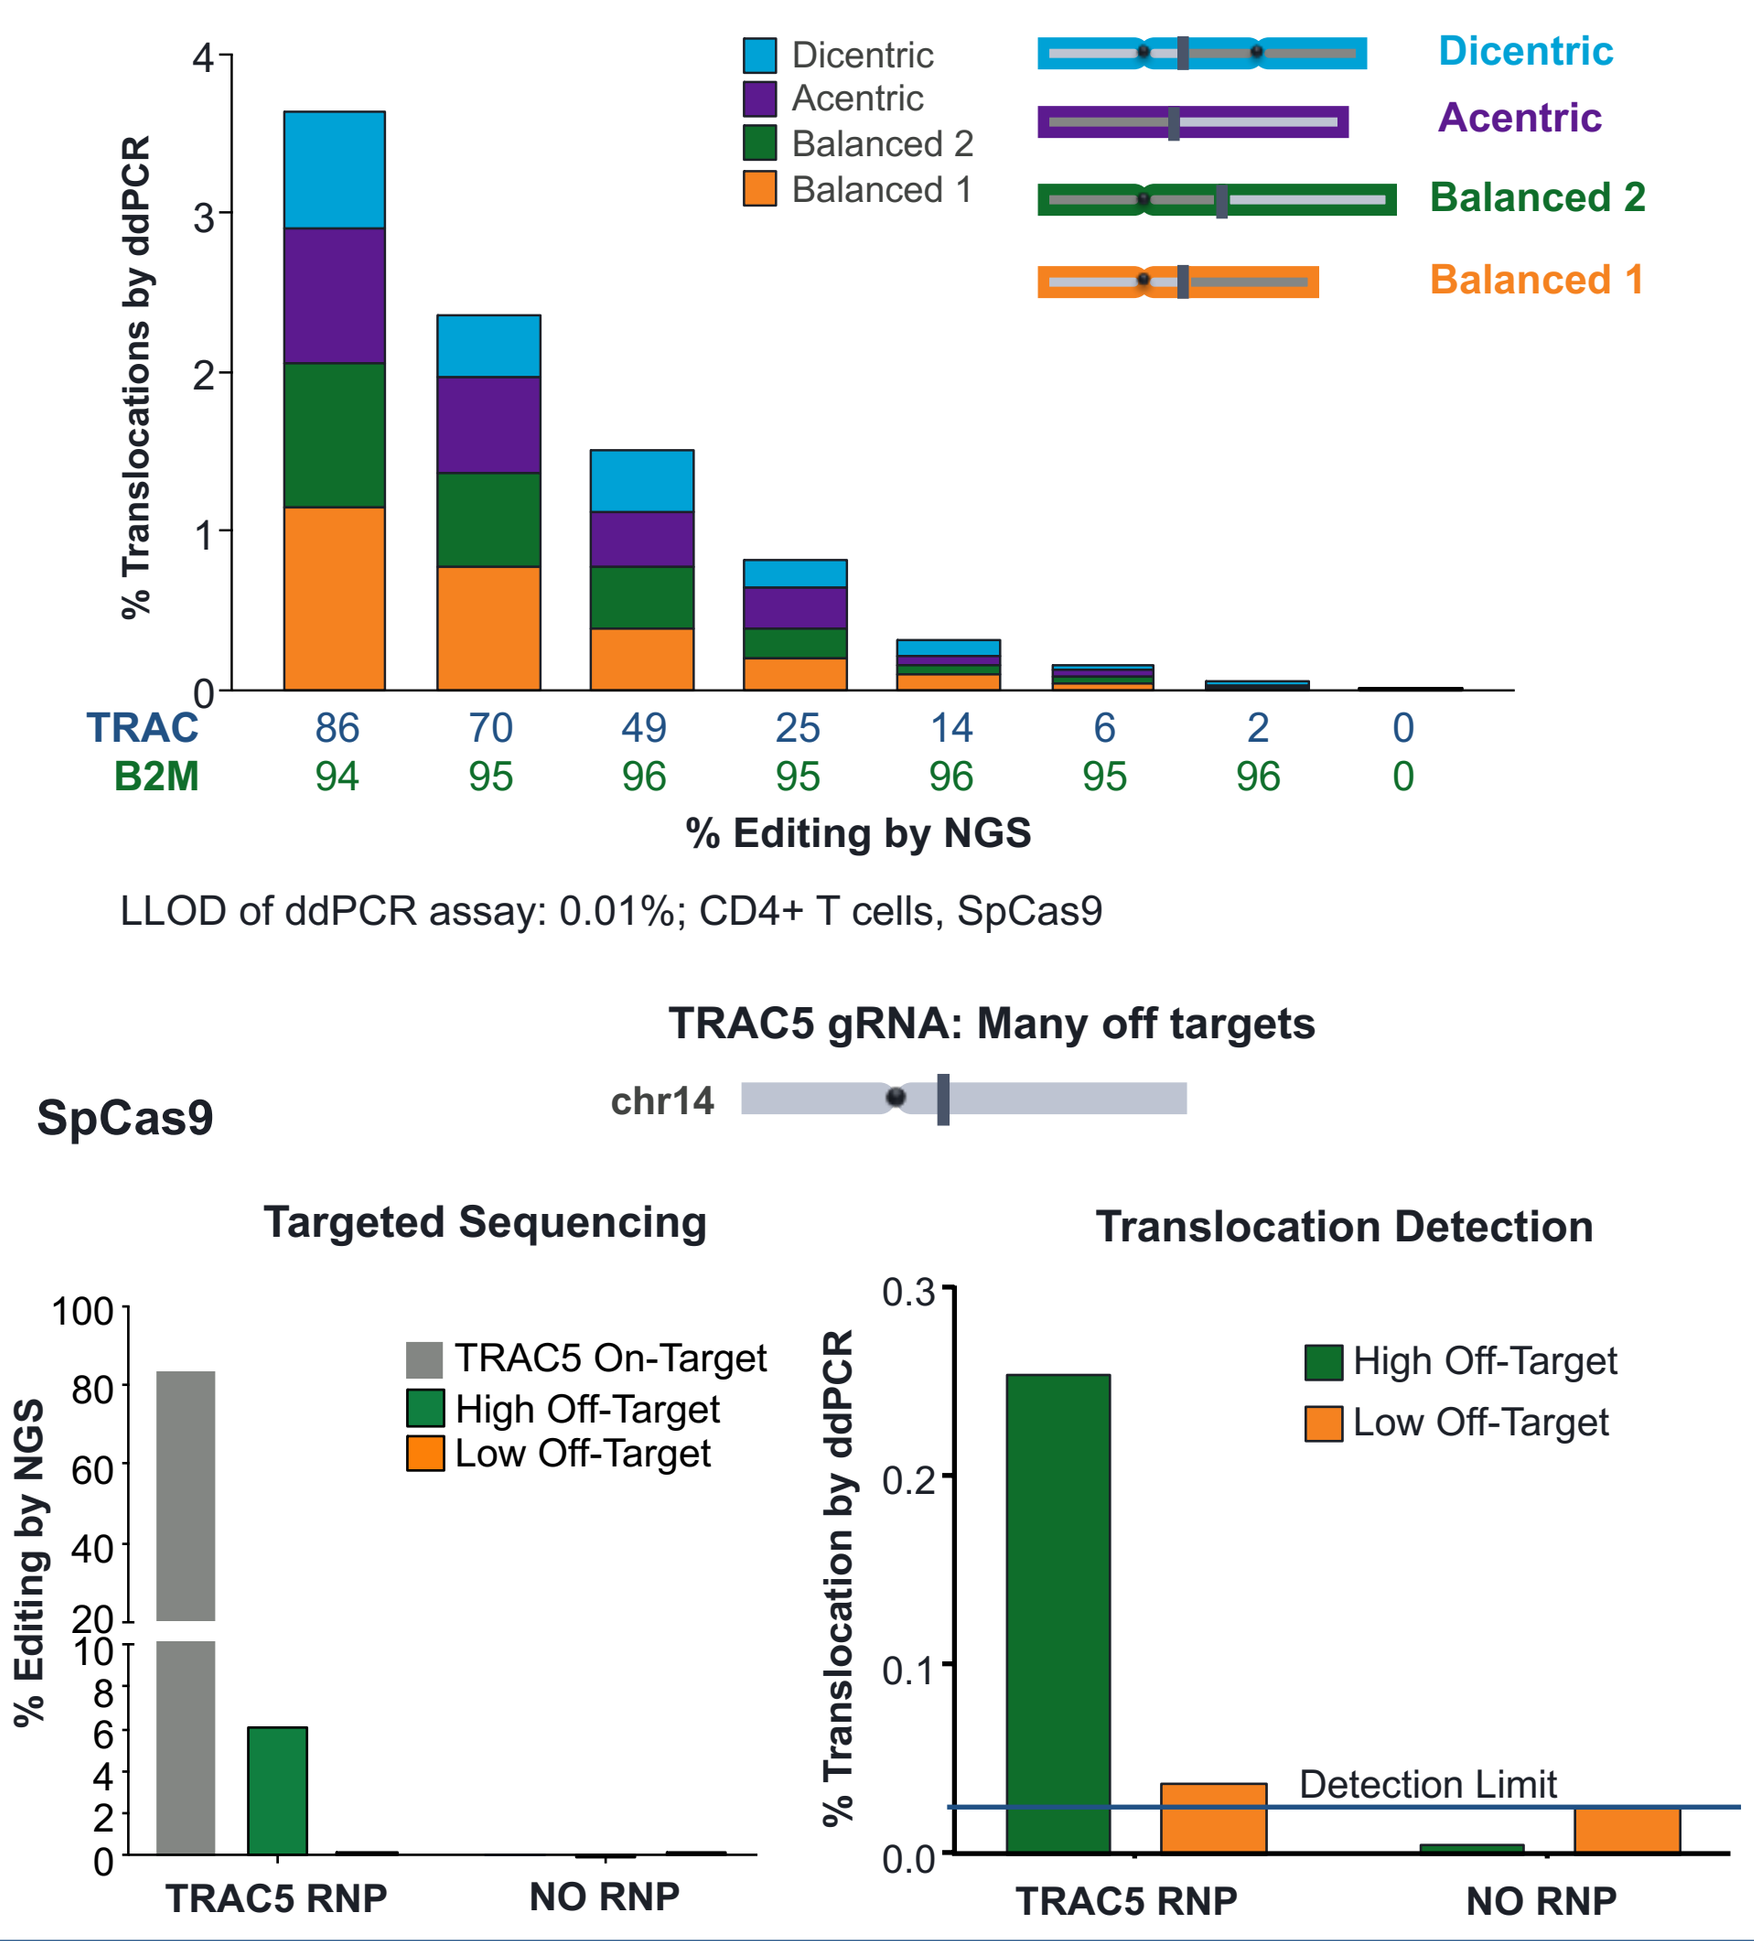
SpCas9
Titrated down TRAC RNP while holding B2M RNP constant
% Translocations by ddPCR
Dicentric
Acentric
Balanced 2
Balanced 1
TRAC 86 70 49 25 14 6 2 0
B2M 94 95 96 95 96 95 96 0
% Editing by NGS
LLOD of ddPCR assay: 0.01%; CD4+ T cells, SpCas9
TRAC5 gRNA: Many off targets
chr14
Targeted Sequencing
Translocation Detection
TRAC5 On-Target
High Off-Target
Low Off-Target
% Translocation by ddPCR
Detection Limit
TRAC5 RNP
NO RNP

Balanced Translocations Persist While Unbalanced Translocations Do Not

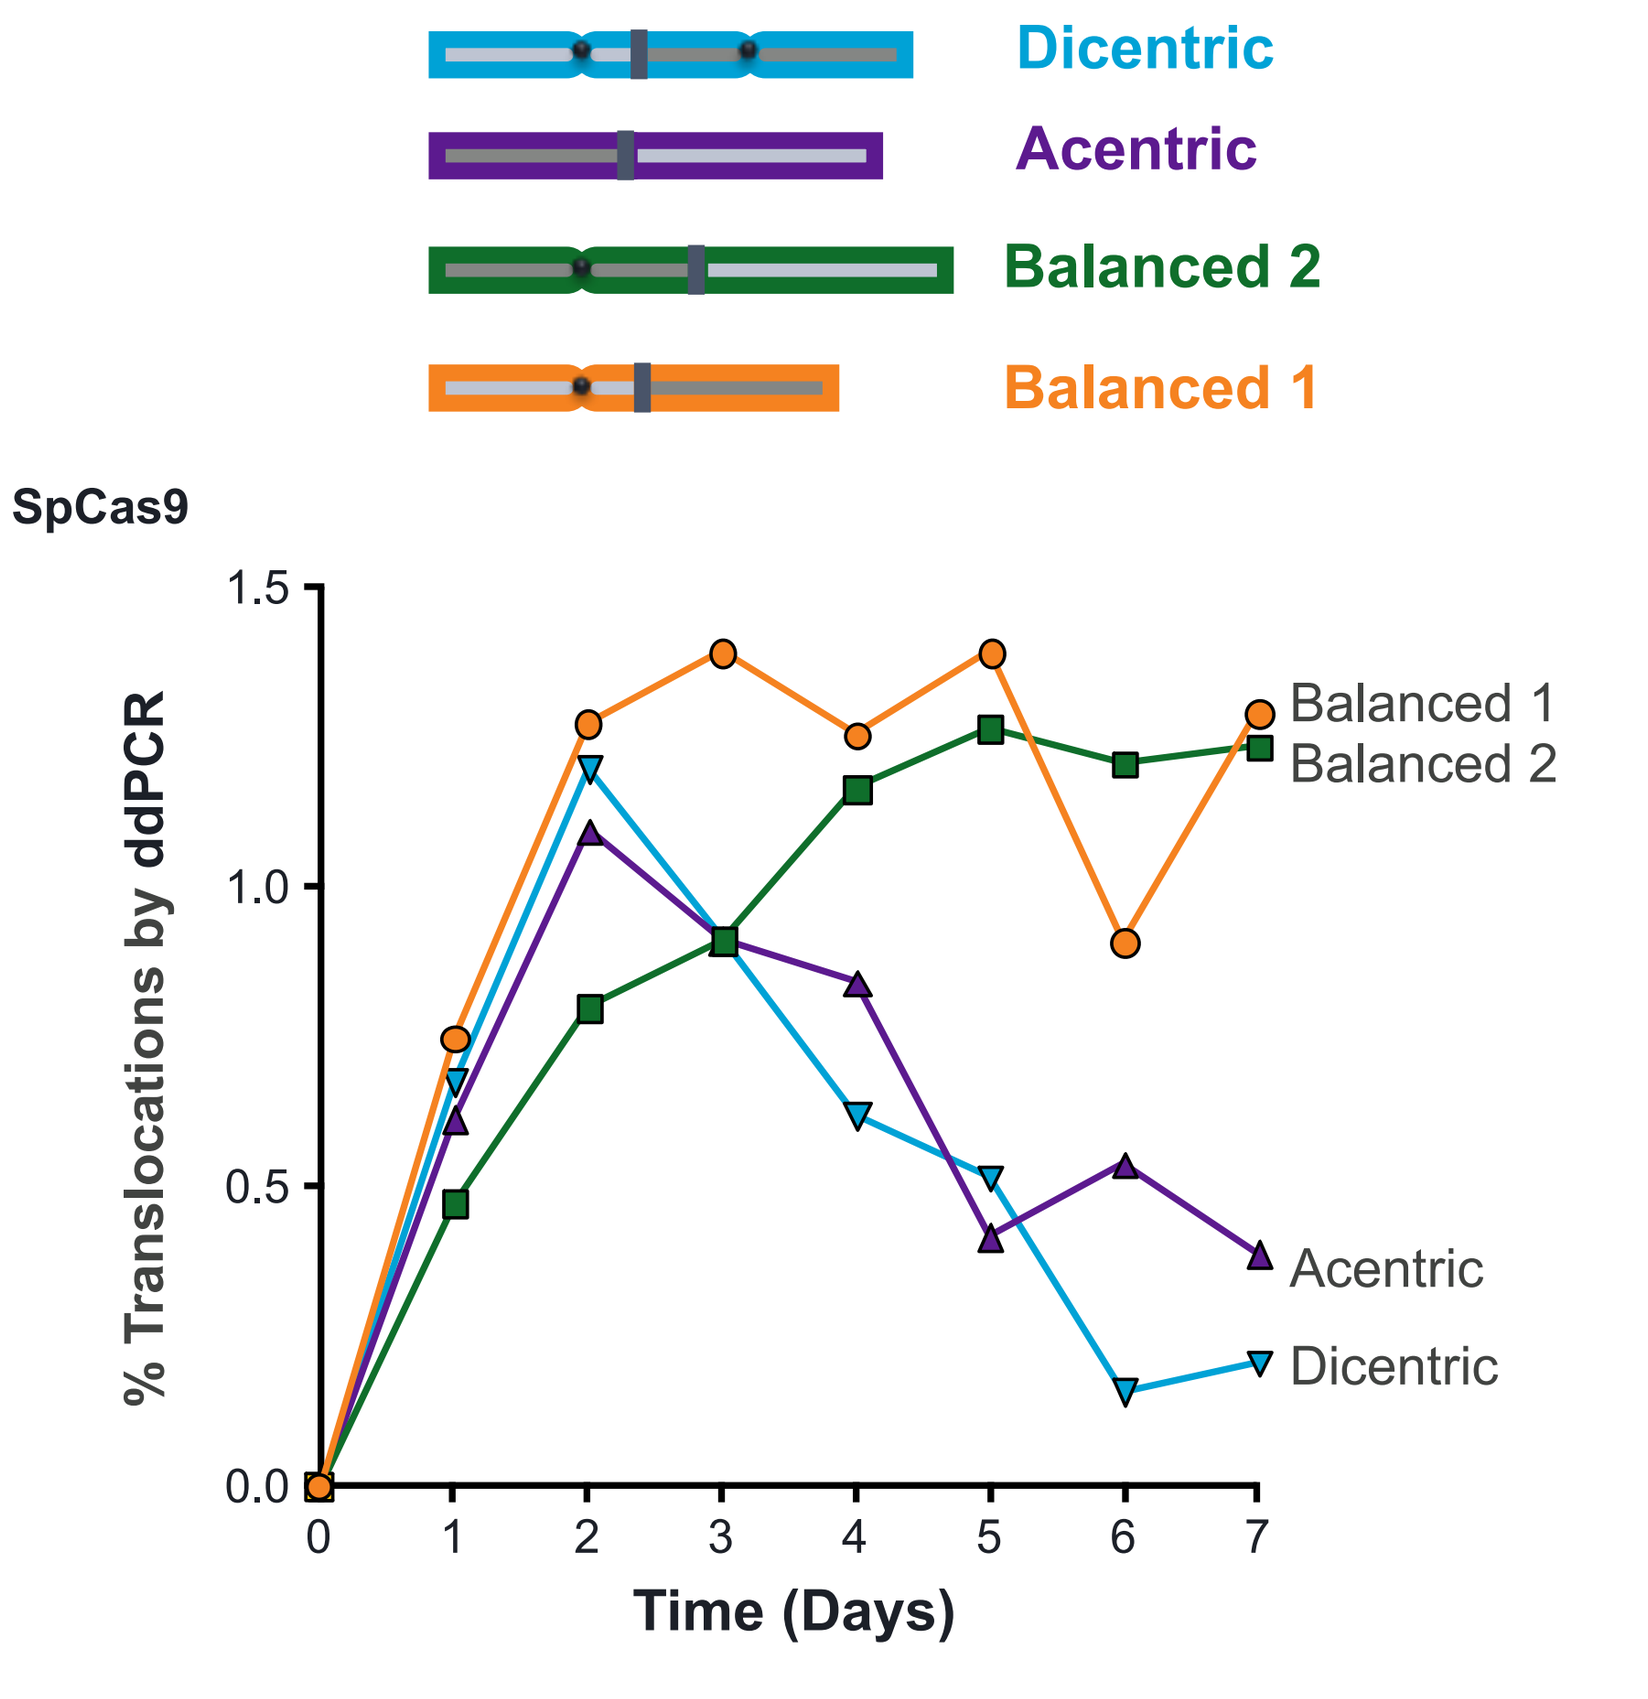
Dicentric
Acentric
Balanced 2
Balanced 1
SpCas9
% Translocations by ddPCR
Time (Days)

CD4+ Human Primary T Cells, B2M and TRAC RNPs, SpCas9

Strategies to Reduce Translocations Using Cpf1 in a Multi-gene Edit

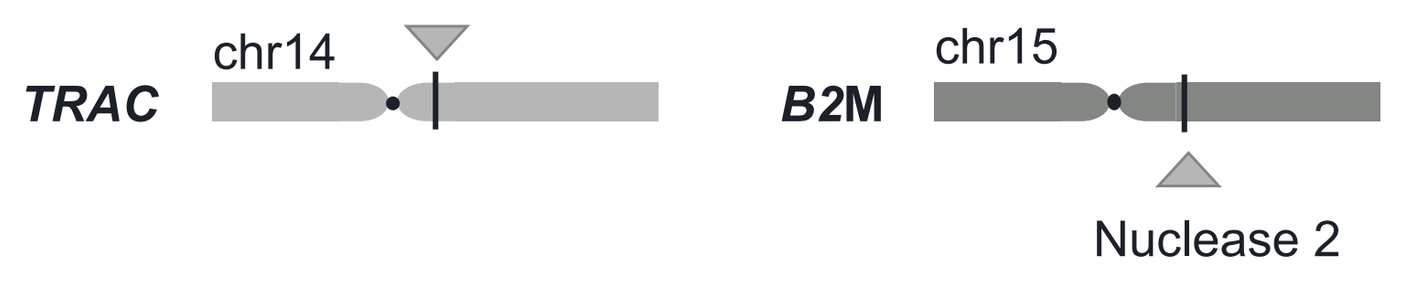
Nuclease 1
TRAC
chr14
B2M
chr15
Nuclease 2

On Target Editing (% by NGS)

Locus	Enzyme: *B2M*	Enzyme: *TRAC*	Percent
TRAC	SpCas9	SpCas9	89
	AsCpf1	AsCpf1	93
	SpCas9	AsCpf1 RR	96
	AsCpf1	SpCas9	91
	AsCpf1	AsCpf1 RR	96

On Target Editing (% by NGS)

Locus	Enzyme: *B2M*	Enzyme: *TRAC*	Percent
B2M	SpCas9	SpCas9	95
	AsCpf1	AsCpf1	92
	SpCas9	AsCpf1 RR	96
	AsCpf1	SpCas9	89
	AsCpf1	AsCpf1 RR	93

Translocation Frequency with Different CRISPR Nuclease Combinations

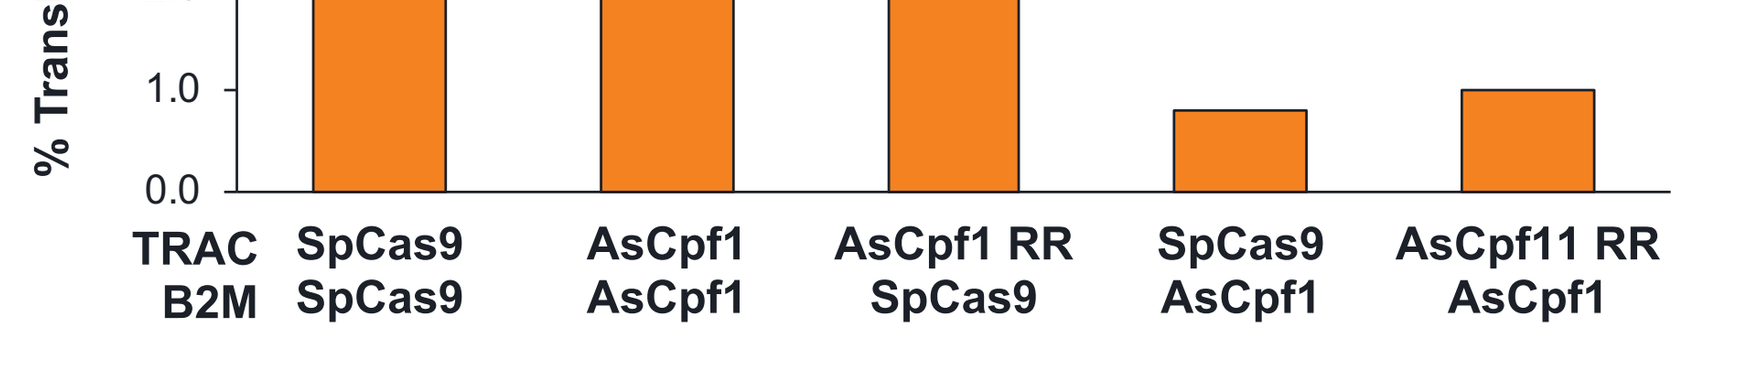
% Translocations by ddPCR
TRAC SpCas9 AsCpf1 AsCpf1 RR SpCas9 AsCpf11 RR
B2M SpCas9 AsCpf1 SpCas9 AsCpf1 AsCpf1

Conclusions

- Multi-gene editing of T cell loci with either the CRISPR-Cas9 or CRISPR-Cpf1 system resulted in translocations which were detected by multiple orthogonal methods. These translocations were dependent on editing rates and persisted over time.
- Multi-gene editing with a CRISPR-Cas9/CRISPR-Cpf1 or CRISPR-Cpf1 with engineered Cpf1 PAM variants reduced translocation frequencies compared to multiplexing with only CRISPR-Cas9.
- For the development of T cell-based medicines, these data suggest that CRISPR-Cpf1 is both robust, specific, and capable of reducing genomic rearrangements when making multiple gene edits compared to the CRISPR-Cas9 system alone.

Acknowledgements

We would like to thank Cecilia Cotta-Ramusino, Sequencing, Screening, Bioinformatics, DNA Damage Response, Computational Biology, Regulators, and Scientific Communication teams for their valuable contributions to this project. Graphic design support was provided by Robert Brown.

Disclosures:
Employees and shareholders of Editas Medicine: J.A.Z., A.B., R.V., H.S.A., K.G., S.N.S., J.W.F., M.S.C., J.A.D., T.W., G.M.G., C.M.B., E.M., C.J.W., G.G.W., V.E.M., C.A.F.

Former employee(s) of Editas Medicine: F.H.

editasmedicine.com
© 2019 Editas Medicine
11 Hurley Street | Cambridge, MA 02141