1106

EDIT-202, a Multiplexed CRISPR-Cas12a Gene-Edited iPSC-Derived NK Cell Therapy, has Prolonged Persistence, Promotes High Cytotoxicity, and Enhances *In Vivo* Tumor Killing

Samia Q. Khan, Alexander G. Allen, Alexandra Gerew, Kaitlyn M. Izzo, Mrunali Jagdale, Jared Getgano, Nadire R. Cochran, Stephen Sherman, Patricia Sousa, Laura Blaha, Michael Nehil, John A. Follit, Mark S. Shearman, Kate Zhang, Kai-Hsin Chang

Editas Medicine, Inc., Cambridge, MA, USA

OBJECTIVE

To evaluate the *in vitro* and *in vivo* anti-tumor efficacy of EDIT-202, an induced pluripotent stem cell (iPSC)-derived natural killer (iNK) cell therapy generated by using Editas' proprietary engineered, highly active and specific AsCas12a to knock-out *CISH* and *TGFBR2* and knock-in CD16 and membrane bound interleukin-15 (mbIL-15).

INTRODUCTION

- Natural killer (NK) cell-based immunotherapy has emerged as a promising therapeutic approach for solid tumors due to their intrinsic tumor killing capacity, few treatment-related toxicities, and ability to be given to patients as an off-the-shelf therapy.
- NK cell effector function is diminished by the lack of functional persistence, as well as tumor-intrinsic immunosuppressive mechanisms, such as production of transforming growth factor beta (TGFβ), a pleiotropic cytokine that inhibits immune effector function.[1] Furthermore, NK cells' ability to exert antibody-dependent cellular cytotoxicity (ADCC) is impaired when CD16 is cleaved off after NK cell activation.[2]
- We established an iNK platform where biological limitations can be overcome simultaneously via multiplexed gene editing of iPSCs. Using an engineered AsCas12a and our proprietary selection by essential-gene exon knock-in (SLEEK™) editing tool, we successfully generated iPSC clones with double knock-out (DKO) of *CISH* and *TGFβR2*, and double knock-in (DKI) of CD16 and mbIL-15.
- DKO/DKI iPSCs were differentiated into DKO/DKI iNK cells, termed EDIT-202, which were characterized in vitro and in vivo to show edit-mediated enhancement of NK effector functions.

Enhanced functions of AsCas12a-edited iNK cells

***CISH**$^{-/-}$/**TGFβR2**$^{-/-}$* **double knock-out (DKO)**

Enhanced survival and/or expansion of iNK cells

Increased **IL-15** signaling through:
- Knock-out of ***CISH***

Increased tumor control

Resistance to ***TGFβ*** inhibition through:
- Knock-out of ***TGFβR2***

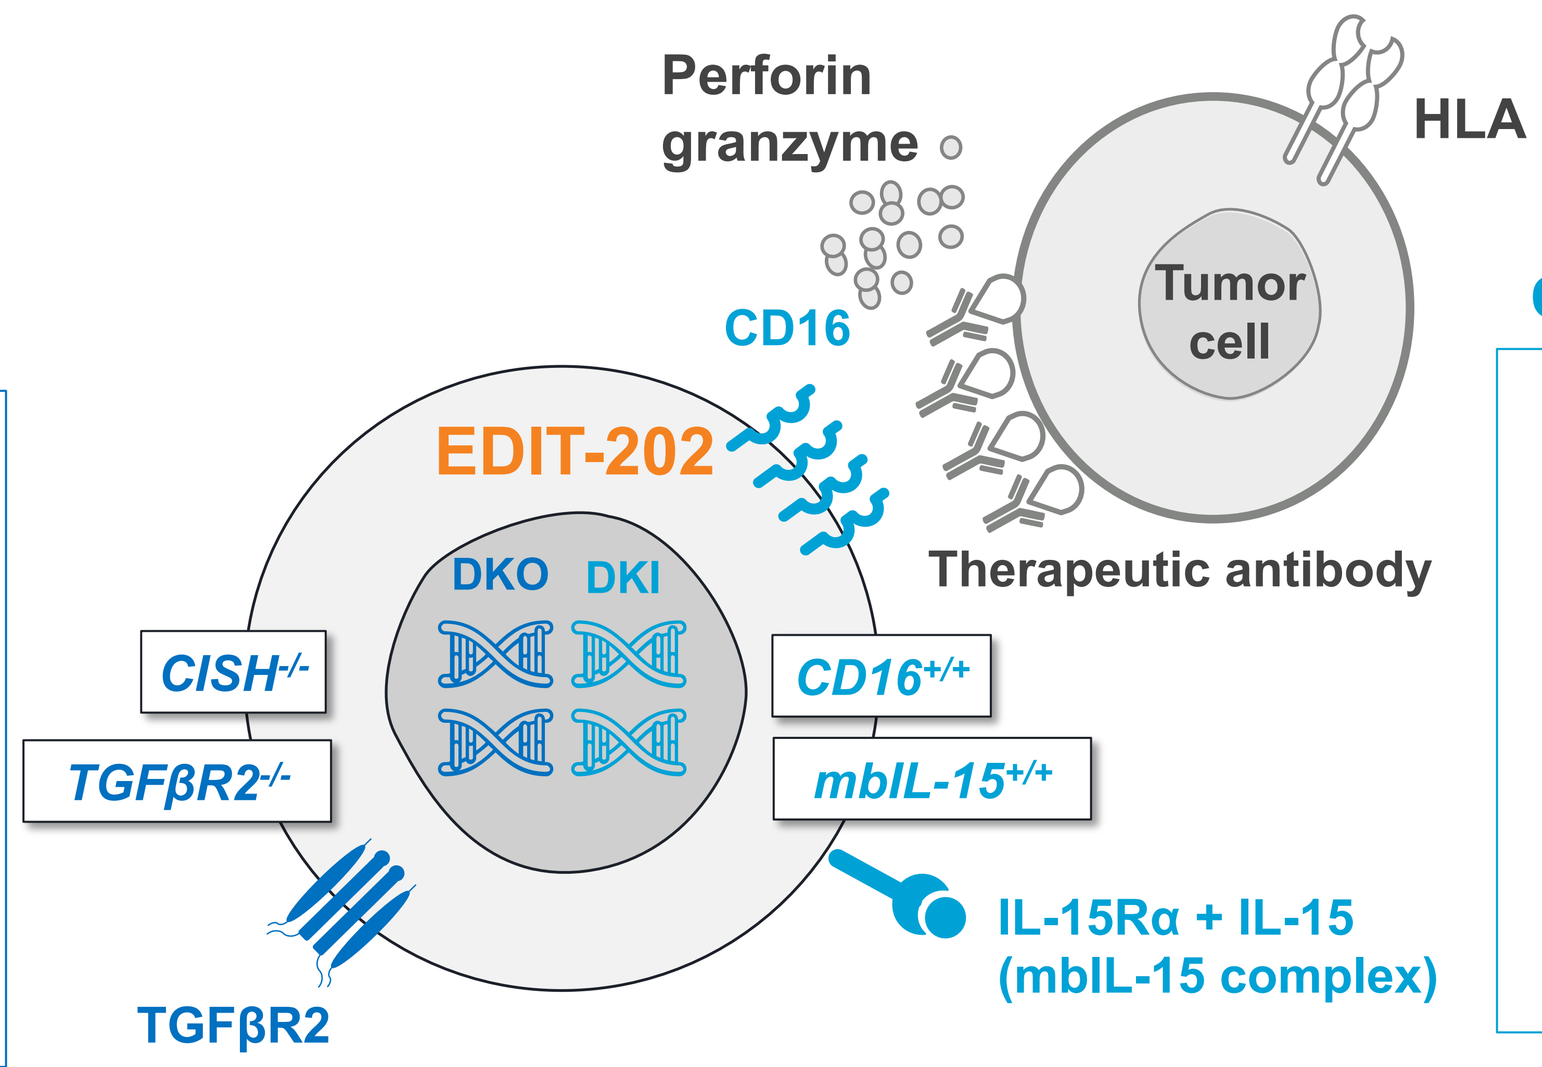
Perforin
granzyme
HLA
Tumor cell
CD16
EDIT-202
DKO
DKI
Therapeutic antibody
CISH-/-
TGFβR2-/-
CD16+/+
mbIL-15+/+
IL-15Rα + IL-15
(mbIL-15 complex)
TGFβR2

***CD16**$^{+/+}$/**mbIL-15**$^{+/+}$* **double knock-in (DKI)**

Enhanced ADCC

Constitutive overexpression of **CD16** through:
- Knock-in of ***CD16***

Enhanced survival and/or expansion of iNK cells

Increased **IL-15** signaling through:
- Knock-in of ***mbIL-15* complex**

ADCC, antibody-dependent cellular cytotoxicity; CISH, cytokine-inducible SH2-containing protein; HLA, human leukocyte antigen; IL-15, interleukin-15; IL-15Rα, interleukin-15 receptor alpha; iNK, induced pluripotent stem cell-derived natural killer; mbIL-15, membrane-bound IL-15; TGFβ, transforming growth factor beta; TGFβR2, transforming growth factor beta receptor 2.

METHODS

- iPSCs were edited with an engineered AsCas12a to knock-in CD16 and mbIL-15 using the SLEEK™ method.[3] Simultaneously, iPSCs were also edited with AsCas12a to knock-out *CISH* and *TGFβR2*. iPSC clones were then differentiated into iNK cells. Constitutive surface expression of CD16 and mbIL-15 by the DKI iNK cells was demonstrated by flow cytometry.
- A 3D tumor spheroid killing assay using Incucyte® imaging of NucLight Red-tagged SK-OV-3 cells was used to assess iNK cell cytotoxicity. In vitro persistence was measured by culturing wild type (WT) and DKI iNK cells in basal media without supporting cytokines for 21 days.
- Nonobese diabetic (NOD) severe combined immunodeficient (scid) gamma (NSG) mice were inoculated with 0.25x 10^6 luciferase (luc)-expressing SKOV-3 cell line (SKOV-3-luc) ovarian tumor cells. Mice received a single intraperitoneal (IP) dose of 5 million WT iNK or EDIT-202 cells with multiple IP doses of 2.5 mg/kg trastuzumab (TRA). Tumor burden was calculated using a Perkin Elmer bioluminescent in vivo imaging system (IVIS).

RESULTS

Figure 1. EDIT-202 cells had increased CD16 and IL-15Rα expression compared with WT iNK cells

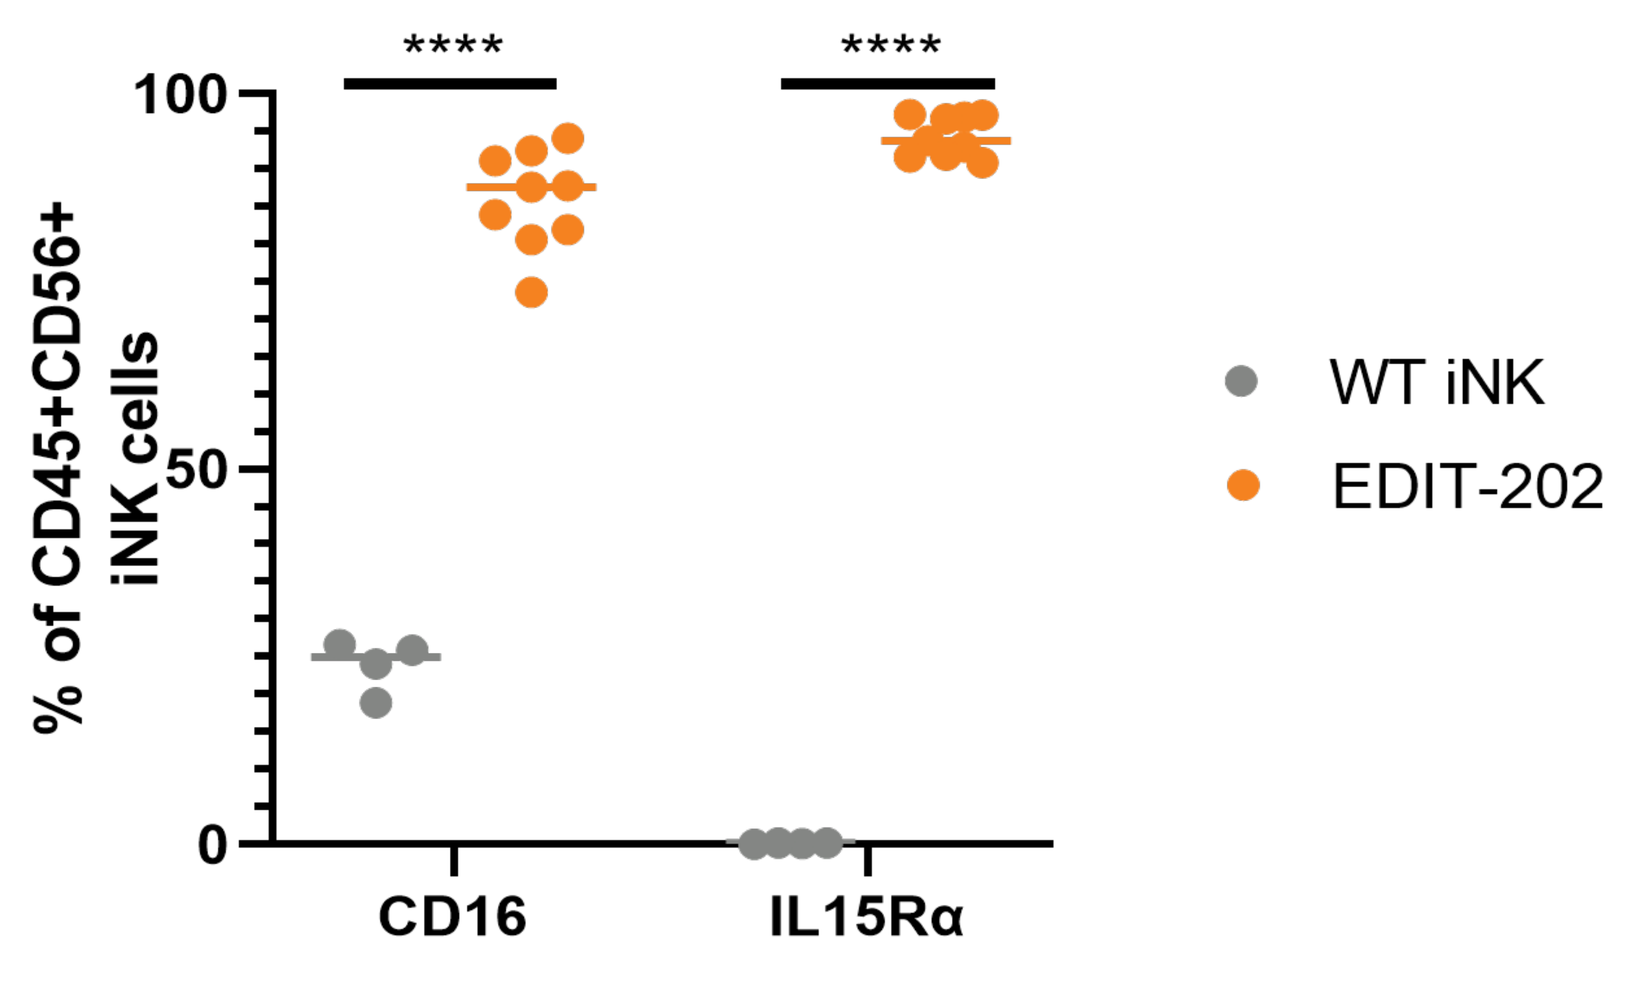
% of CD45+CD56+ iNK cells
100
50
0

CD16
IL15Rα
WT iNK
EDIT-202

Number of independent experiments=1; ****p≤0.0001 (two-way ANOVA).

Figure 2. EDIT-202 cells showed enhanced ADCC-mediated tumor killing and resistance to TGFβ-induced immunosuppression compared with WT iNK cells in a 3D ovarian SKOV-3 spheroid assay

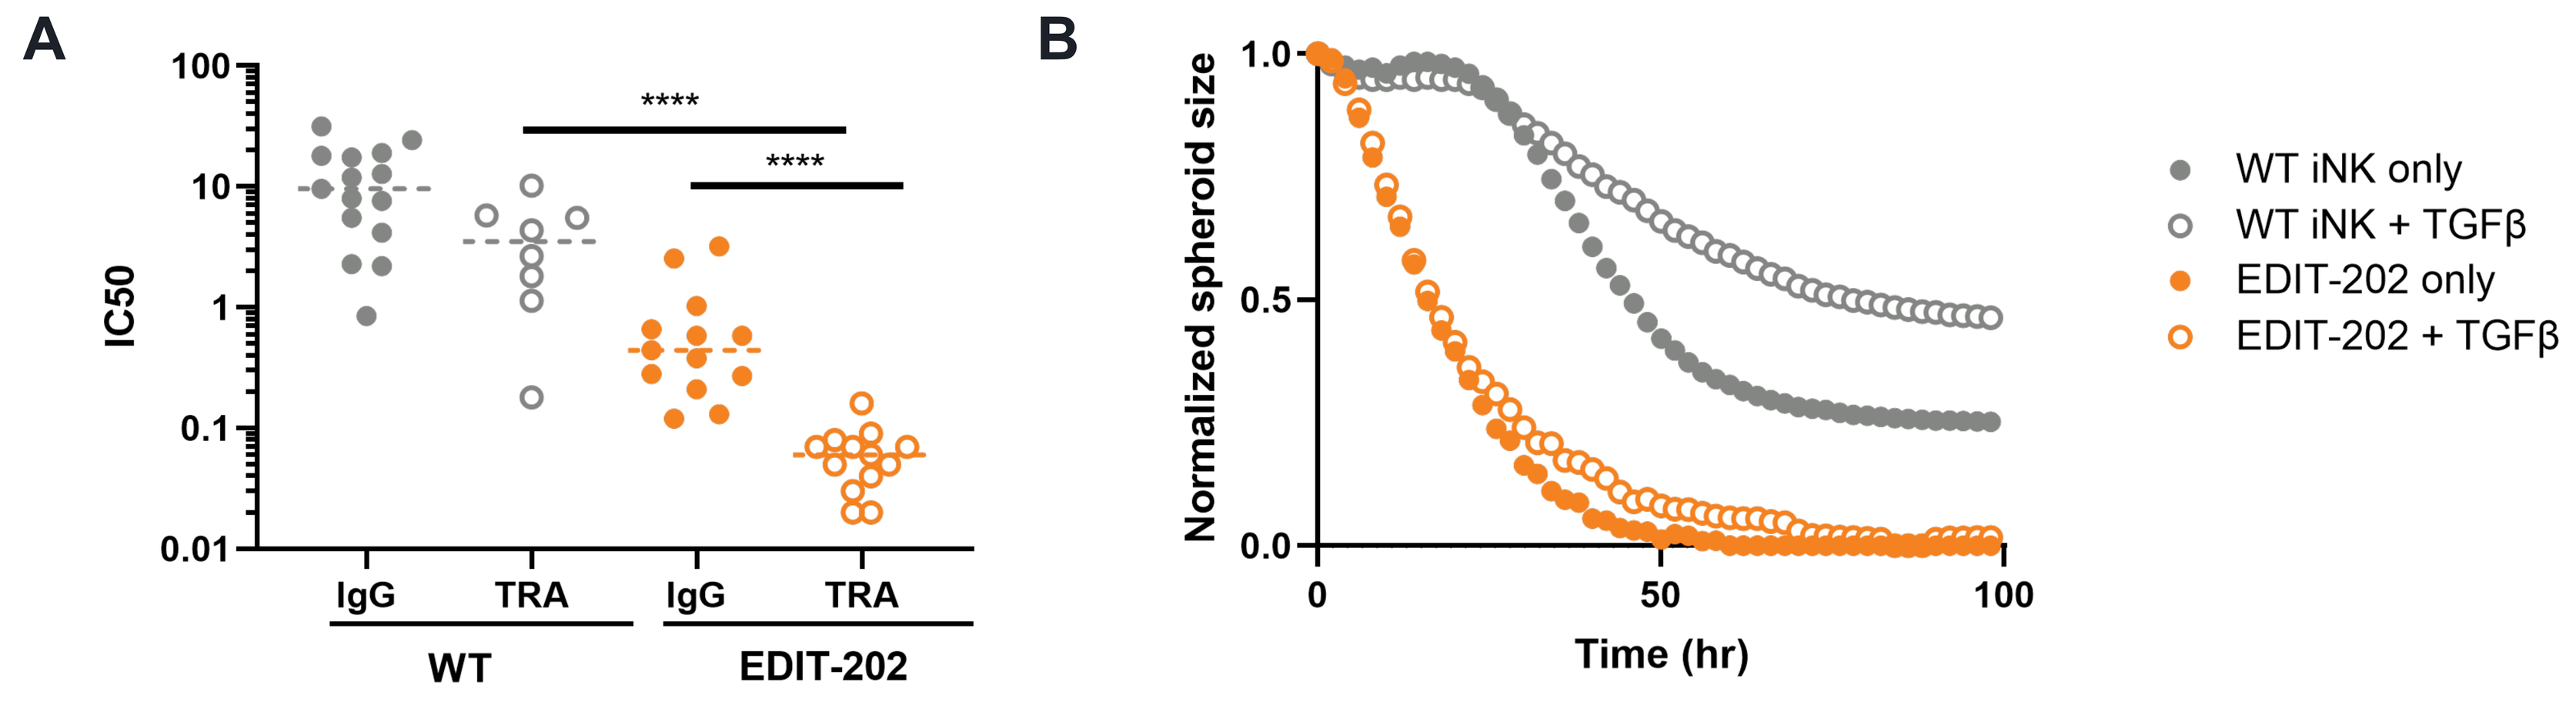
A
IC50
100
10
1
0.1
0.01

IgG
TRA
IgG
TRA
WT
EDIT-202
B
Normalized spheroid size
1.0
0.5
0.0
0
50
100
Time (hr)
WT iNK only
WT iNK + TGFβ
EDIT-202 only
EDIT-202 + TGFβ

A) Number of independent experiments=11; TRA, trastuzumab; IC50, half-maximal inhibitory concentration; ****p≤0.0001 (unpaired t-test); B) Number of independent experiments=2; Normalized spheroid size, spheroid size after 100hrs of co-culture with NKs at E:T=31.6; TGFβ, transforming growth factor beta.

Figure 3. EDIT-202 showed upregulated and continuous replenishment of CD16 on the cell surface after tumor exposure

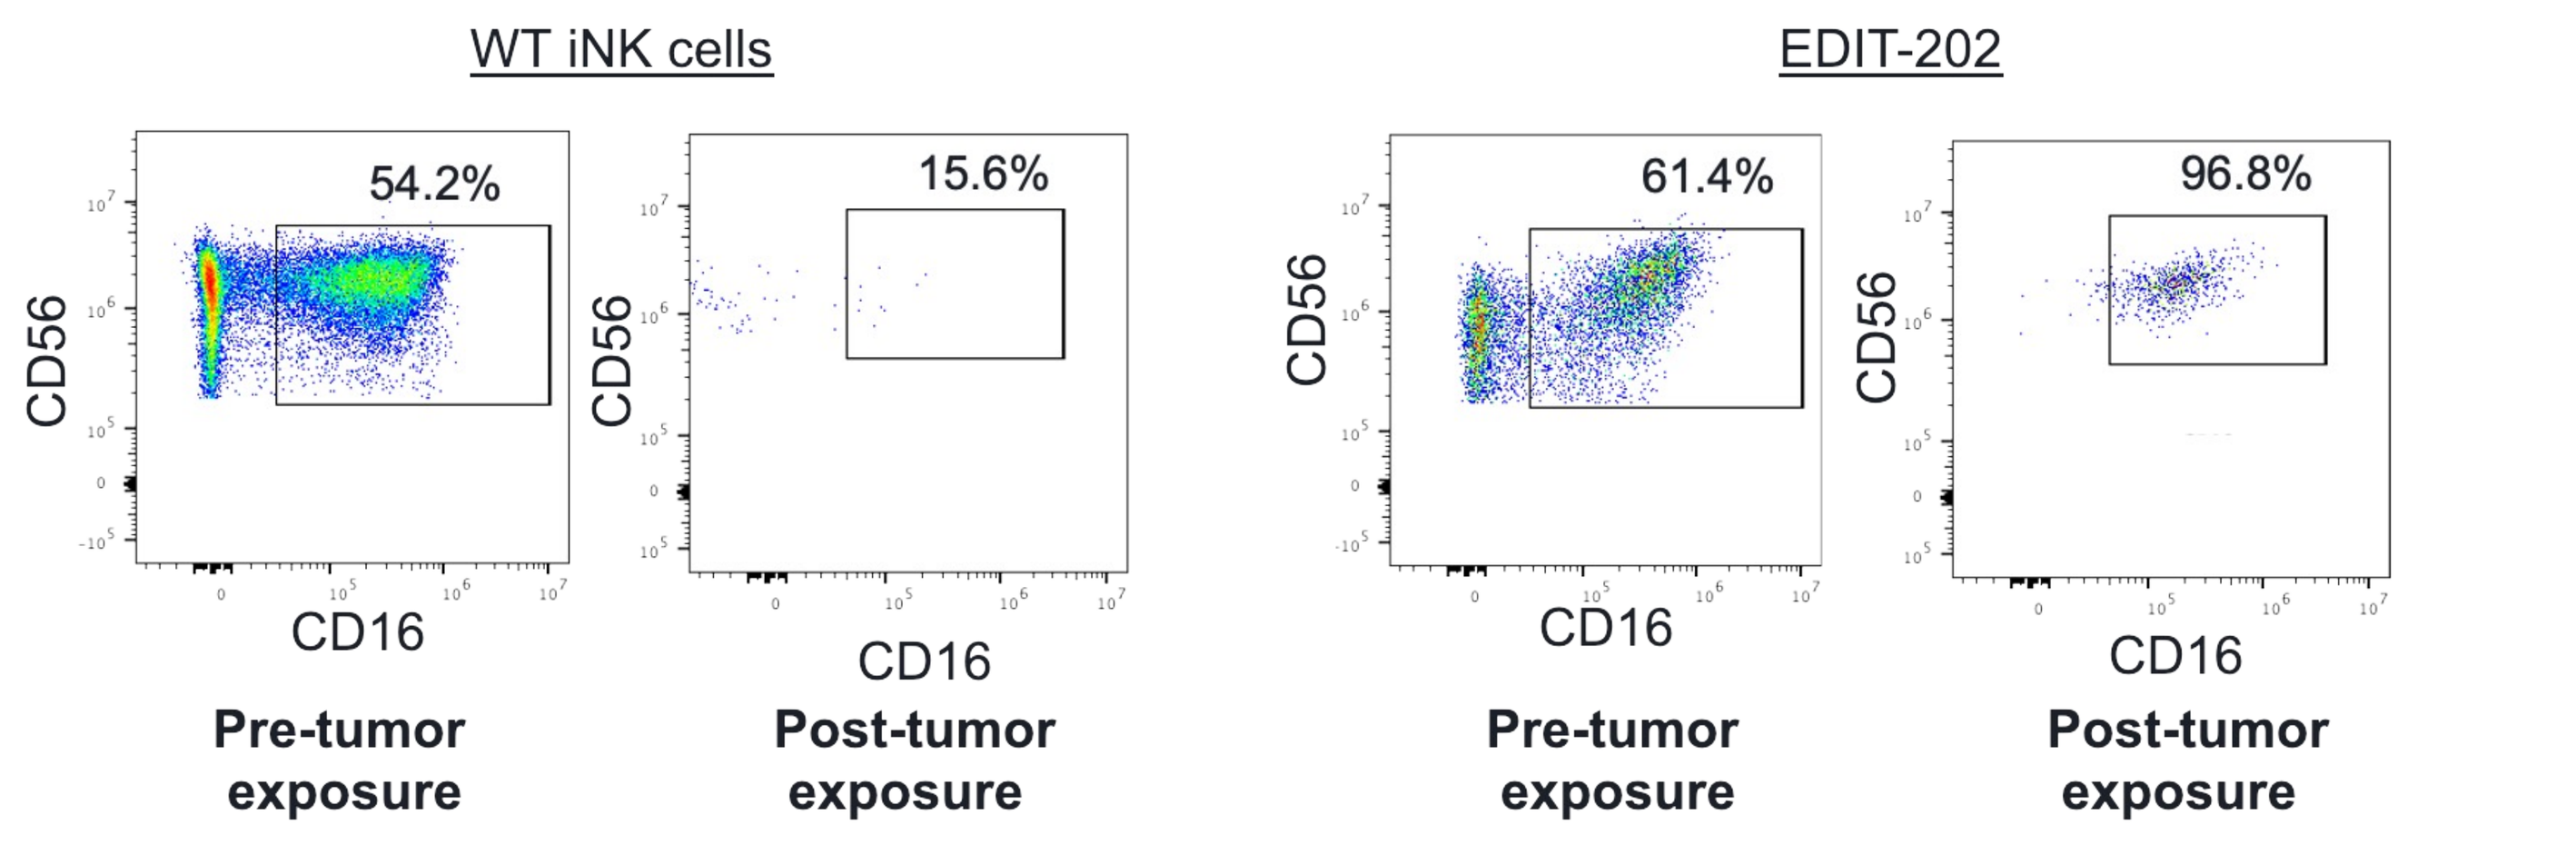
WT iNK cells
EDIT-202
54.2%
15.6%
61.4%
96.8%
CD56
CD16
Pre-tumor exposure
Post-tumor exposure
Pre-tumor exposure
Post-tumor exposure

Number of independent experiments=1. Representative flow plots gated on alive CD56+ NK cells analyzed after 4 days of co-culture with HT29 colorectal tumor cells at E:T=1:1.

Figure 4. mbIL-15 KI resulted in prolonged persistence of EDIT-202 cells in the absence of cytokines

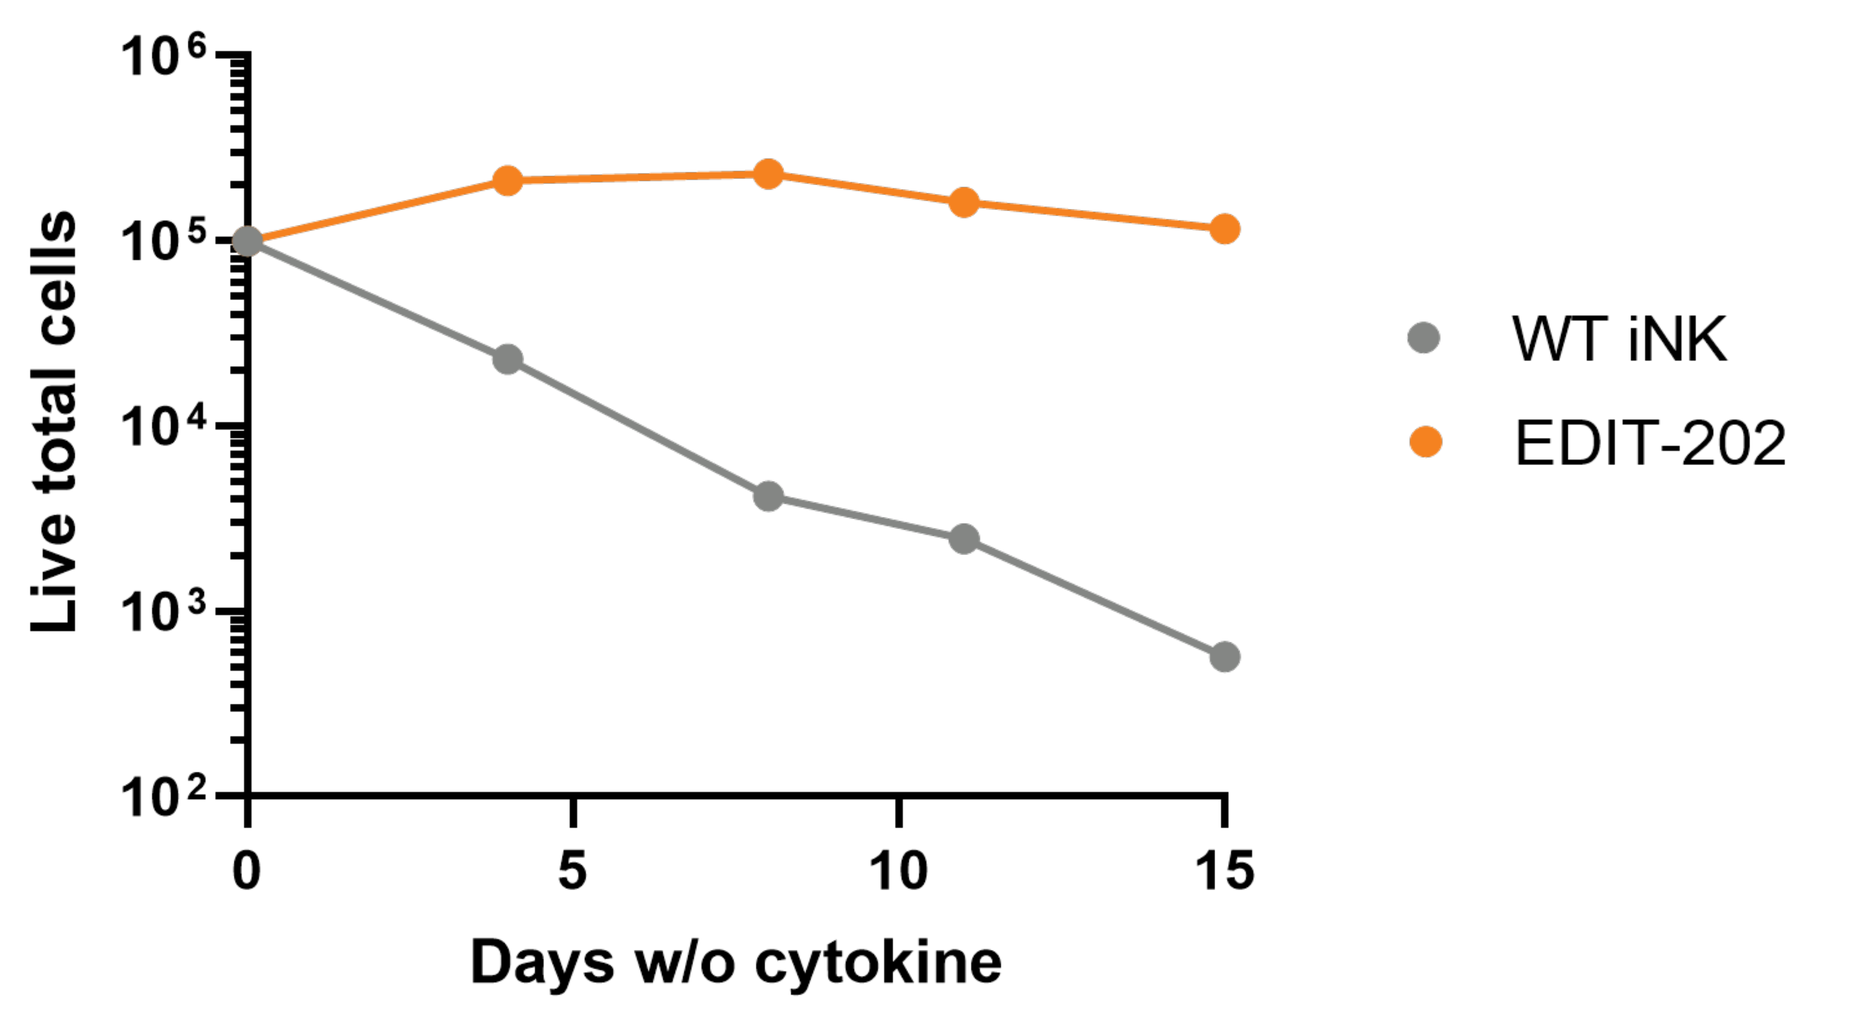
Live total cells
10^6
10^5
10^4
10^3
10^2
0
5
10
15
Days w/o cytokine
WT iNK
EDIT-202

KI, knock-in; mbIL-15, membrane-bound IL-15; w/o, without.

Figure 5. EDIT-202 administered in combination with 3 doses of TRA induced significant tumor reduction to complete clearance in an ovarian SKOV-3-luc IP solid tumor model.

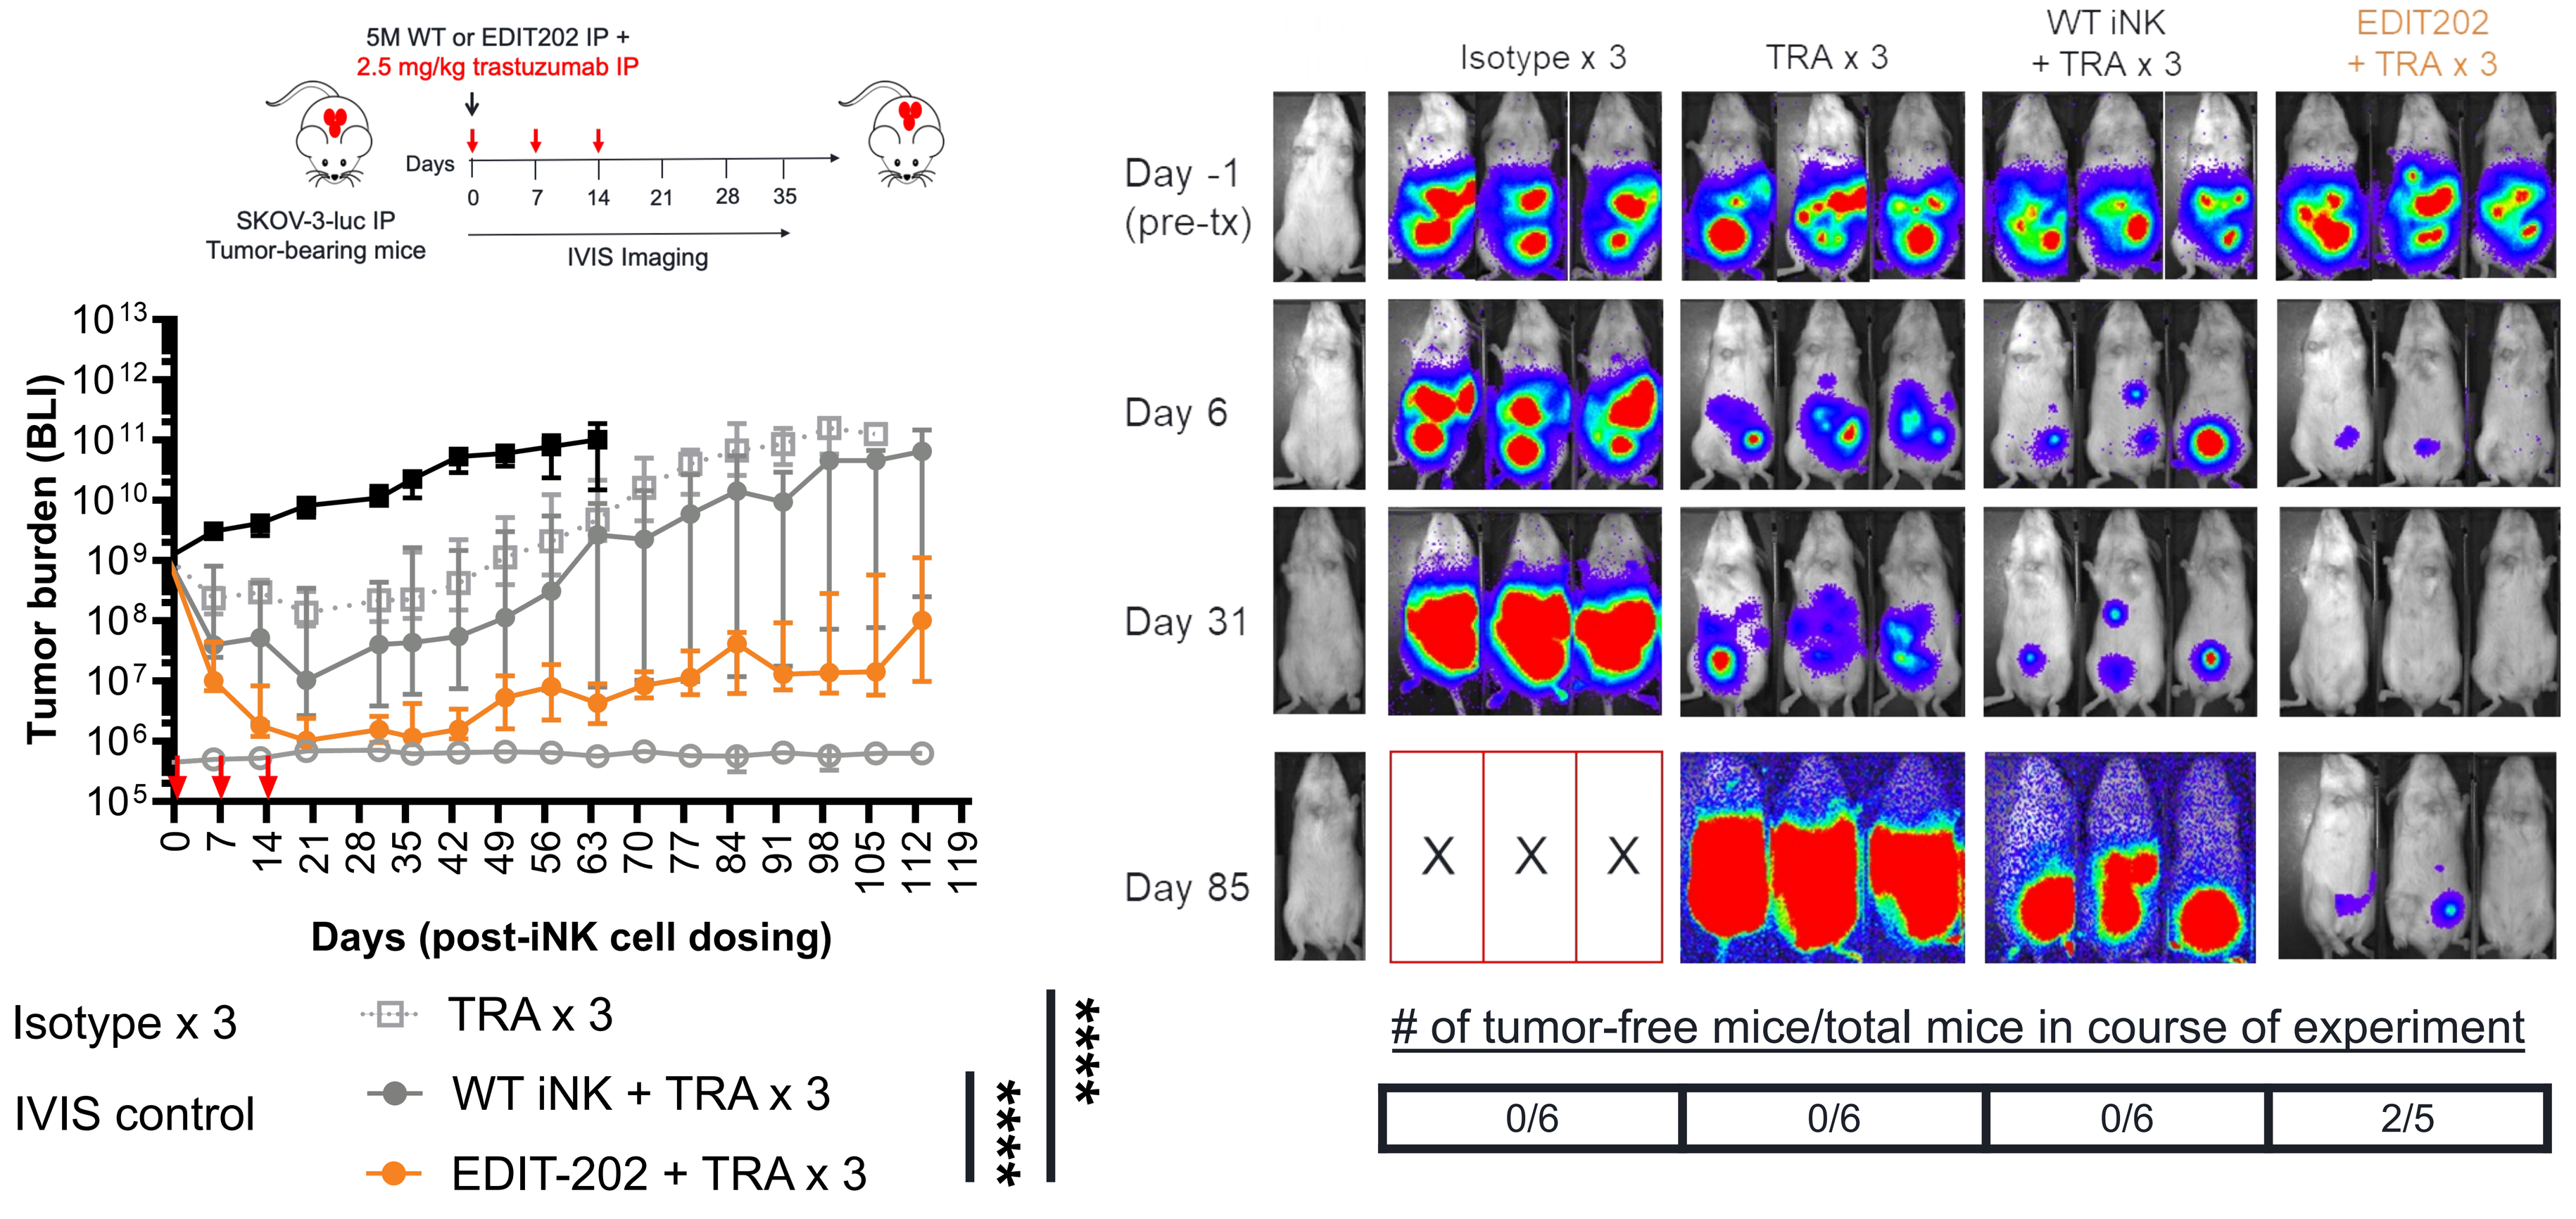
5M WT or EDIT202 IP +
2.5 mg/kg trastuzumab IP
Days
0
7
14
21
28
35
SKOV-3-luc IP
Tumor-bearing mice
IVIS Imaging
Tumor burden (BLI)
Days (post-iNK cell dosing)
Isotype x 3
IVIS control
TRA x 3
WT iNK + TRA x 3
EDIT-202 + TRA x 3

Isotype x 3
TRA x 3
WT iNK
+ TRA x 3
EDIT202
+ TRA x 3
Day -1
(pre-tx)
Day 6
Day 31
Day 85
X
X
X
of tumor-free mice/total mice in course of experiment
0/6
0/6
0/6
2/5

Number of mice per group=5-6; ****p<0.0001; TRA, trastuzumab; BLI, bioluminescent imaging.

Figure 6: EDIT-202 treatment with 3 doses of TRA significantly increased survival over WT iNK cells + TRA in an ovarian SKOV-3-luc IP solid tumor model.

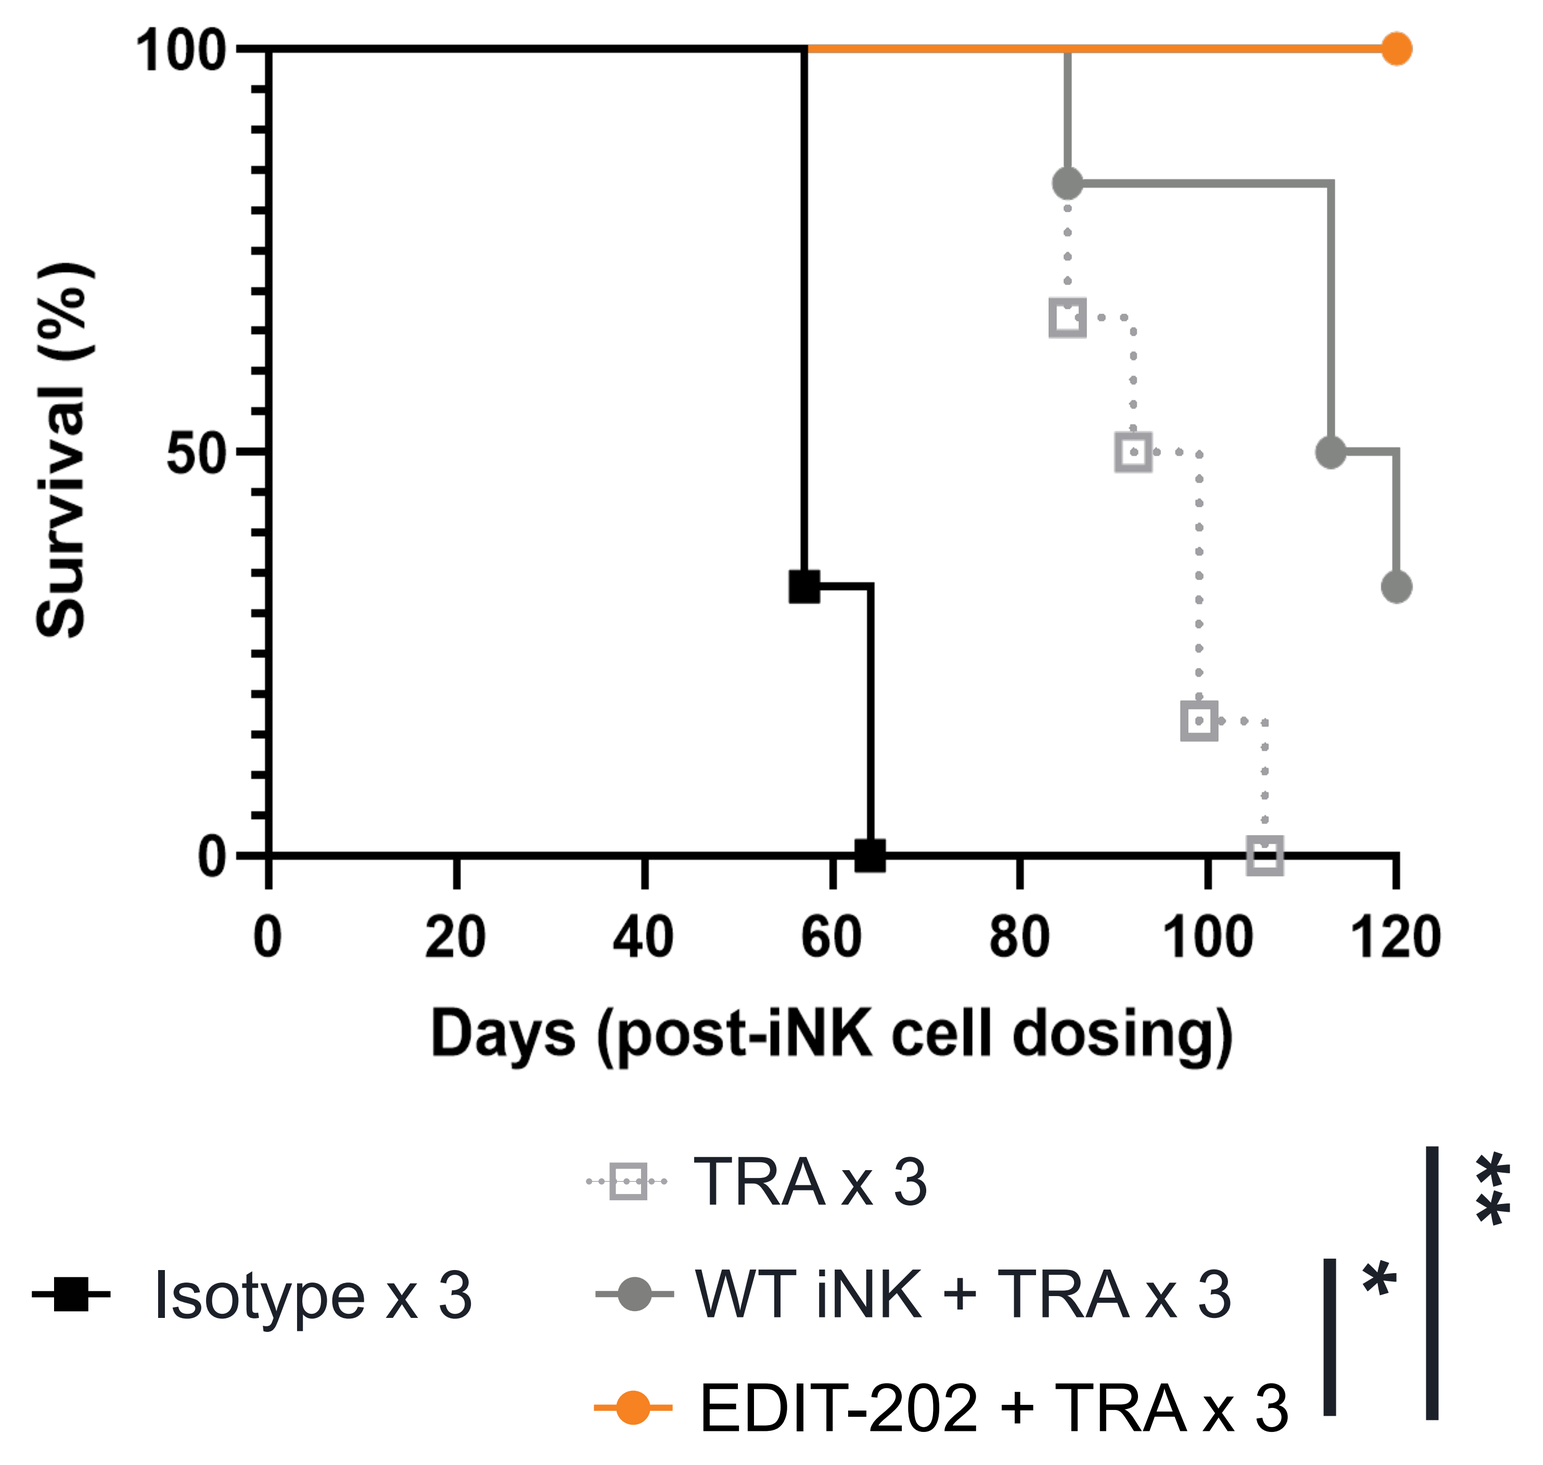
Survival (%)
100
50
0
0
20
40
60
80
100
120
Days (post-iNK cell dosing)
Isotype x 3
TRA x 3
WT iNK + TRA x 3
EDIT-202 + TRA x 3
*
**

Number of mice per group=5-6; TRA, trastuzumab; *p<0.05, **p<0.01

DISCLOSURES

Employees and shareholders of Editas Medicine: S.Q.K, A.G.A, A.G, K.M.I, M.J, J.G, N.R.C, S.S, P.S, L.B, M.N, J.A.F, M.S.S, K.Z, and K.-H.C.

Acknowledgments:
The authors would like to thank all their Editas colleagues for helping to plan, perform, analyze, and present this work. Editorial assistance was provided by Bertha Vandegrift, PhD, Porterhouse Medical, funded by Editas Medicine.

CONCLUSIONS

- EDIT-202 had significantly higher levels of CD16 and mbIL15 compared with WT iNK cells indicating that our SLEEK™ method, powered by our proprietary AsCas12a, is a robust method for editing iPSCs (Figure 1).
- EDIT-202 showed significantly higher natural cytotoxicity and augmented ADCC-mediated killing against 3D SKOV-3 spheroids compared with WT iNK cells. EDIT-202 induced reduction in tumor burden in the absence or presence of TGFβ, indicating that the TGFβR2 KO makes EDIT-202 resistant to TGFβ induced immunosuppression (Figure 2).
- Due to the CD16 KI, EDIT-202 cells had upregulated and continuous expression of CD16 compared with WT iNK cells after tumor exposure (Figure 3).
- *In vitro*, EDIT-202 had prolonged cytokine independent persistence compared with WT iNK cells indicating that the mbIL15 KI in EDIT-202 is sufficient for cell survival and maintenance (Figure 4).
- In an *in vivo* ovarian SKOV-3-luc solid tumor model, EDIT-202 treatment with 3 doses of TRA induced significant reduction in tumor burden compared with WT iNK cells plus TRA and TRA only, resulting in complete tumor clearance in 40% of mice over the course of the experiment (Figure 5).
- When combined with TRA, EDIT-202 significantly increased mouse survival over WT iNK cells in the solid tumor SKOV-3 model. At day 120, 100% of EDIT202-treated mice were alive, compared with 33.3% of WT iNK-treated mice (Figure 6).
- These data support the development of EDIT-202 as a potential allogeneic cell-based medicine for treating solid tumors.

REFERENCES

1. Wu Y, et al. Front Immunol 2017;8:930
2. Snyder KM, et al. Front Immunol 2018; 9:2873
3. Editas Medicine. CRISPR Frontiers 2021. https://www.editasmedicine.com/wp-content/uploads/2021/08/2021_CSHL-CRISPR-Frontiers-SLEEK-Zuris_FINAL.pdf

© 2022 Editas Medicine