EDIT-202, an AsCas12a and SLEEK™ gene-edited iPSC-derived NK cell therapy maintains prolonged persistence, high cytotoxicity, and enhanced *in vivo* control of solid tumors

Samia Q. Khan, Alexander G. Allen, Skyler Accomando, Robert Ford, Alexandra Gerew, Kaitlyn M. Izzo, Mrunali Jagdale, Andrew Barros, Jared Getgano, Aishwarya Ramanujan, Nadire R. Cochran, Stephen Sherman, Tusneem Janoudi, Patricia Sousa, Laura Blaha, Michael Nehil, John A. Follit, Abhijit Dandapat, Mark S. Shearman, Kai-Hsin Chang

Editas Medicine, Inc., Cambridge, MA, USA.

Poster P368

OBJECTIVE

To characterize and evaluate the anti-tumor efficacy of EDIT-202, an induced pluripotent stem cell (iPSC)–derived natural killer (iNK) cell therapy that has CD16 and membrane-bound interleukin-15 (mbIL-15) knocked in and cytokine-inducible SH2-containing protein (CISH) and transforming growth factor β receptor 2 (TGFβR2) knocked out using Editas' proprietary engineered, highly active, and highly specific AsCas12a and SLEEK™ technology.

INTRODUCTION

- EDIT-202 is an engineered iNK cell product under development to address the unmet need for treating solid tumors.
- Natural killer (NK) cell–based immunotherapy is a promising therapeutic approach for solid tumors because of its intrinsic tumor-killing capacity, minimal treatment-related toxicities, and ability to be administered to patients as an off-the-shelf therapy.
- NK cell effector function is diminished by a lack of functional persistence and tumor-intrinsic immunosuppressive mechanisms, such as the production of the pleiotropic cytokine transforming growth factor beta (TGF-β).[1] Furthermore, the ability of NK cells to exert antibody-dependent cellular cytotoxicity (ADCC) is impaired once CD16 is cleaved off after NK cell activation.[2] These biological limitations can be overcome with powerful editing tools that enable multiplexed gene editing of cells.
- EDIT-202 cells have been engineered with Editas' proprietary AsCas12a and the SeLection by Essential-gene Exon Knock-in (SLEEK™) gene editing tool to knock in CD16 and mbIL-15 and knock out CISH and TGFβR2. This prolongs persistence, enhances ADCC, and resists TGF-β-mediated immunosuppression in the tumor microenvironment.

Engineered AsCas12a and SLEEK™ enable the generation of iNK cells with enhanced functions against solid tumors

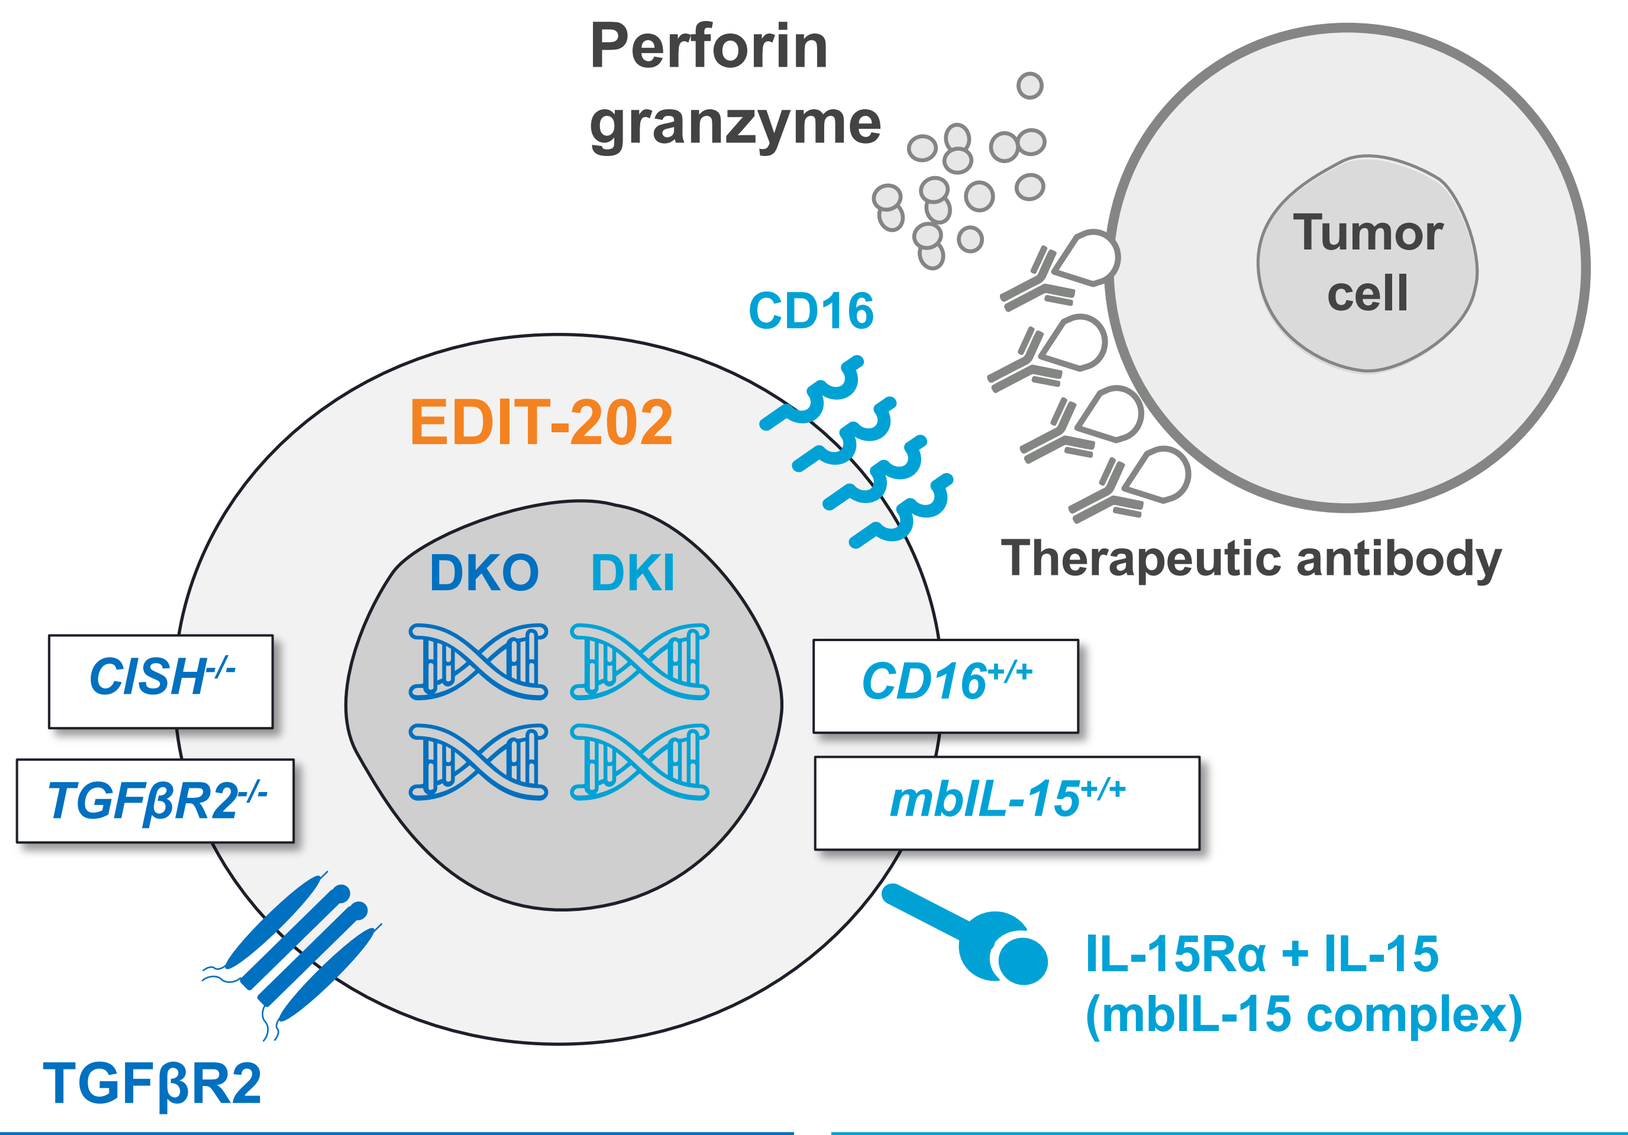

Perforin
granzyme
Tumor cell
CD16
EDIT-202
Therapeutic antibody
DKO DKI
CISH-/-
TGFβR2-/-
CD16+/+
mbIL-15+/+
IL-15Rα + IL-15
(mbIL-15 complex)
TGFβR2

$CISH^{-/-}/TGF\beta R2^{-/-}$ double knock-out (DKO)	$CD16^{+/+}/mbIL\text{-}15^{+/+}$ double knock-in (DKI)
Enhanced survival and/or expansion of iNK cells Increased IL-15 signaling through knock-out of CISH	**Enhanced survival and/or expansion of iNK cells** Increased IL-15 signaling through knock-in of mbIL-15 complex
Increased tumor control Resistance to TGF-β inhibition through knock-out of TGFβR2	**Enhanced ADCC** Constitutive overexpression of CD16 through knock-in of CD16

ADCC, antibody-dependent cellular cytotoxicity; CISH, cytokine-inducible SH2-containing protein; IL-15, interleukin-15; IL-15Rα, interleukin-15 receptor alpha; iNK, induced pluripotent stem cell-derived natural killer; mbIL-15, membrane-bound IL-15; SLEEK™, SeLection by Essential-gene Exon Knock-in: TGF-β, transforming growth factor beta; TGFβR2, transforming growth factor beta receptor 2.

METHODS

- iPSCs were edited with an engineered AsCas12a to knock in CD16 and mbIL-15 using the SLEEK™ method.[3] Simultaneously, iPSCs were also edited with AsCas12a to knock out CISH and TGFβR2. iPSC clones were selected and differentiated into iNK cells. Surface expression of CD16 and mbIL-15 by EDIT-202 cells was shown by flow cytometry (Fig. 1).
- *In vitro* persistence was measured by culturing wild-type (WT) and EDIT-202 cells in basal media without supporting cytokines for 21 days (Fig. 2).
- A fluorescent lactate dehydrogenase (LDH) release assay using SKOV-3, FaDu, or A549 tumor cells was performed to assess 2D cell tumor killing (Fig. 3). CD16 shedding was analyzed by extracting iNK cells from the 2D killing assay after 48 hours and running flow cytometry (Fig. 4).
- A 3D tumor spheroid killing assay using Incucyte® imaging of NucLight Red–tagged SKOV-3 cells was used to determine iNK cell cytotoxicity (Fig. 3 & 4).
- Non-obese diabetic (NOD) severe combined immunodeficient (scid) gamma (NSG) mice were inoculated intravenously with 0.125–1 × 10^6 luciferase (luc)–expressing SKOV-3 ovarian tumor cells (SKOV-3-luc). Mice received a single intravenous (IV) dose of 1.7–20 × 10^6 EDIT-202 cells with an IV dose of 2.5 mg/kg trastuzumab (TRA). Tumor burden was calculated using a PerkinElmer bioluminescent *in vivo* imaging system (IVIS) (Fig. 5).

RESULTS

Fig 1. EDIT-202 cells had increased CD16 and IL-15Rα expression compared with WT iNK cells

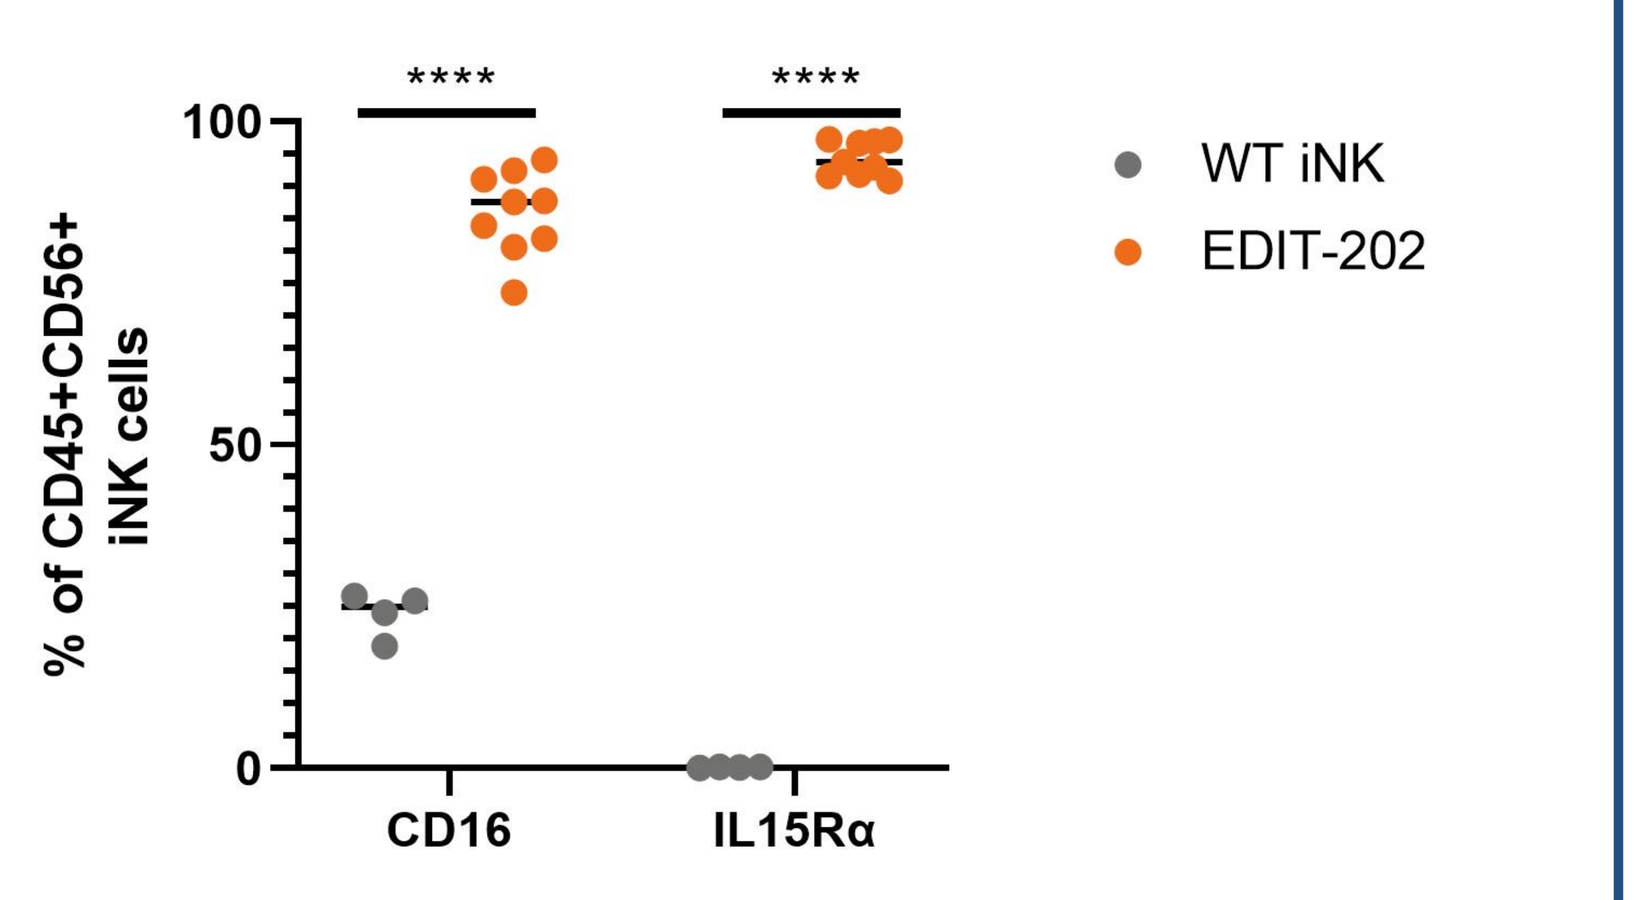

% of CD45+CD56+ iNK cells

WT iNK
EDIT-202
CD16
IL15Rα

Number of independent experiments: 3; ****$P \le 0.0001$ (two-way ANOVA).
IL-15Rα, interleukin-15 receptor alpha; iNK, induced pluripotent stem cell-derived natural killer; WT, wild-type.

Fig 2. mbIL-15 knock-in resulted in prolonged persistence of EDIT-202 cells in the absence of cytokines

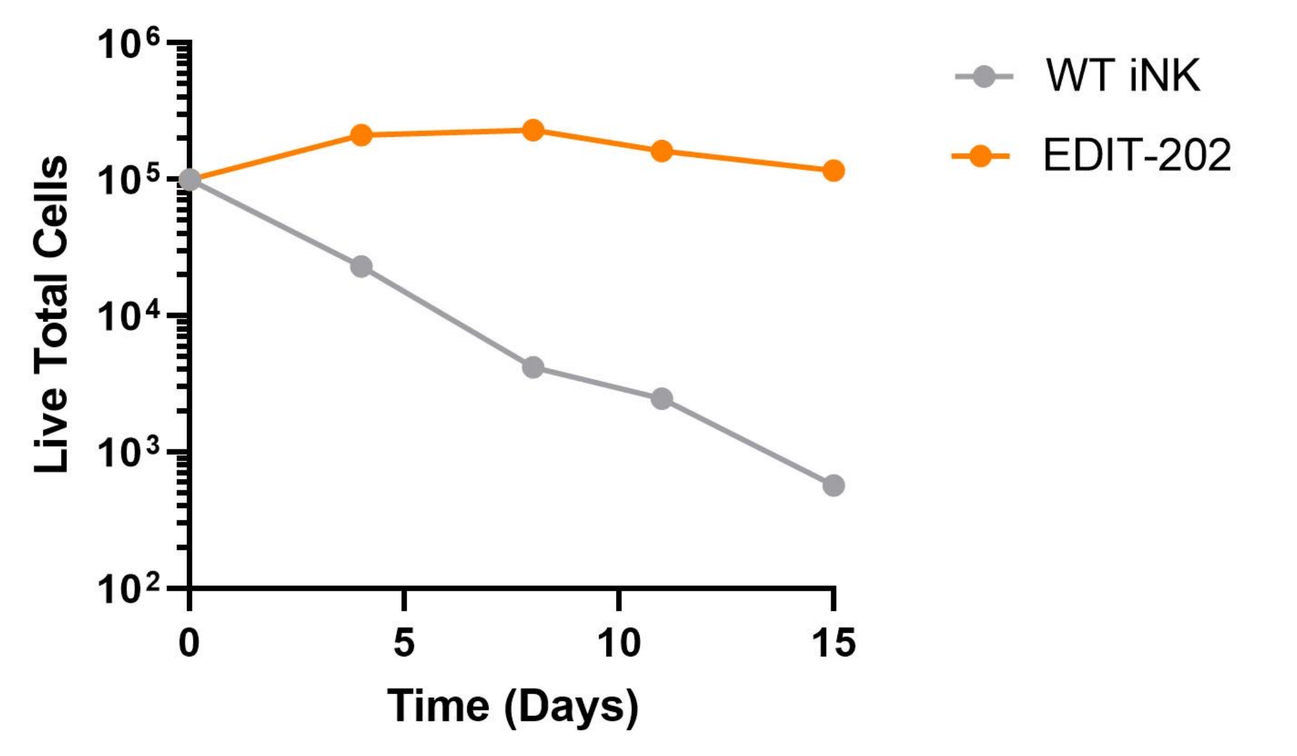

Live Total Cells
Time (Days)
WT iNK
EDIT-202

Number of independent experiments: 1.
iNK, induced pluripotent stem cell-derived natural killer; mbIL-15, membrane-bound interleukin-15; WT, wild-type.

Fig 3. EDIT-202 had enhanced ADCC-mediated cytotoxicity against multiple solid tumor cell lines and produced elevated levels of IFN-γ

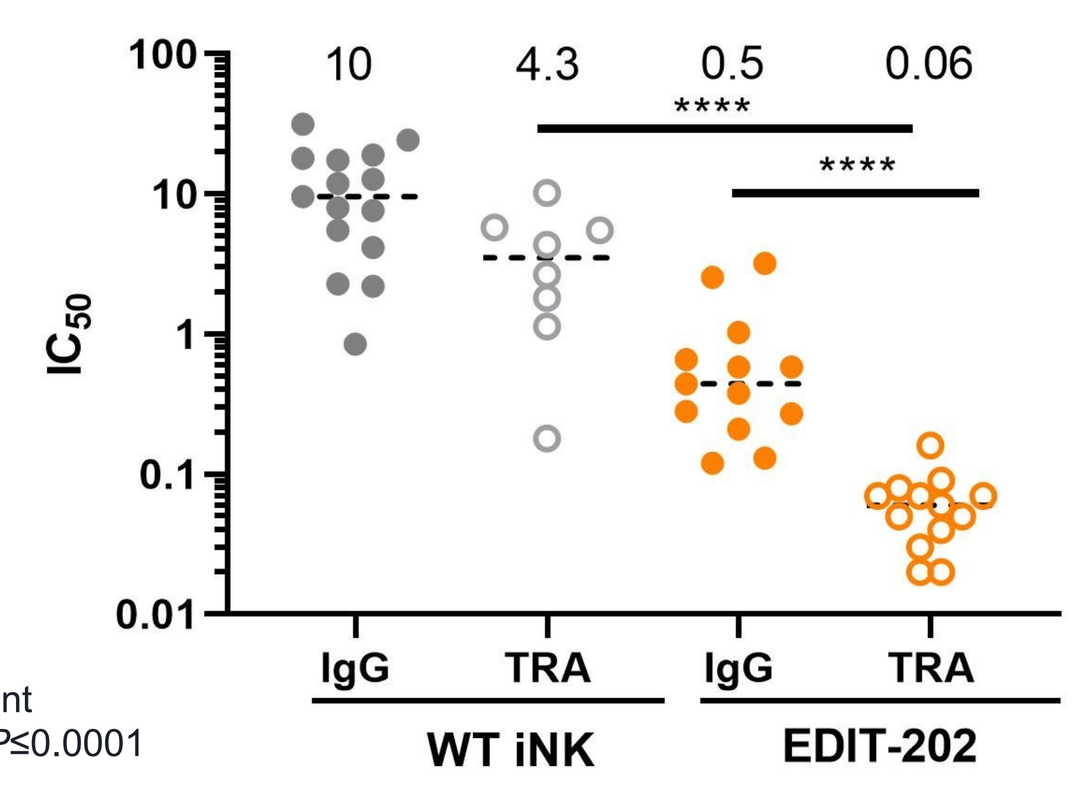

3D SKOV-3 spheroid killing
10
4.3
0.5
0.06
IC50
IgG
TRA
IgG
TRA
WT iNK
EDIT-202

Number of independent experiments: 11; ****$P \le 0.0001$ (unpaired t-test).

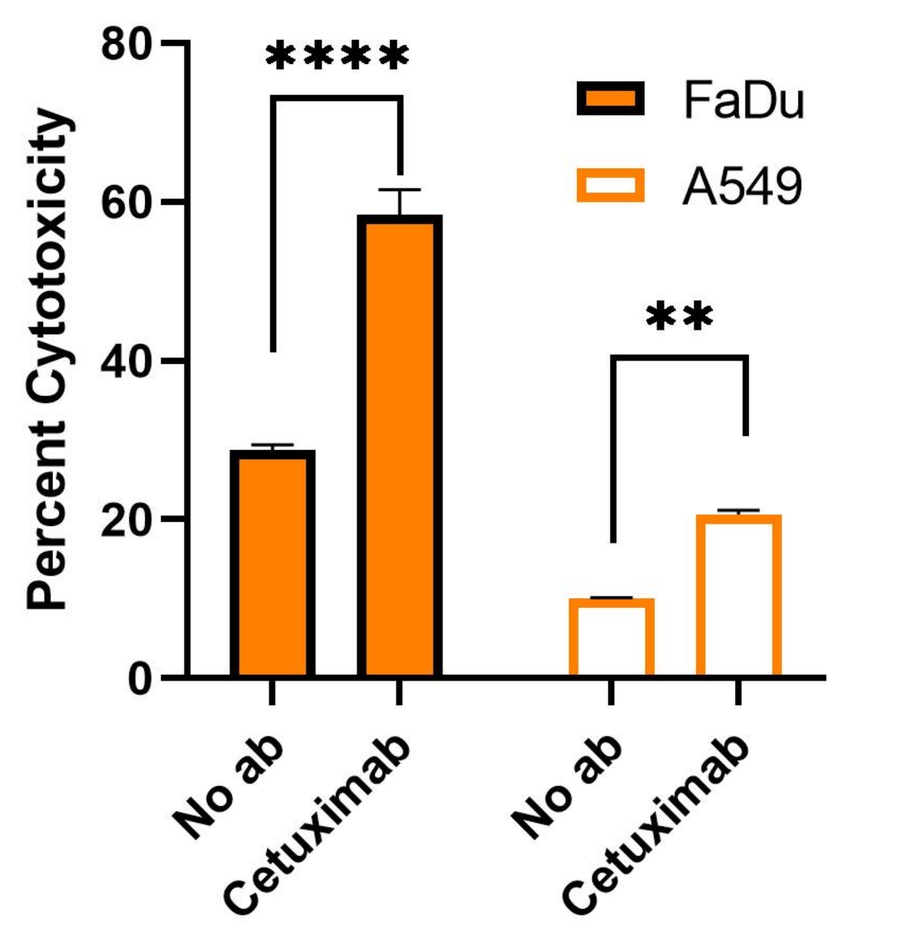

LDH release assay
Percent Cytotoxicity
FaDu
A549
No ab
Cetuximab
No ab
Cetuximab

Number of independent experiments: 2; ****$P \le 0.0001$; **$P<0.01$ (unpaired t-test).

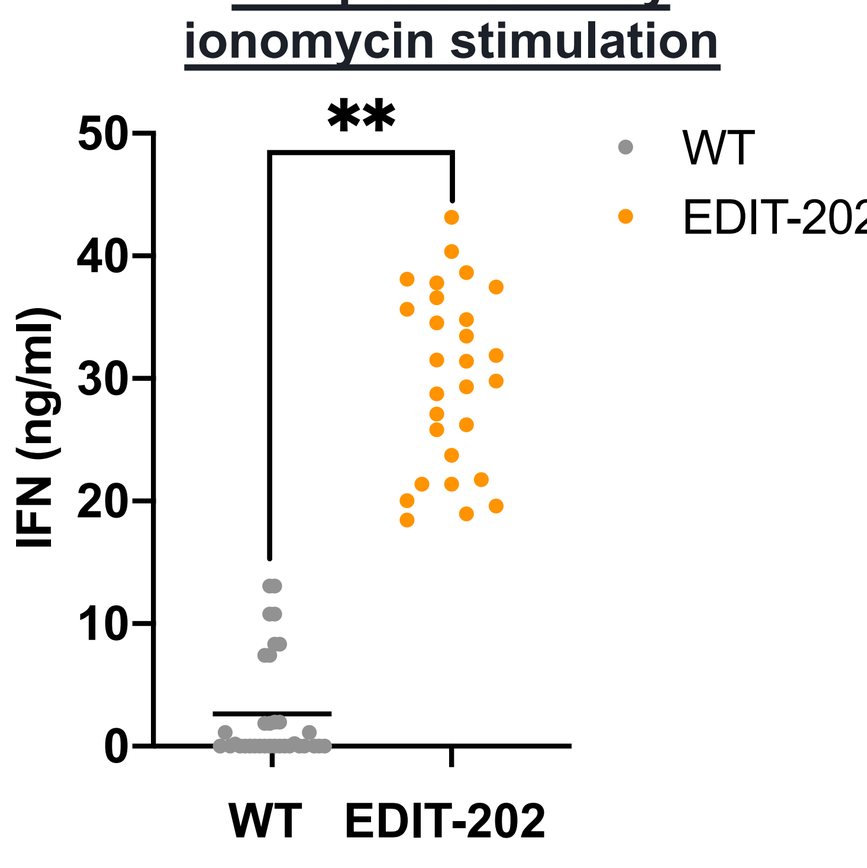

IFN-γ secretion by ionomycin stimulation
IFN (ng/ml)
WT
EDIT-202

Number of independent experiments: 3–9; **$P=0.0067$ (paired t-test)

3D, three-dimensional; ab, antibody; ADCC, antibody-dependent cellular cytotoxicity; IC_{50}, half maximal inhibitory concentration; IFN-γ, interferon gamma; IgA, immunoglobin A; iNK, induced pluripotent stem cell-derived natural killer; LDH, Lactate dehydrogenase; TRA, trastuzumab; WT, wild-type.

Fig 4. Constitutive surface expression of CD16 enabled EDIT-202 cells to induce enhanced serial killing of 3D SKOV-3 tumors, which was maintained in the presence of TGF-β

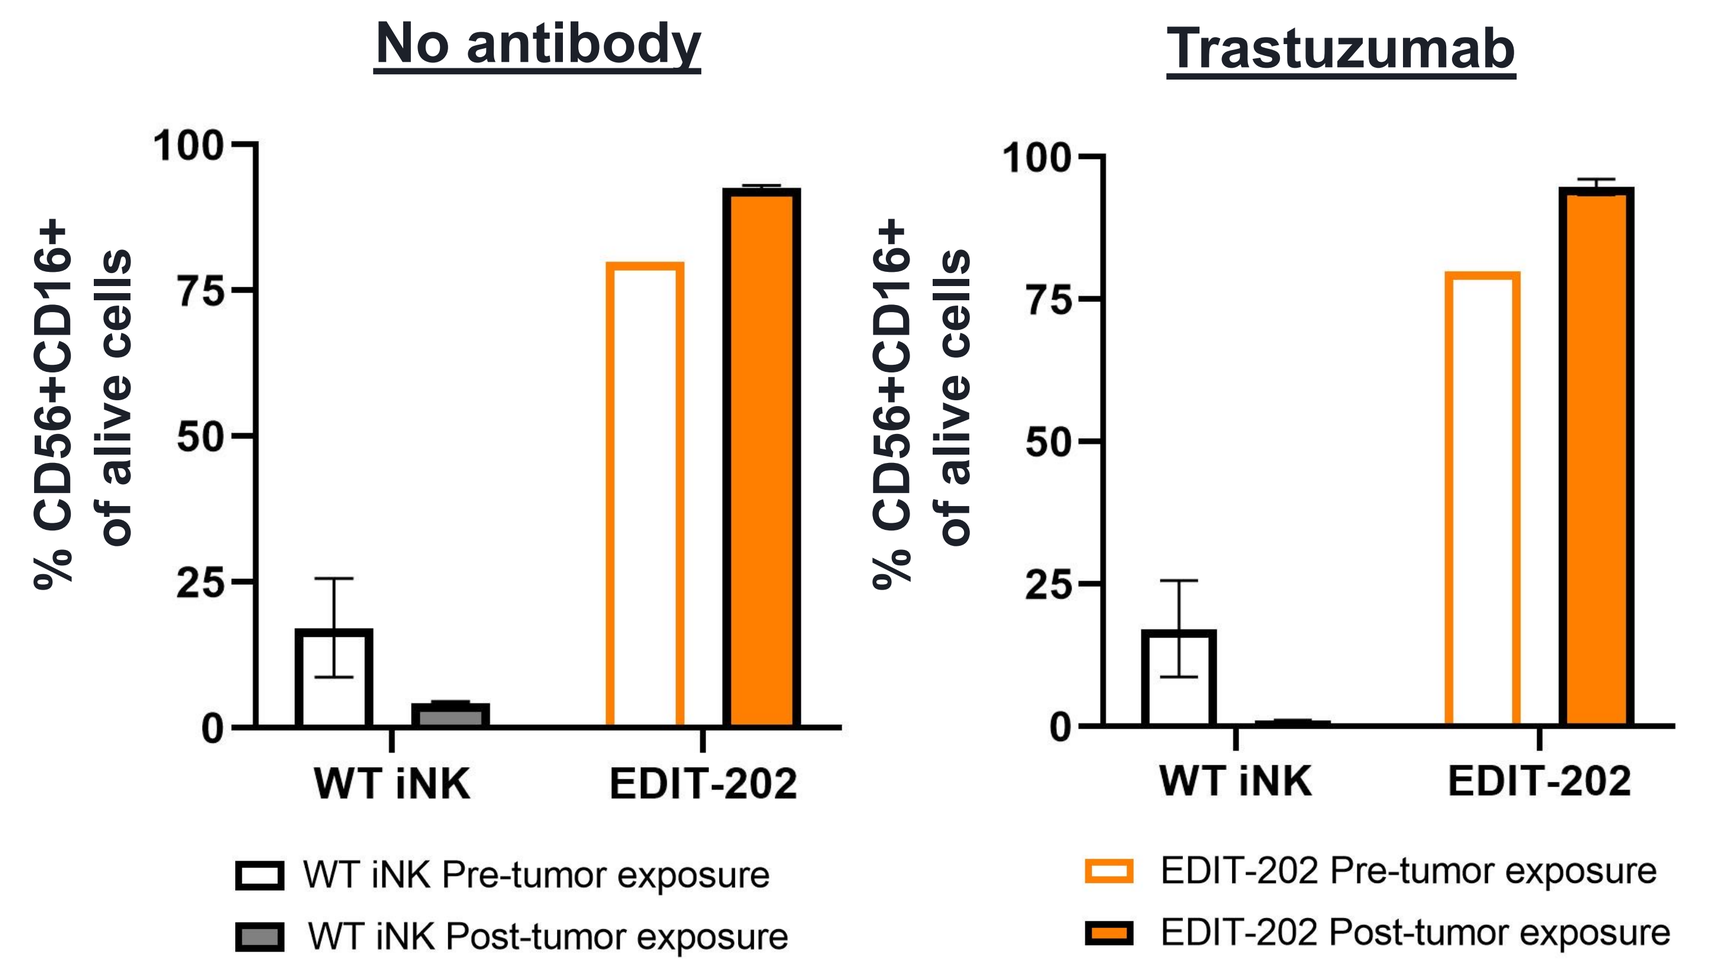

No antibody
Trastuzumab
% CD56+CD16+ of alive cells
WT iNK
EDIT-202
WT iNK Pre-tumor exposure
WT iNK Post-tumor exposure
EDIT-202 Pre-tumor exposure
EDIT-202 Post-tumor exposure

Number of independent experiments: 1.

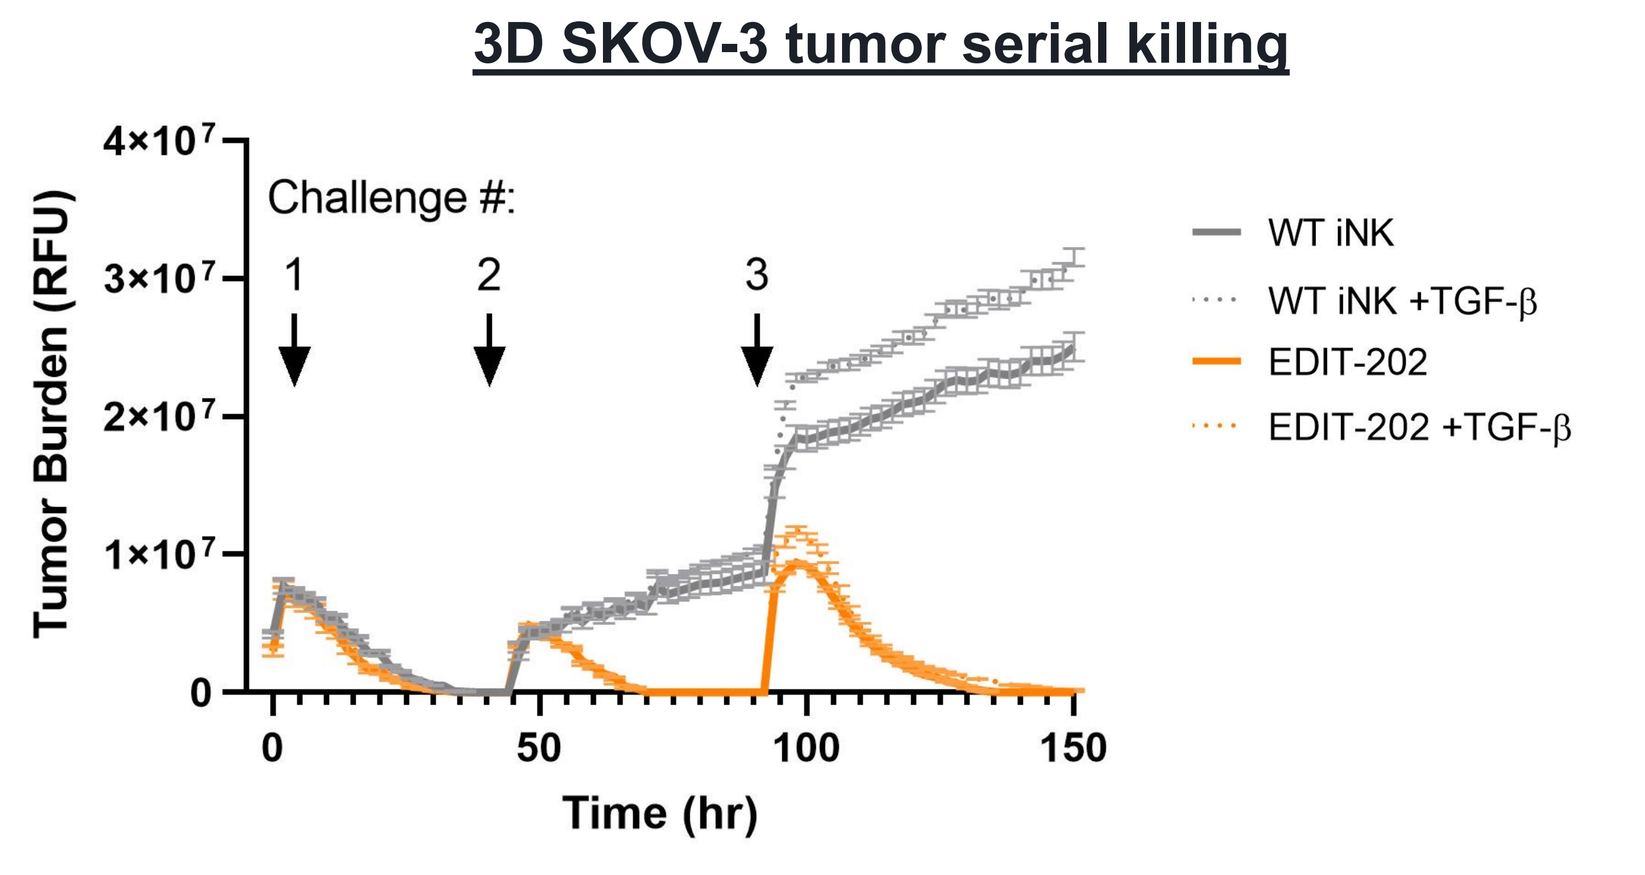

3D SKOV-3 tumor serial killing
Challenge #:
Tumor Burden (RFU)
Time (hr)
WT iNK
WT iNK +TGF-β
EDIT-202
EDIT-202 +TGF-β

Number of independent experiments: 1.

iNK, induced pluripotent stem cell-derived natural killer; LDH, Lactate dehydrogenase; RFU, relative fluorescence units; TGF-β, transforming growth factor beta; WT, wild-type

Fig 5. EDIT-202 administered in combination with TRA induced significant reduction in tumor burden to complete tumor clearance in an ovarian SKOV-3-luc IV solid tumor model

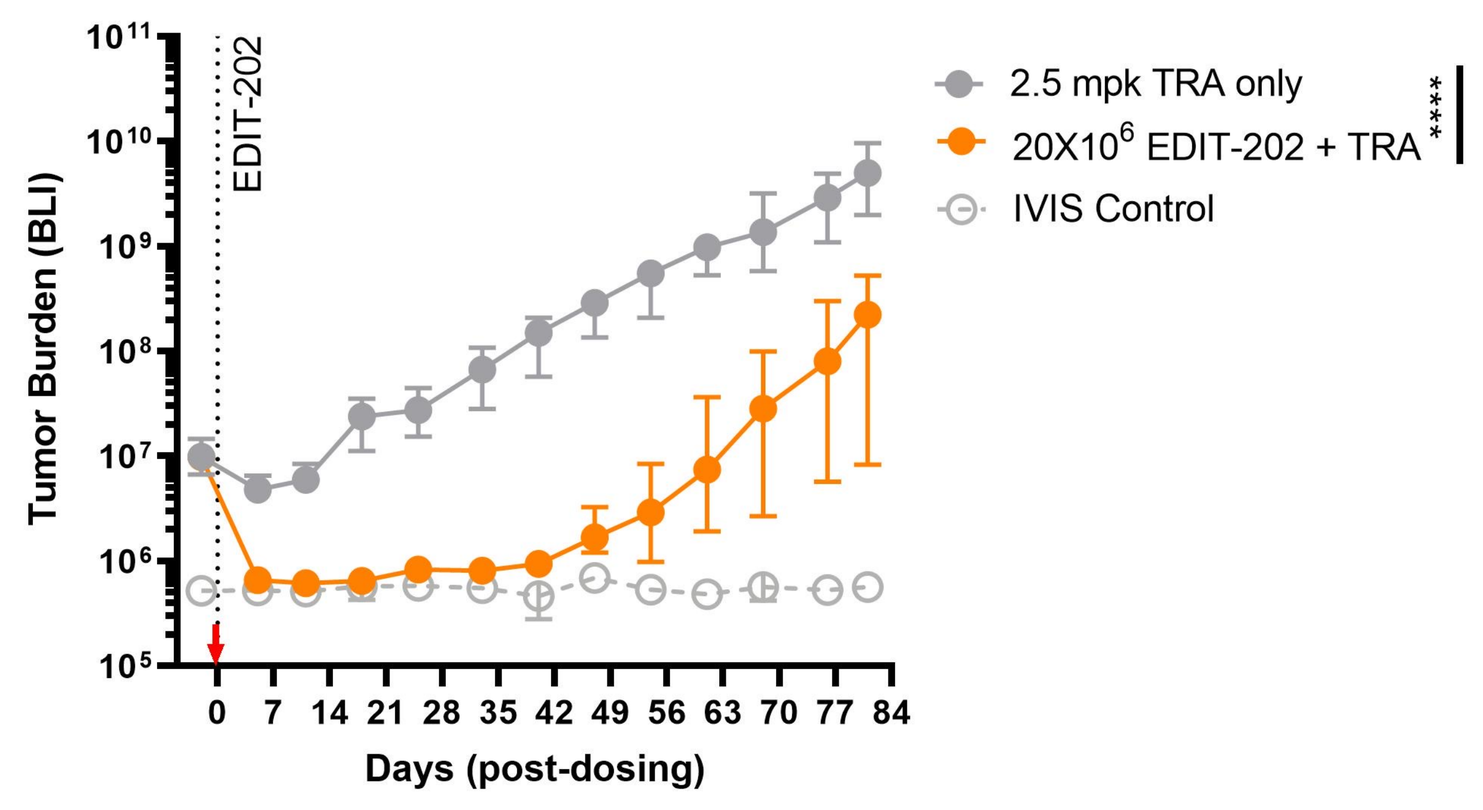

Tumor Burden (BLI)
EDIT-202
Days (post-dosing)
2.5 mpk TRA only
20X10^6 EDIT-202 + TRA
IVIS Control

Number of mice per group: 4; ****$P<0.0001$ (one-way ANOVA).

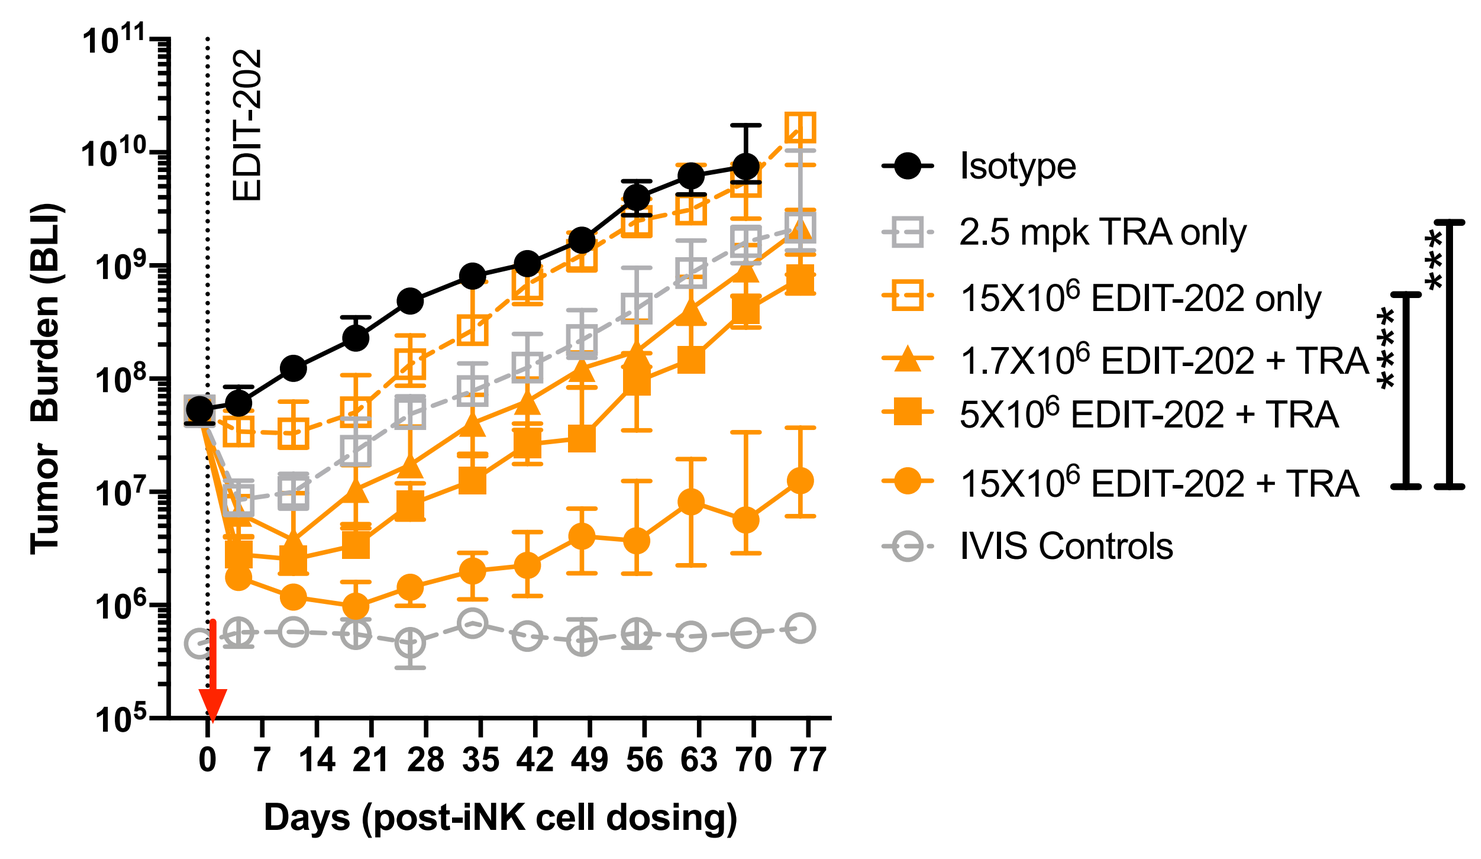

Tumor Burden (BLI)
EDIT-202
Days (post-iNK cell dosing)
Isotype
2.5 mpk TRA only
15X10^6 EDIT-202 only
1.7X10^6 EDIT-202 + TRA
5X10^6 EDIT-202 + TRA
15X10^6 EDIT-202 + TRA
IVIS Controls

Number of mice per group: 5; ****$P<0.0001$; ***$P=0.008$ (one-way ANOVA).

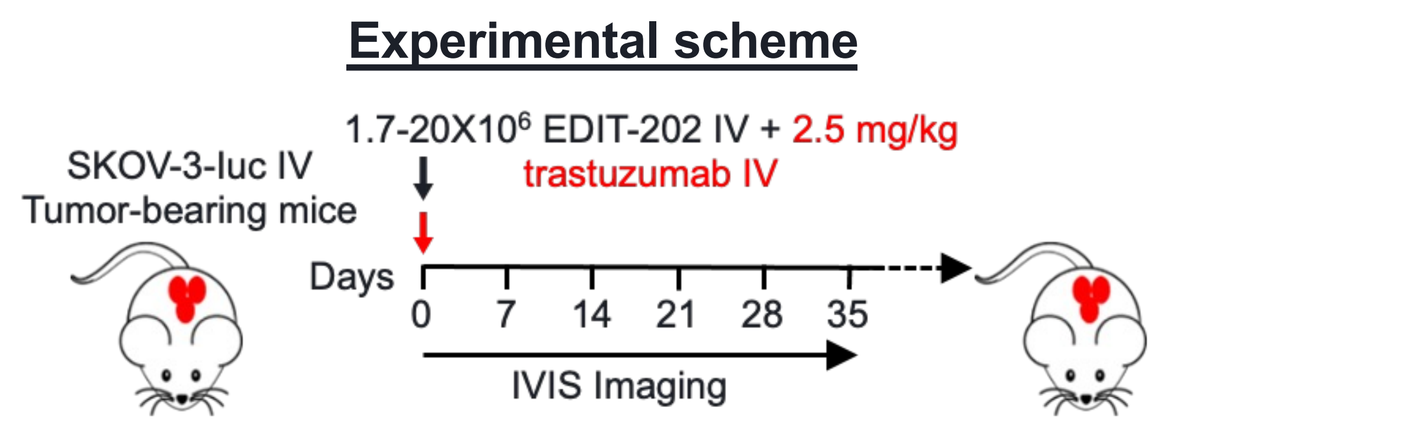

Experimental scheme
SKOV-3-luc IV Tumor-bearing mice
1.7-20X10^6 EDIT-202 IV + 2.5 mg/kg trastuzumab IV
Days
IVIS Imaging

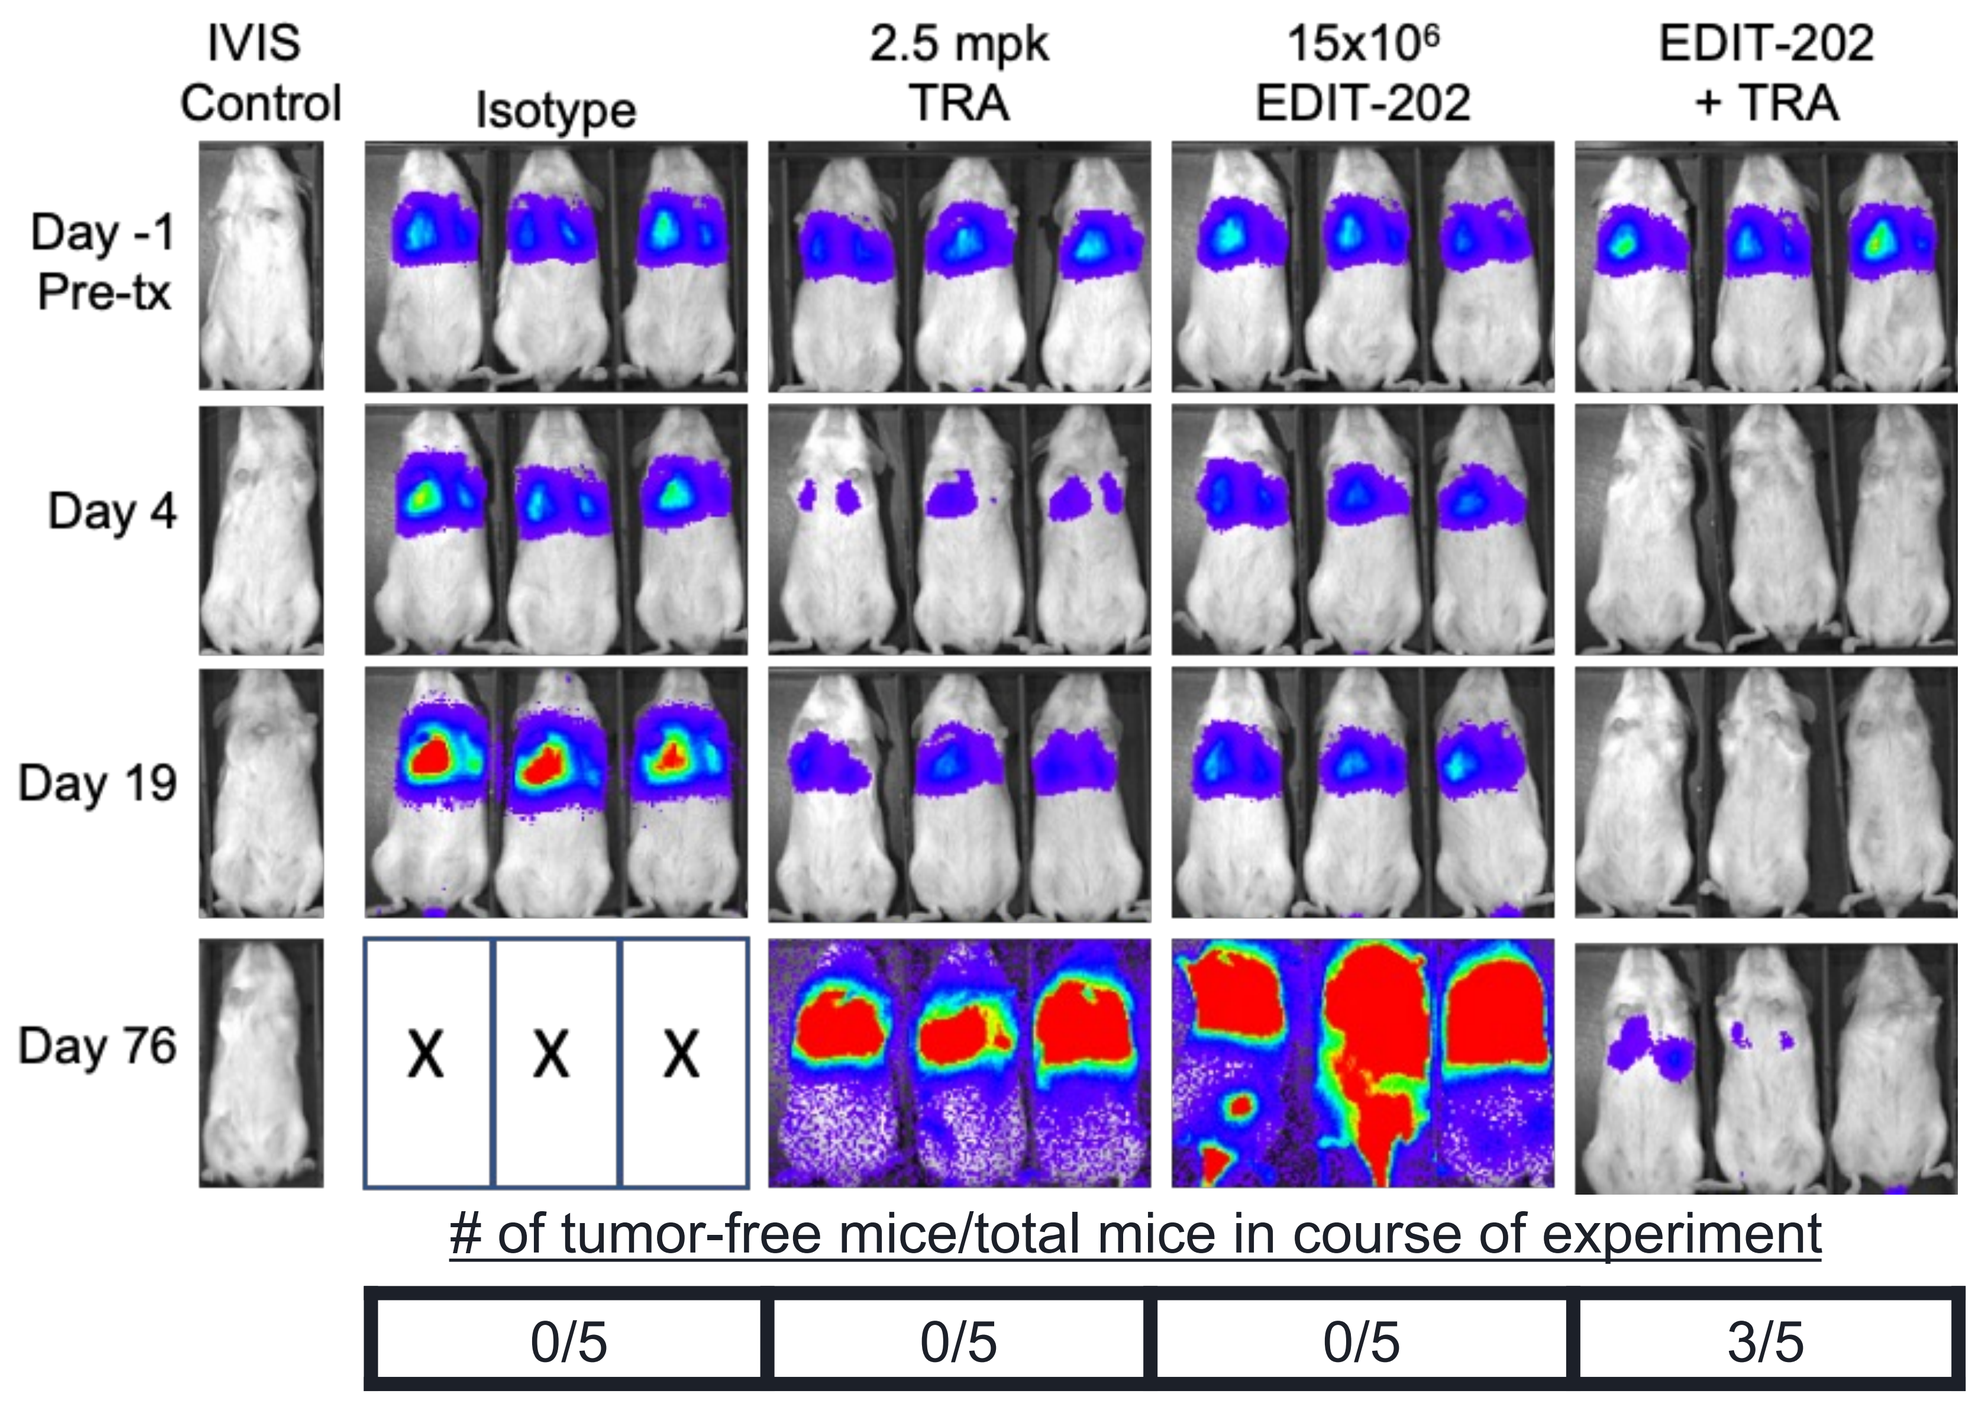

IVIS Control
Isotype
2.5 mpk TRA
15x10^6 EDIT-202
EDIT-202 + TRA
Day -1 Pre-tx
Day 4
Day 19
Day 76
X
X
X

of tumor-free mice/total mice in course of experiment

0/5	0/5	0/5	3/5

BLI, bioluminescent imaging; iNK, induced pluripotent stem cell-derived natural killer; IV, intravenous; IVIS, *in vivo* imaging system; SKOV-3-luc, luciferase-expressing SKOV-3 cell line; TRA, trastuzumab; tx, treatment; WT, wild-type

Fig 6. EDIT-202 treatment with TRA significantly increased overall survival in an ovarian SKOV-3-luc IV solid tumor model

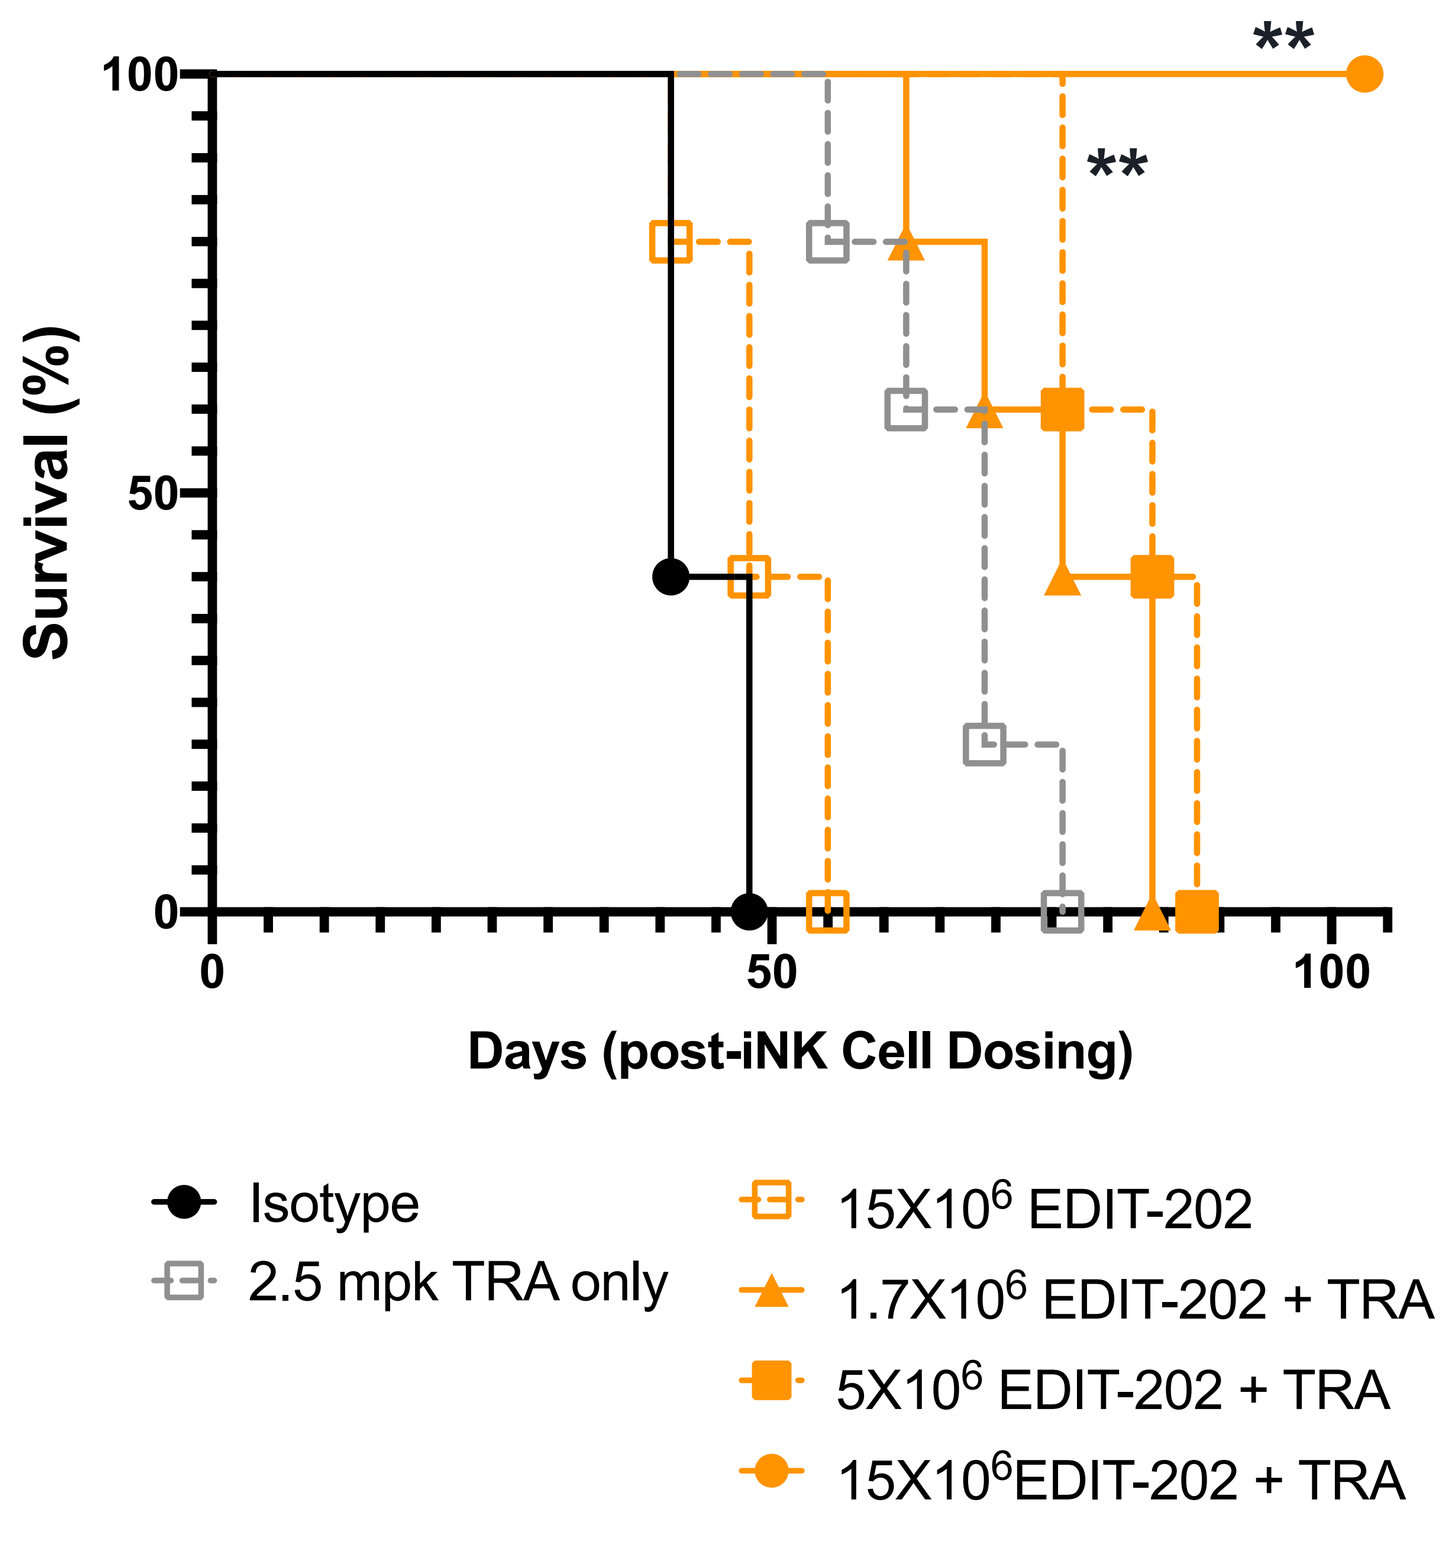

Survival (%)
Days (post-iNK Cell Dosing)
**
**
Isotype
2.5 mpk TRA only
15X10^6 EDIT-202
1.7X10^6 EDIT-202 + TRA
5X10^6 EDIT-202 + TRA
15X10^6EDIT-202 + TRA

Number of mice per group: 5; **$P<0.01$ (Log-rank test, compared with TRA only).
iNK, induced pluripotent stem cell-derived natural killer; IV, intravenous; SKOV-3-luc, luciferase-expressing SKOV-3 cell line; TRA, trastuzumab

CONCLUSIONS

- EDIT-202 had significantly higher levels of CD16 and mbIL-15 compared with WT iNK cells, indicating that our SLEEK™ technology is a robust method for editing iPSCs.
- EDIT-202 had prolonged cytokine-independent persistence *in vitro* compared with WT iNK cells, indicating that the mbIL-15 knock-in in EDIT-202 cells is sufficient for survival and maintenance.
- EDIT-202 showed significantly higher ADCC-mediated cytotoxicities against ovarian SKOV-3, head and neck FaDu, and lung adenocarcinoma A549 tumor cells. Activated EDIT-202 induced significantly higher levels of IFN-γ.
- EDIT-202 cells had upregulated and continuous expression of CD16 after tumor exposure, which enabled the cells to significantly induce serial killing of SKOV-3 tumor cells compared with WT iNK cells. EDIT-202 serial killing capacity was not affected in the presence of immunosuppressive TGF-β.
- In an *in vivo* SKOV-3-luc solid tumor model, EDIT-202 + TRA treatment led to a significant reduction in lung tumor burden compared with TRA only and resulted in complete tumor clearance in multiple mice.
- When combined with TRA, EDIT-202 significantly increased survival over TRA only treatment in the solid tumor SKOV-3 model. At Day 100, 100% of 15 × 10^6 EDIT-202 + TRA treated mice were alive, whereas there was 0% survival with TRA only.
- These data support the development of EDIT-202 as a potential allogeneic cell-based medicine for treating solid tumors.

REFERENCES

1. Wu Y *et al*. *Front Immunol* 2017; 8: 930.
2. Snyder KM *et al*. *Front Immunol* 2018; 9: 2873.
3. A Method for Highly Efficient Knock-in and Expression of Transgene Cargos for NextGeneration Cell-based Medicines. Available at: https://www.editasmedicine.com/wp-content/uploads/2021/08/2021_CSHL-CRISPR-Frontiers-SLEEK-Zuris_FINAL.pdf. Accessed September 2022.

DISCLOSURES

Employees and shareholders of Editas Medicine: S.Q.K., A.G.A., A.G., S.A., K.M.I., M.J., J.G., A.R., N.R.C., S.S., P.S., M.N., J.A.F., A.D., M.S.S., K-H.C.

Former employees of Editas Medicine: A.G, J.G.,L.B.

ACKNOWLEDGMENTS

This work was funded by Editas Medicine. The authors would like to thank all their Editas colleagues for helping to plan, perform, analyze, and present this work. Editorial assistance was provided by Shervonne Poleon, PhD, and Tony Ferrar, MSc, ISMPP CMPP™ of Porterhouse Medical, and was funded by Editas Medicine in accordance with Good Publication Practice (GPP) guidelines.

© 2022 Editas Medicine