Elucidating the impact of RNA secondary structure on RNP behavior

Chrysa Latrick[1], Bridget Moffet[1], Pallavi Gambhire[2], Pranjali Ghude[1], Jean-Noel Lemercier[1], Steven Wolk[1]

[1]Analytical Chemistry Department, Editas Medicine, Boulder, CO, USA
[2]Process and Analytical Department, Editas Medicine, Cambridge, MA, USA

INTRODUCTION

RNAs are known to form different secondary structures. The Cas12a RNA is composed of a pseudoknot-hairpin region and a targeting region. The pseudoknot is required for binding to the Cas12a protein, while the targeting region pairs with the target strand DNA. Any variations in the gRNA structure can impact ribonucleoprotein (RNP) formation and activity. We show here three different Cas12a RNPs that have different behaviors by SEC. By native gel electrophoresis mobility shift assay (EMSA) the guide RNAs alone migrate as either monomers or dimers. As RNPs by EMSA, while some of these RNAs migrate as either a 1:1 or 2:1 RNA:protein complex, others multimerize, indicating that a complex set of guide-specific interactions drive this process. By calculating theoretical secondary structure predictions, the mechanism of dimerization or multimerization was elucidated, showing that guide-guide interactions play a critical role.

RESULTS

Figure 1: SEC chromatograms of three different RNPs and RNAs

A)

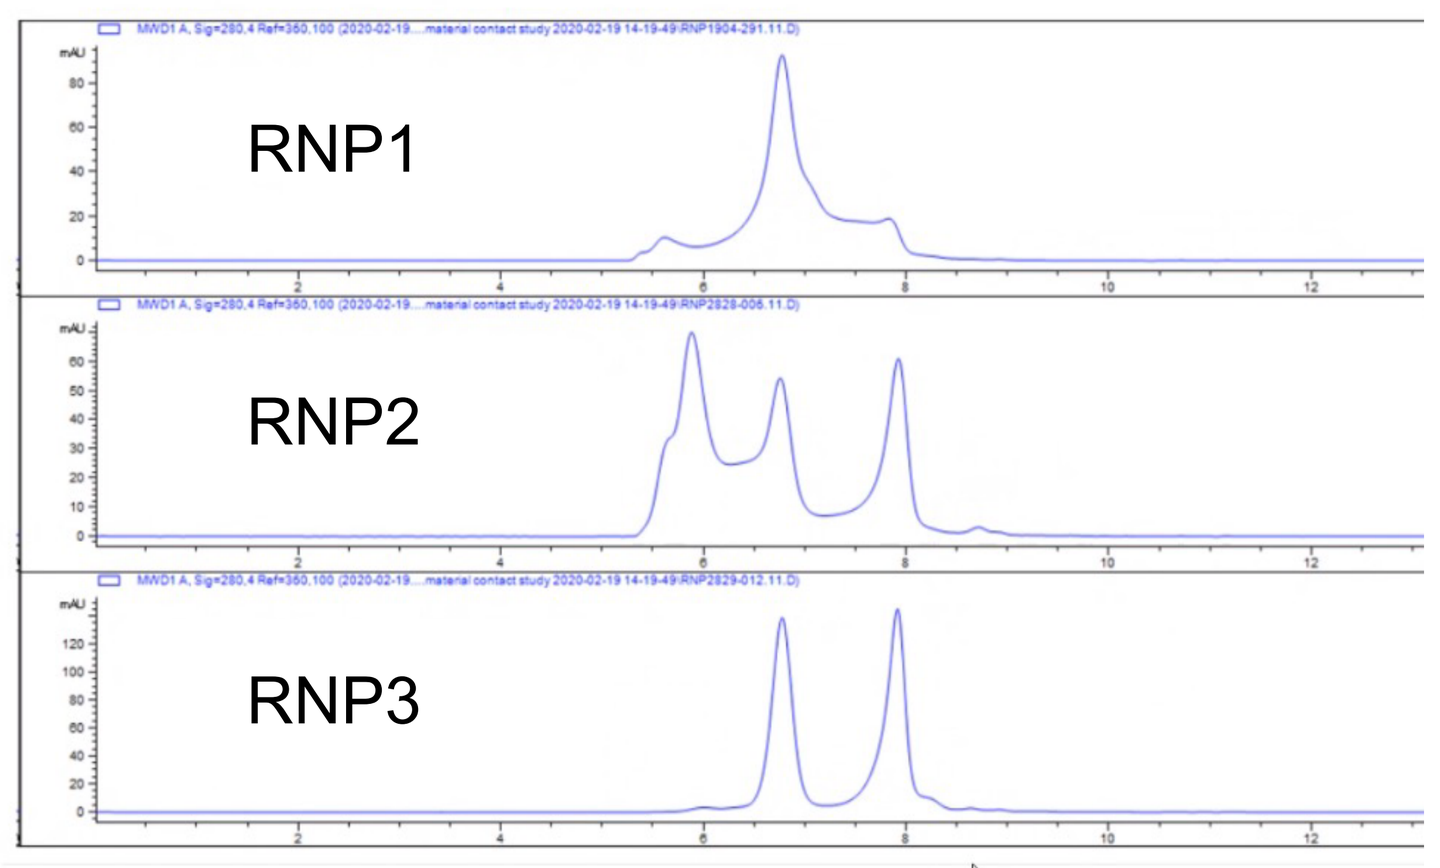
RNP1
RNP2
RNP3

B)

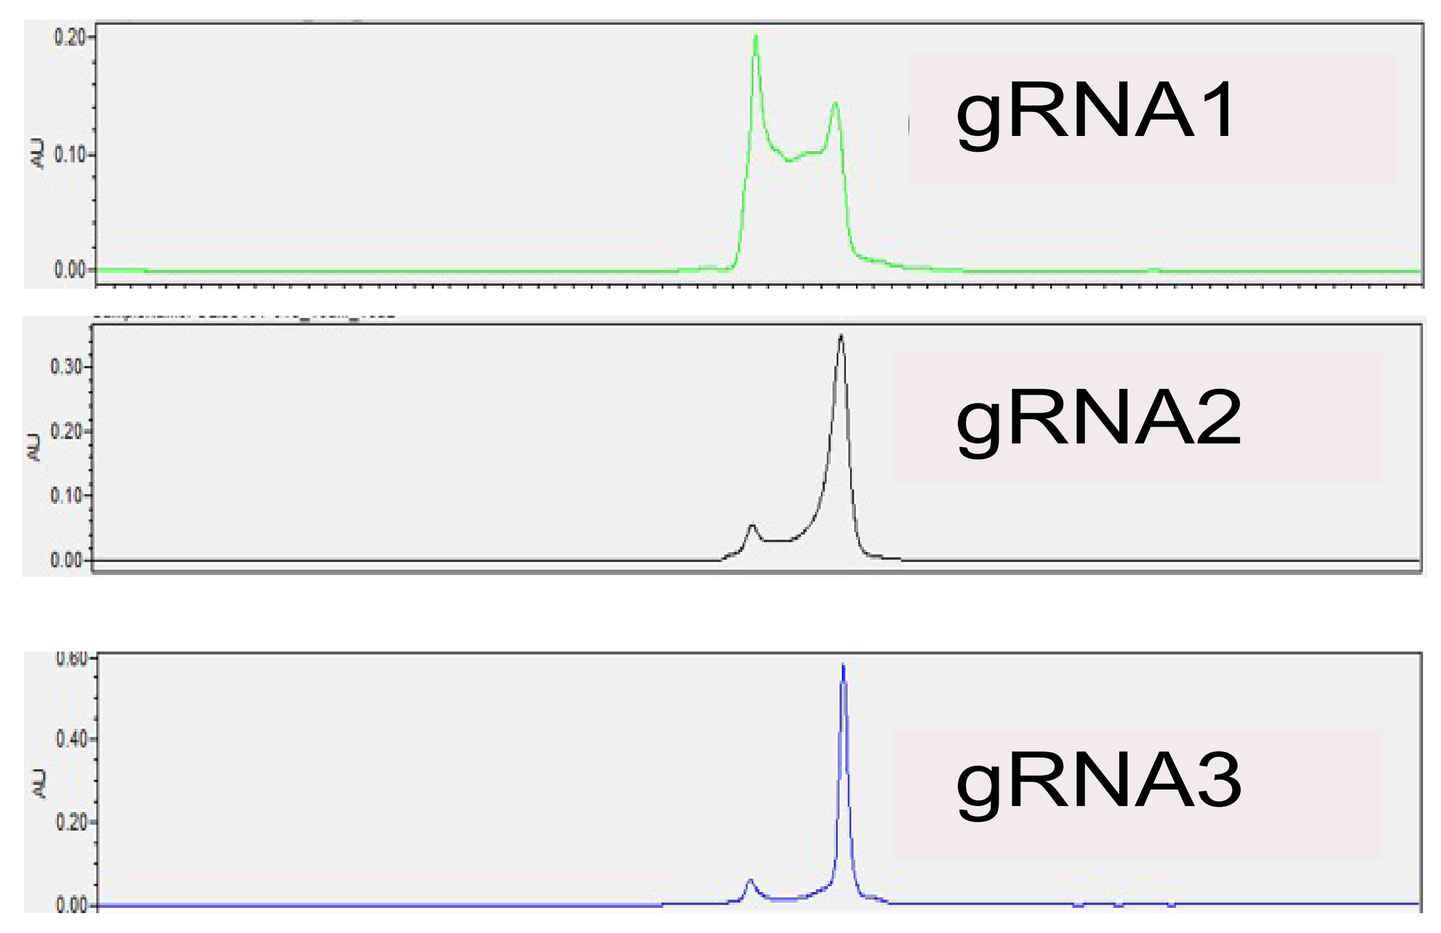
gRNA1
gRNA2
gRNA3

A) SEC chromatograms for RNP1, RNP2, and RNP3 show different behaviors. RNPs were complexed with excess 70-mer guide. RNP1 shows a single peak, with no free excess guide. RNP2 shows a higher MW species, as well as the expected peak, and free excess guide. RNP3 shows only two peaks, one for RNP, and a second for free excess guide.

B) SEC chromatograms for 70-mer gRNA1, gRNA2, gRNA3 under the same conditions as the RNP. gRNA1 demonstrates two peaks, corresponding to a monomer and a dimer in solution. The dimer runs at a similar retention time as the RNP peak. Both gRNA2 and gRNA3 show largely monomer behavior.

Figure 2: Native gel of gRNAs shows different propensities to dimerize

A)

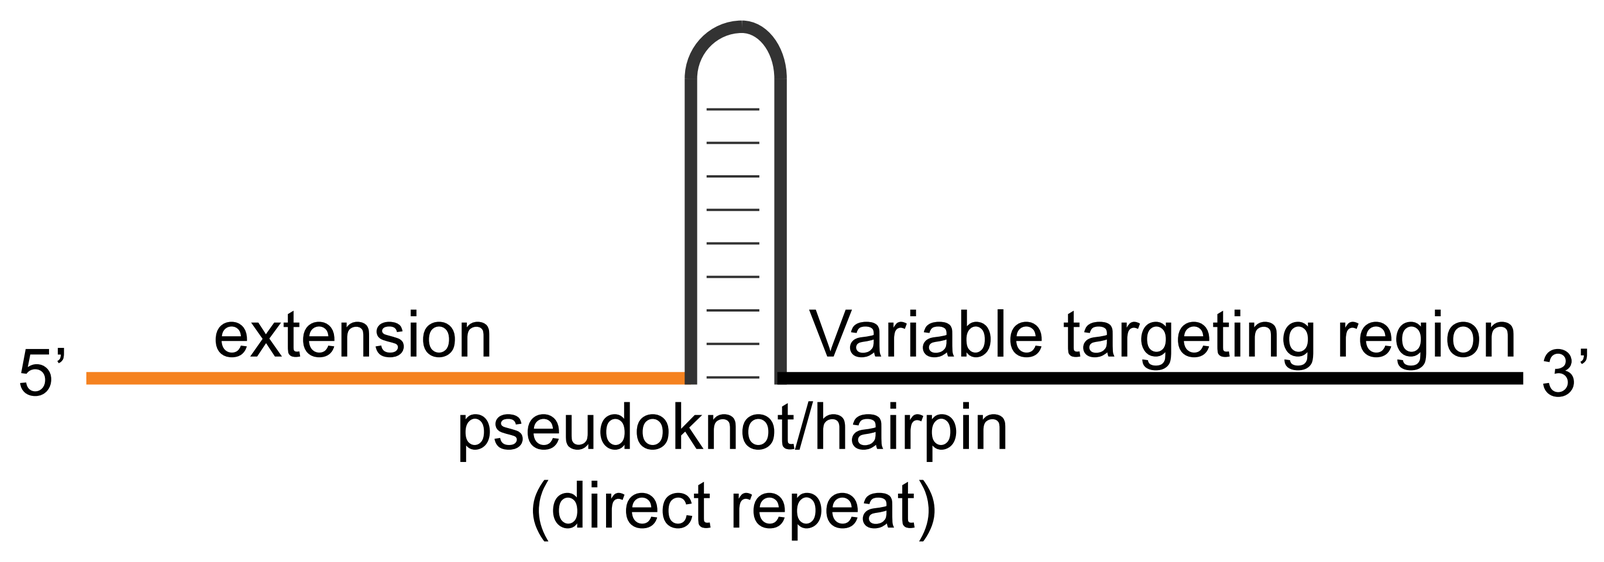
5'
extension
Variable targeting region
3'
pseudoknot/hairpin
(direct repeat)

B)

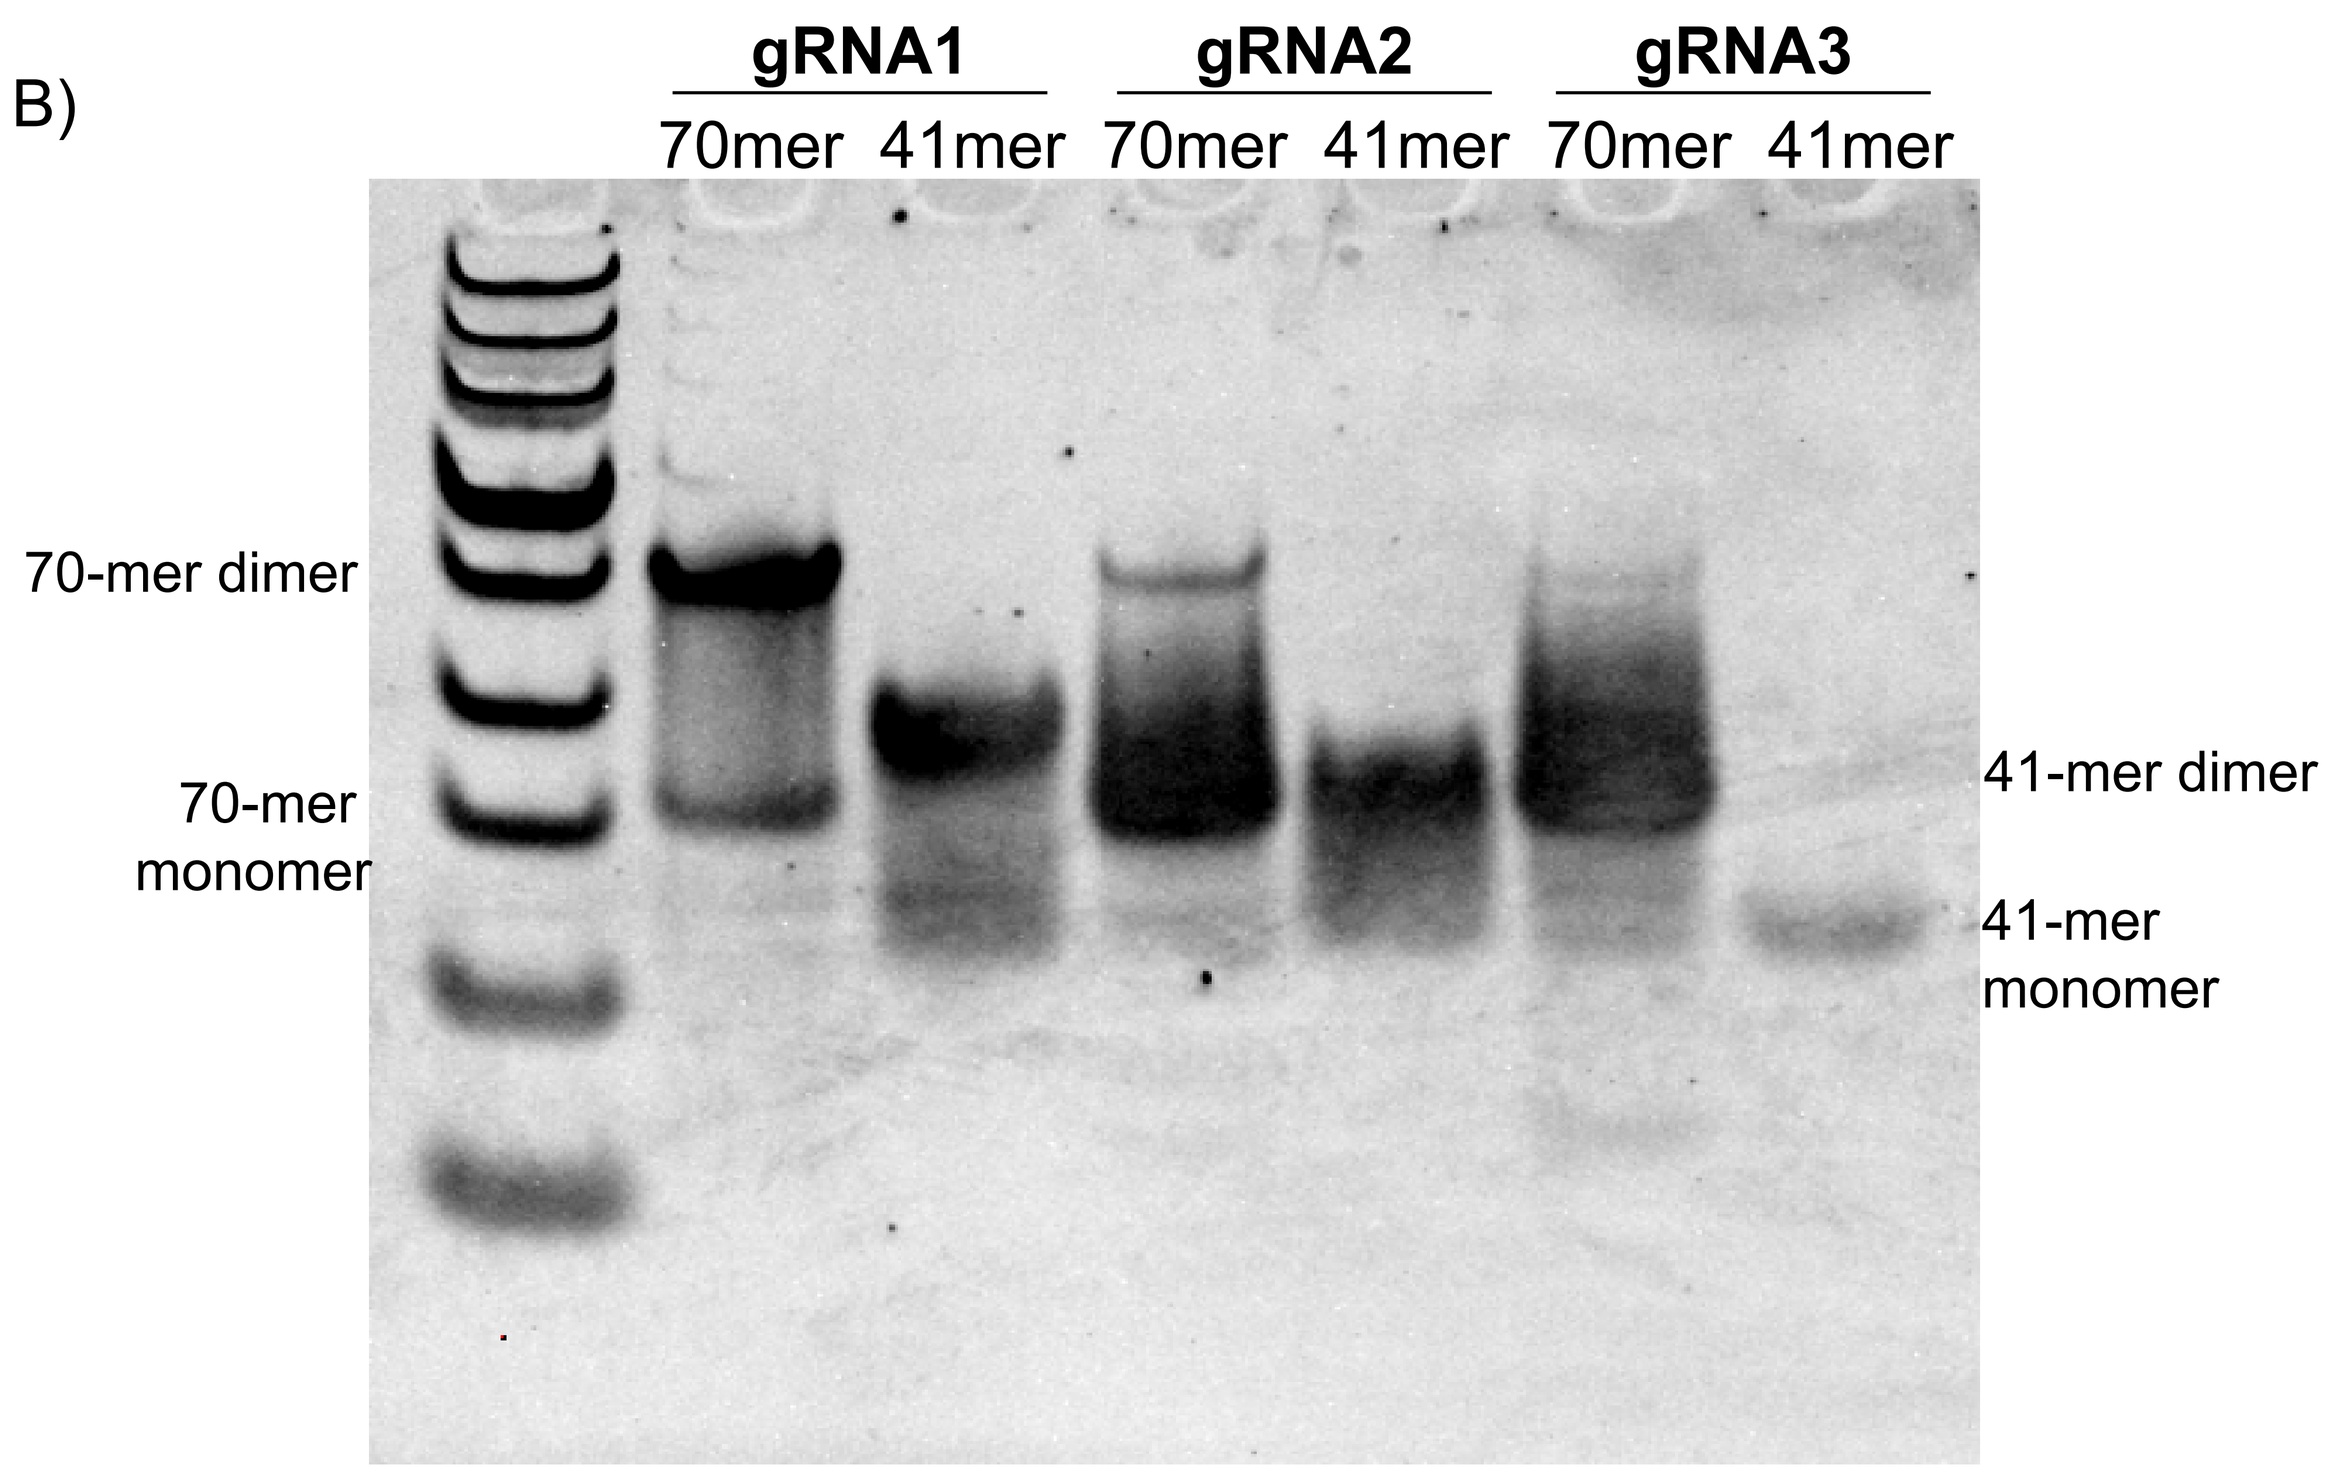
gRNA1
gRNA2
gRNA3
70mer 41mer 70mer 41mer 70mer 41mer
70-mer dimer
70-mer monomer
41-mer dimer
41-mer monomer

A) Some guide RNAs feature a 5' extension (orange).

B) Native gel EMSA shows that the three different guides exhibit different behaviors either with or without the extension. Guide RNA1 is a dimer both as a 70-mer and a 41-mer. gRNA2 is a monomer as a 70-mer, but a dimer as a 41-mer, indicating that the targeting region is responsible for the dimerization. gRNA3 is a monomer regardless of the presence or absence of the extension.

Figure 3: EMSA shows different RNP multimerization behaviors

A)

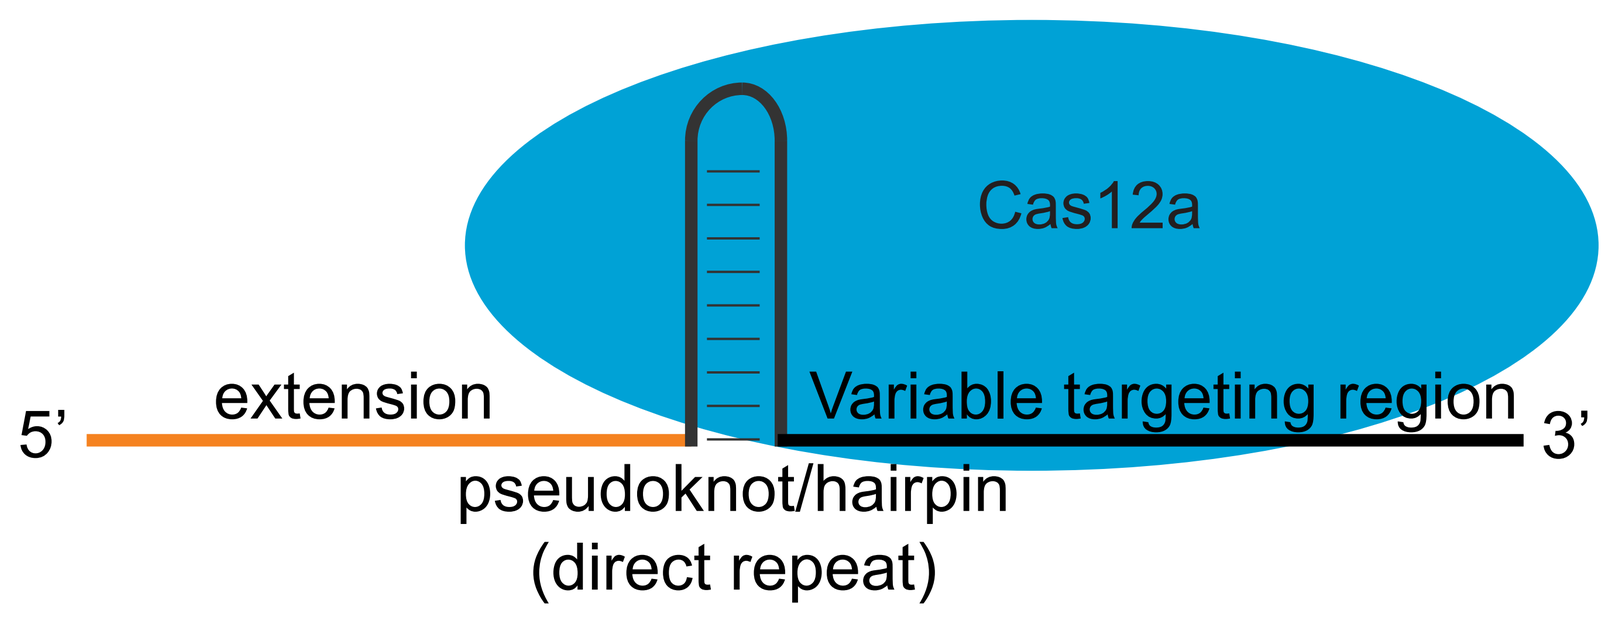
Cas12a
5'
extension
Variable targeting region
3'
pseudoknot/hairpin
(direct repeat)

B)

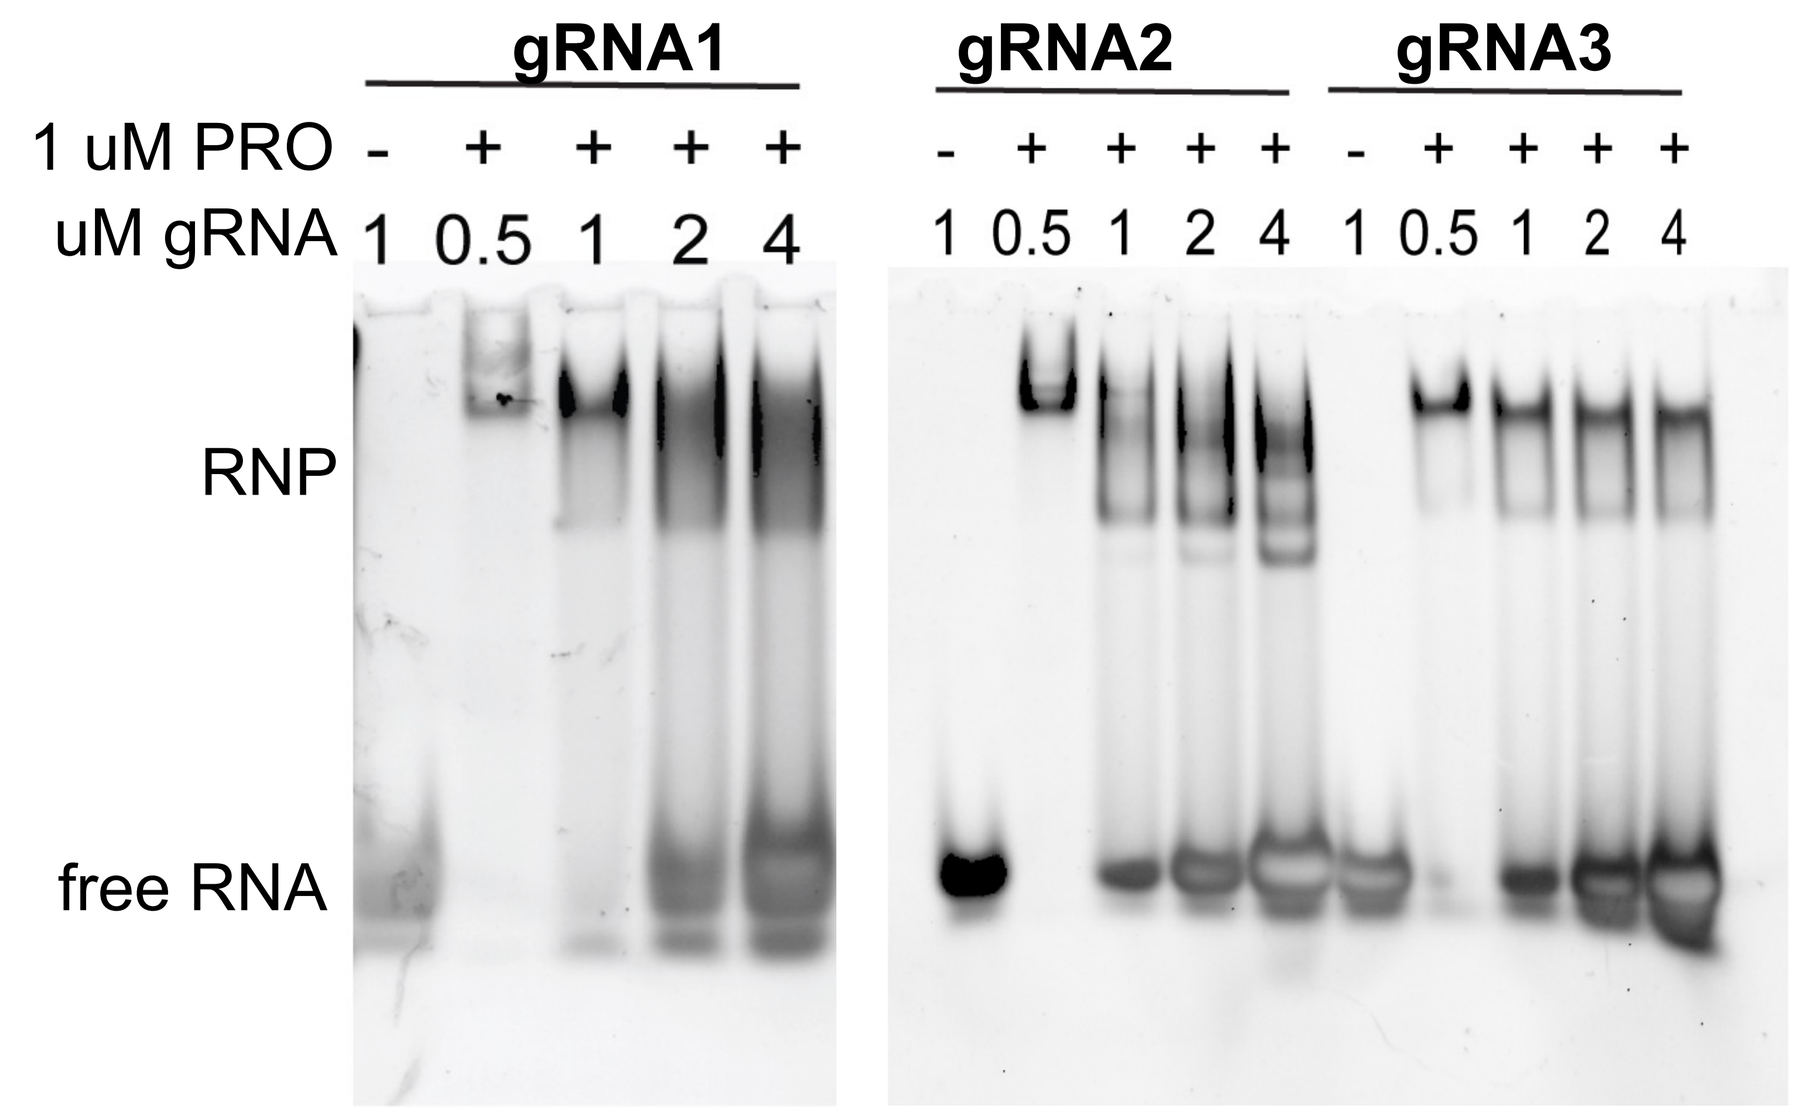
gRNA1
gRNA2
gRNA3
1 uM PRO - + + + + - + + + + - + + + +
uM gRNA 1 0.5 1 2 4 1 0.5 1 2 4 1 0.5 1 2 4
RNP
free RNA

A) Cas12 specifically recognizes the pseudoknot/hairpin region of the gRNA and interacts with the variable targeting region.

B) gRNA1 readily forms an RNP complex with 1:1 stoichiometry and equimolar ratios of protein:gRNA. However, upon further addition of gRNA, additional guide associates with the RNP complex. The smear indicates that the complex dissociates on a time scale faster than that of the gel separation. gRNA2 forms a 1:1 complex only when guide is substoichiometric to the protein. Upon addition of equimolar or superstoichiometric amounts of gRNA2, several discreet species of RNP-gRNA form. gRNA3 is not prone to multimerization at any stoichiometries.

Figure 4: Secondary structure predictions for gRNA1 and gRNA3

A)

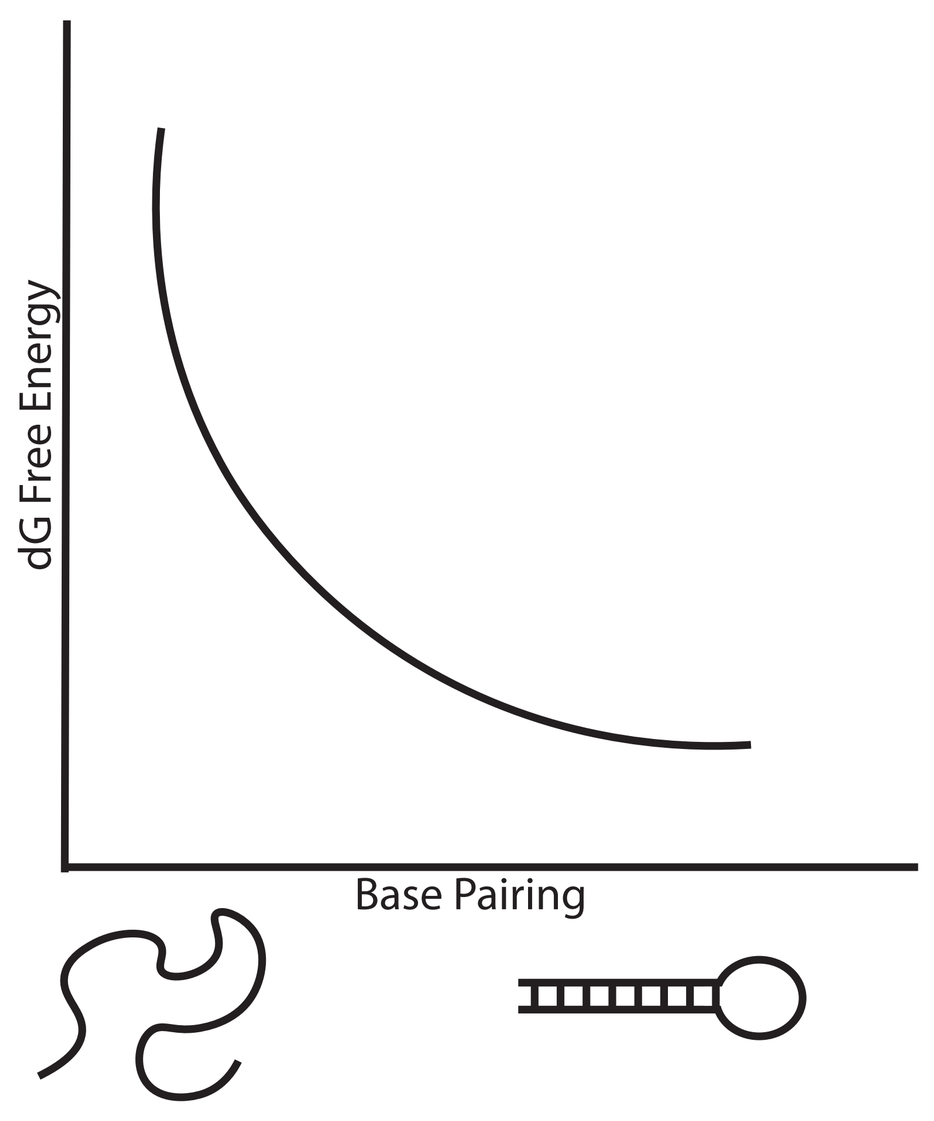
dG Free Energy
Base Pairing

B)

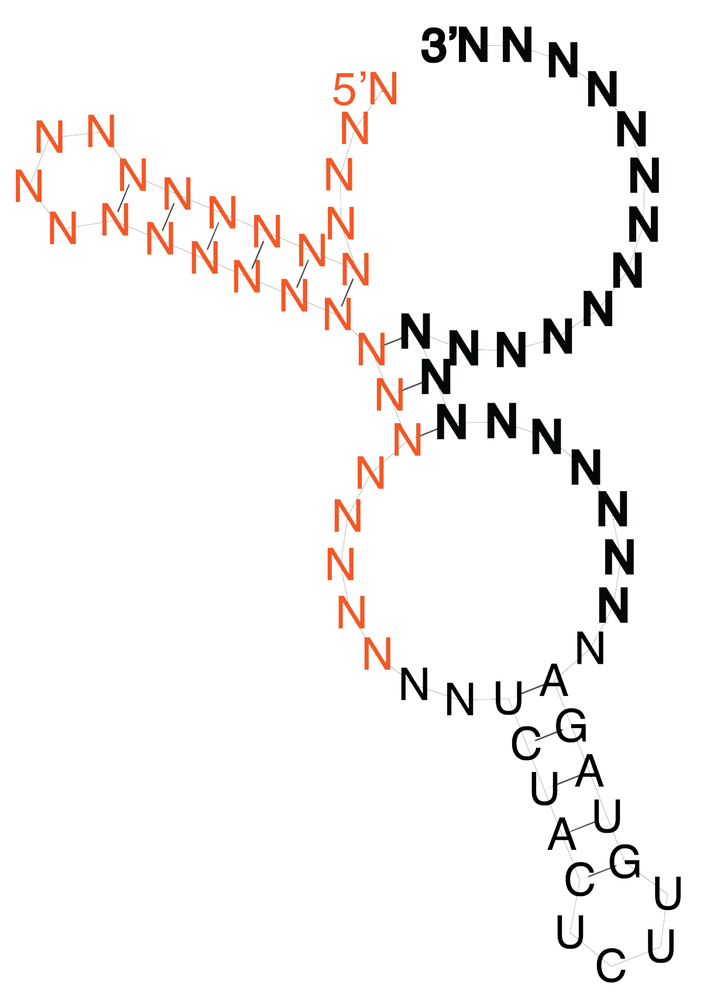

C)

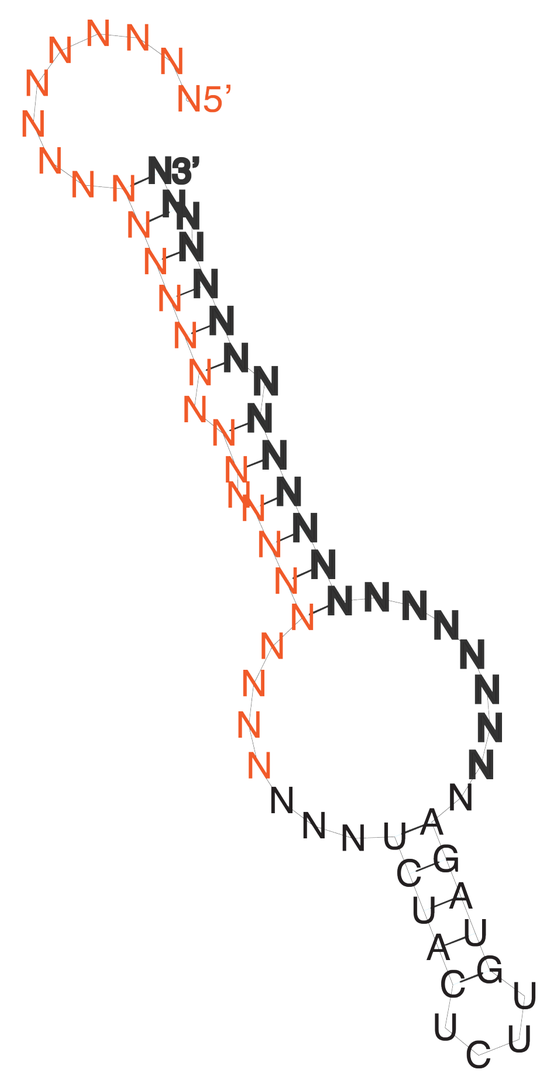

D)

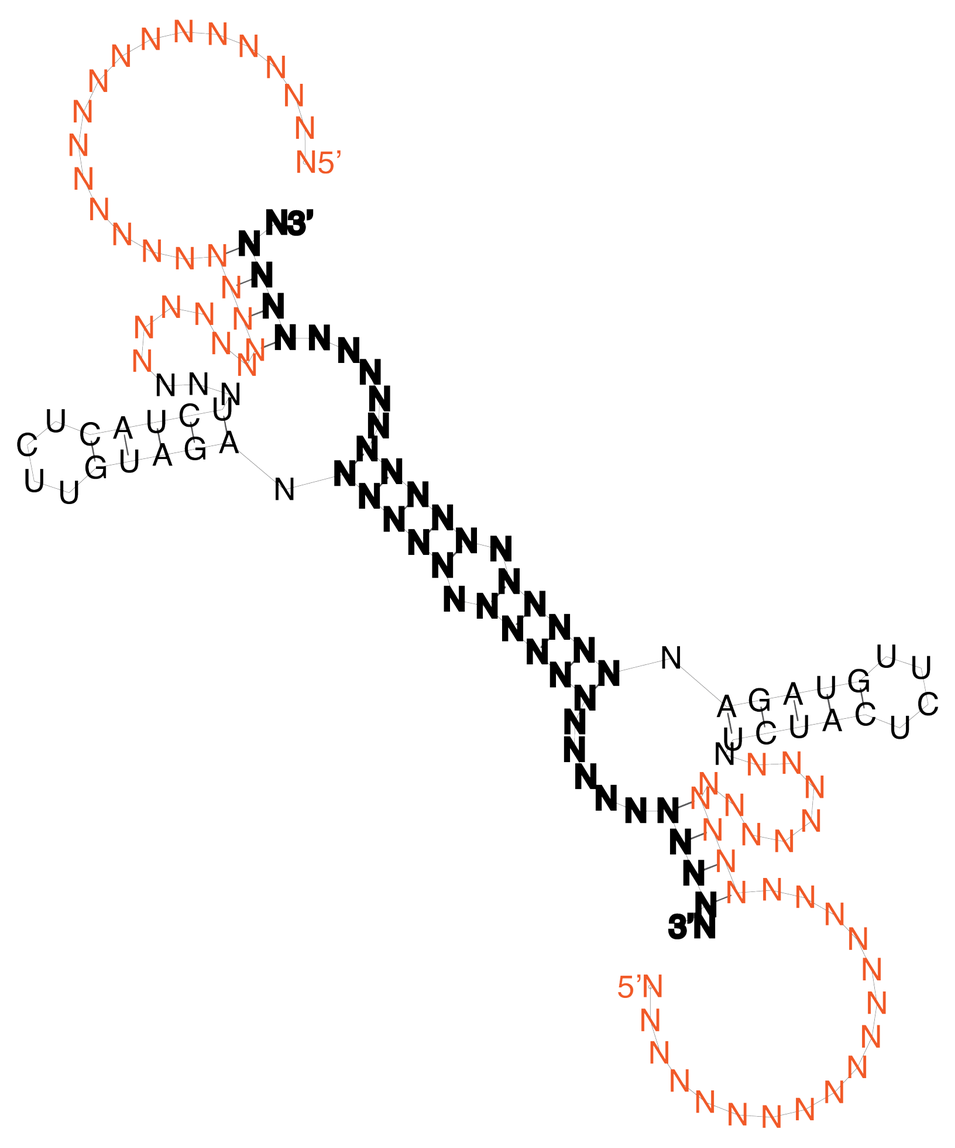

A) Graph showing minimization of free energy during RNA folding calculations
B) Secondary structure prediction of gRNA3, which has minimal extension-targeting region interactions and also is not predicted to be a dimer.
C) Secondary structure prediction for gRNA1, predicts extension-targeting region interactions.
D) Secondary structure prediction for gRNA1, when folded in the presence of itself, is predicted to form a dimer which is mediated by targeting region-targeting region interactions.

Extensions are shown in orange, targeting regions in bold, hairpin in regular font.

Figure 5: Secondary structure predictions for gRNA2

A)

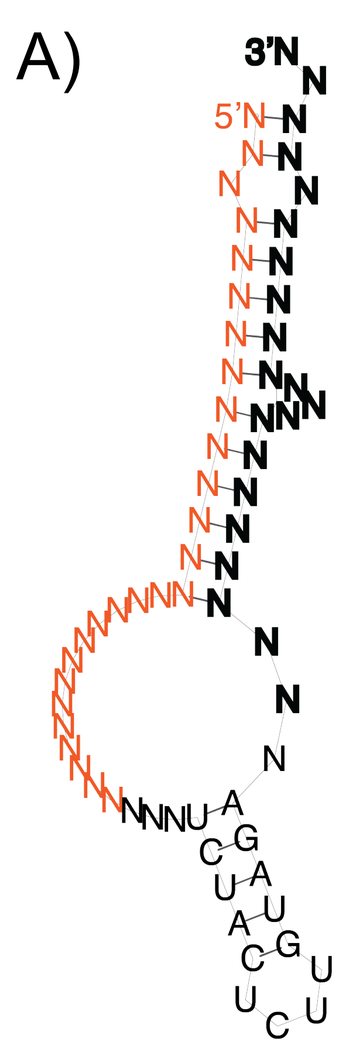

B)

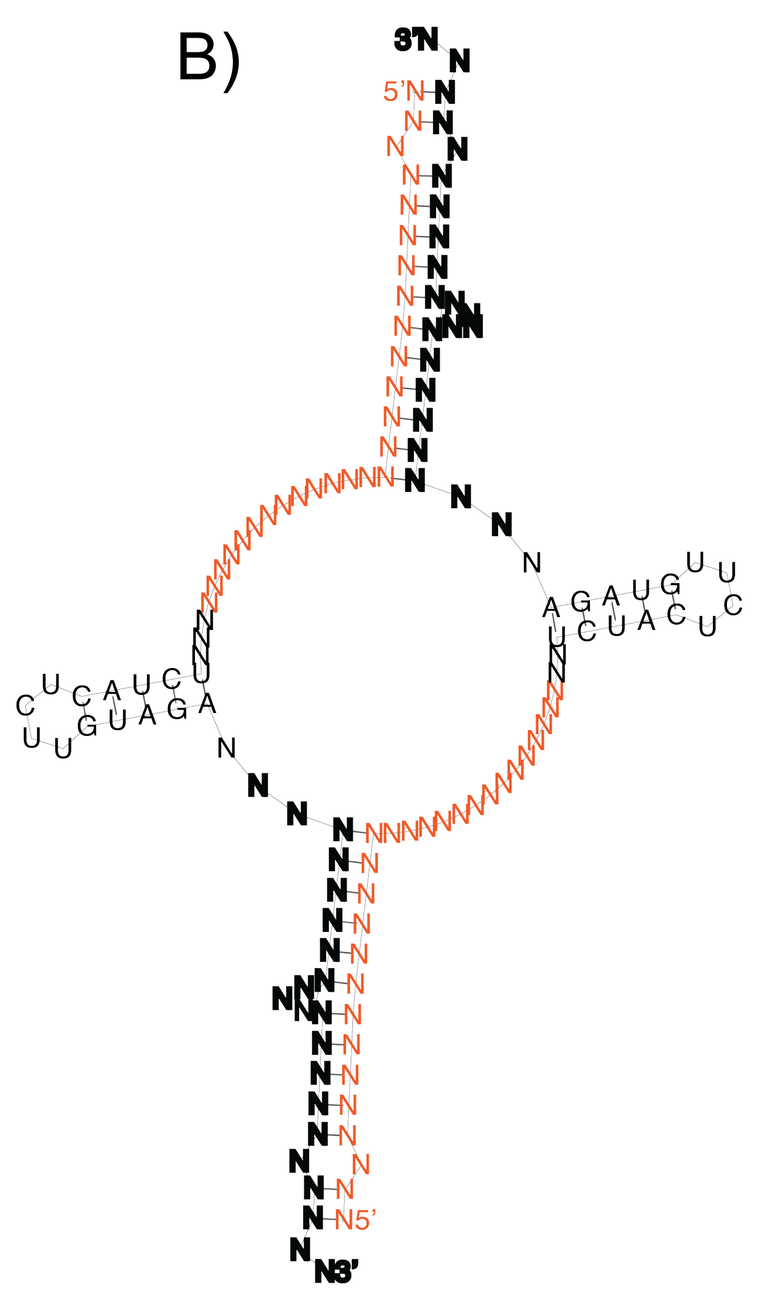

C)

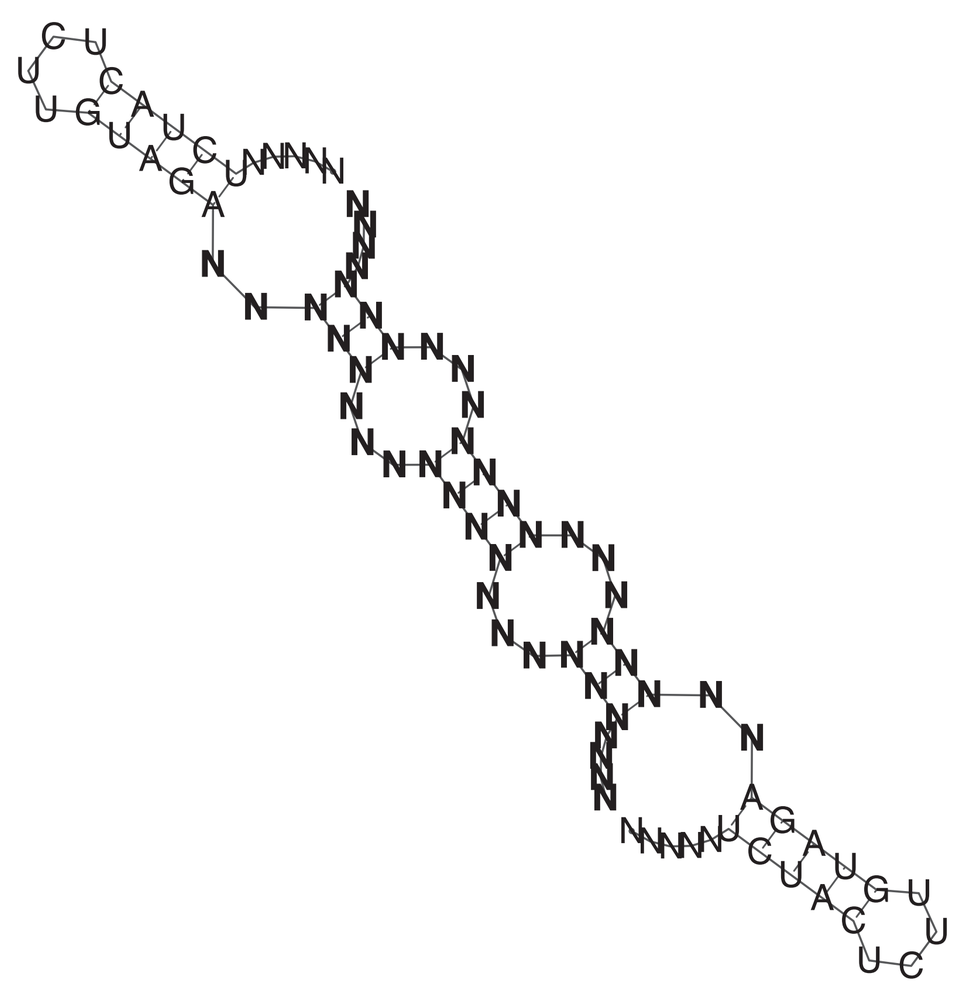

D)

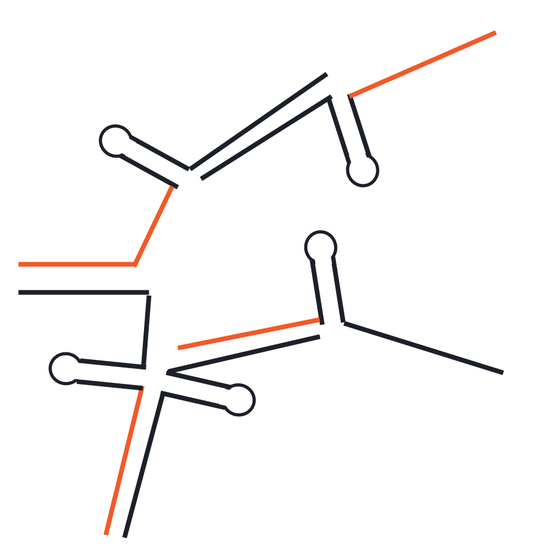

E)

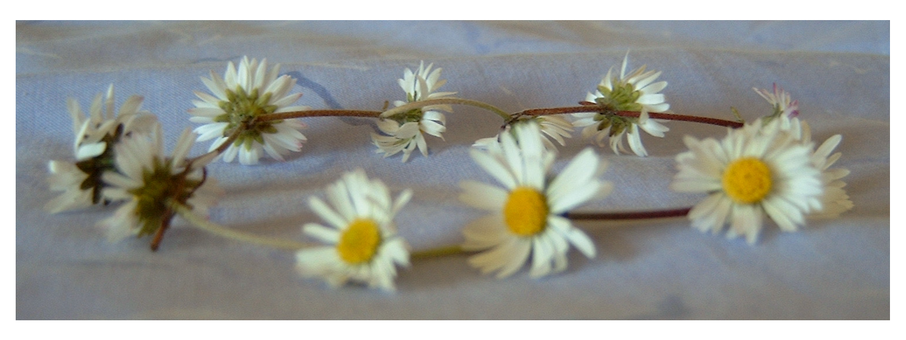

A) Secondary structure prediction for gRNA2 shows that the extension and targeting region are predicted to interact with one another.

B) Secondary structure prediction for the dimerization of gRNA2 with itself predicts extension-targeting region interactions.

C) Secondary structure prediction for the 41-mer of gRNA2 shows targeting region interactions as the mechanism for dimerization.

D) Cartoon showing how multiple gRNAs could interact with one another using primarily extension-targeting region interactions. Note that targeting region interactions are also possible, although not predicted to be the lowest energy structures.

E) Daisy chain model of 'head to tail' interactions.

Figure 6: Binding affinities derived from filter binding experiments

A)

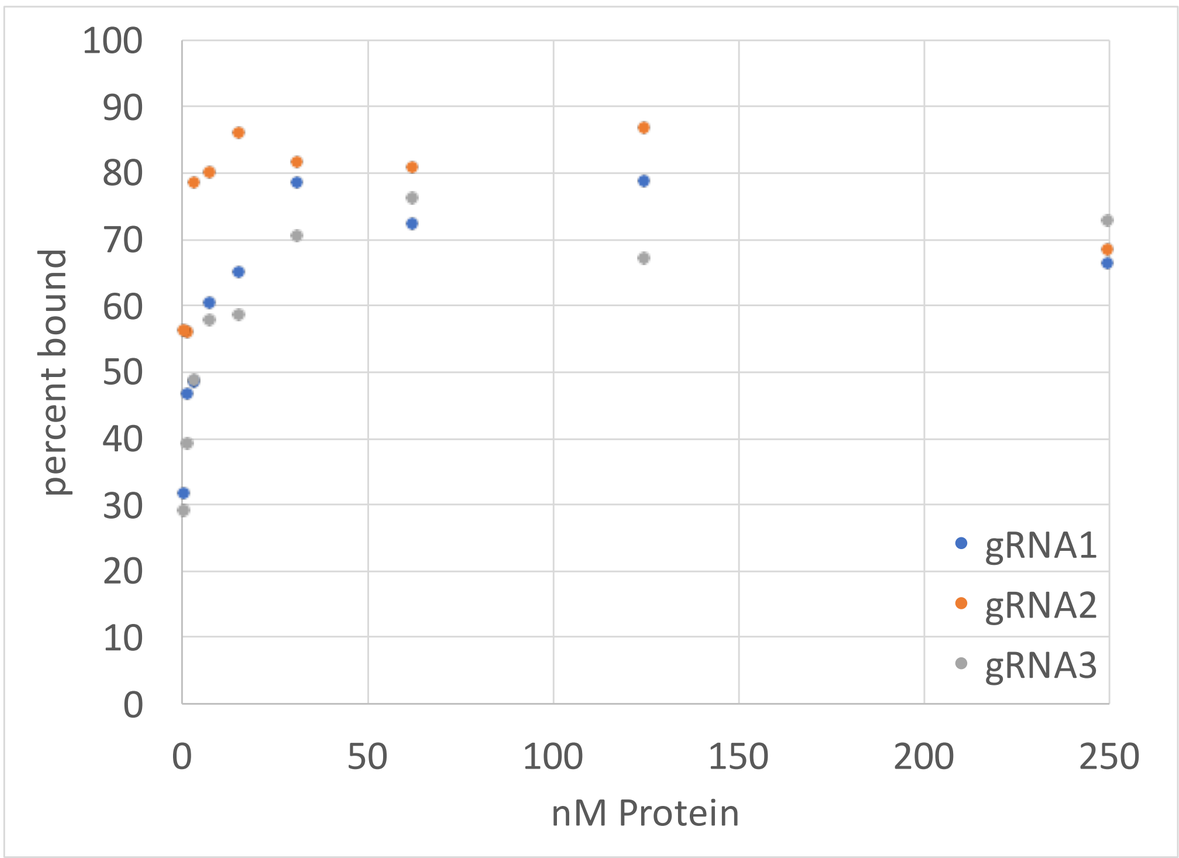
percent bound
nM Protein
gRNA1
gRNA2
gRNA3

B)

	gRNA1	gRNA2	gRNA3
Kd	1.4 nM	0.5 nM	1.7 nM

A) Graph showing percent bound derived from filter binding experiment with labeled gRNA at 250 pM and the concentration of protein varying up to 250 nM.
B) Binding curves were fit to a single site binding model and Kds were derived from the fit. All gRNAs bind relatively tightly. It is important to note that during these experiments, the gRNA is at a very low concentration, and this will drive the equilibrium to the monomer form.

CONCLUSION

gRNAs for different targets differ by only the variable targeting region. We show here three examples of gRNAs that have different behaviors as RNAs alone, and as RNPs. gRNA1 shows a strong propensity to dimerize via the targeting region, and this dimerization continues as an RNP. gRNA3 is a monomer as RNA alone, and as an RNP. The behavior for gRNA2 is complex. As a 41-mer, gRNA2 is a dimer, indicating that the dimerization is driven by targeting region interactions. However, as a 70-mer, the dimerization now disappears as extension-targeting interactions dominate the secondary structure and force the gRNA to behave as a monomer. As an RNP, gRNA2 drives multimerization as now 'daisy-chain' interactions, where the extension of one guide can interact with the targeting region of another, can cause several gRNAs or RNPs to associate with one another. It should not be overlooked that targeting region dimerization can also potentially play a role, but that targeting region interactions alone cannot explain the multimerization observed.

Acknowledgments:
We would like to thank the Process Chemistry Department at Editas for synthesizing the gRNAs.
Secondary structure predictions were made using ViennaRNA Package 2.0; Lorenz, Ronny and Bernhart, Stephan H. and Höner zu Siederdissen, Christian and Tafer, Hakim and Flamm, Christoph and Stadler, Peter F. and Hofacker, Ivo L. ViennaRNA Package 2.0, Algorithms for Molecular Biology, 6:1 26, 2011, doi:10.1186/1748-7188-6-26

DISCLOSURES

Chrysa Latrick is an employee and a stock holder of Editas Medicine, Inc.

Presented at: CSHL Genome Engineering: CRISPR Frontiers 2022

© 2022 Editas Medicine