Highly efficient CRISPR/Cas9 mediated gene editing in long-term engrafting human hematopoietic stem progenitor cells

J.M. Heath, A. Chalishazar, C.S. Lee, W. Selleck,
C. Cotta-Ramusino, D. Bumcrot, J.L. Gori

Introduction

- Gene-modified CD34⁺ hematopoietic stem progenitor cell (HSPC) transplantation is curative for select hematologic diseases
- Electroporation of human HSPCs with Cas9 ribonucleoprotein complex (RNP) supports effective gene editing *ex vivo*

Goals of Study

- Evaluate reproducibility of Cas9 RNP gene editing across human CD34⁺ cells donors (*HBB, AAVS1, CXCR4* loci)
- Compare engraftment of human CD34⁺ cells electroporated with D10A nickase RNPs targeting *HBB*

Summary

- Cas9 RNP supports efficient, reproducible gene editing in human CD34⁺ cells across multiple donors and experiments
- Gene-edited human long-term repopulating CD34⁺ cells retain engraftment capability and multipotency *in vivo*

In vivo Experimental Design

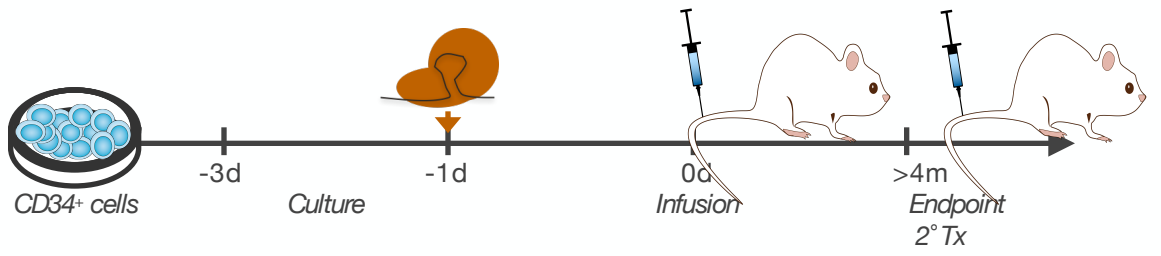
CD34+ cells
-3d
Culture
-1d
0d
Infusion
>4m
Endpoint
2° Tx

- CD34⁺ cells were cultured for 2 days, electroporated with RNPs targeting *HBB*, and transplanted into immunodeficient (NSG) mice
- ≥4 months after transplantation, human blood reconstitution of NSG mice and gene editing in human blood cells were evaluated

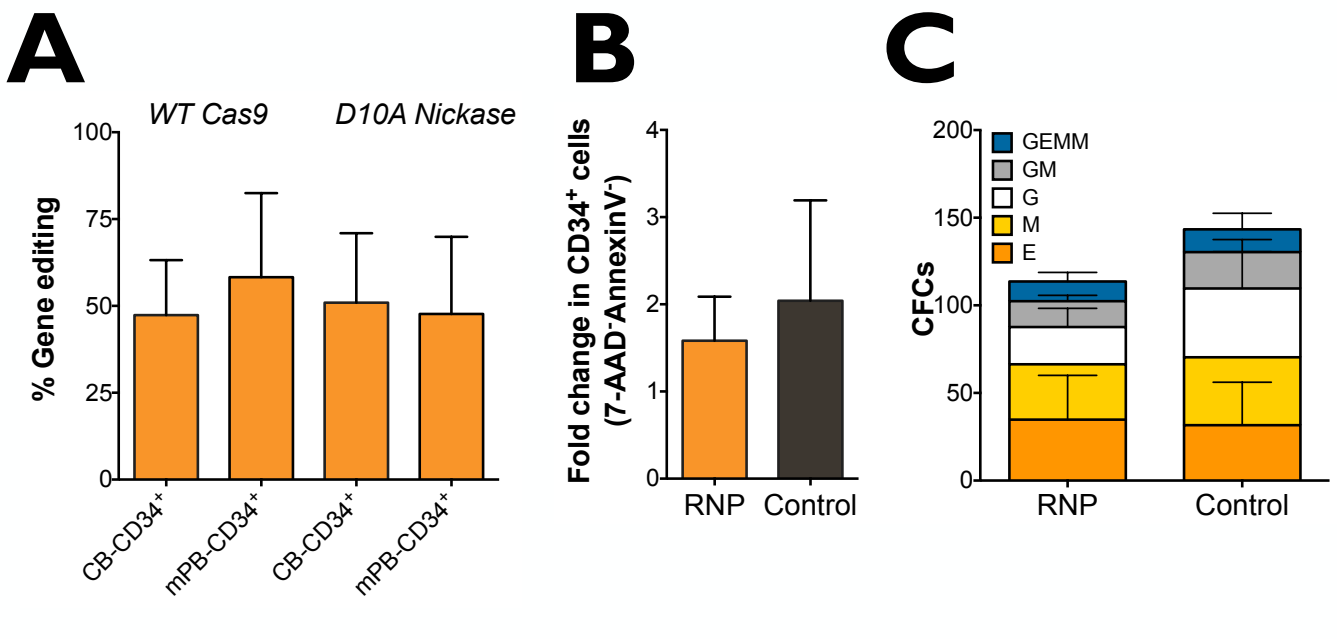
A
WT Cas9
D10A Nickase
% Gene editing
CB-CD34+
mPB-CD34+
CB-CD34+
mPB-CD34+
B
Fold change in CD34+ cells (7-AAD-AnnexinV-)
RNP
Control
C
CFCs
GEMM
GM
G
M
E
RNP
Control

FIGURE 1. Cas9 RNP treated human CD34⁺ cells maintain viability and hematopoietic activity *ex vivo*. A. Gene editing (determined by sequencing) in cord blood (CB) and adult mobilized peripheral blood (mPB) CD34⁺ cells after electroporation with wild-type (WT) Cas9 or D10A paired nickases targeting *HBB* (n=20 donors, 15 experiments). **B.** Fold change of viable RNP-treated or donor matched untreated control CD34⁺ cells. **C.** Hematopoietic activity in RNP-treated and control CD34⁺ cells (colony forming cell [CFC] assays). GEMM: Granulocyte Erythroid Macrophage Monocyte. Statistics: Mean + S.D. For panels B, C, P-values were calculated using a paired t-test (all P-values n.s.).

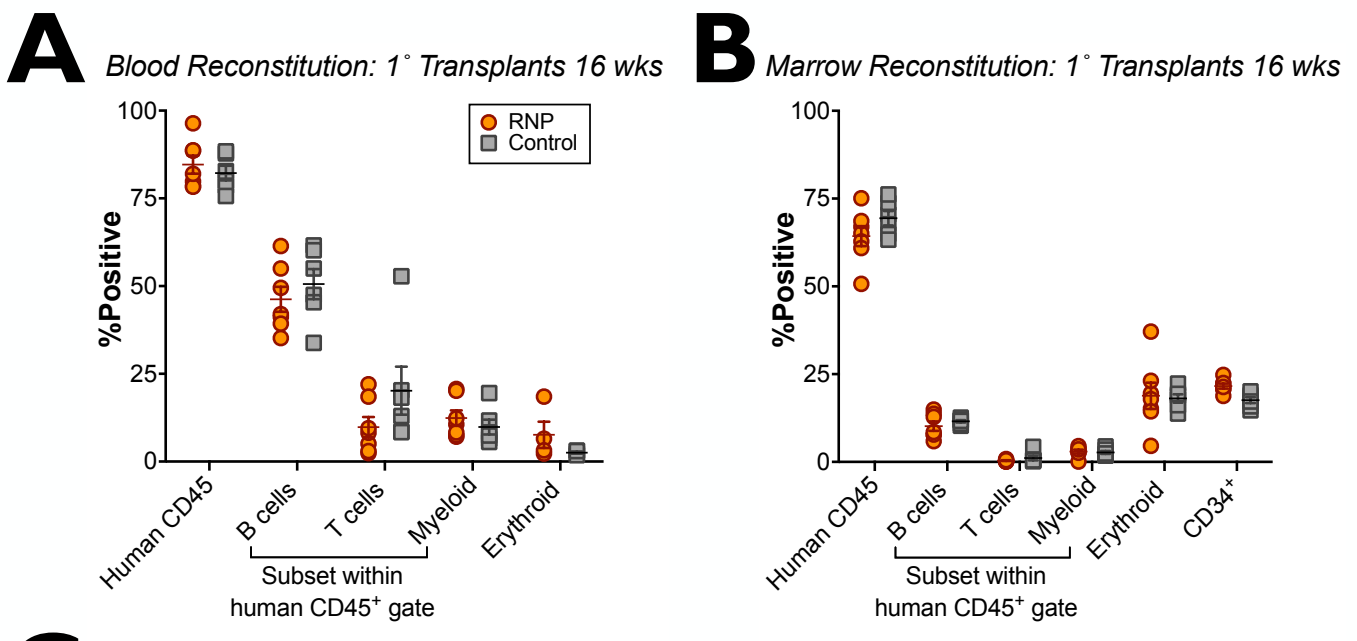
A
Blood Reconstitution: 1° Transplants 16 wks
%Positive
RNP
Control
Human CD45
B cells
T cells
Myeloid
Erythroid
Subset within human CD45+ gate
B
Marrow Reconstitution: 1° Transplants 16 wks
%Positive
Human CD45
B cells
T cells
Myeloid
Erythroid
CD34+
Subset within human CD45+ gate

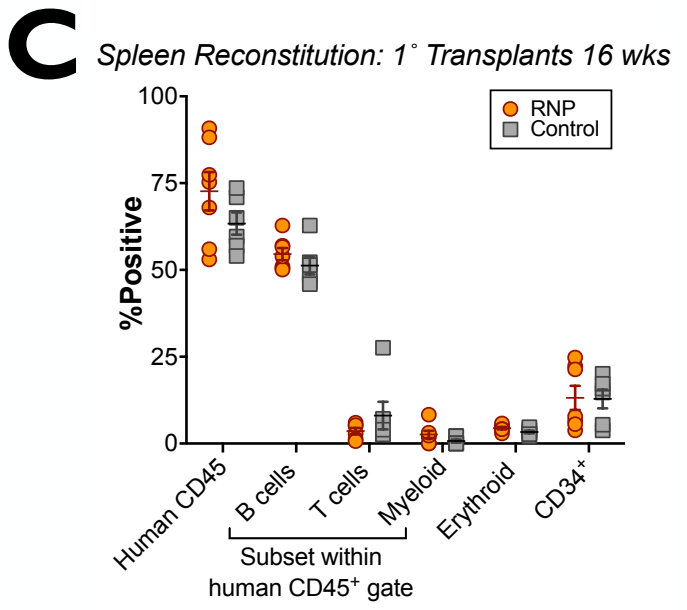
C
Spleen Reconstitution: 1° Transplants 16 wks
%Positive
RNP
Control
Human CD45
B cells
T cells
Myeloid
Erythroid
CD34+
Subset within human CD45+ gate

FIGURE 2. Cas9 RNP-treated human CD34⁺ cells support long-term blood reconstitution in NSG mice. Human hematopoiesis *in vivo* as determined by flow cytometry analysis in the **A.** blood, **B.** bone marrow, and **C.** Spleen of 1° recipients 16 weeks after RNP-treated CD34⁺ cell transplantation.

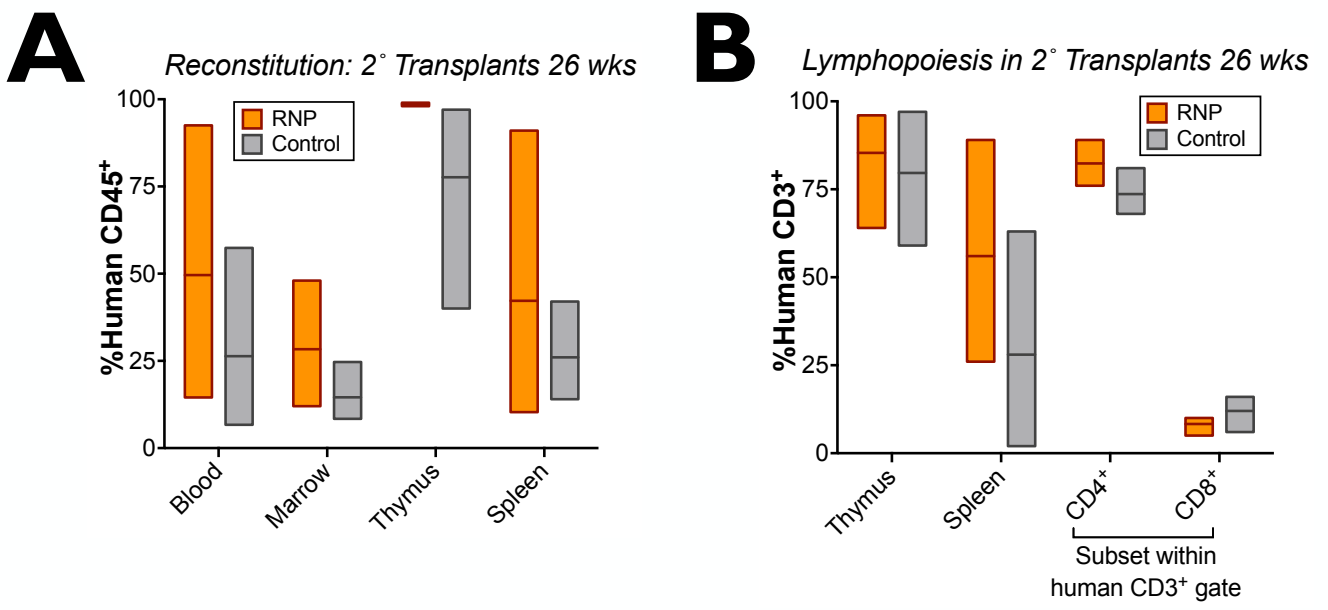
A
Reconstitution: 2° Transplants 26 wks
%Human CD45+
RNP
Control
Blood
Marrow
Thymus
Spleen
B
Lymphopoiesis in 2° Transplants 26 wks
%Human CD3+
Thymus
Spleen
CD4+
CD8+
Subset within human CD3+ gate

FIGURE 3. Cas9 RNP-treated human CD34⁺ cells repopulate hematopoiesis in secondary recipient mice. A. Reconstitution of human blood in hematopoietic organs and **B.** T cell lymphopoiesis in the thymus and spleen of 2° transplant recipient mice.

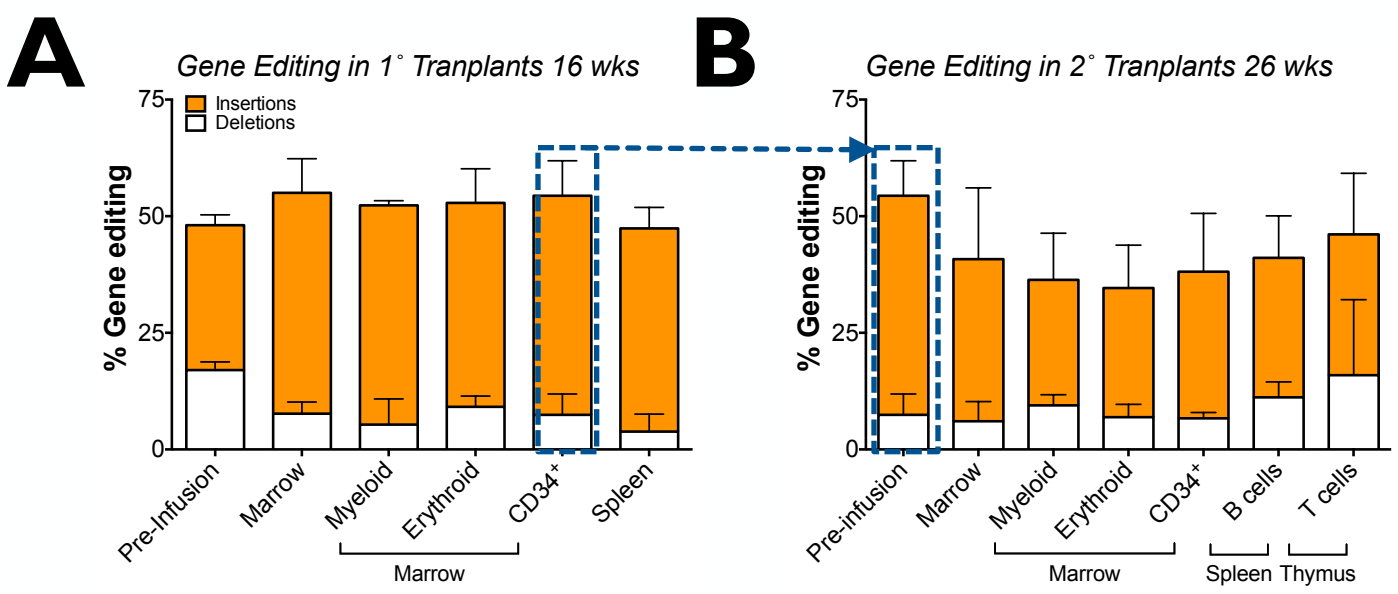
A
Gene Editing in 1° Tranplants 16 wks
% Gene editing
Insertions
Deletions
Pre-Infusion
Marrow
Myeloid
Erythroid
CD34+
Spleen
Marrow
B
Gene Editing in 2° Tranplants 26 wks
Pre-infusion
Marrow
Myeloid
Erythroid
CD34+
B cells
T cells
Marrow
Spleen
Thymus

FIGURE 4. Gene-edited human CD34⁺ cells differentiate into gene-edited myeloid, erythroid, and lymphoid progeny *in vivo*. Gene editing levels detected in the human cells that repopulated the indicated hematopoietic organs and human blood subsets in 1° (NSG) and 2° (NSG-SGM3) mice.

Additional acknowledgements: Thanks to Luis Barrera (Bioinformatics), Carrie Margulies, Tanushree Phadke (DNA library preparation), Georgia Giannoukos, Dawn Ciulla (DNA sequencing), Anne Bothmer.