Knock-out of *CISH* and *TGFβR2* or knock-in of *CD16* and *mbIL-15* in iPSC-derived NK cells promotes high cytotoxicity and enhances *in vivo* tumor killing

POSTER 2780

Alexandra Gerew, Alexander G. Allen, Laura Blaha, Kaitlyn Izzo, Jared Getgano, Nadire R. Cochran, Barsha Pokharel, Patricia Sousa, Stephen Sherman, Tusneem Janoudi, Steven Sexton, Kevin Wasko, Mark S. Shearman, Kate Zhang, Kai-Hsin Chang, and Samia Q. Khan

Editas Medicine, Inc., Cambridge, MA, USA

OBJECTIVE

- To evaluate the *in vitro* and *in vivo* anti-tumor efficacy of induced pluripotent stem cell (iPSC)-derived natural killer (iNK) cell therapies modified using Editas' engineered highly active and specific AsCas12a to double knock-out (DKO) *CISH* and *TGFβR2* or double knock-in (DKI) *CD16* and *mbIL-15*.

INTRODUCTION

- Natural killer (NK) cells are strong effectors of the innate immune system that can directly recognize and kill tumor cells, hence making them attractive for use as cellular therapies in cancer.
- NK cell effectiveness is diminished by the lack of persistence due to low levels of critical cytokines required for NK survival, such as interleukin-15 (IL-15), as well as tumor-intrinsic immunosuppressive mechanisms, such as high levels of transforming growth factor beta (TGFβ) within the tumor microenvironment.[1] Furthermore, NK cells' ability to exert antibody-dependent cellular cytotoxicity (ADCC) is impaired when CD16 is cleaved off after NK cell activation.[2] Editas' proprietary AsCas12a-based gene editing method was used to modify iNK cells to overcome these biologic limitations and enhance NK cell function.
- We hypothesized that DKO of the cytokine-inducible SH2-containing protein (*CISH*) gene, a negative regulator of IL-2/IL-15 signaling, and of the TGFβ-receptor 2 (*TGFβR2*) gene would improve NK cell effector function and make them resistant to TGFβ-mediated suppression. Additionally, we hypothesized that DKI of membrane-bound IL-15 (*mbIL-15*) and *CD16* into NK cells would prolong NK persistence and increase cytotoxicity when combined with ADCC-inducing tumor-targeting antibodies, such as trastuzumab.

Enhanced functions of AsCas12a-edited iNK cells

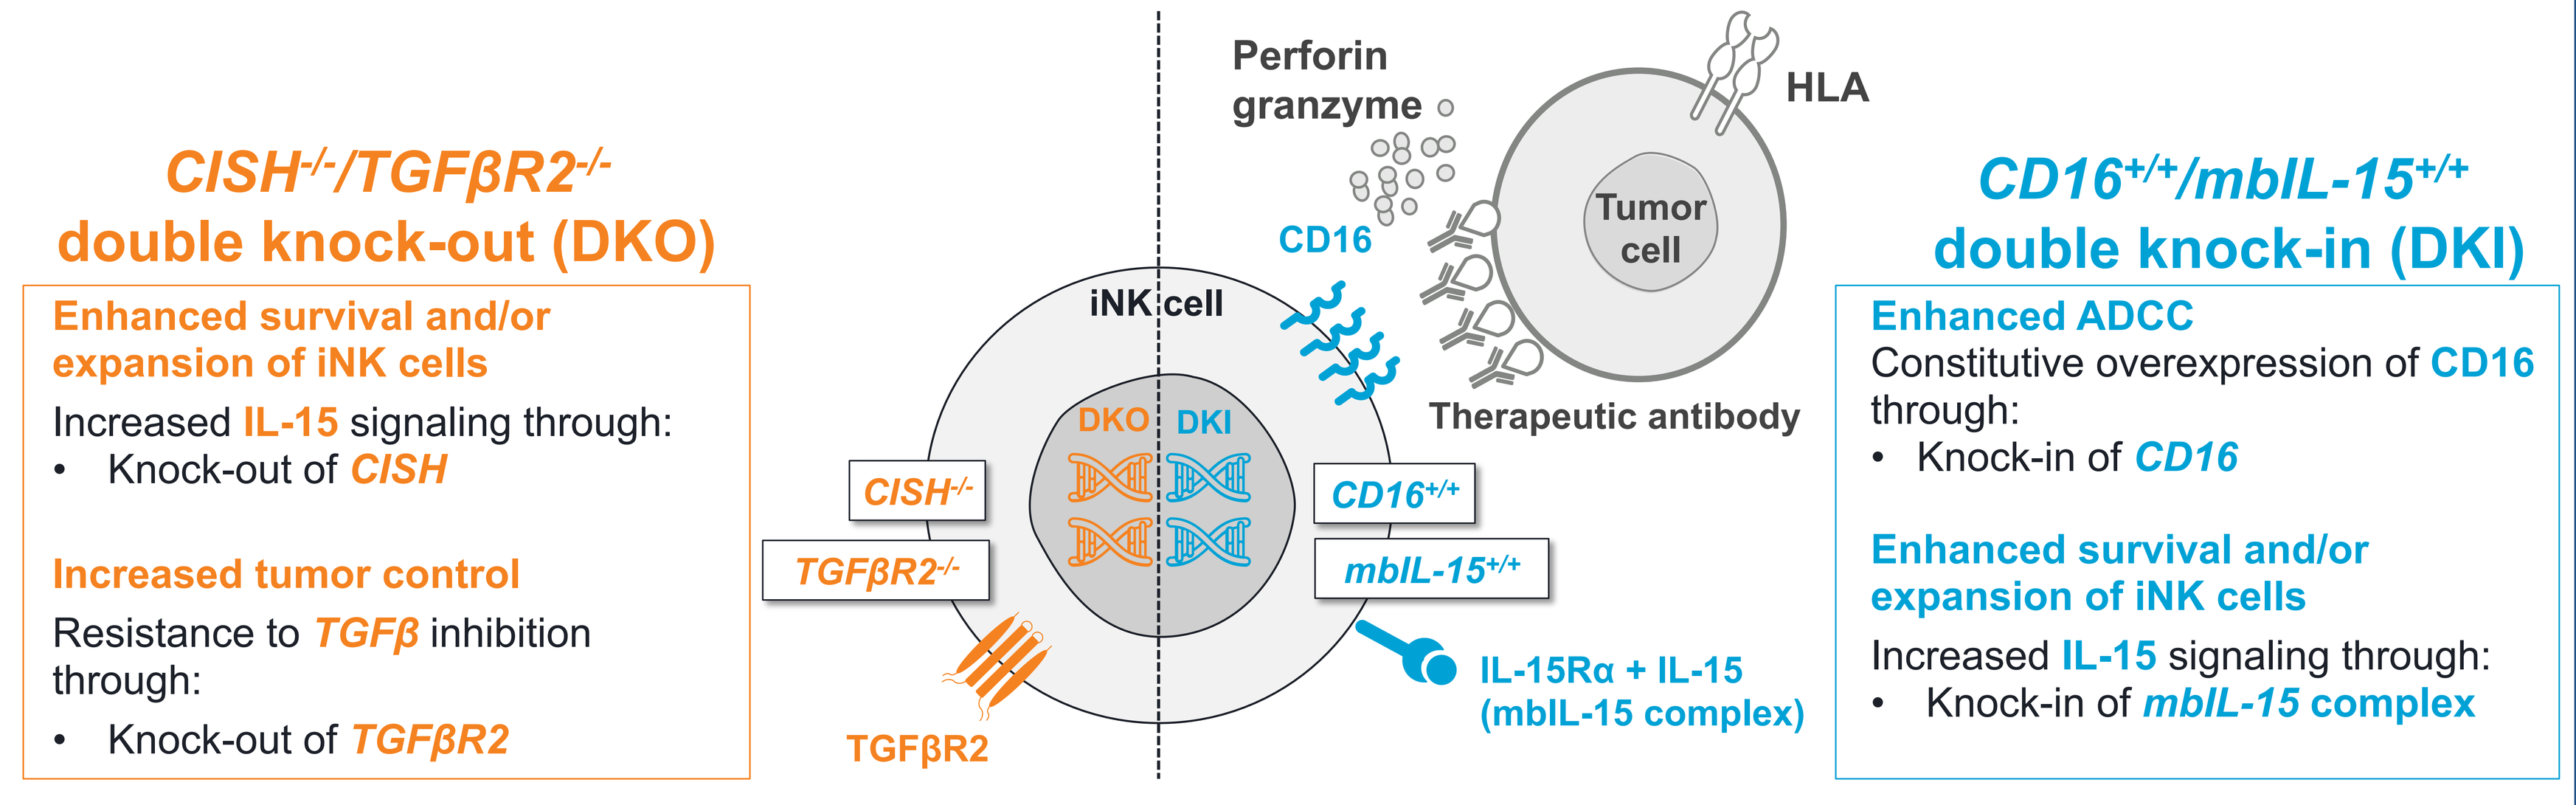
CISH-/-/TGFβR2-/-
double knock-out (DKO)
Enhanced survival and/or expansion of iNK cells
Increased IL-15 signaling through:
• Knock-out of CISH
Increased tumor control
Resistance to TGFβ inhibition through:
• Knock-out of TGFβR2
Perforin granzyme
HLA
Tumor cell
CD16
iNK cell
DKO
DKI
Therapeutic antibody
CISH-/-
TGFβR2-/-
CD16+/+
mbIL-15+/+
TGFβR2
IL-15Rα + IL-15 (mbIL-15 complex)
CD16+/+/mbIL-15+/+
double knock-in (DKI)
Enhanced ADCC
Constitutive overexpression of CD16 through:
• Knock-in of CD16
Enhanced survival and/or expansion of iNK cells
Increased IL-15 signaling through:
• Knock-in of mbIL-15 complex

ADCC, antibody-dependent cellular cytotoxicity; CISH, cytokine-inducible SH2-containing protein; DKI, double knock-in; DKO, double knock-out; HLA, human leukocyte antigen; IL-15, interleukin-15; IL-15Rα, interleukin-15 receptor alpha; iNK, induced pluripotent stem cell-derived natural killer; mbIL-15, membrane-bound IL-15; TGFβ, transforming growth factor beta; TGFβR2, transforming growth factor beta receptor 2.

METHODS

- iPSCs were edited at the glyceraldehyde-3-phosphate dehydrogenase (GAPDH) locus with an engineered AsCas12a to knock-in *CD16* and *mbIL-15* using the SeLection by Essential-gene Exon Knock-in (SLEEK) method[3]. Separate iPSCs were edited with AsCas12a to knock-out *CISH* and *TGFβR2*. iPSC clones were then differentiated into iPSC-derived NK (iNK) cells.
- A 3D tumor spheroid killing assay using Incucyte® imaging of NucLight Red (NLR)-tagged SKOV-3 cells was performed to assess *in vitro* NK cell cytotoxicity.
- Non-obese diabetic (NOD) severe combined immunodeficient (scid) gamma (NSG) mice were inoculated with 0.25–1.00 x 10^6 luciferase (luc)-expressing SKOV-3 cell line (SKOV-3-luc) ovarian tumor cells. Mice received a single intraperitoneal (IP) dose of 5–20 million wild-type (WT) iNK, DKO iNK, or DKI iNK cells with or without a single or multi-dose IP of 2.5 mg/kg trastuzumab. Tumor burden was calculated using a Perkin Elmer bioluminescent *in vivo* imaging system (IVIS). Mice were sacrificed at days 90 and 118 to assess iNK persistence and CD16 expression in the peritoneal cavity by flow cytometry.

RESULTS

Figure 1. DKO iNK cells induced enhanced anti-tumor activity against 3D SKOV-3 ovarian tumor spheroids and increased inflammatory cytokine production

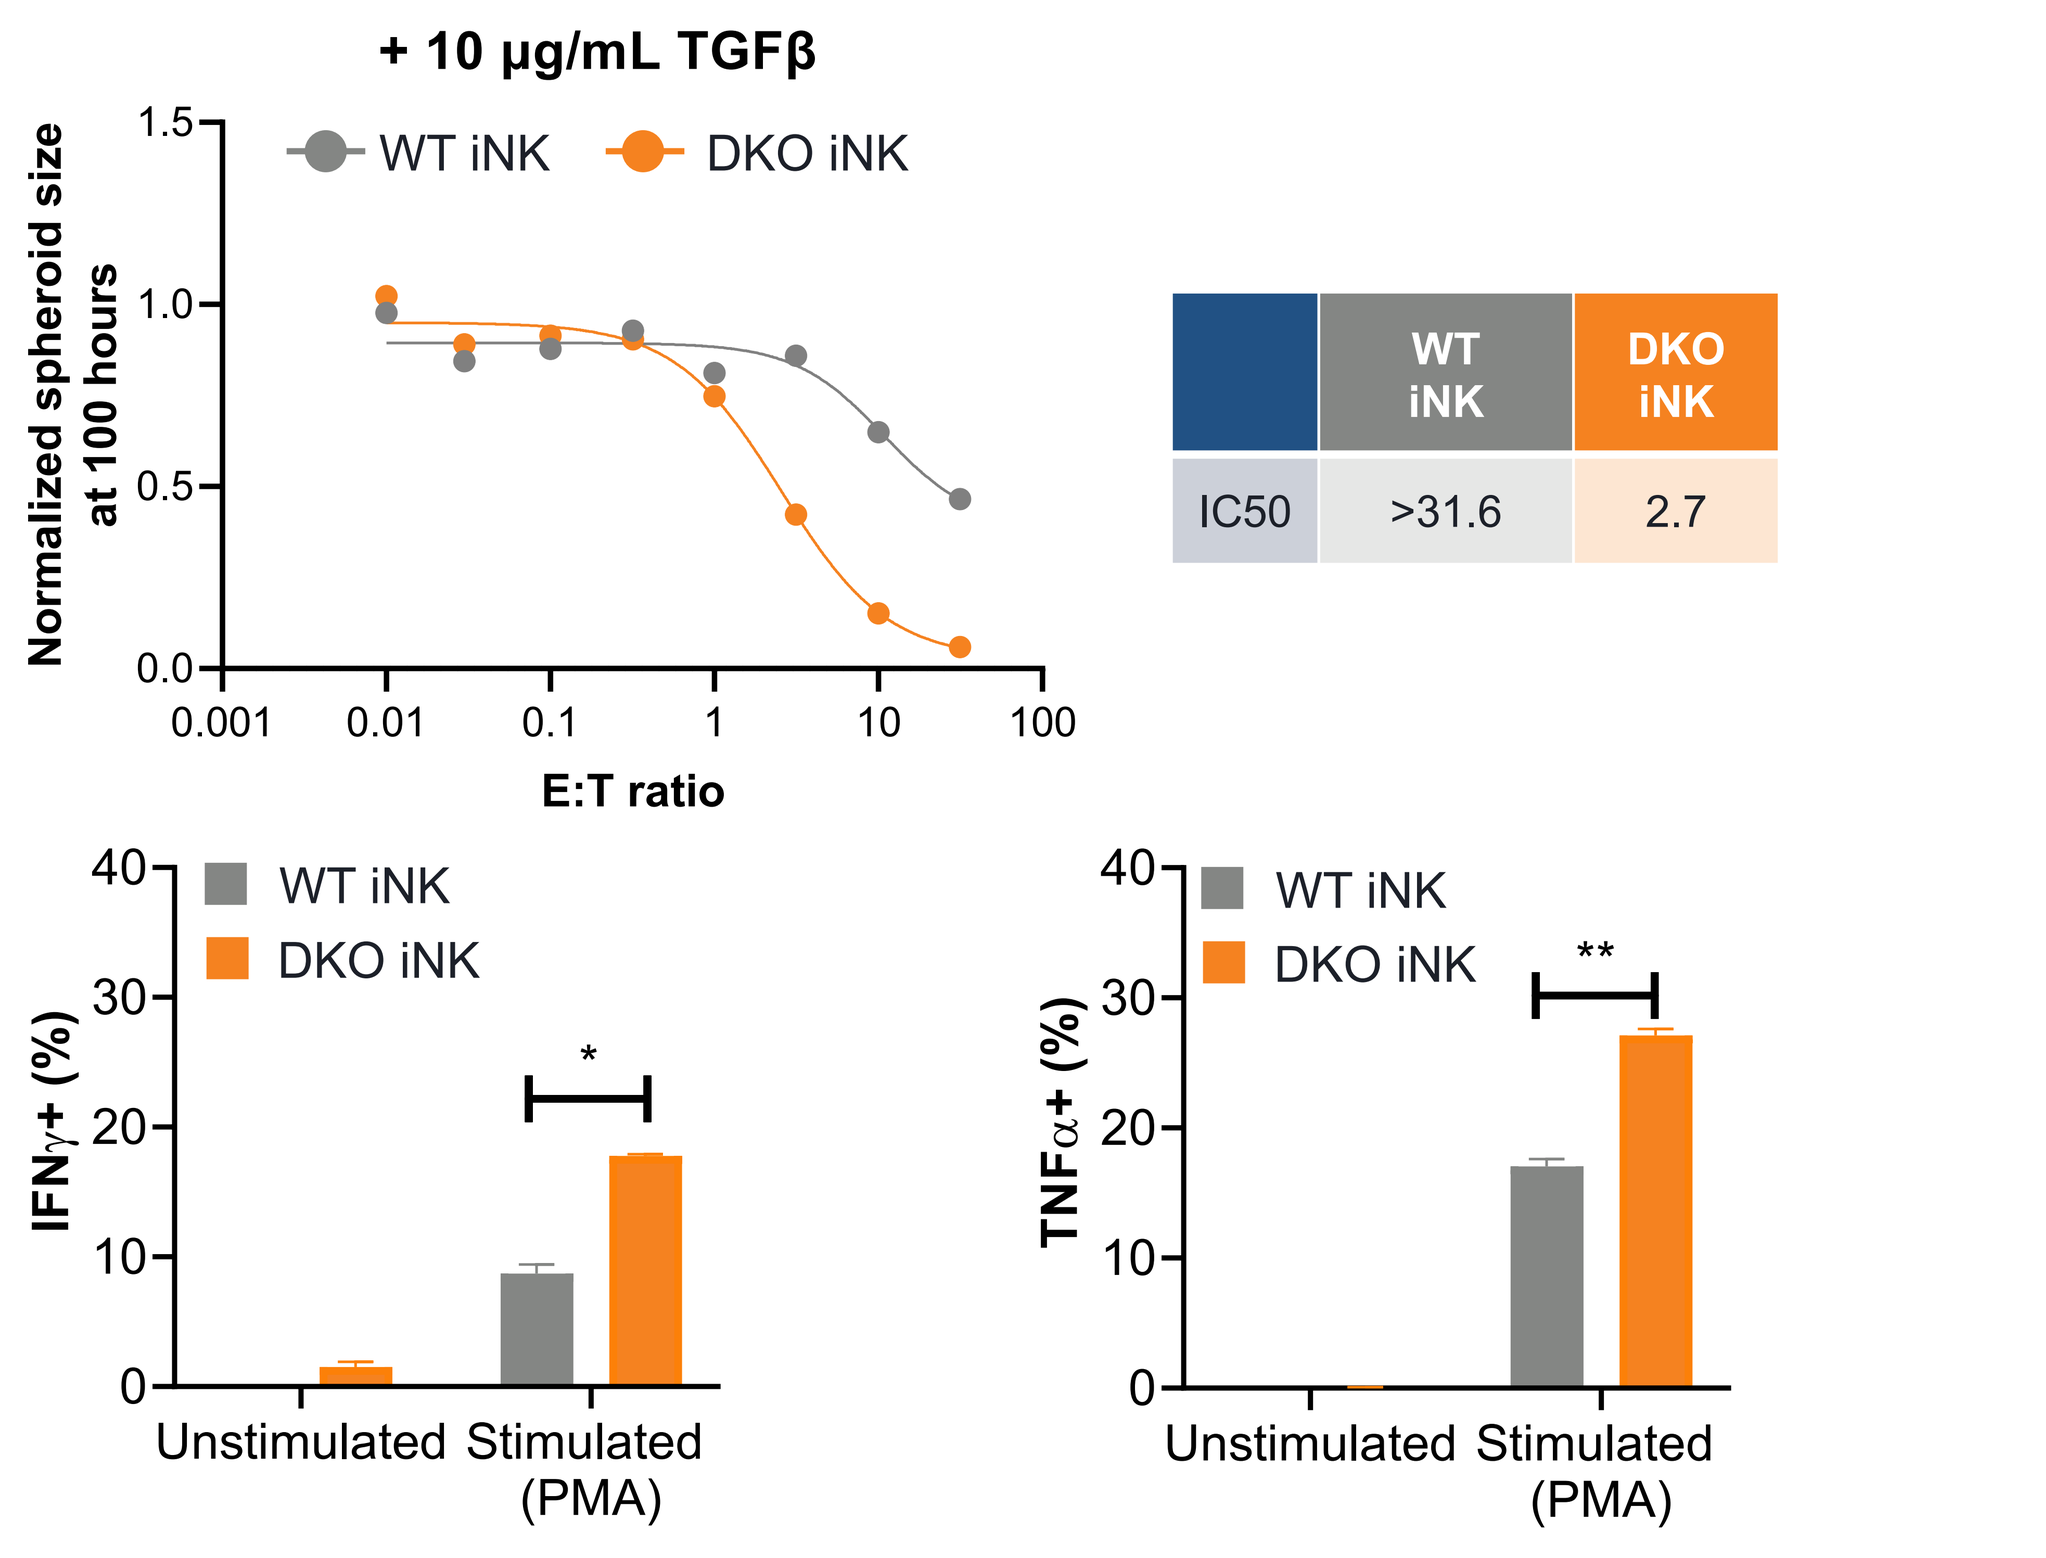
+ 10 µg/mL TGFβ
WT iNK
DKO iNK
Normalized spheroid size at 100 hours
E:T ratio
IC50: WT iNK >31.6; DKO iNK 2.7
IFNγ+ (%)
TNFα+ (%)
Unstimulated
Stimulated (PMA)

Number of independent experiments=2 ; *p<0.05, **p<0.01. 3D, 3 dimensional; DKO, double knock-out; E:T, effector-to-target; IC50, half maximal inhibitory concentration; IFNγ, interferon gamma; iNK, induced pluripotent stem cell-derived natural killer; PMA, phorbol 12-myristate 13-acetate; TGFβ, transforming growth factor beta; TNFα, tumor necrosis factor alpha; WT, wild-type.

Figure 2. DKI iNK cells induced enhanced anti-tumor activity against 3D SKOV-3 ovarian tumor spheroids compared with WT iNK cells

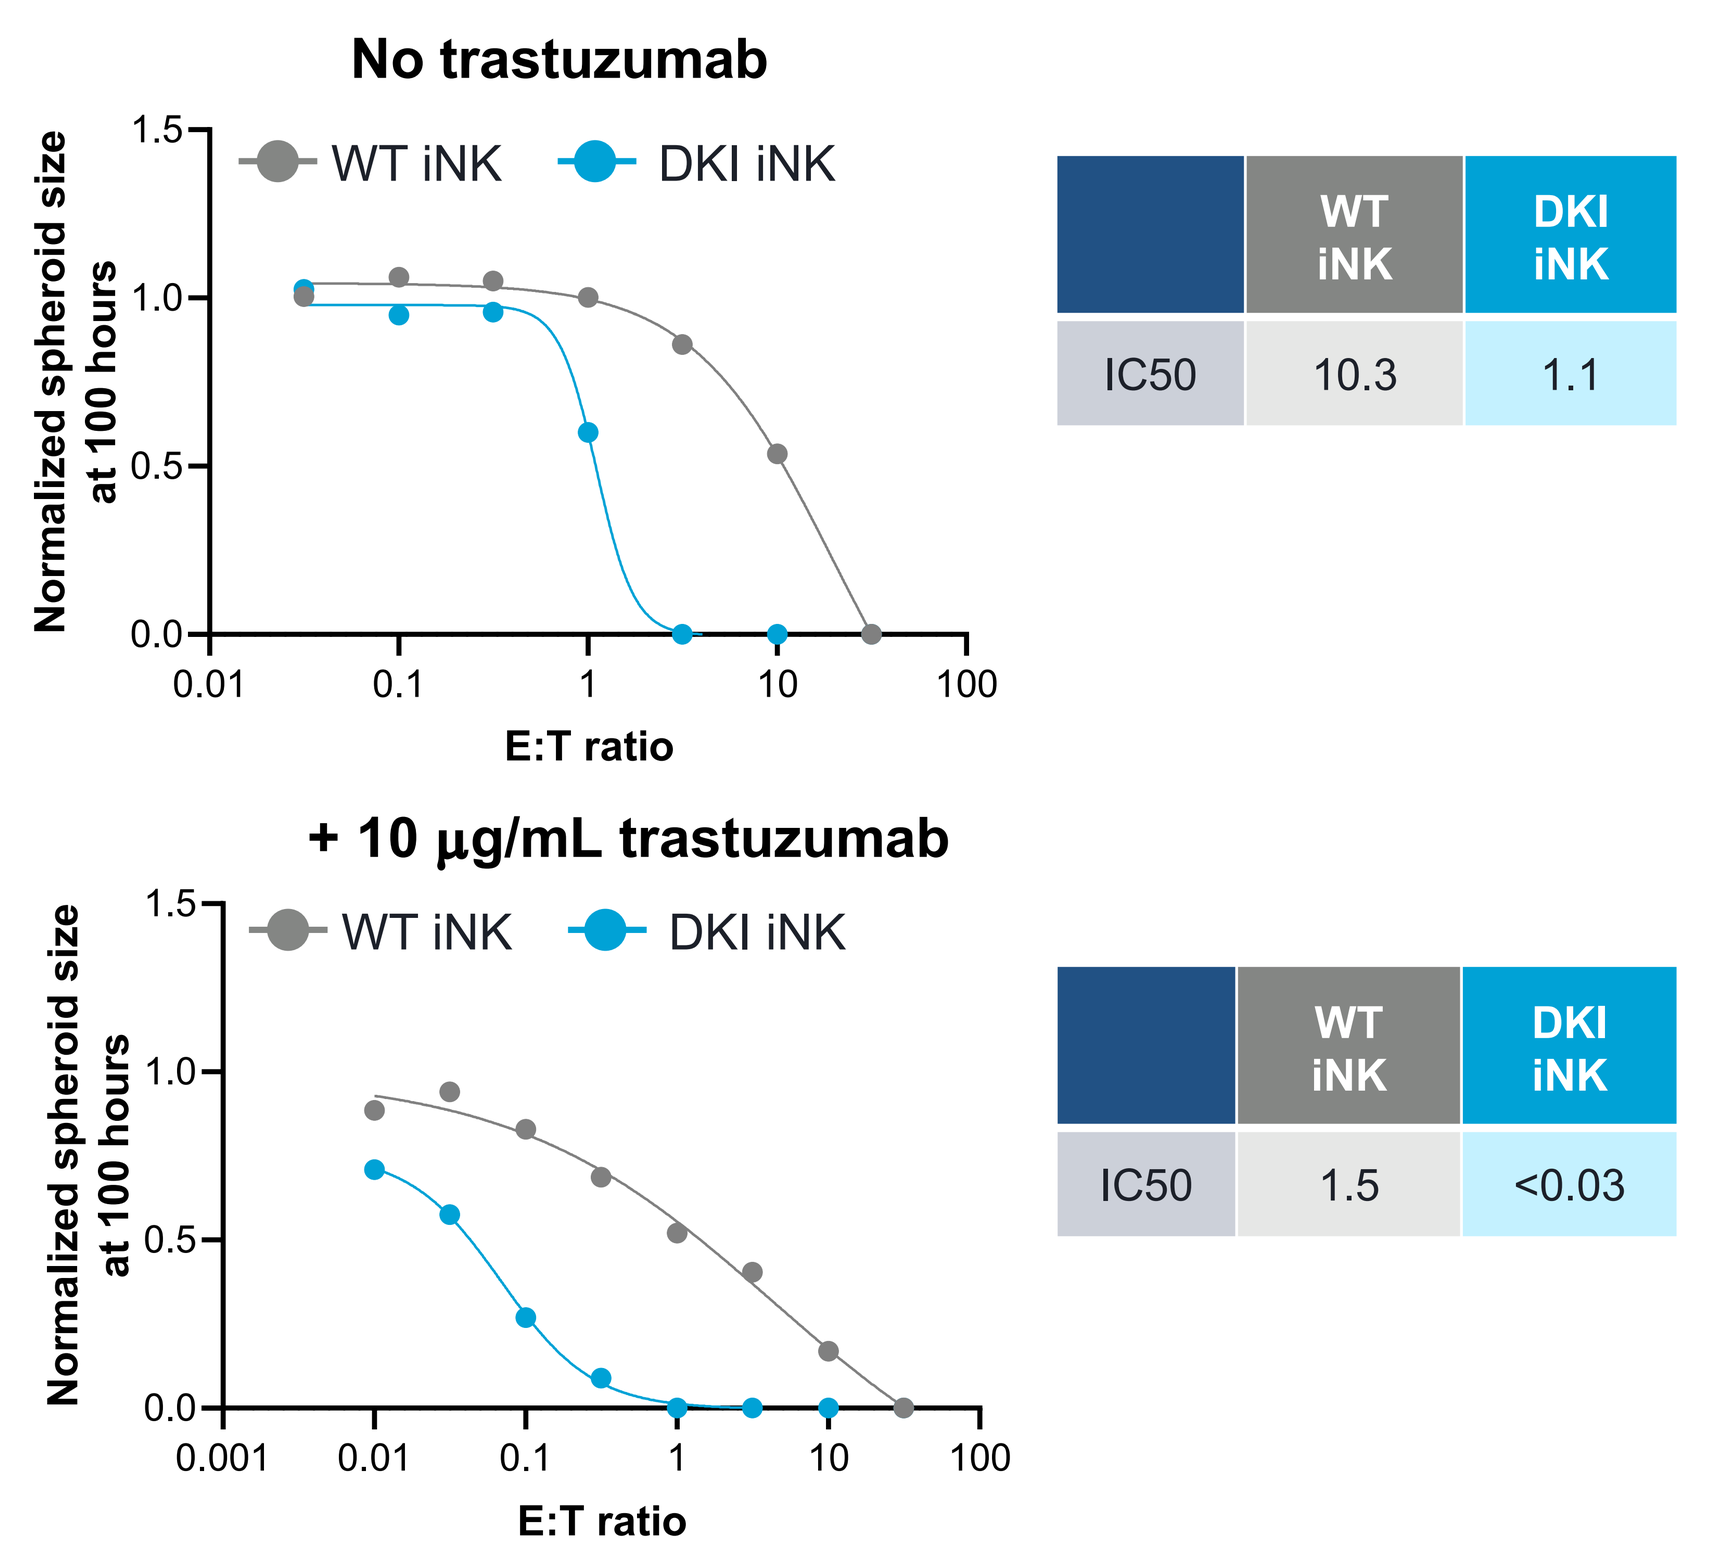
No trastuzumab
WT iNK
DKI iNK
IC50: WT iNK 10.3; DKI iNK 1.1
+ 10 µg/mL trastuzumab
IC50: WT iNK 1.5; DKI iNK <0.03
Normalized spheroid size at 100 hours
E:T ratio

Number of independent experiments=2. 3D, 3 dimensional; DKI, double knock-in; E:T, effector-to-target; IC50, half maximal inhibitory concentration; iNK, induced pluripotent stem cell-derived natural killer; WT, wild-type.

Figure 3. DKO iNK cells induced significant reduction in tumor burden compared with WT iNK cells in SKOV-3-luc IP tumor-bearing mice

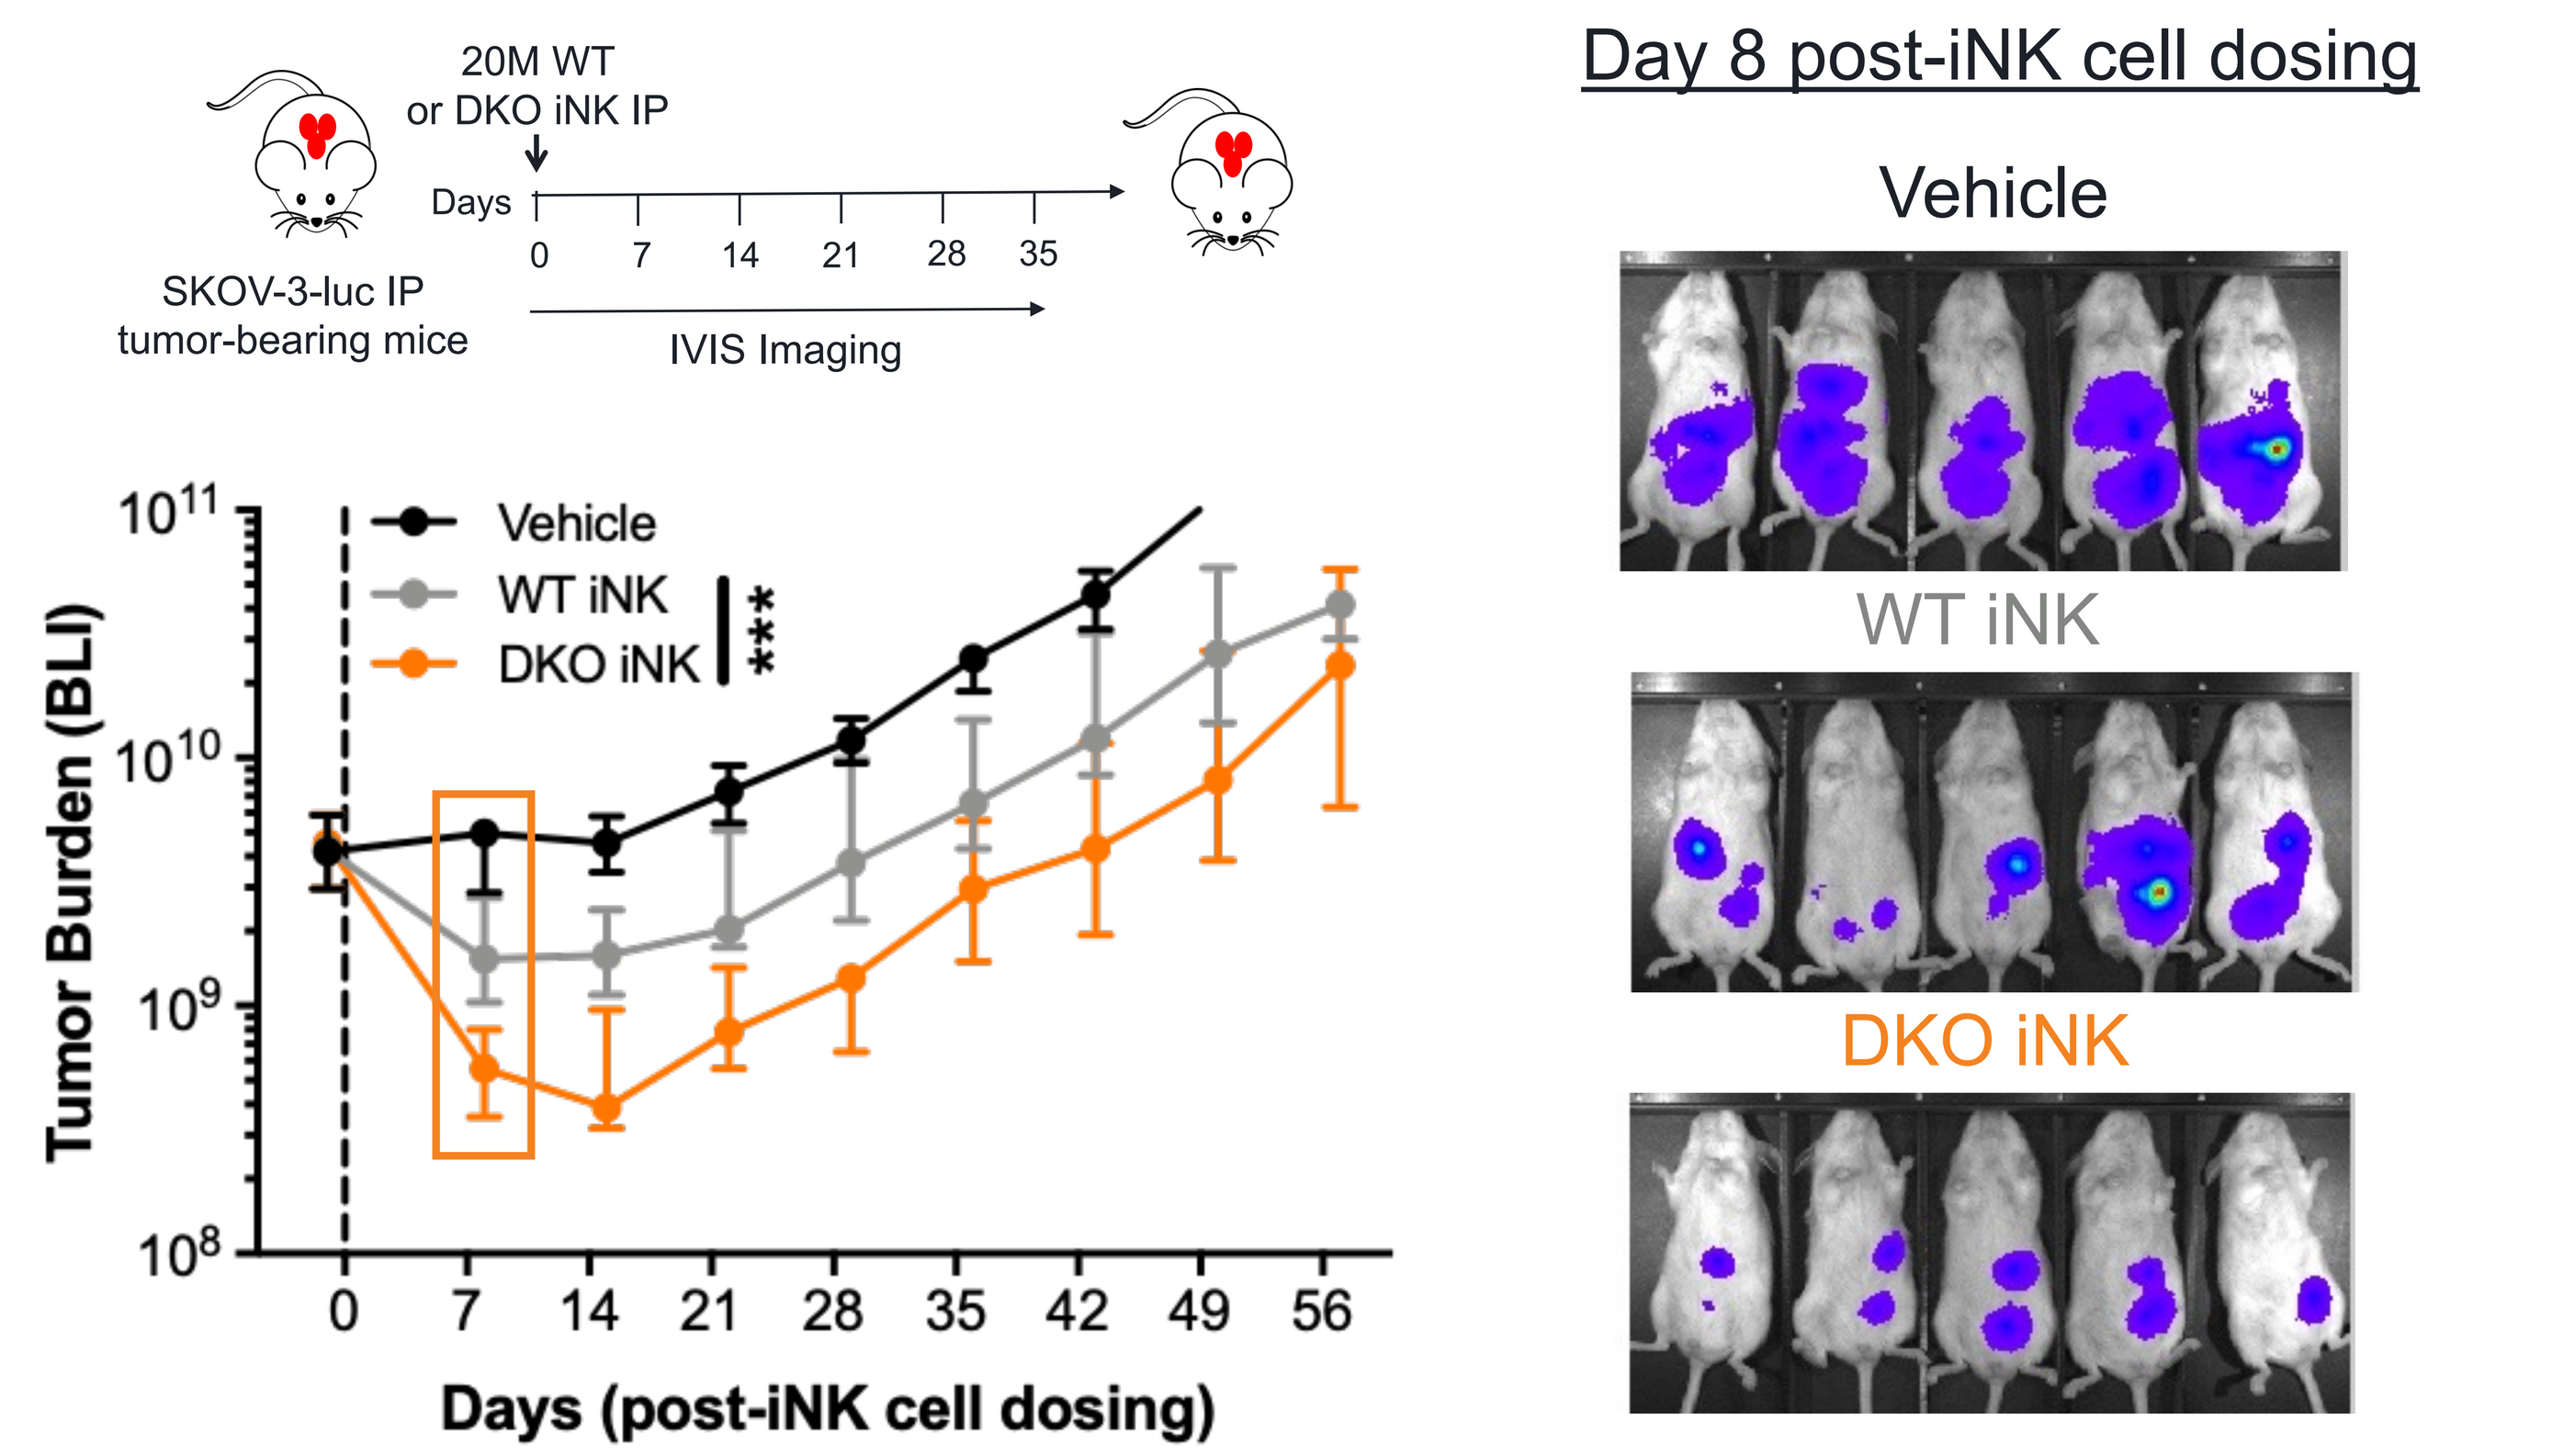
20M WT or DKO iNK IP
Days
SKOV-3-luc IP tumor-bearing mice
IVIS Imaging
Vehicle
WT iNK
DKO iNK
Tumor Burden (BLI)
Days (post-iNK cell dosing)
Day 8 post-iNK cell dosing

Number of independent experiments=1; ***p<0.001. BLI, bioluminescent imaging; DKO, double knock-out; IP, iNK, induced pluripotent stem cell-derived natural killer; SKOV-3-luc, luciferase-expressing SKOV-3 cell line; IP, intraperitoneal; WT, wild-type.

Figure 4. DKI iNK cells administered in combination with a single dose or multiple doses of trastuzumab induced significant to complete tumor clearance in SKOV-3-luc IP tumor-bearing mice compared with WT iNK cells

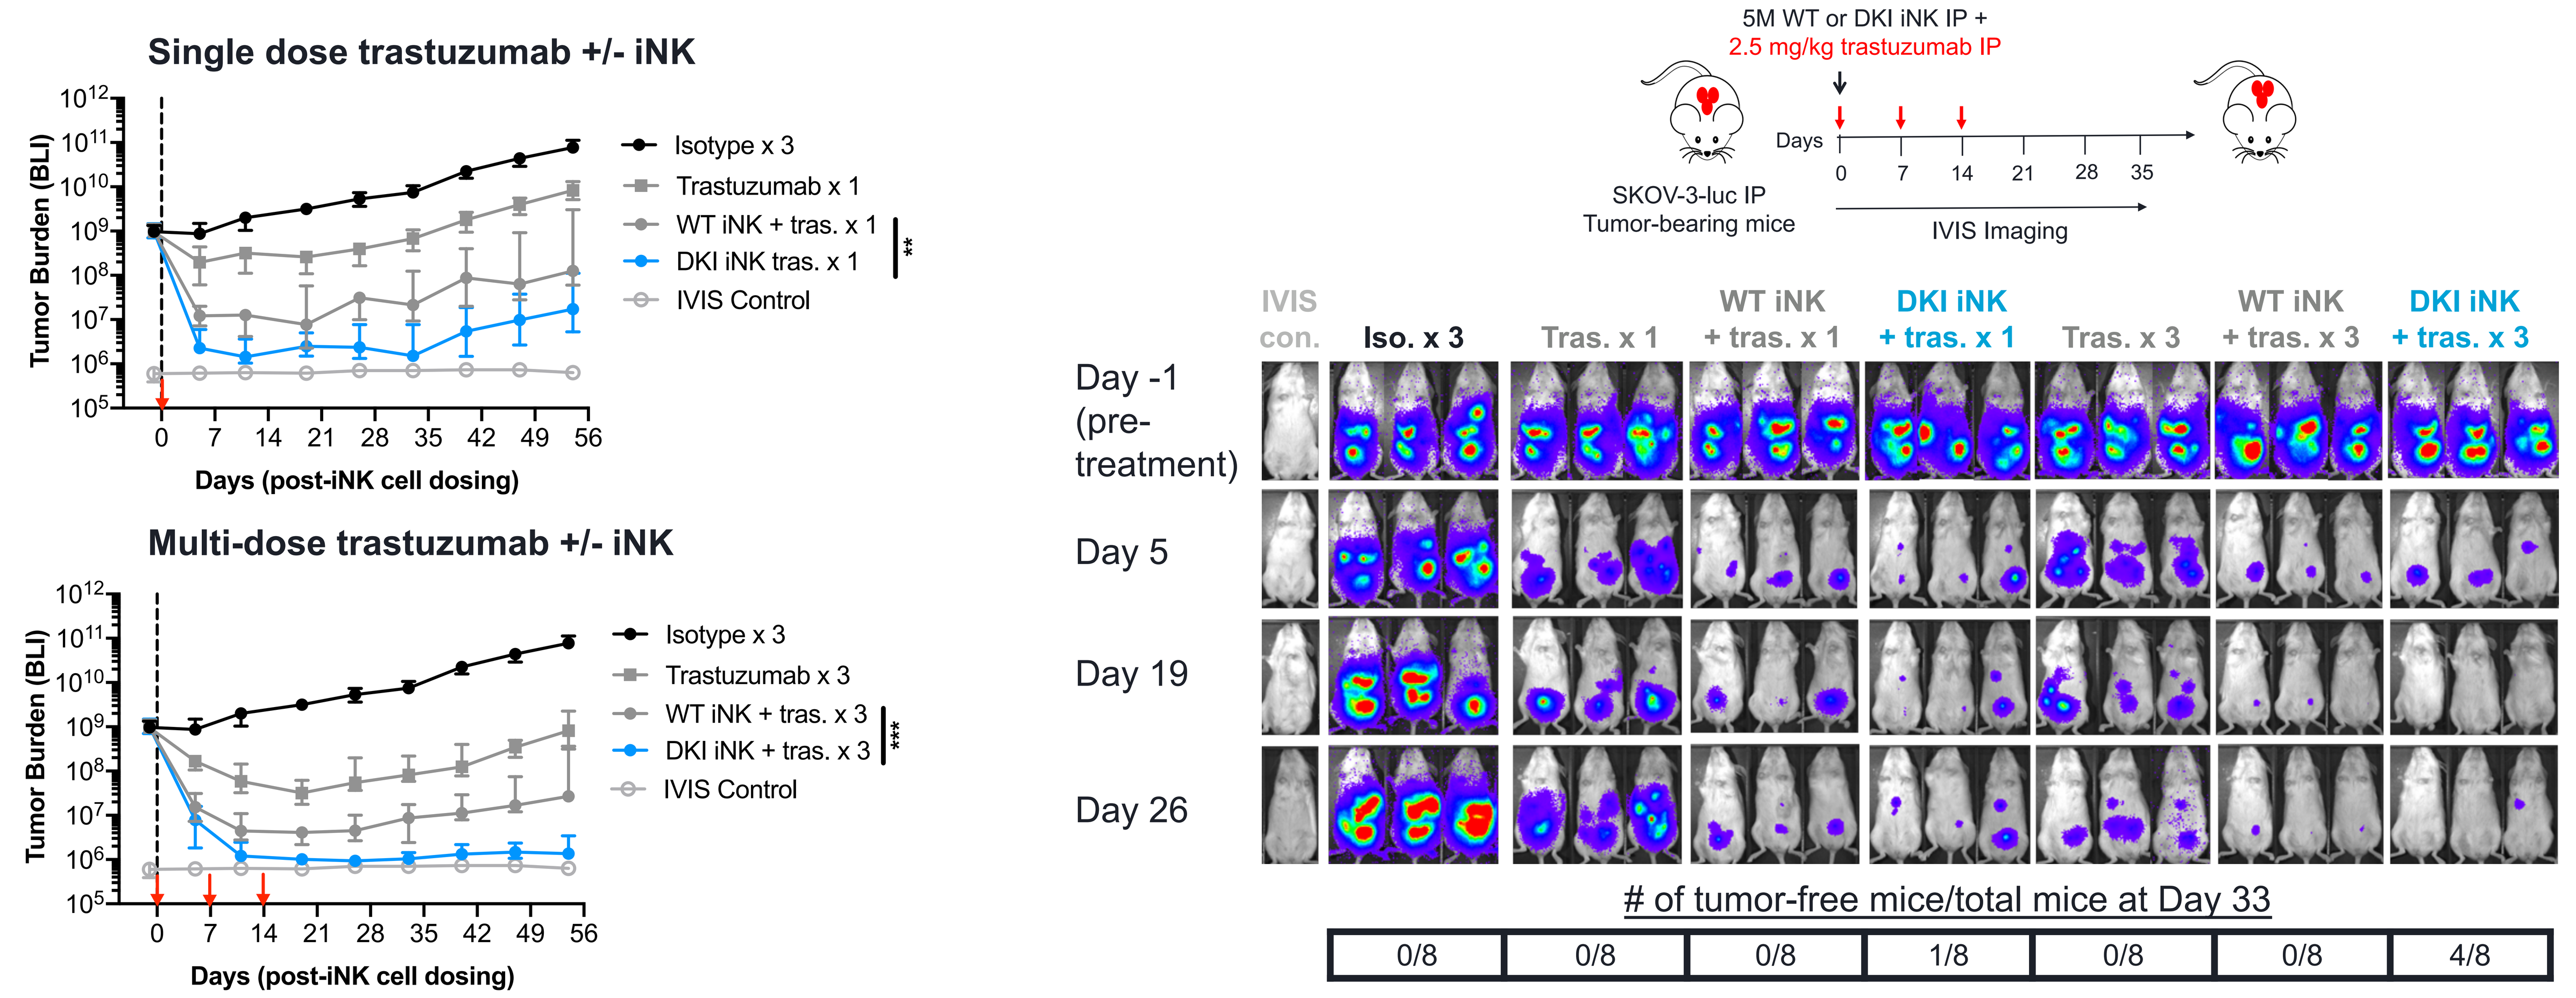
Single dose trastuzumab +/- iNK
Isotype x 3
Trastuzumab x 1
WT iNK + tras. x 1
DKI iNK tras. x 1
IVIS Control
Multi-dose trastuzumab +/- iNK
Isotype x 3
Trastuzumab x 3
WT iNK + tras. x 3
DKI iNK + tras. x 3
IVIS Control
Tumor Burden (BLI)
Days (post-iNK cell dosing)
5M WT or DKI iNK IP +
2.5 mg/kg trastuzumab IP
SKOV-3-luc IP Tumor-bearing mice
IVIS Imaging
IVIS con.
Iso. x 3
Tras. x 1
WT iNK + tras. x 1
DKI iNK + tras. x 1
Tras. x 3
WT iNK + tras. x 3
DKI iNK + tras. x 3
Day -1 (pre-treatment)
Day 5
Day 19
Day 26
of tumor-free mice/total mice at Day 33
0/8 | 0/8 | 0/8 | 1/8 | 0/8 | 0/8 | 4/8

Number of independent experiments=2; ns, p>0.05; **p<0.01; ***p<0.001. BLI, bioluminescent imaging; con., control; DKI, double knock-in; iNK, induced pluripotent stem cell-derived natural killer; IP, intraperitoneal; iso., isotype; IVIS, *in vivo* imaging system; SKOV-3-luc, luciferase-expressing SKOV-3 cell line; tras., trastuzumab; WT, wild-type.

Figure 5. DKI iNK cells had prolonged *in vivo* persistence and expressed high levels of CD16 up to Day 118 post-dosing without exogenous cytokine support

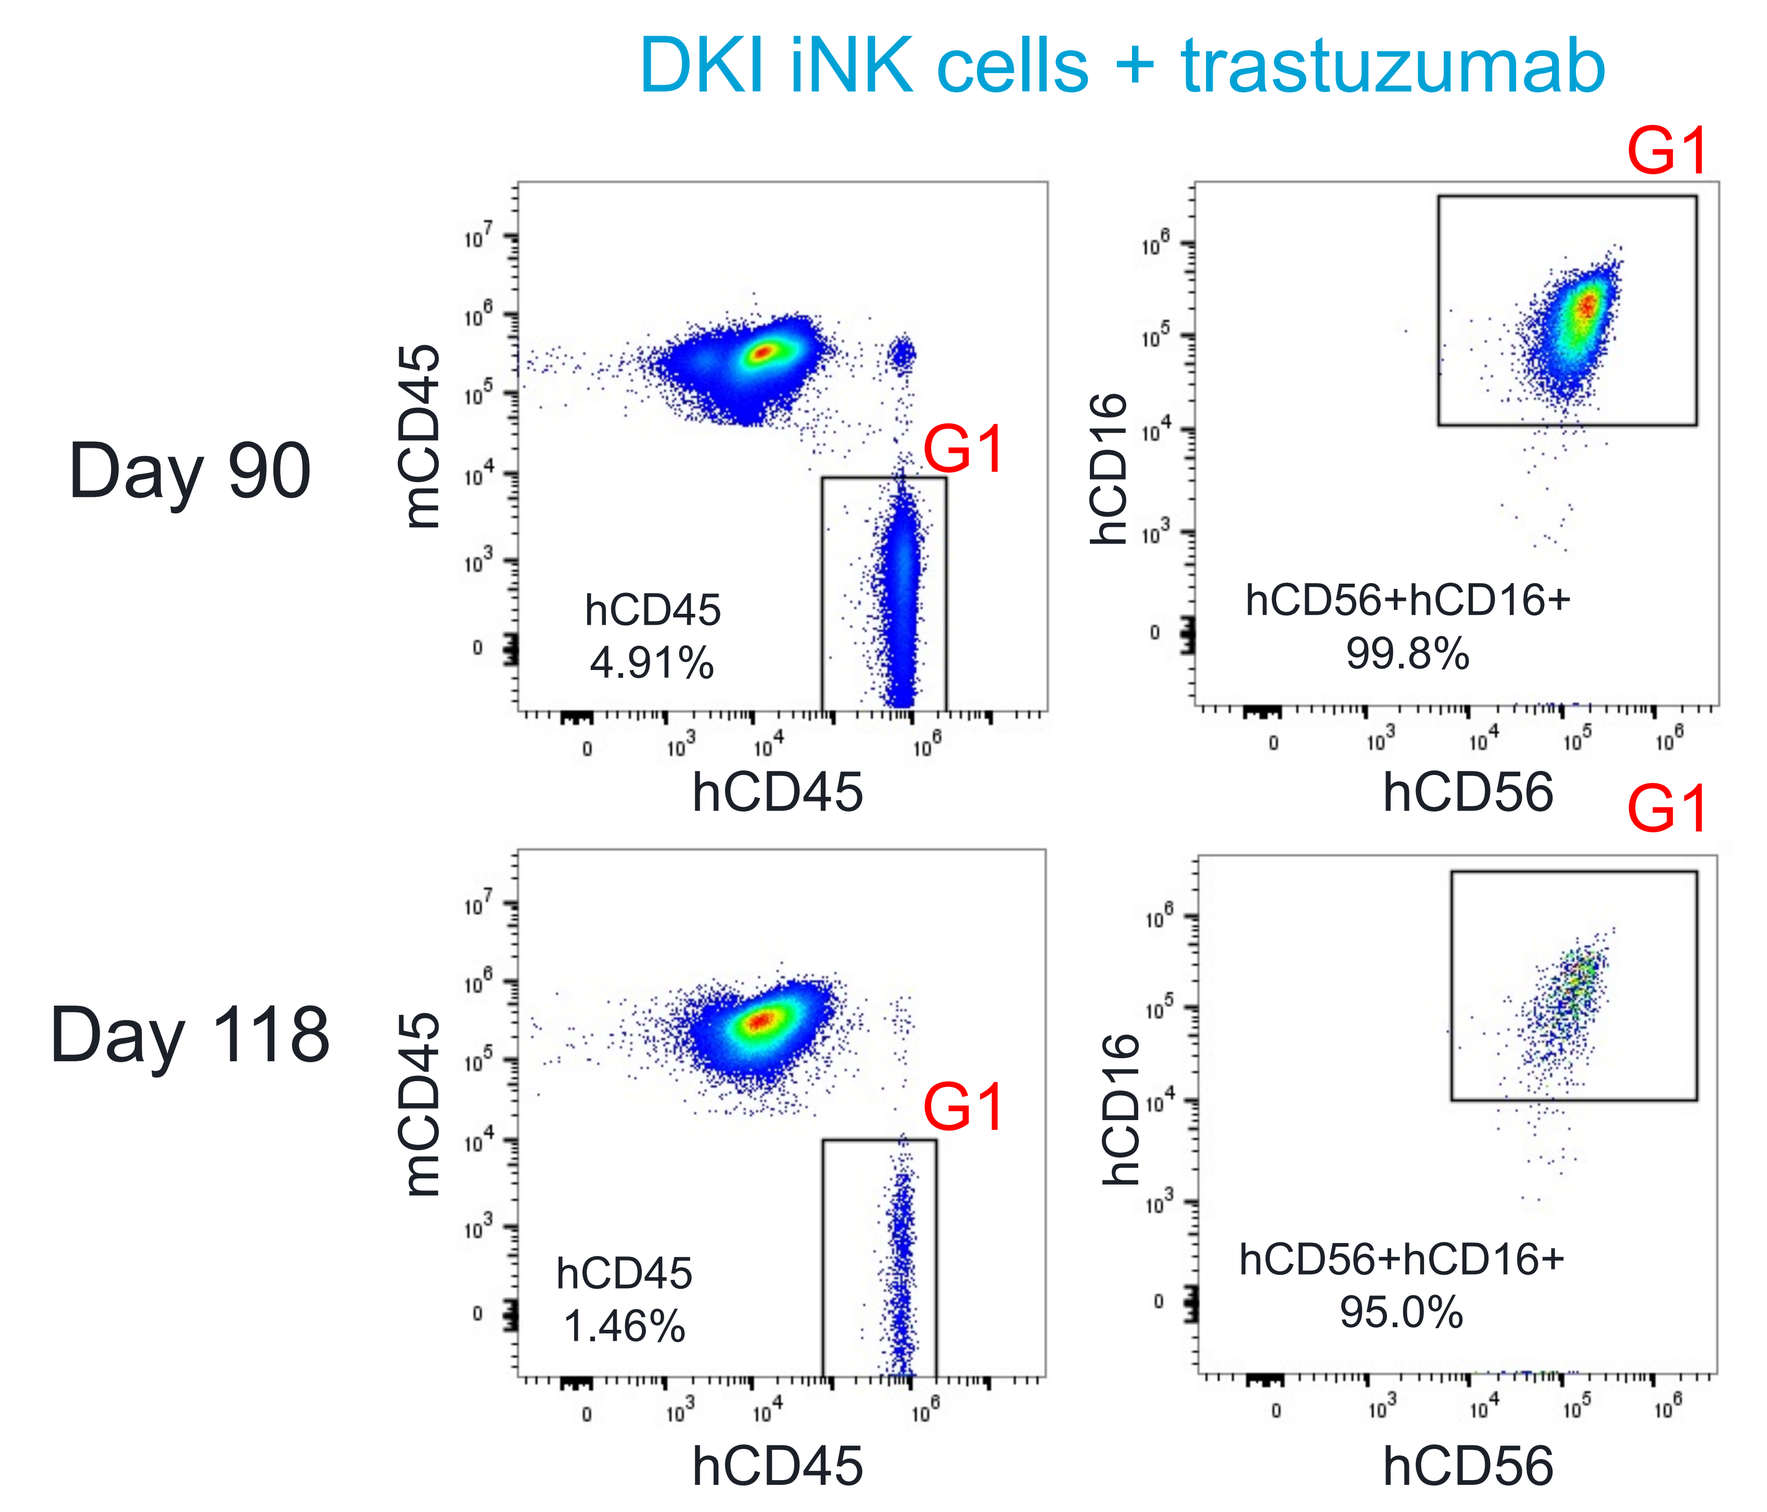
Peritoneal cavity
DKI iNK cells + trastuzumab
Day 90
Day 118
mCD45
hCD45
hCD16
hCD56
G1
hCD45 4.91%
hCD56+hCD16+ 99.8%
hCD45 1.46%
hCD56+hCD16+ 95.0%

Number of independent experiments =2. DKI, double knock-in; iNK, induced pluripotent stem cell-derived natural killer.

CONCLUSIONS

- DKO and DKI iNK cells induced enhanced anti-tumor activity against 3D SKOV-3 tumor spheroids compared with WT iNK cells (Figure 1 and 2). DKO iNK cells had enhanced resistance to TGFβ and, when stimulated, produced elevated levels of cytotoxic inflammatory cytokines, including IFN gamma and TNF alpha (Figure 1).
- DKO iNK cells, administered IP (20M cells) as monotherapy, induced significant reduction in tumor burden compared with WT iNK cells in SKOV-3-luc tumor-bearing mice (Figure 3).
- DKI iNK cells, administered IP (5M cells) in combination with single or multiple doses of trastuzumab, induced significant to complete tumor clearance in multiple mice (Figure 4).
- DKI iNK cells were detected *in vivo* for more than 3 months, indicating that mbIL-15 is expressed at a level sufficient to maintain iNK survival for a prolonged time without exogeneous cytokine support (Figure 5).

REFERENCES

1. Wu Y, et al. Front Immunol 2017;8:930
2. Snyder KM, et al. Front Immunol 2018; 9:2873
3. Editas Medicine. CRISPR Frontiers 2021. https://www.editasmedicine.com/wp-content/uploads/2021/08/2021_CSHL-CRISPR-Frontiers-SLEEK-Zuris_FINAL.pdf

DISCLOSURES

Employees and shareholders of Editas Medicine: A.G., A.G.A., L.B., K.I., J.G., N.R.C., B.P., P.S., S.Sh., T.J., M.S.S., K.Z., K.-H.C. & S.Q.K. Former employees of Editas Medicine: S.S. and K.W.

Acknowledgments:
This work was funded by Editas Medicine. The authors would like to thank all their Editas colleagues for helping to plan, perform, analyze, and present this work. Editorial assistance was provided by Parastoo Momeni, PhD, of 2 the Nth (Cheshire, UK), funded by Editas Medicine.

Presented at: The 63rd Annual Meeting of the American Society of Hematology (ASH 2021)

© 2021 Editas Medicine