Staphyloccocus aureus Cas9:

an alternative Cas9 for genome editing applications

A.E. Friedland, A. Sousa, M. Collins, M.L. Maeder, G.G. Welstead, H. Jayaram, S. Gloskowski, D. Bumcrot

Editas Medicine, Cambridge, MA 02142

Summary

S. aureus Cas9:

- Is encoded by 3159 nucleotides, compared to 4104 nucleotides for *S. pyogenes* Cas9.
- Recognizes an NNGRRT PAM.
- Cleaves dsDNA with efficiency comparable to *S. pyogenes* Cas9.
- Cleaves dsDNA with gRNAs of different spacer lengths, with shorter guides generally less tolerant of sequence mismatches.

S. aureus Cas9 cleavage efficiency is comparable to *S. pyogenes* Cas9

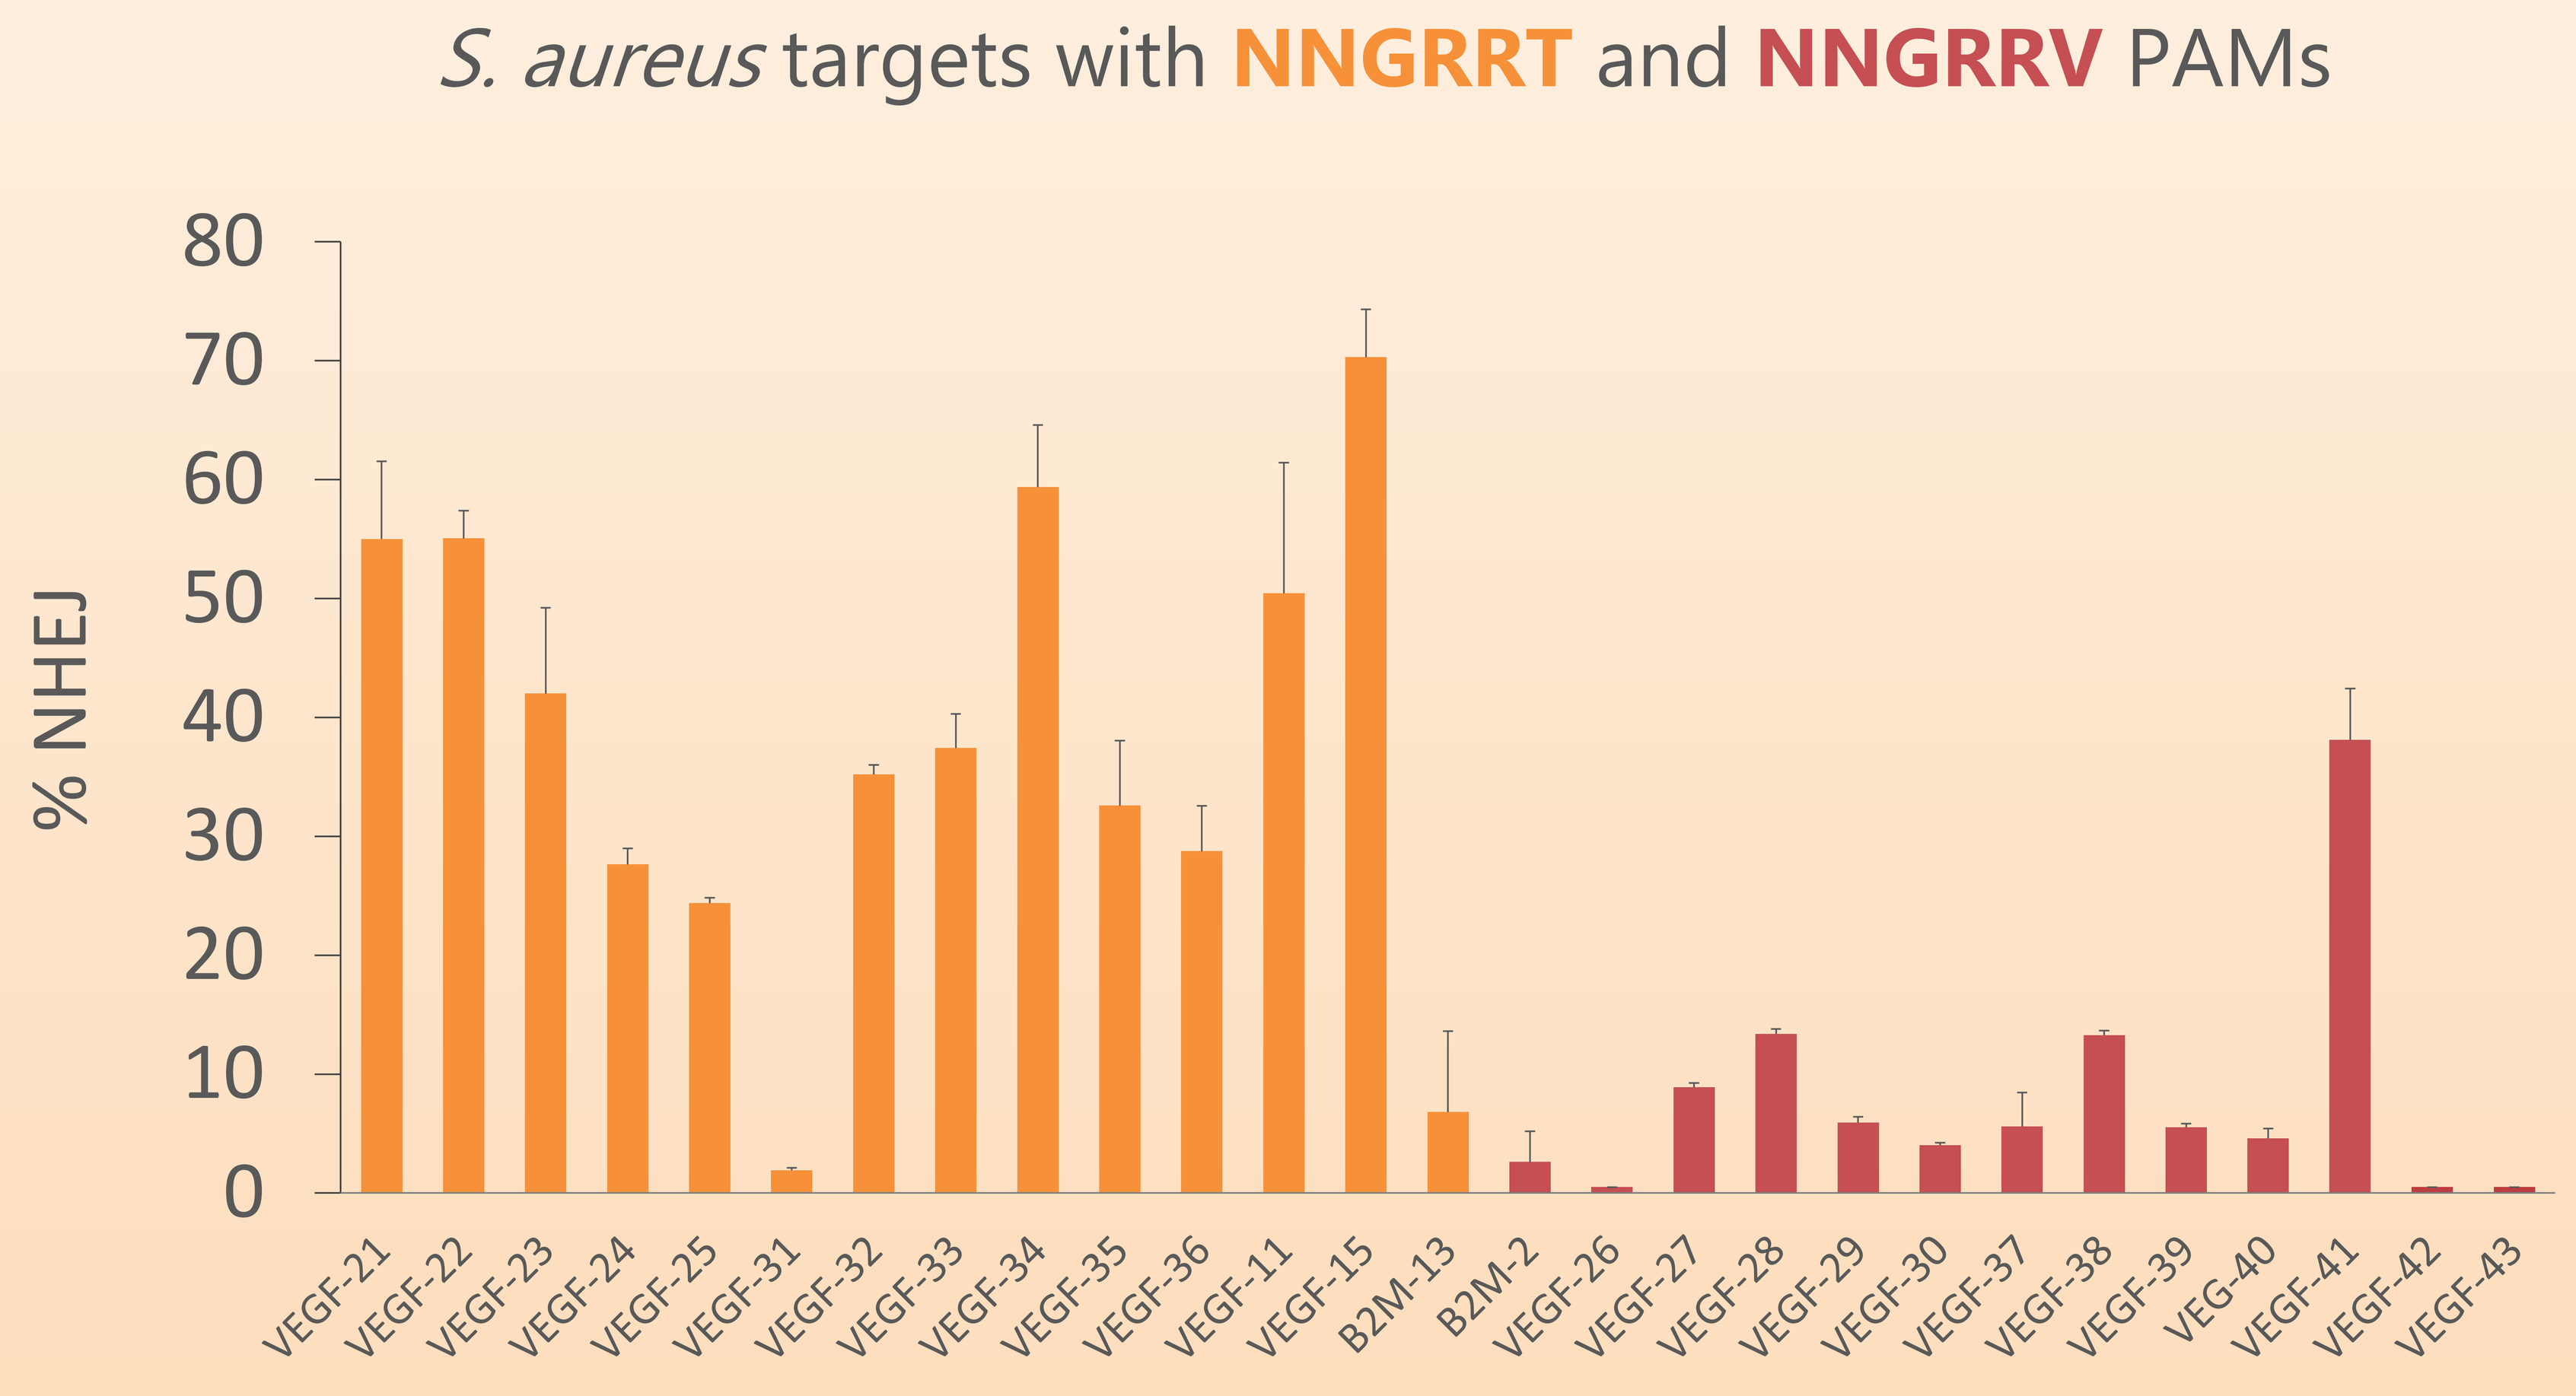
S. aureus targets with NNGRRT and NNGRRV PAMs
% NHEJ
VEGF-21 VEGF-22 VEGF-23 VEGF-24 VEGF-25 VEGF-31 VEGF-32 VEGF-33 VEGF-34 VEGF-35 VEGF-36 VEGF-11 VEGF-15 B2M-13 B2M-2 VEGF-26 VEGF-27 VEGF-28 VEGF-29 VEGF-30 VEGF-37 VEGF-38 VEGF-39 VEG-40 VEGF-41 VEGF-42 VEGF-43

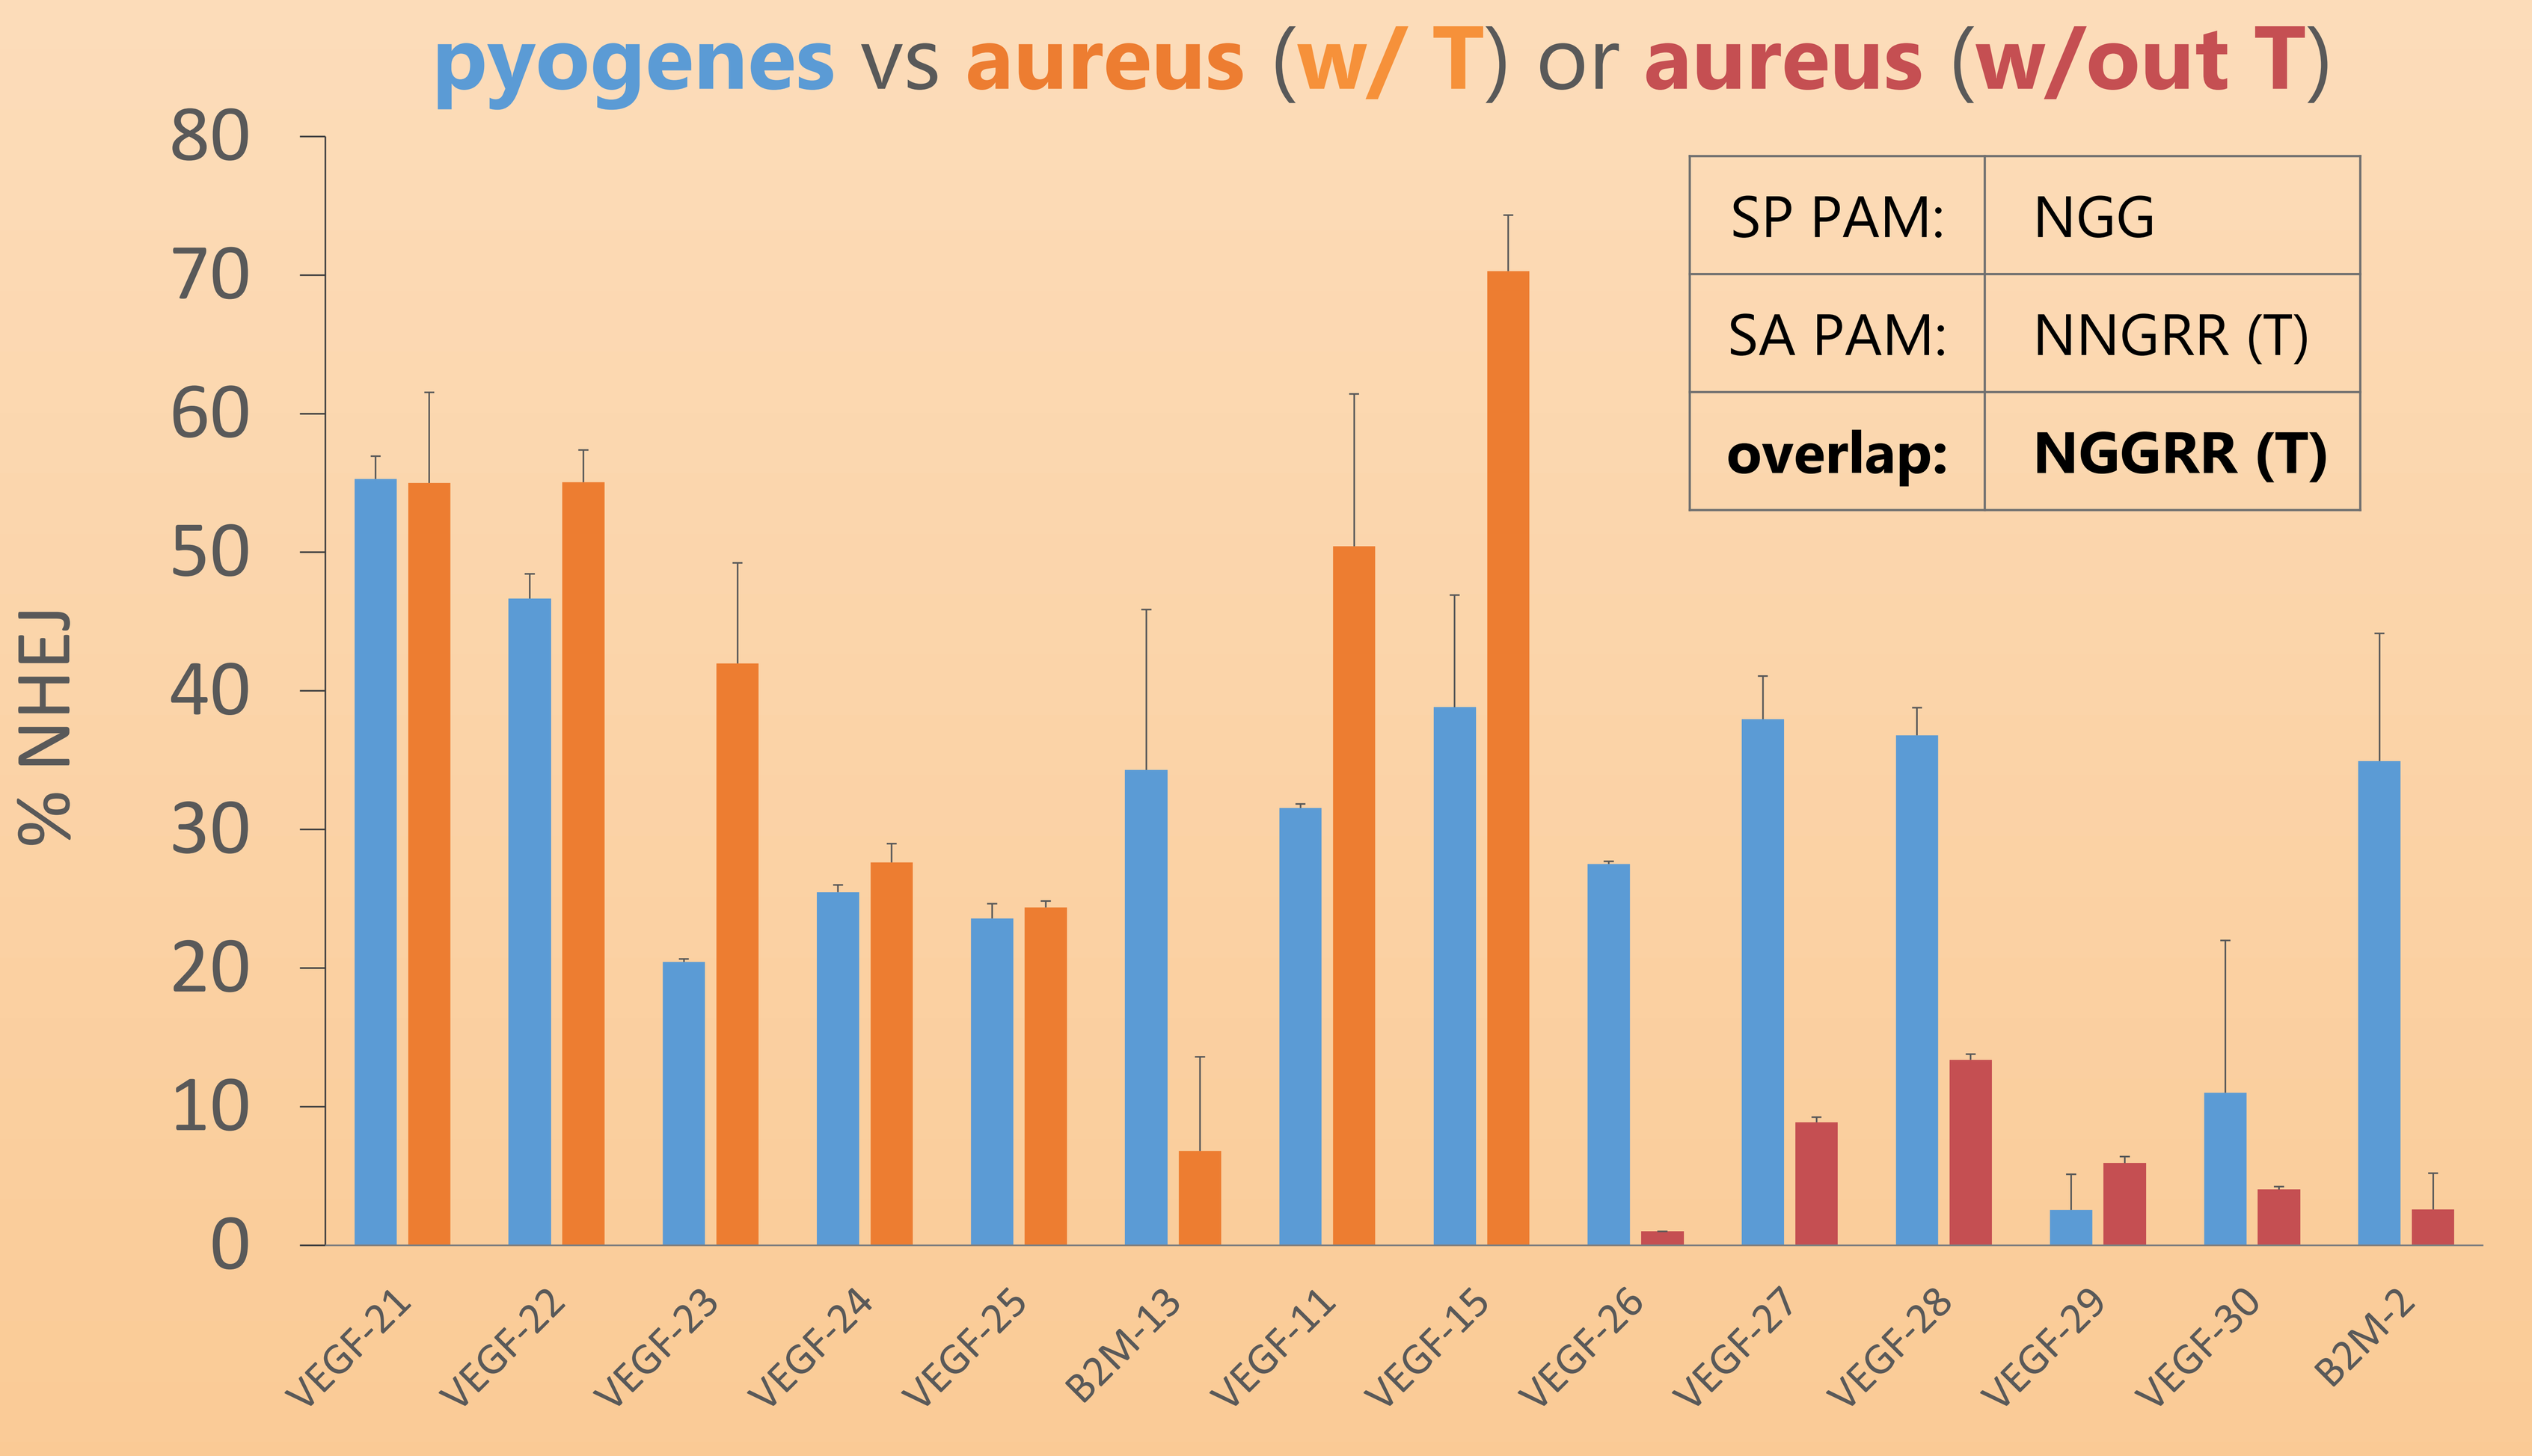
pyogenes vs aureus (w/ T) or aureus (w/out T)
SP PAM: NGG
SA PAM: NNGRR (T)
overlap: NGGRR (T)
% NHEJ
VEGF-21 VEGF-22 VEGF-23 VEGF-24 VEGF-25 B2M-13 VEGF-11 VEGF-15 VEGF-26 VEGF-27 VEGF-28 VEGF-29 VEGF-30 B2M-2

Figure 1. *(Above) S. aureus* Cas9 with gRNAs targeting loci with different PAMs. NHEJ % on the Y axis represents on-target cleavage rates as measured by T7E1 assay. (*Below*) Comparison of *S. aureus* Cas9 and *S. pyogenes* Cas9 DNA cleavage at identical target sites, with dual-compatible NNGRR(T) PAMs.

Cleavage efficiency depends on gRNA spacer length

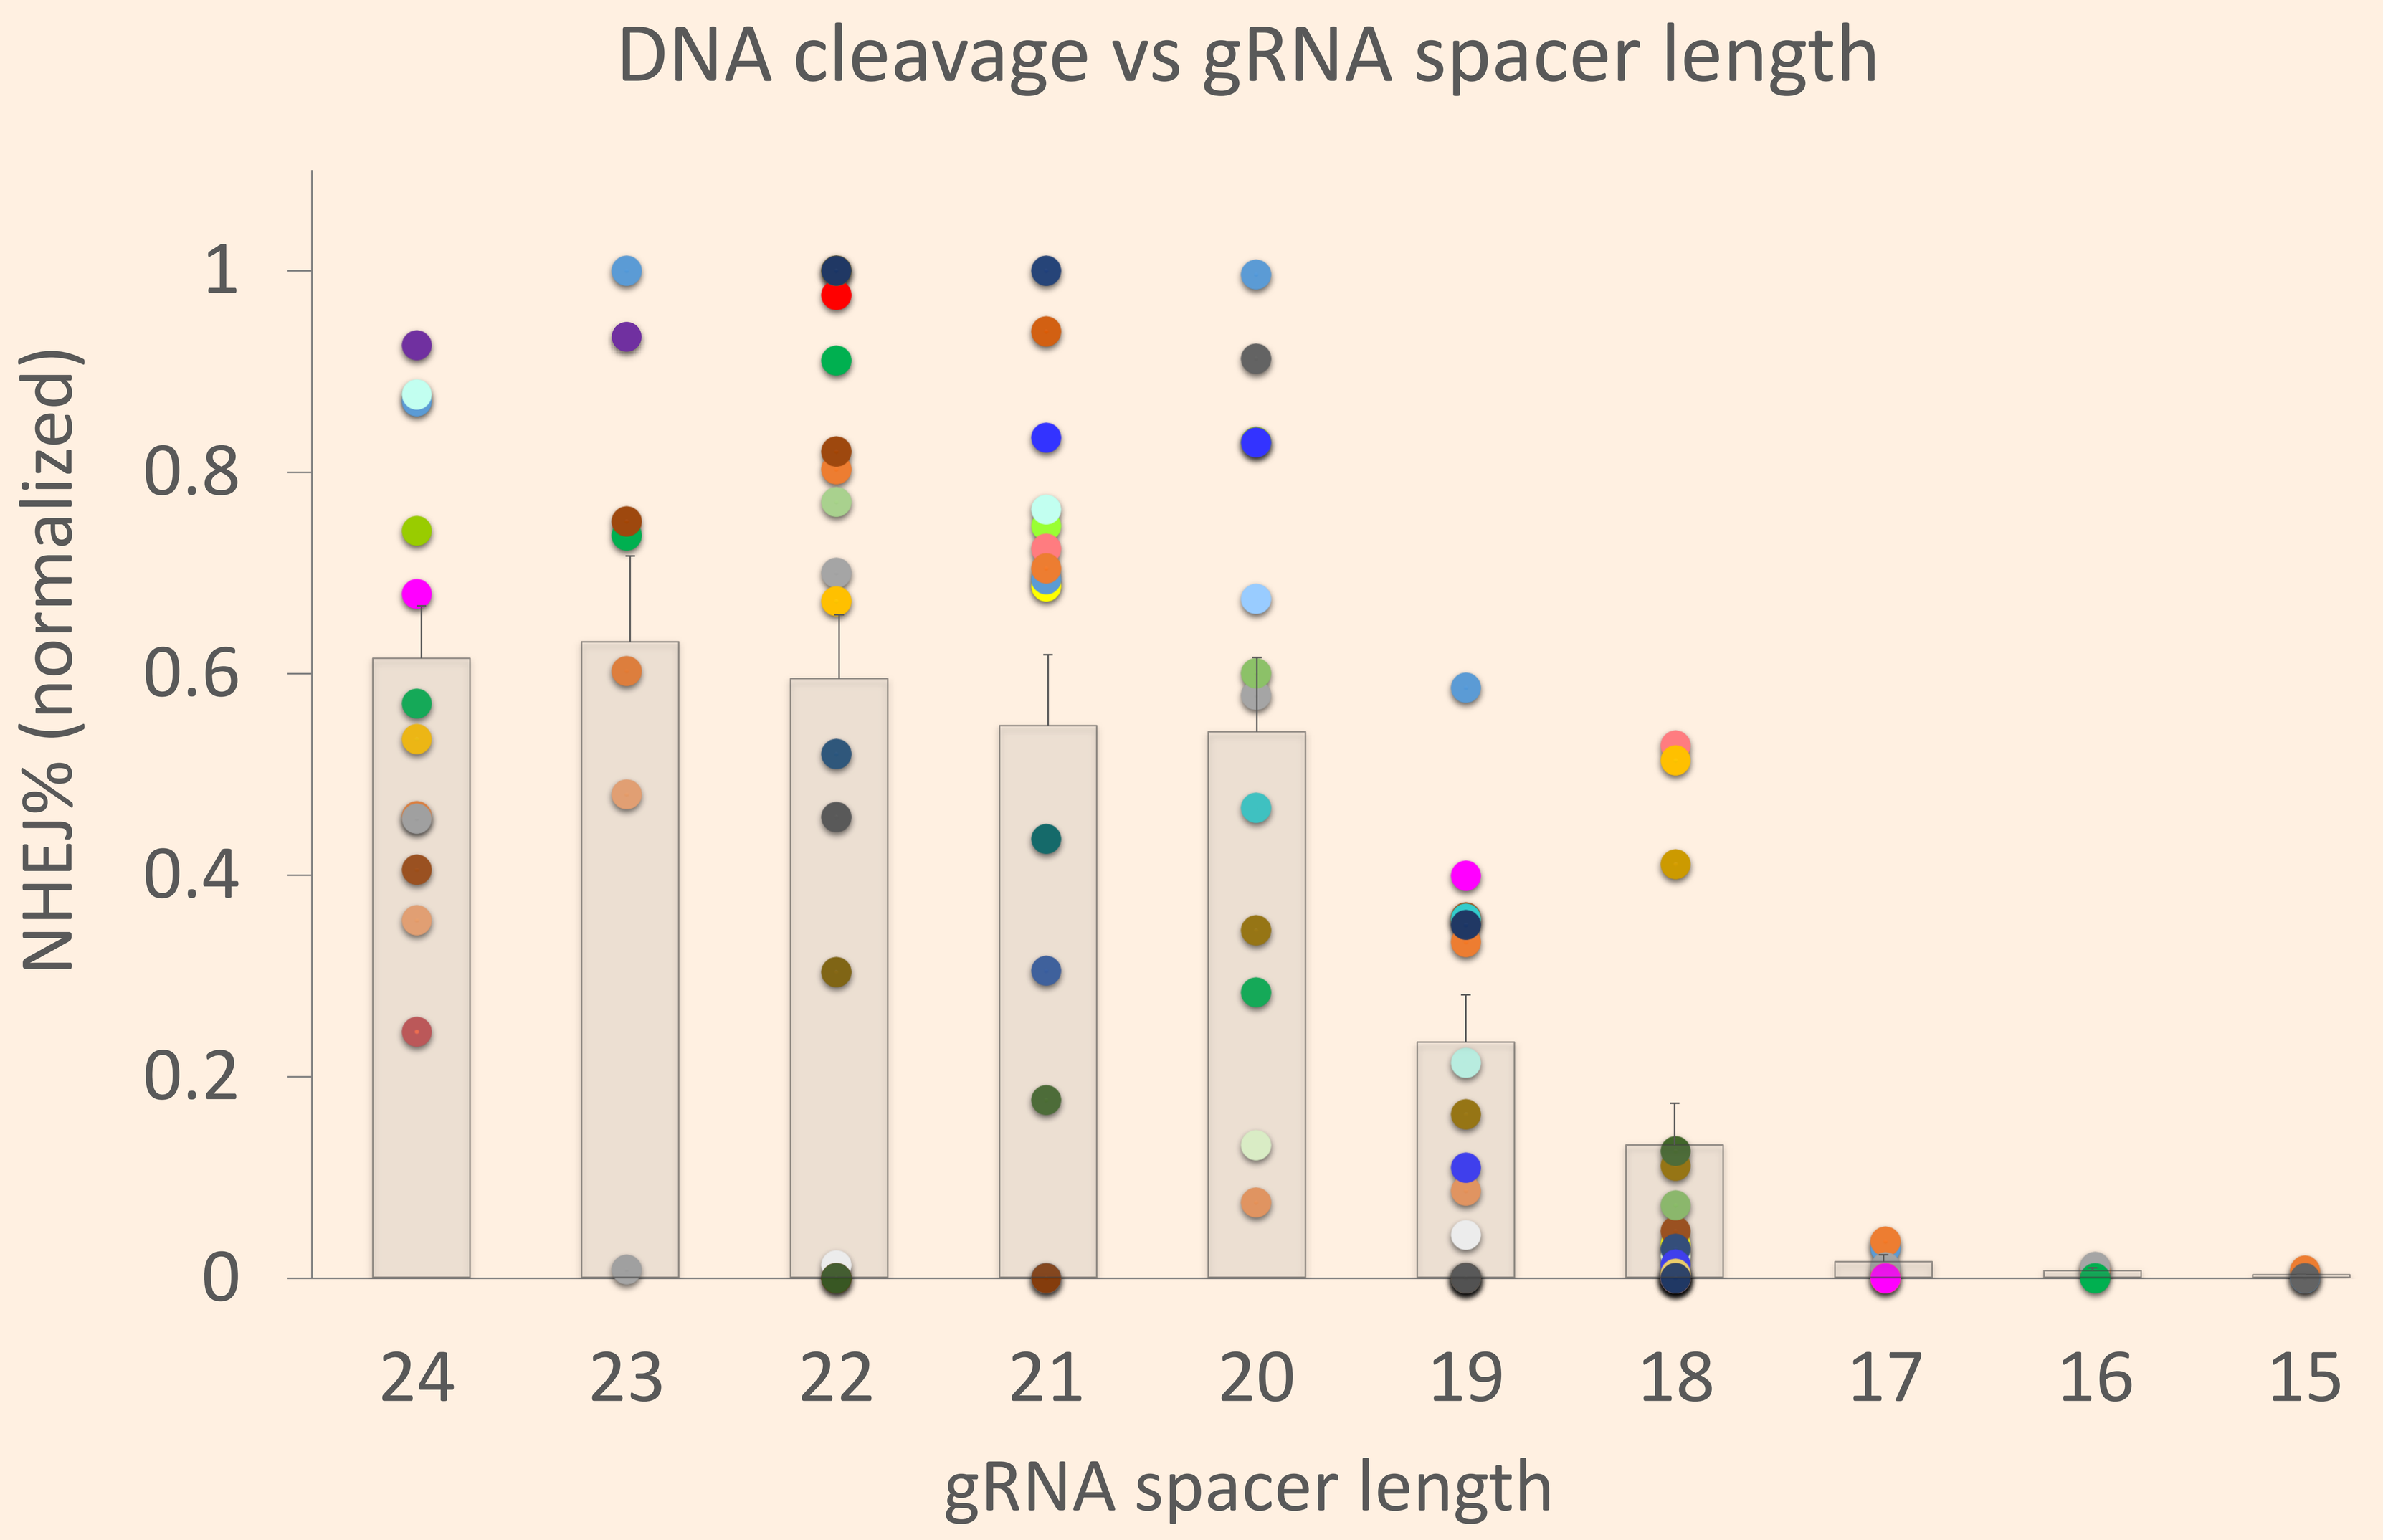
DNA cleavage vs gRNA spacer length
NHEJ% (normalized)
gRNA spacer length

Figure 2. *S. aureus* Cas9 cleavage of target DNA with "sibling" gRNAs, which have spacers of different lengths but target the same precise locus. Same-colored dots represent gRNA siblings, all of which initiate with a G for optimal U6 promoter expression. Data points come from multiple experiments, with maximum cleavage in each experiment set to 1 and all other data points normalized. Grey bars represent mean cleavage for gRNAs of that length.

Shorter gRNAs are generally less tolerant of mismatches

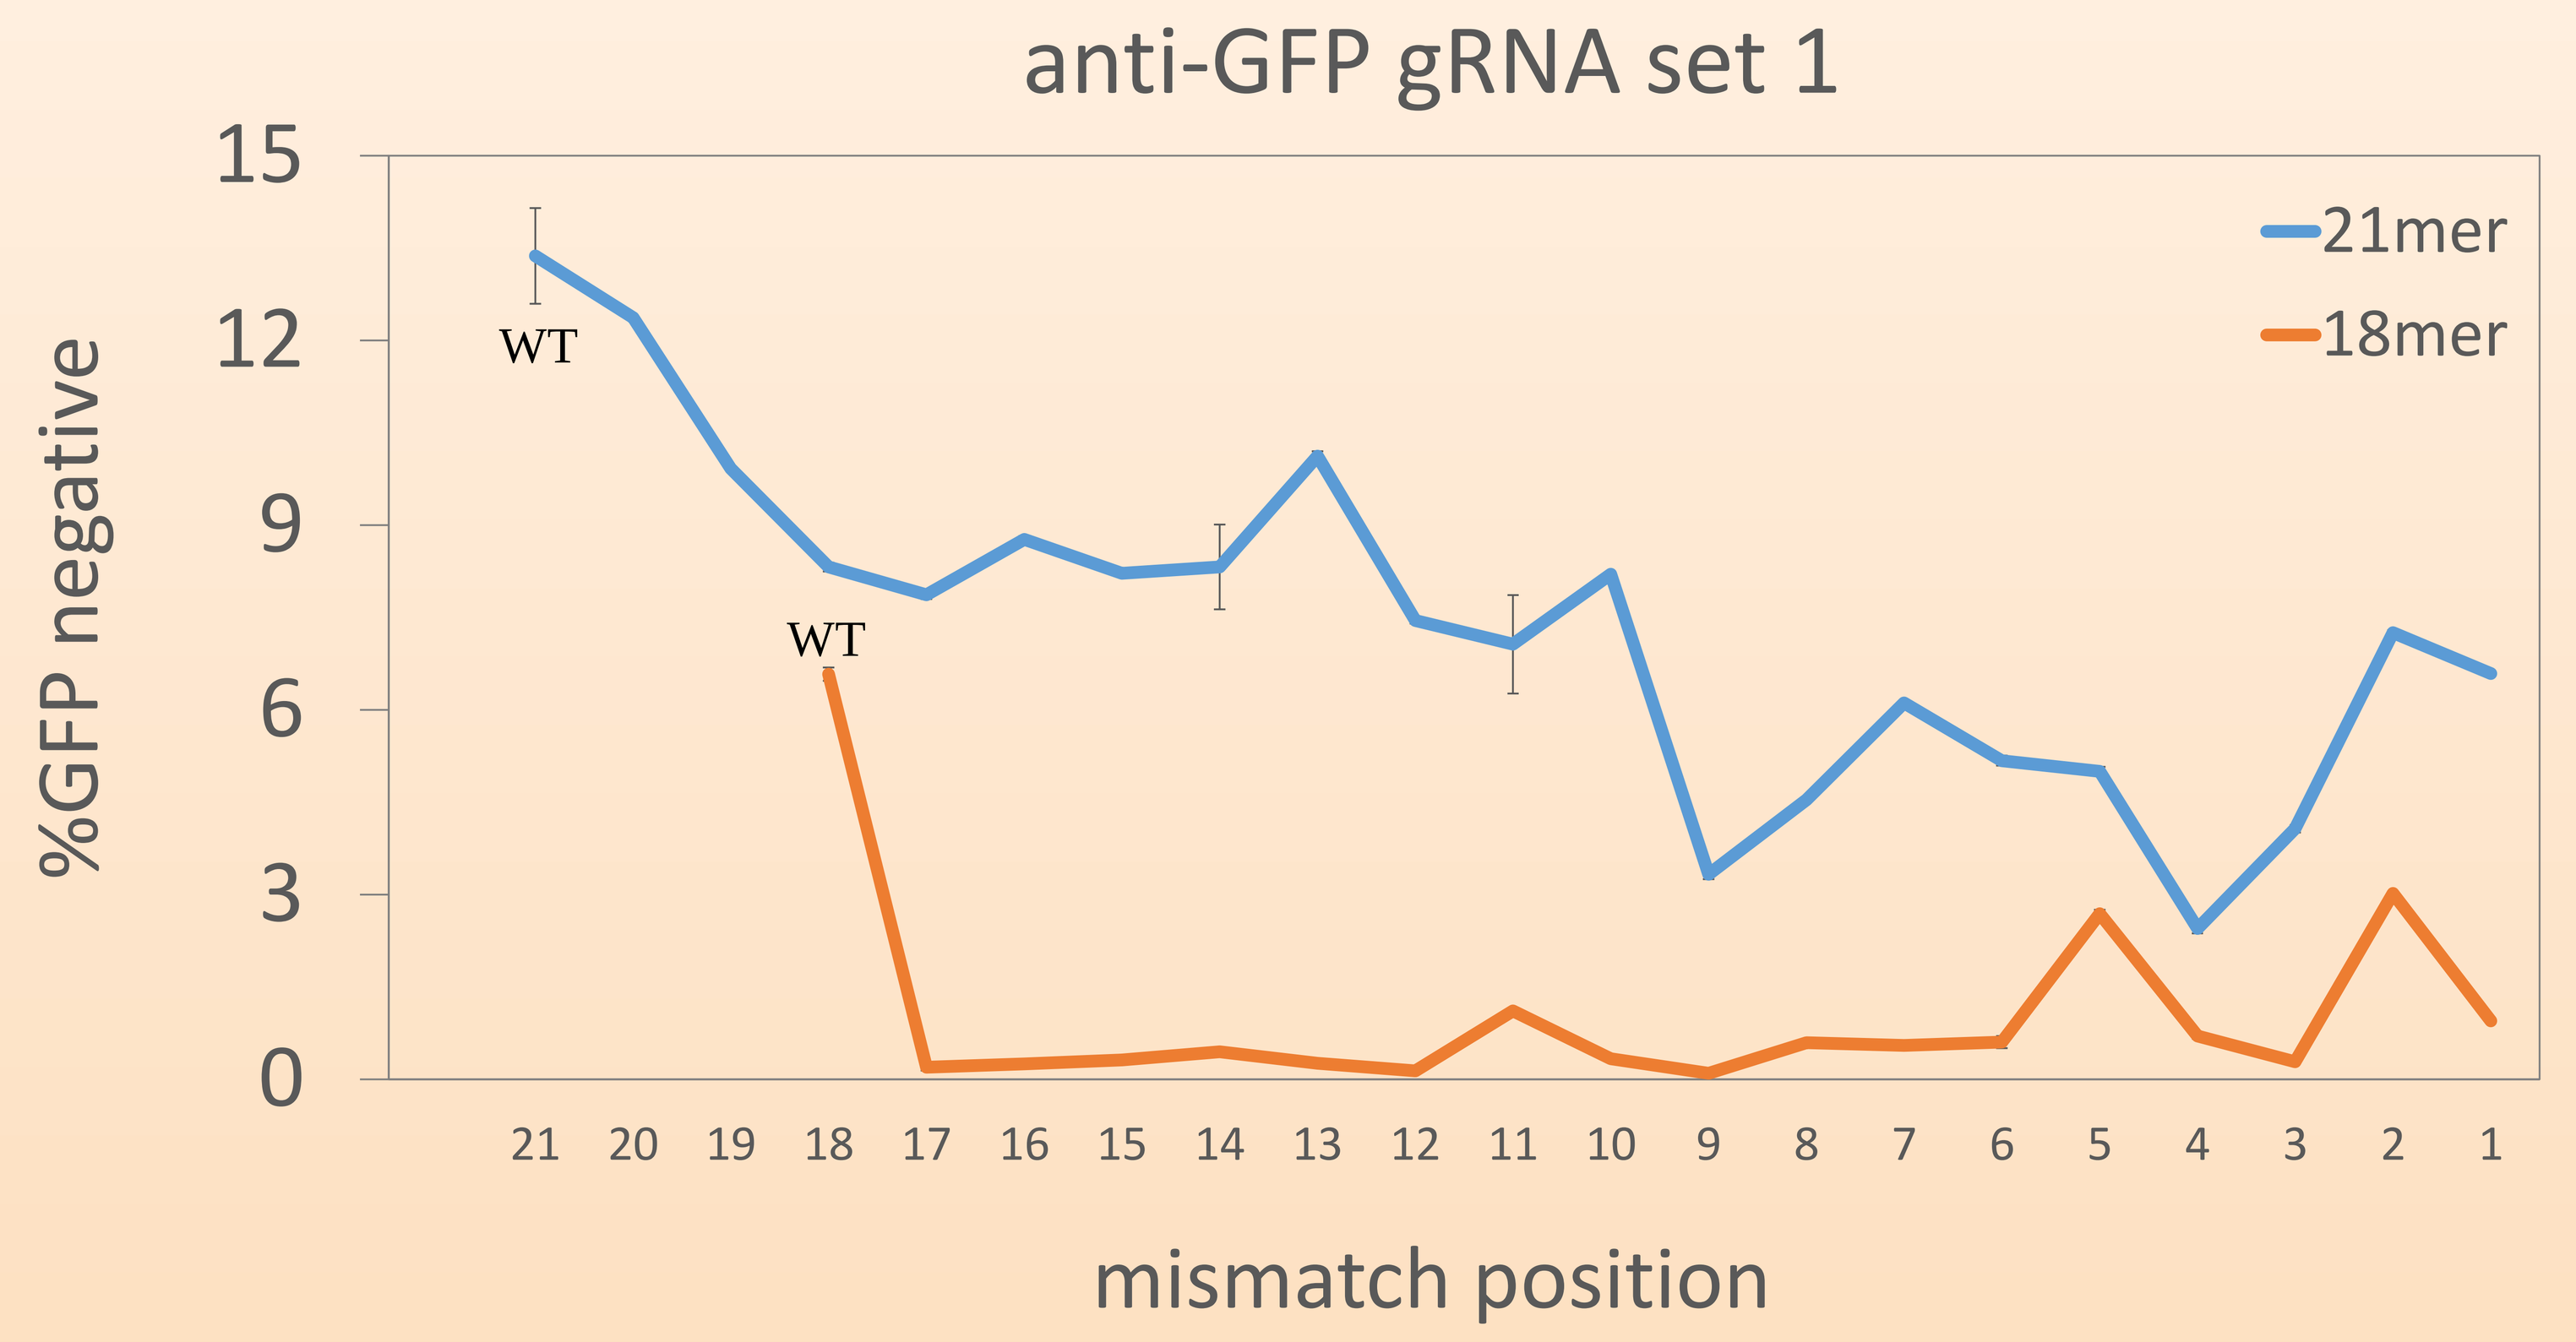
anti-GFP gRNA set 1
21mer
18mer
WT
WT
%GFP negative
mismatch position

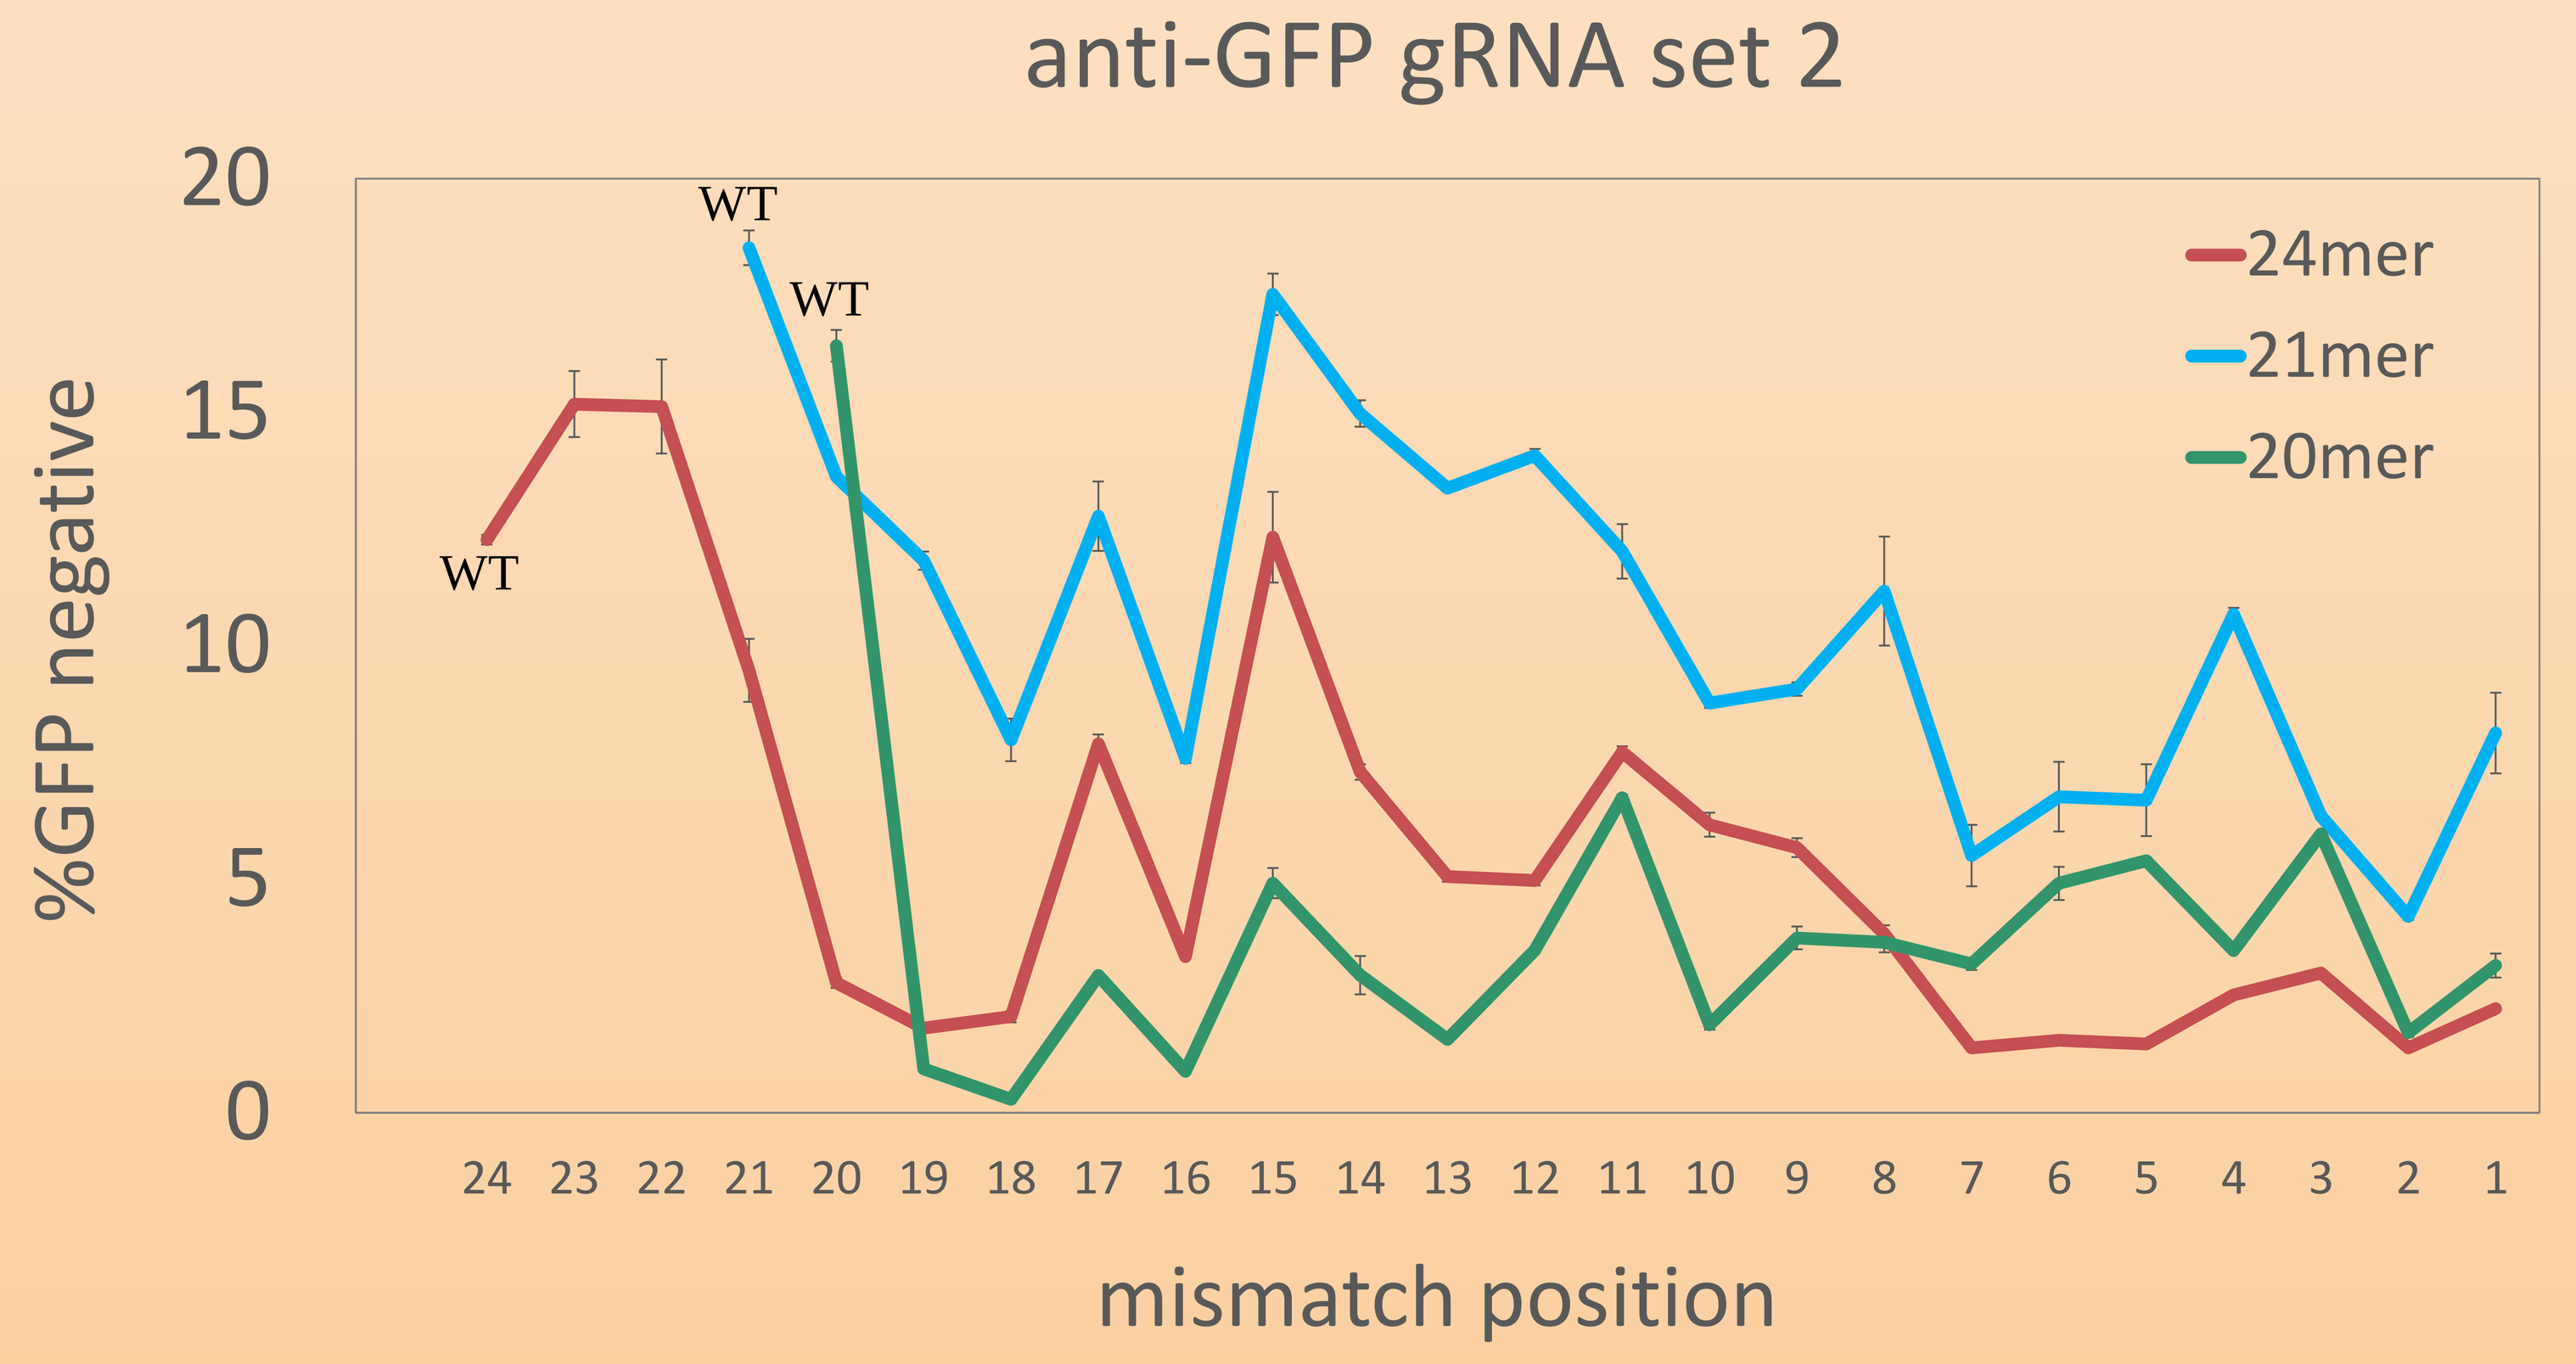
anti-GFP gRNA set 2
24mer
21mer
20mer
WT
WT
WT
%GFP negative
mismatch position

Figure 3. Knockdown of GFP with *S. aureus* Cas9 and gRNAs containing single mismatches. WT (wild type) gRNAs are perfect matches to the target sequence. *(Above)* gRNAs targeting GFP site 1 with spacer lengths of 21 and 18 bases. (Below) gRNAs targeting GFP site 2 with spacer lengths of 24, 21, and 20 bases.

Methods: All experiments were performed via transfection into HEK293T cells using Lipofectamine 3000 in 24-well format. Transfections consisted of 750 ng/well of Cas9 (driven by CMV promoter) plasmid and 250 ng/well of gRNA (driven by U6 promoter) construct. For most of these experiments, total DNA was harvested from cells 72 hours post transfection, and NHEJ % was determined by target locus PCR followed by T7E1 cleavage assay. Alternatively, for assays involving fluorescence, GFP knockdown was measured 3.5 days post transfection.