Characterization of Cas9-mediated genome editing in human T cells

G. Grant Welstead[1], Jennifer L. Gori[1], Justin Fang[1], McKensie Collins[1], Will Selleck[1], Ari Friedland[1], Hari Jayaram[1], David Bumcrot[1]

[1]Editas Medicine, 300 3rd Street, Cambridge, MA 02142

Abstract

Genome editing via CRISPR/Cas9 promises to provide a novel class of therapies for a variety of human diseases. To unlock the potential of the CRISPR/Cas9 technology, a deeper understanding of its efficacy in different primary cell types is required. A cell type of particular interest for gene editing is the human T cell due to its central role in the evolving cancer immunotherapy field. To better understand the utility of Cas9-mediated gene editing for engineering human T cells, we surveyed a variety of delivery modalities including electroporation of RNA and administration of ribonucleic acid-protein complexes. Additionally, we assessed the functionality of different Cas9 variants in human T cells. Here we report our findings including genome editing in human T cells using CRISPR/Cas technology.

Identification of efficient gRNAs in 293s

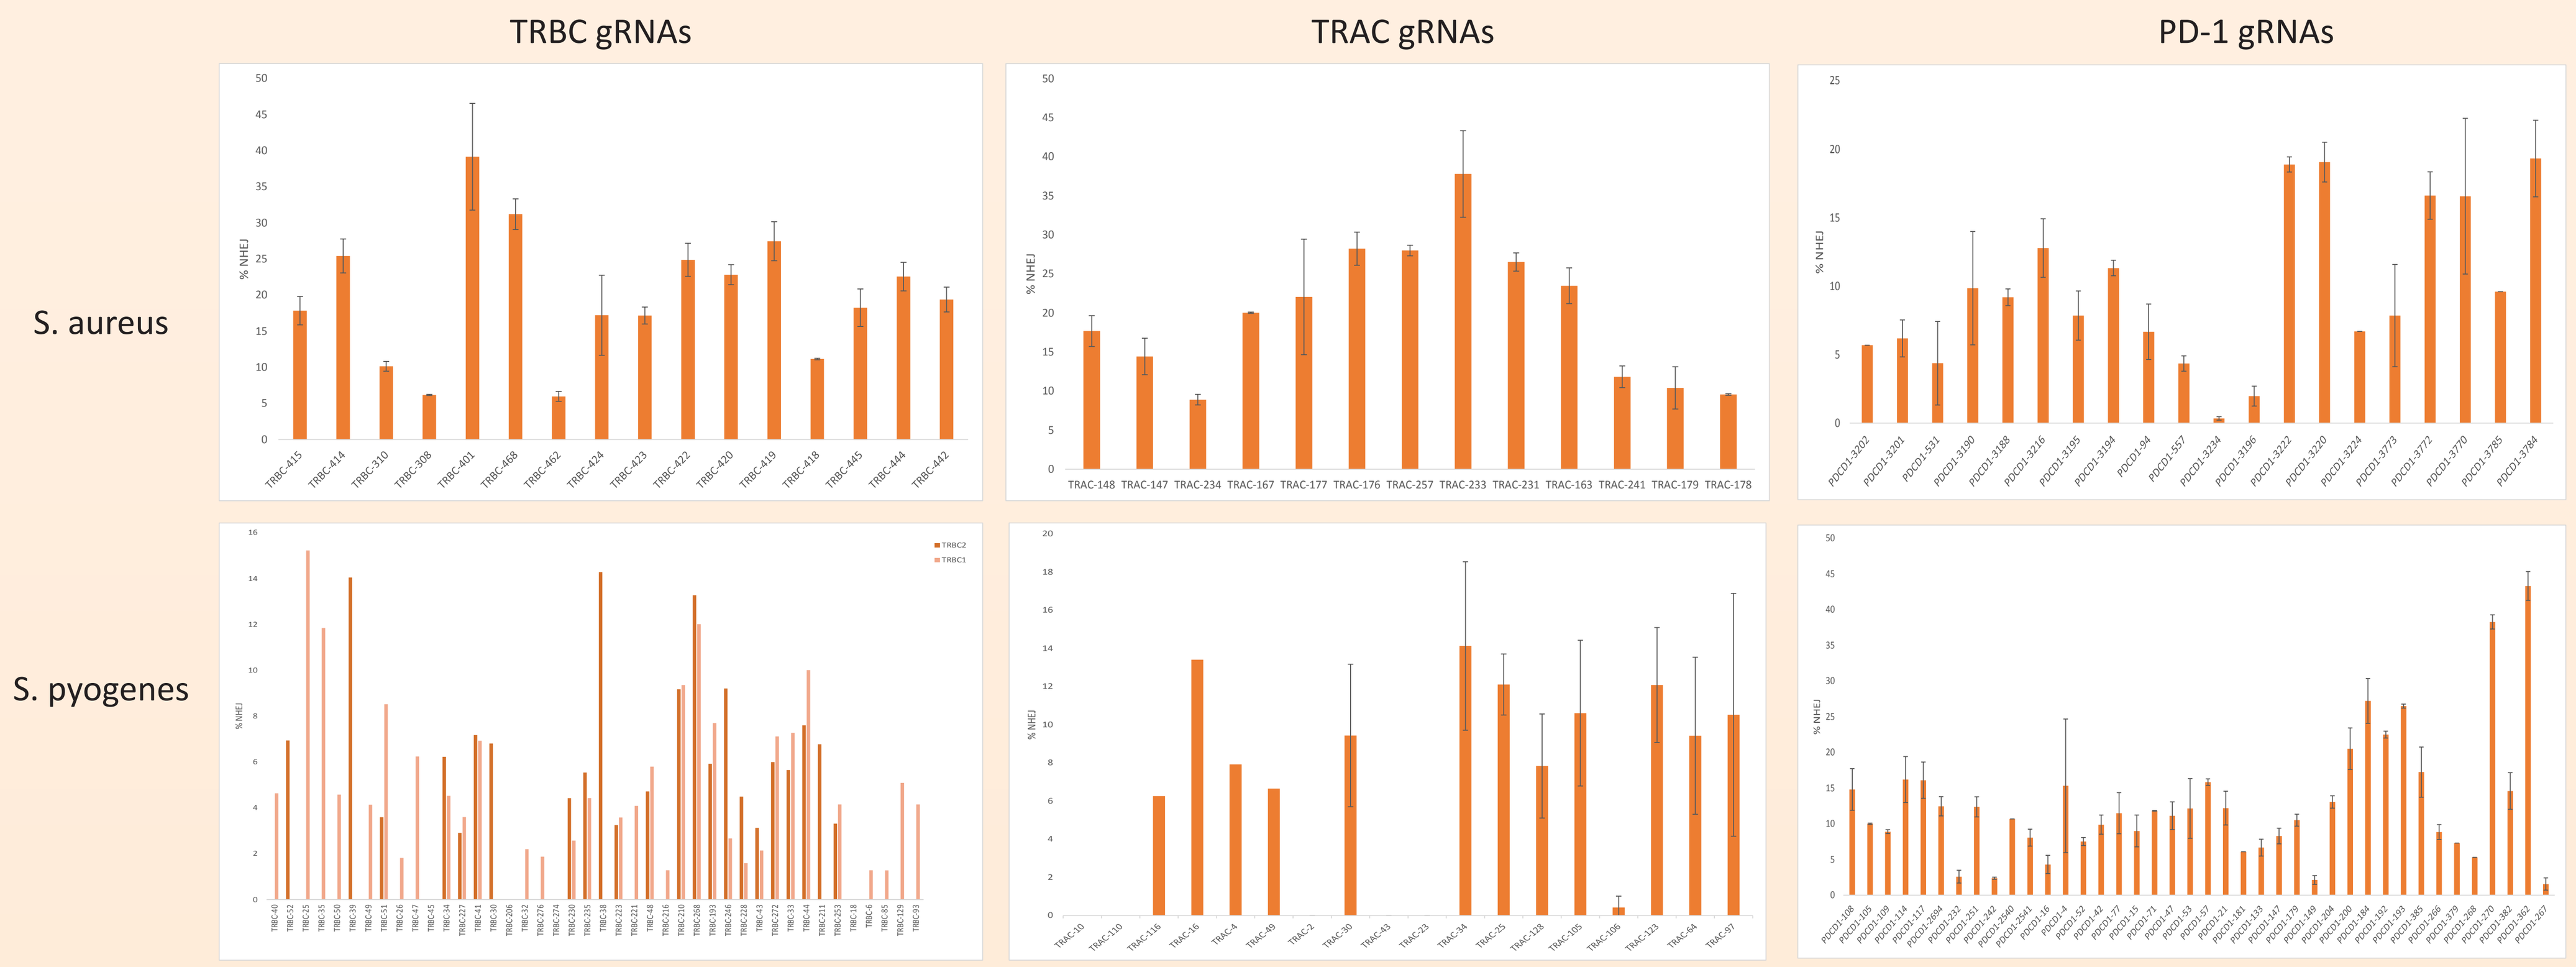
TRBC gRNAs
TRAC gRNAs
PD-1 gRNAs
S. aureus
S. pyogenes
% NHEJ

293s were transfected with two plasmids – one encoding S. aureus or S. pyogenes Cas9 and the other encoding the listed gRNA. The graph summarizes the average %NHEJ observed at the specificied locus for each gRNA, which was calculated from a T7E1 assay performed on genomic DNA isolated from duplicate samples.

Generation of CD3⁻ cells by mRNA/gRNA delivery to activated CD4⁺ T cells

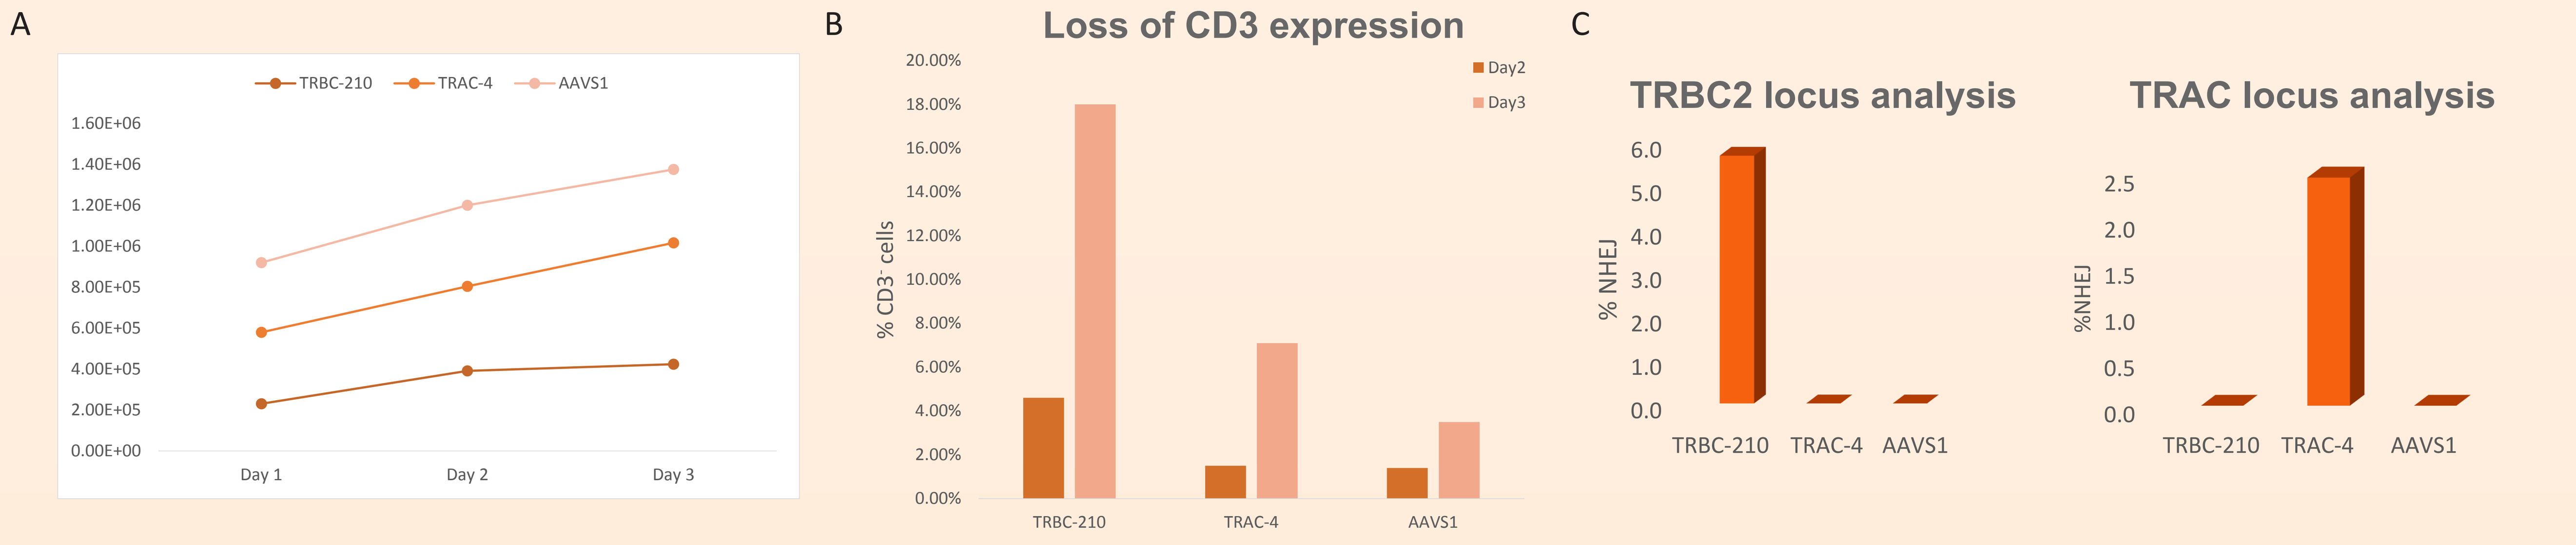
A
TRBC-210
TRAC-4
AAVS1
Day 1
Day 2
Day 3
B
Loss of CD3 expression
Day2
Day3
% CD3⁻ cells
C
TRBC2 locus analysis
TRAC locus analysis
% NHEJ

A) Activated CD4+ T cells were electroporated with S. pyogenes Cas9 mRNA and the gRNA indicated (TRBC-210, TRAC-4 or AAVS1). Cells were replated in RPMI1640 + IL-2 and counted for 3 consecutive days after staining with trypan blue. B) The electroporated cells were collected at day 2 and day 3 post electroporation and were stained with an APC-CD3 antibody and analyzed by FACS. Quantification of the CD3 negative population is shown. C) %NHEJ results from the T7E1 assay performed on TRBC2 and TRAC loci.

Comparison of Cas9-mediated editing by RNP and mRNA in Jurkat T cells

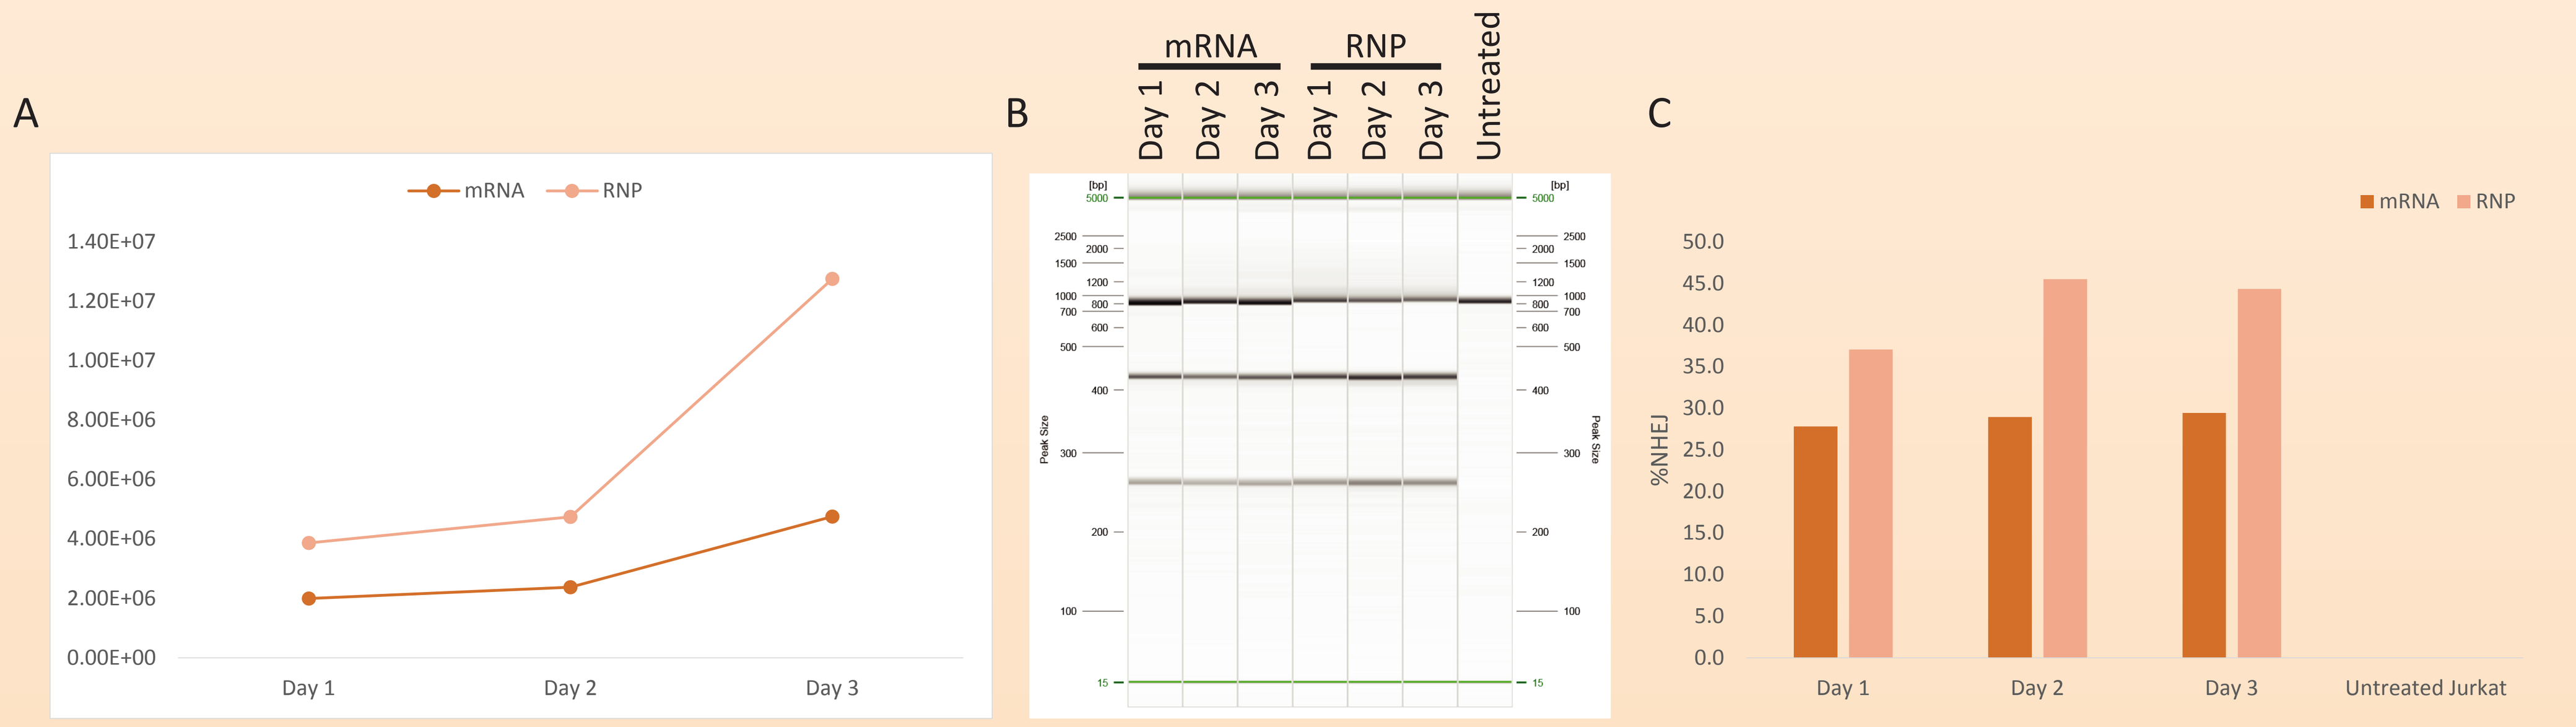
A
mRNA
RNP
Day 1
Day 2
Day 3
B
mRNA
RNP
Untreated
C
%NHEJ
Untreated Jurkat

A) Jurkat T cells were electroporated with either S. pyogenes Cas9 mRNA and PDCD1 gRNA (PDCD1-108) or S. pyogenes Cas9/gRNA (PDCD1-108) RNP targeting PDCD1. Cells were replated in RPMI1640 and counted for 3 consecutive days after staining with trypan blue. B) Gel image of the DNA resulting from the T7E1 assay performed on the PDCD1 locus at 24, 48, and 72 hours. C) Quantification of %NHEJ results from the gel depicted in (B). Higher levels of %NHEJ were detected with RNP vs mRNA delivery. As expected, there was undetectable levels of NHEJ in untreated Jurkat cells.

Successful editing of naive T cells by RNP delivery

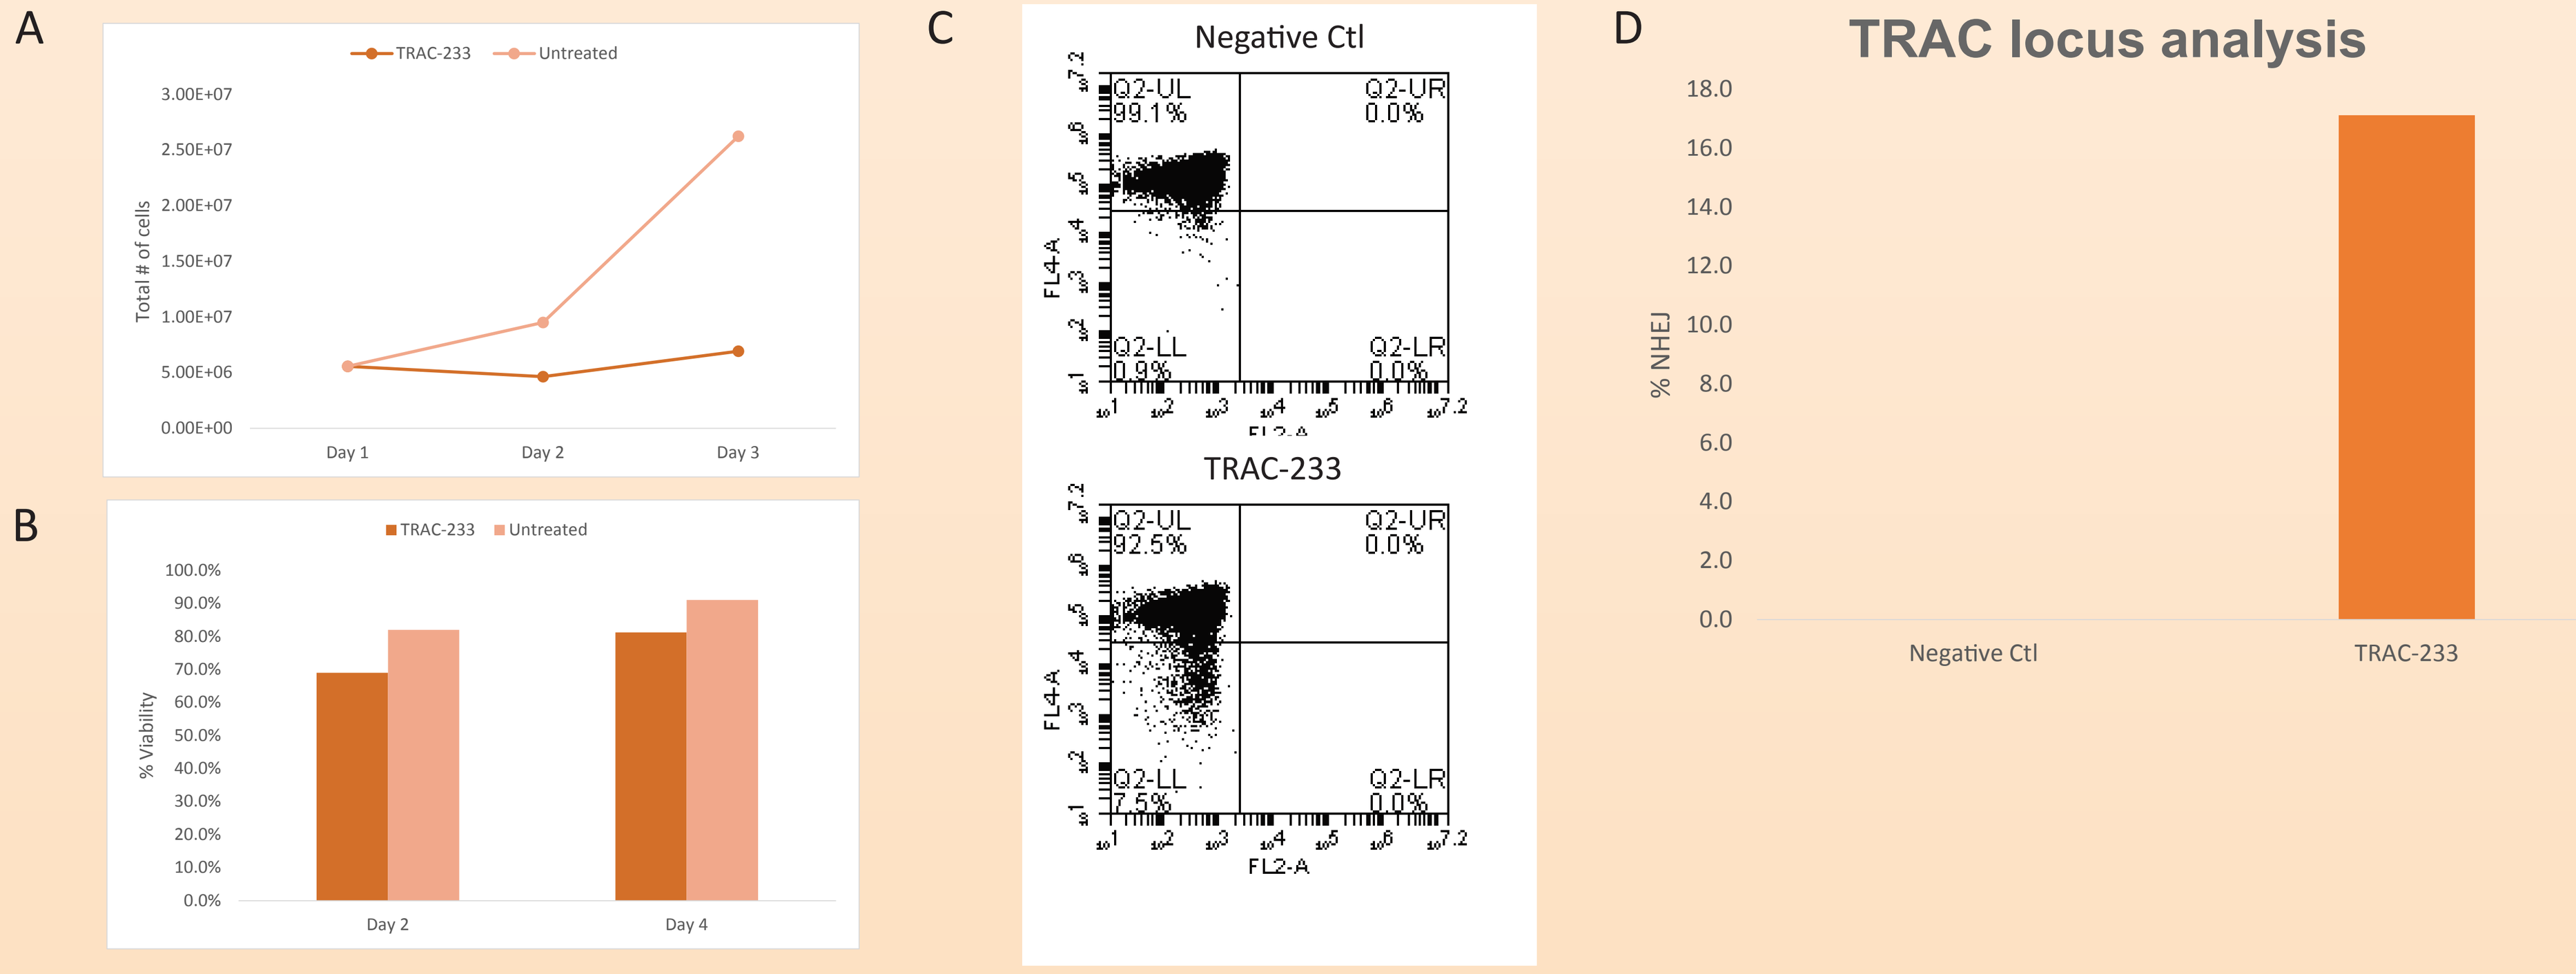
A
TRAC-233
Untreated
Total # of cells
B
% Viability
Day 2
Day 4
C
Negative Ctl
TRAC-233
D
TRAC locus analysis
% NHEJ
Negative Ctl
TRAC-233

A) 4x 10^6 Naïve CD3+ T cells ewere lectroporated with S. aureus Cas9/gRNA (TRAC-233) RNPs targeting TRAC according to the Maxcyte protocol. After electroporation the cells were plated in RPMI1640 + IL-7/IL-15 and counted for 3 consecutive days staining with trypan blue. B) Cells were stained with Annexin V and Priopidium Iodide and analyzed by FACS. Cells that were negative for annexin and PI were considered viable and are plotted for Day 2 and Day 3. C) 4 days after electroporation the cells were stained with an APC-CD3 antibody and analyzed by FACS. The negative control are cells with the TRAC-233 gRNA without a functional Cas9 D) % NHEJ results from the T7E1 assay performed on the TRAC locus.

Generation of CD3⁻ cells by RNP delivery to Jurkat T cells

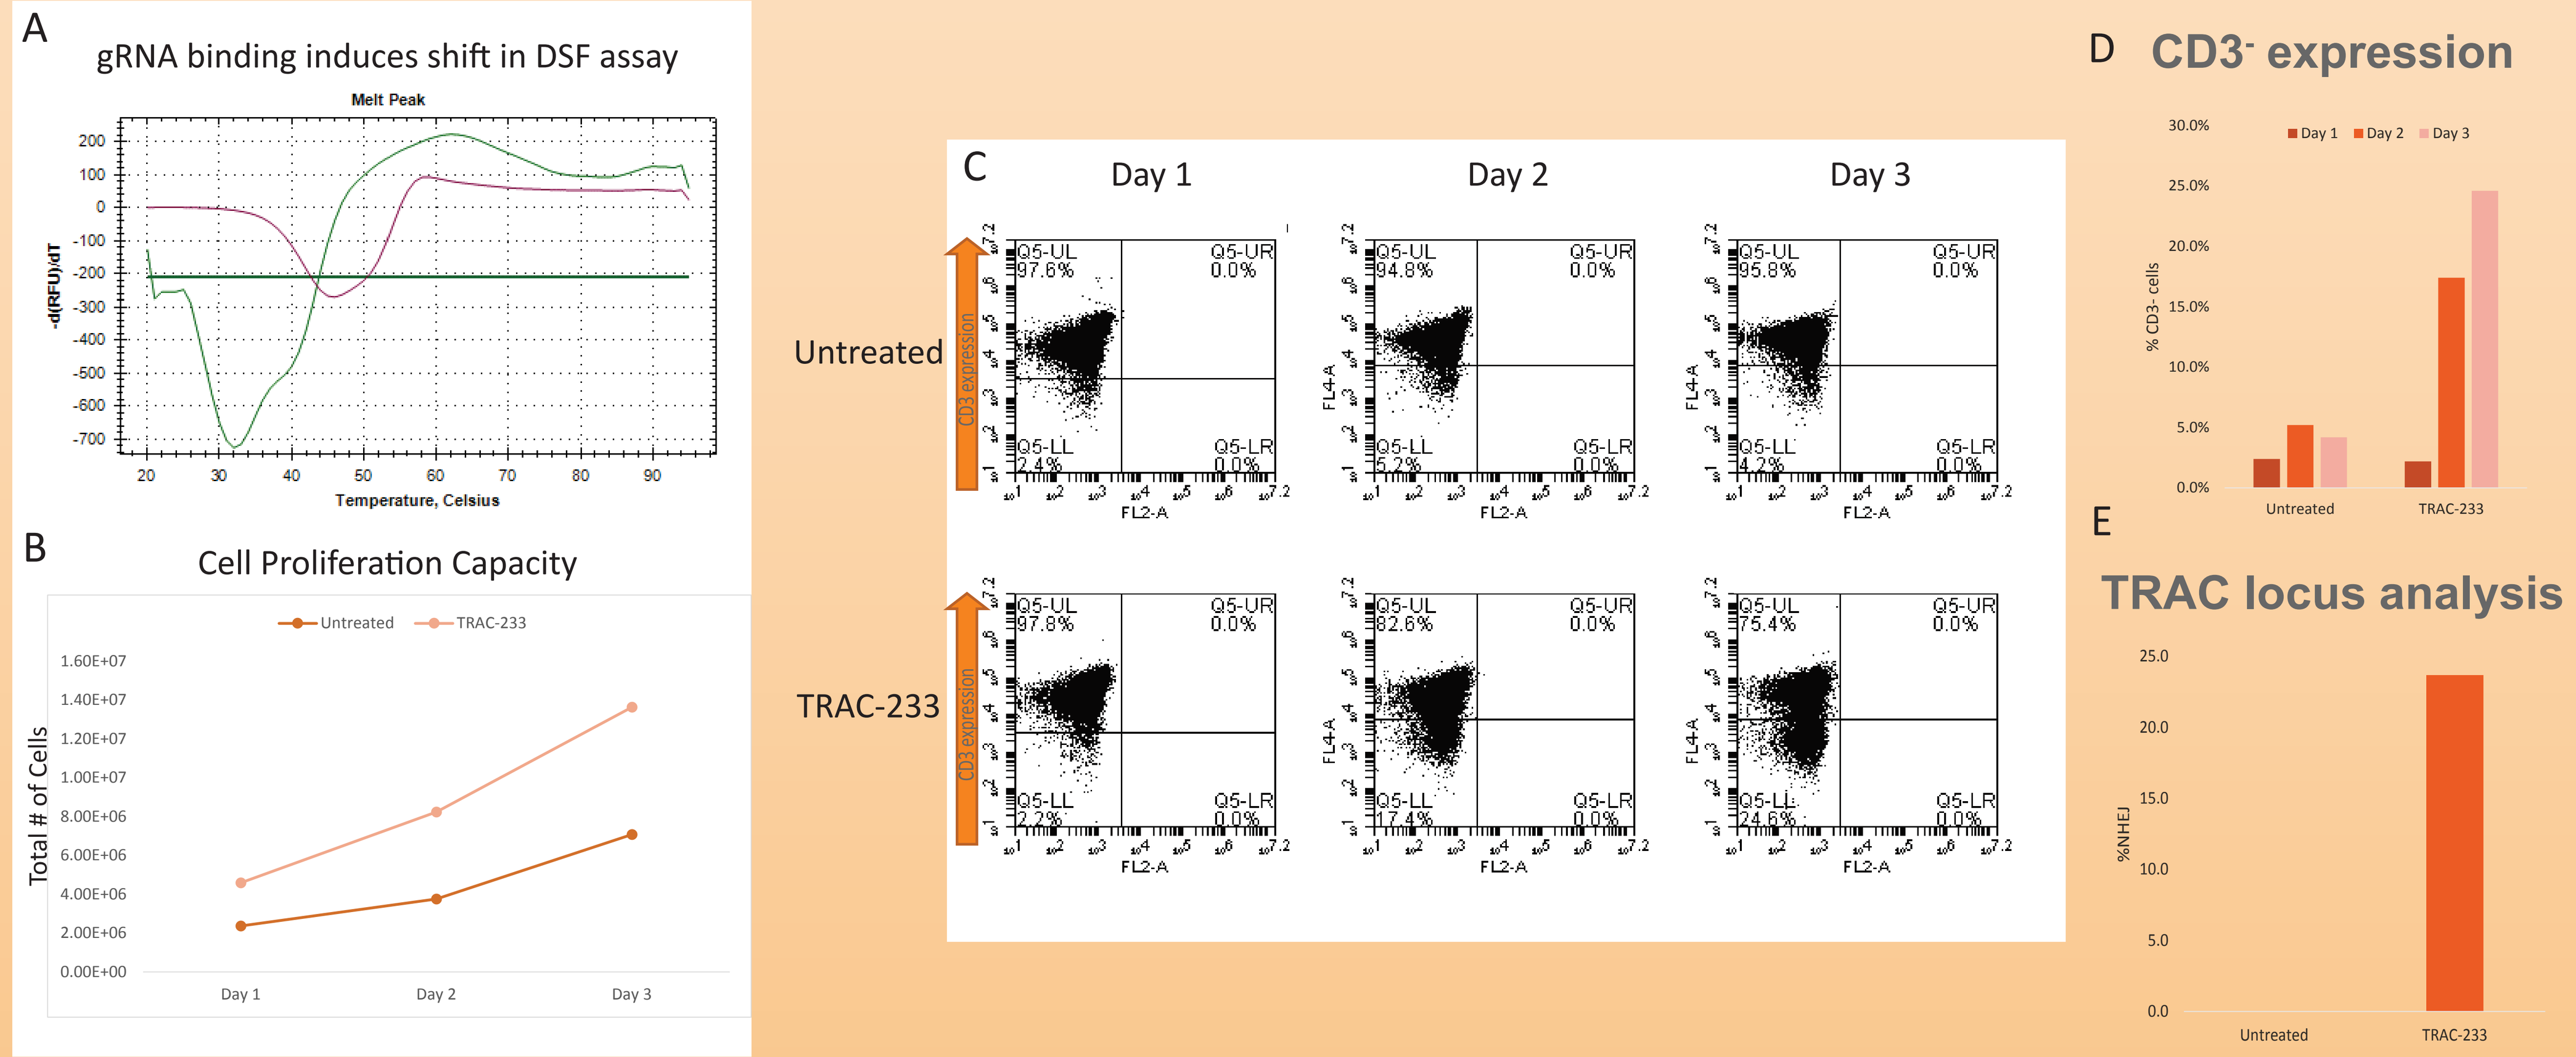
A
gRNA binding induces shift in DSF assay
Melt Peak
Temperature, Celsius
B
Cell Proliferation Capacity
Untreated
TRAC-233
Total # of Cells
C
Day 1
Day 2
Day 3
Untreated
TRAC-233
D
CD3⁻ expression
E
TRAC locus analysis
%NHEJ

A) S. aureus Cas9 and TRAC-233 gRNA were mixed and allowed to form complexes for 10 min. Prior to introduction to the cells, a small amount was removed to analyze in a DSF assay. B) Jurkat T cells electroporated with S. aureus Cas9/gRNA (TRAC-233) RNPs targeting TRAC were cultured in RPMI1640 and counted for 3 consecutive days following staining with trypan blue. C) Transduced Jurkat cells were stained with an APC-CD3 antibody and analyzed by FACS. The cells were analyzed on day 1, day 2 and day 3 after the electroporation. D) Quantification of the CD3 negative population from the plots in (C). E) % NHEJ results from the T7E1 assay performed on the TRAC locus.

Summary

Cas9-mediated T cell editing using Cas9 from both S. aureus and S. pyogenes.

Delivery of Cas9 as an RNP yields better viability and cutting efficiency for the PDCD1 gRNA, PDCD1-108.

Nearly 45% gene editing in Jurkat T cells at the PDCD1 locus

Generation of nearly 25% CD3 Jurkat T cells using RNP delivery

Generation of approximately 18% CD3⁻ T cells when targeting the TRBC locus in activated T CD4 T cells

Over 15% gene editing in naïve T cells was achieved using RNP against TRAC

Acknowledgements

All electroporation experiments in T cells were performed using the Maxcyte system. The authors would like to thank members of the Maxcyte team that facilitated our experiments - Linhong Li and Madhusudan V. Peshwa.

We would also like to thank Shondra Miller at the Washington University Genome Engineering and IPSC Center for providing Editas with some reagents used in our experiments.