Design of Chemically Modified AsCas12a Guide RNAs for Increased Potency of LNP-Delivered Gene Editing Cargos

Bryant Chica, Alexis Burns, Mark Jones, Chrysa Latrick, Stephen Sherman, Jill Fletcher, McKenzie Weiss, Steve Wolk, Paul Wrighton, Vikram Soman, Swarali Lele, Judith Newmark, Stephen Pietrasiewicz, Katerina Tsiounis, Sheryl Bowley

Editas Medicine, Cambridge, MA, United States

Poster #623

Introduction

Modified gRNAs for Cas12a *in vivo* programs

- *In vivo* gene editing via delivery of CRISPR-Cas mRNA and guide RNA (gRNA)-containing lipid nanoparticles (LNPs) is emerging as a powerful technology to treat genetic diseases. Establishing a favorable therapeutic index for this technology depends on engineering the gRNA and mRNA payloads for optimal potency. Optimizing potency of the gRNA can require sequence and chemical modifications, leading to a rapidly expanding number of variants to evaluate. Here, we present the iterative incorporation of chemical modification motifs into AsCas12a gRNA that increases the potency of LNP gene editing cargos across multiple cell types and gRNA target sequences.

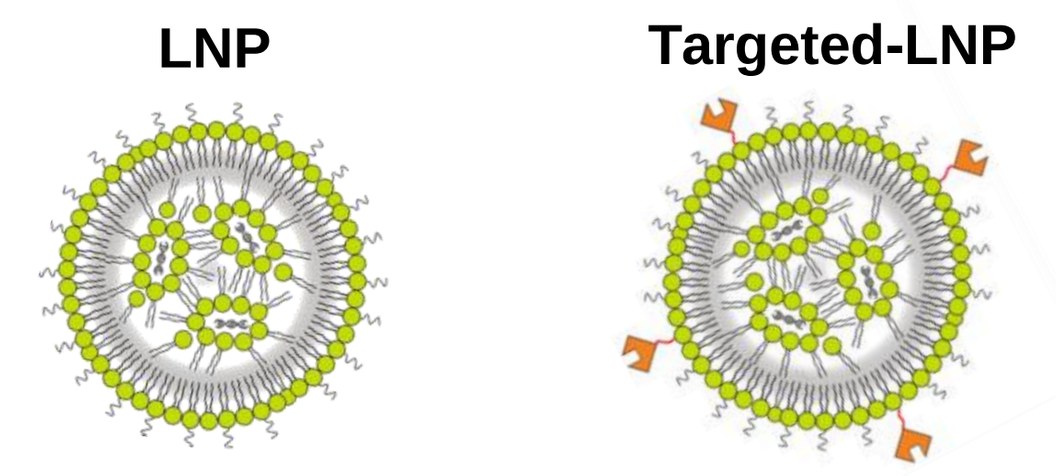
LNP
Targeted-LNP

Model of the *in cellulo* pathway to editing

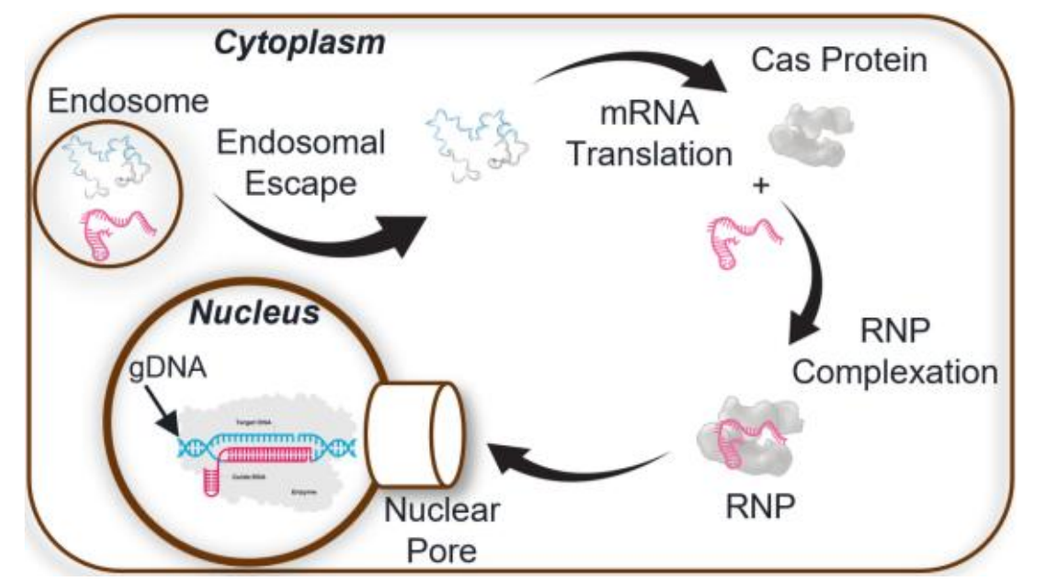
Cytoplasm
Endosome
Endosomal Escape
mRNA Translation
Cas Protein
RNP Complexation
RNP
Nucleus
gDNA
Nuclear Pore

Incorporating chemical modifications into CRISPR gRNA:

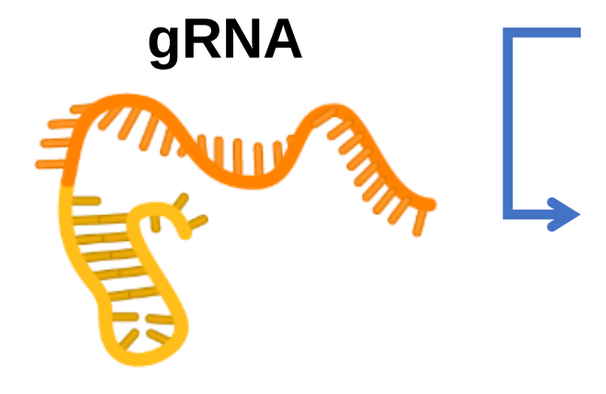
gRNA

- RNA
- Selected RNA positions replaced with patterns of chemical modifications

gRNA optimization workflow

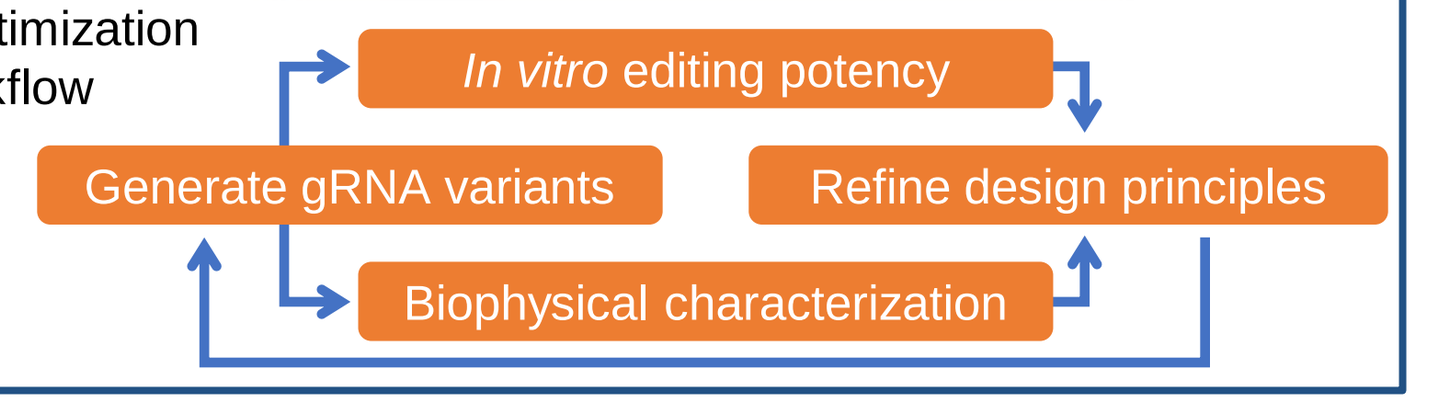
In vitro editing potency
Generate gRNA variants
Refine design principles
Biophysical characterization

Methods

- gRNAs were synthesized via standard phosphoramidite chemistry and purified by ion-pair reversed-phase preparative high-performance liquid chromatography (HPLC), followed by desalt and sequence analysis.
- LNPs were formulated with commercially available lipids using a NanoAssemblr™ Ignite™ (Cytiva). LNP cargo was mRNA encoding engineered AsCas12a[1] (enCas12a) nuclease and gRNA at a 1:1 ratio by weight. LNPs were evaluated for percent encapsulation >80% by RiboGreen assay (ThermoFisher Scientific), polydispersity index (PDI) <0.2, and average diameter size <105 nm by dynamic light scattering using the Zetasizer Ultra (Malvern Panalytical).
- For *in vitro* binding affinity measurements, labeled, unmodified gRNA was incubated with recombinant AsCas12a and increasing concentrations of the modified “test” guide at room temperature for 3 hours, and then double filter separated on nitrocellulose blotting membrane (Cytiva) and Hybond N+ membrane (Cytiva).
- For *in vitro* editing, cell cultures were treated with LNPs at indicated concentrations of enCas12a mRNA and included human apolipoprotein E. gDNA was isolated at 72 hours post-transfection. Percent editing was determined by amplicon-based next-generation sequencing (NGS).
- For *in vivo* editing, LNPs were delivered via intravenous tail vein injection to hMYOC(Y437H) transgenic mice (initial results) or NSBGW hematopoietic stem and progenitor cell (HSPC) mice (translation to editing potency). For the initial results panel, 1 week post-injection, the livers were dissected, gDNA was isolated, and NGS was performed to determine the percentage of editing.

Results: Initial gRNA modification patterns (1/2)

gRNA modification patterns improve editing *in vitro* and *in vivo*

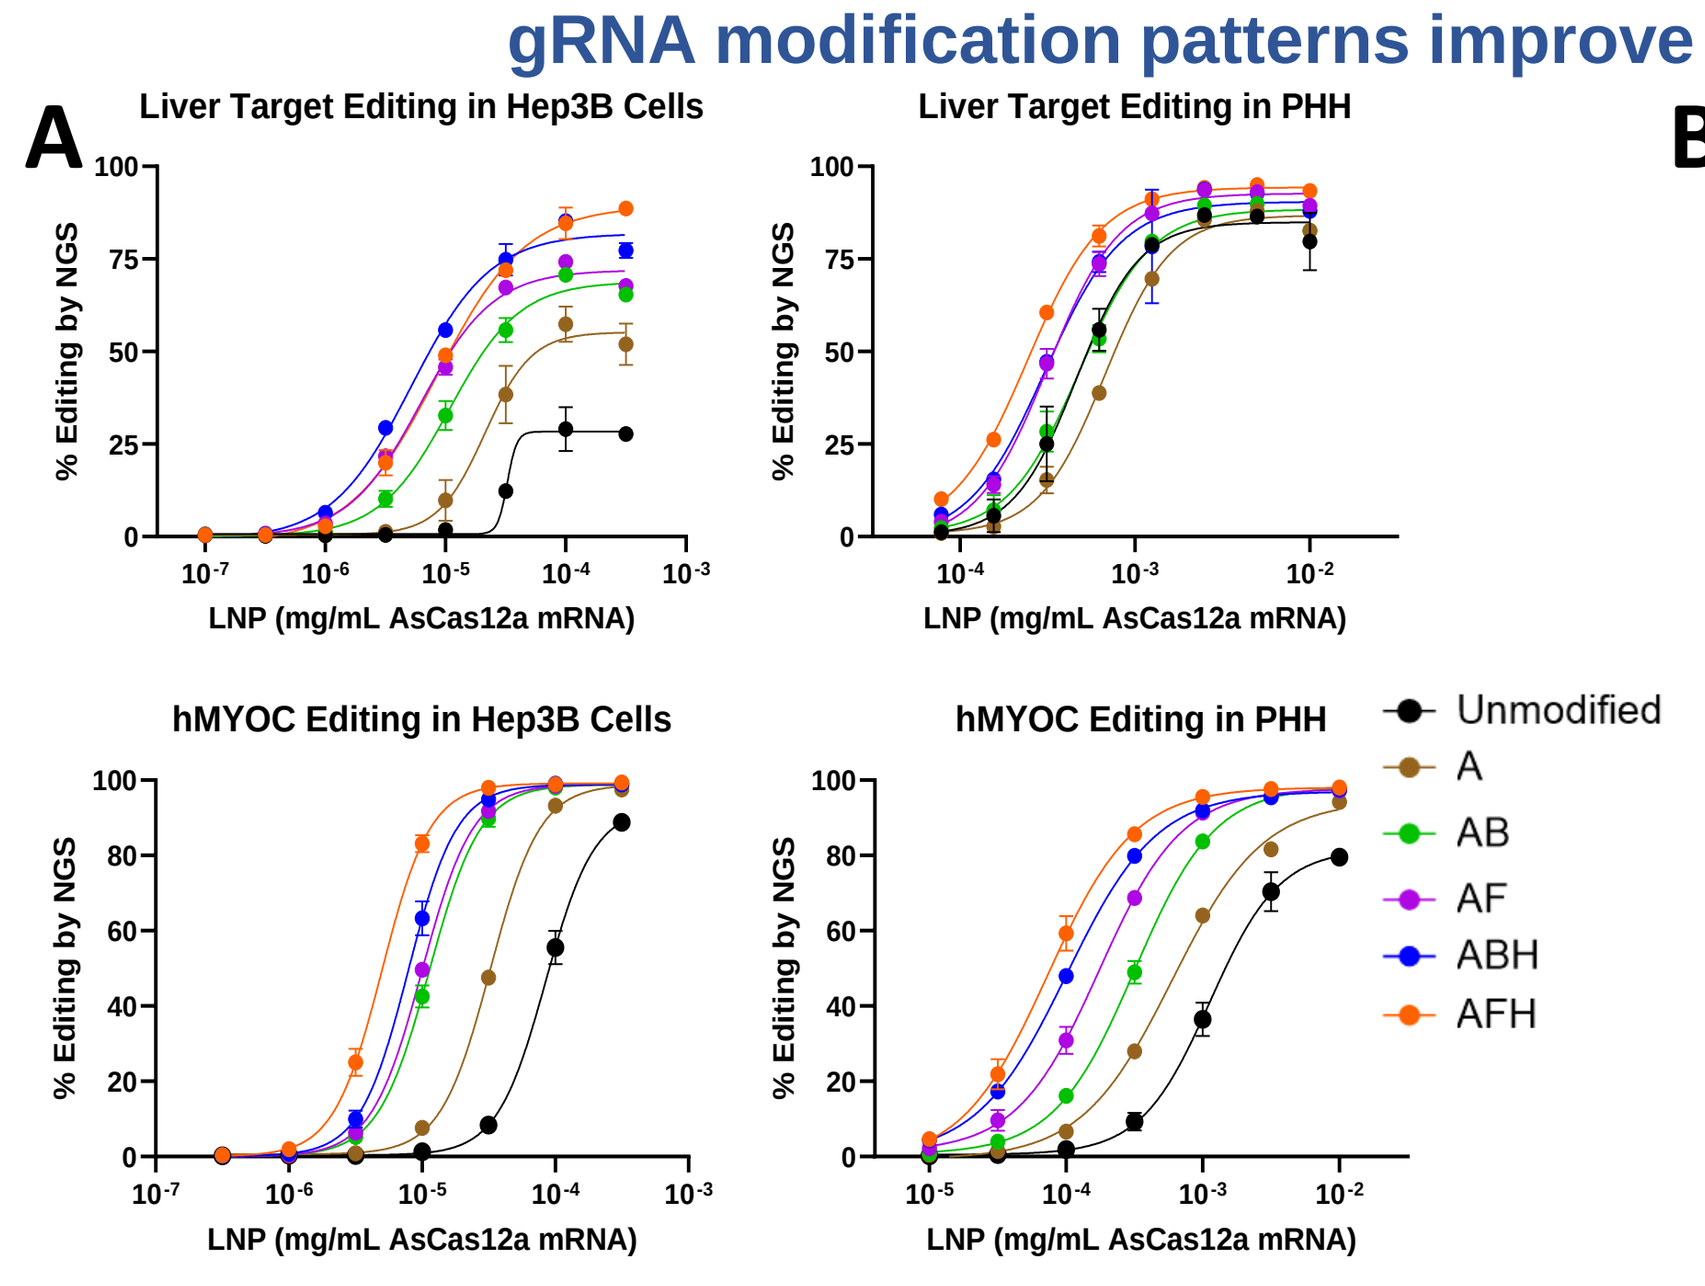
A
Liver Target Editing in Hep3B Cells
Liver Target Editing in PHH
hMYOC Editing in Hep3B Cells
hMYOC Editing in PHH
% Editing by NGS
LNP (mg/mL AsCas12a mRNA)
Unmodified
A
AB
AF
ABH
AFH

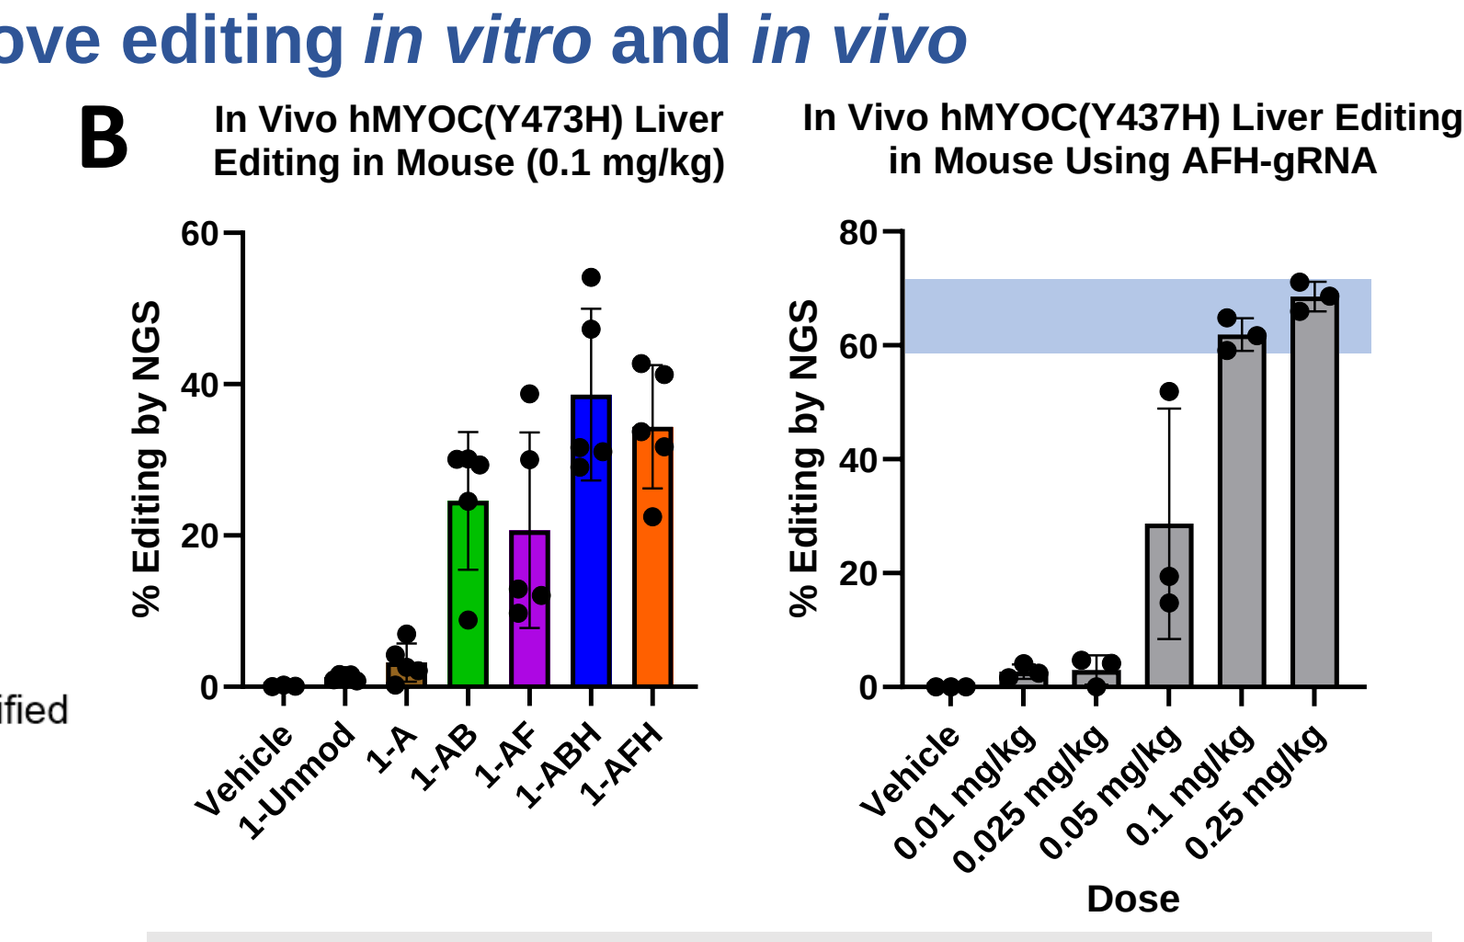
B
In Vivo hMYOC(Y473H) Liver Editing in Mouse (0.1 mg/kg)
In Vivo hMYOC(Y437H) Liver Editing in Mouse Using AFH-gRNA
% Editing by NGS
Vehicle
1-Unmod
1-A
1-AB
1-AF
1-ABH
1-AFH
0.01 mg/kg
0.025 mg/kg
0.05 mg/kg
0.1 mg/kg
0.25 mg/kg
Dose

- Distinct modification motifs can have additive effects on potency.
- Potency improvements can be sequence dependent.

To investigate the effects of incorporating chemical modification motifs into AsCas12a gRNAs, a series of guides that target the same genomic sequence were synthesized with various chemical modifications (anonymized with letters “A” through “H”). **(A)** gRNAs were formulated into LNPs along with mRNA encoding engineered AsCas12a and transfected into various cell types, and editing was evaluated 3 days post-transfection. **(B)** To explore *in vivo* editing, LNPs were delivered into hMYOC(Y437H) mice. Blue bar represents the hepatocyte content of mouse liver. MYOC, myocilin; PHH, primary human hepatocytes.

Results: Initial gRNA modification patterns (2/2)

gRNA modification patterns improve editing regardless of cell type *in vitro*

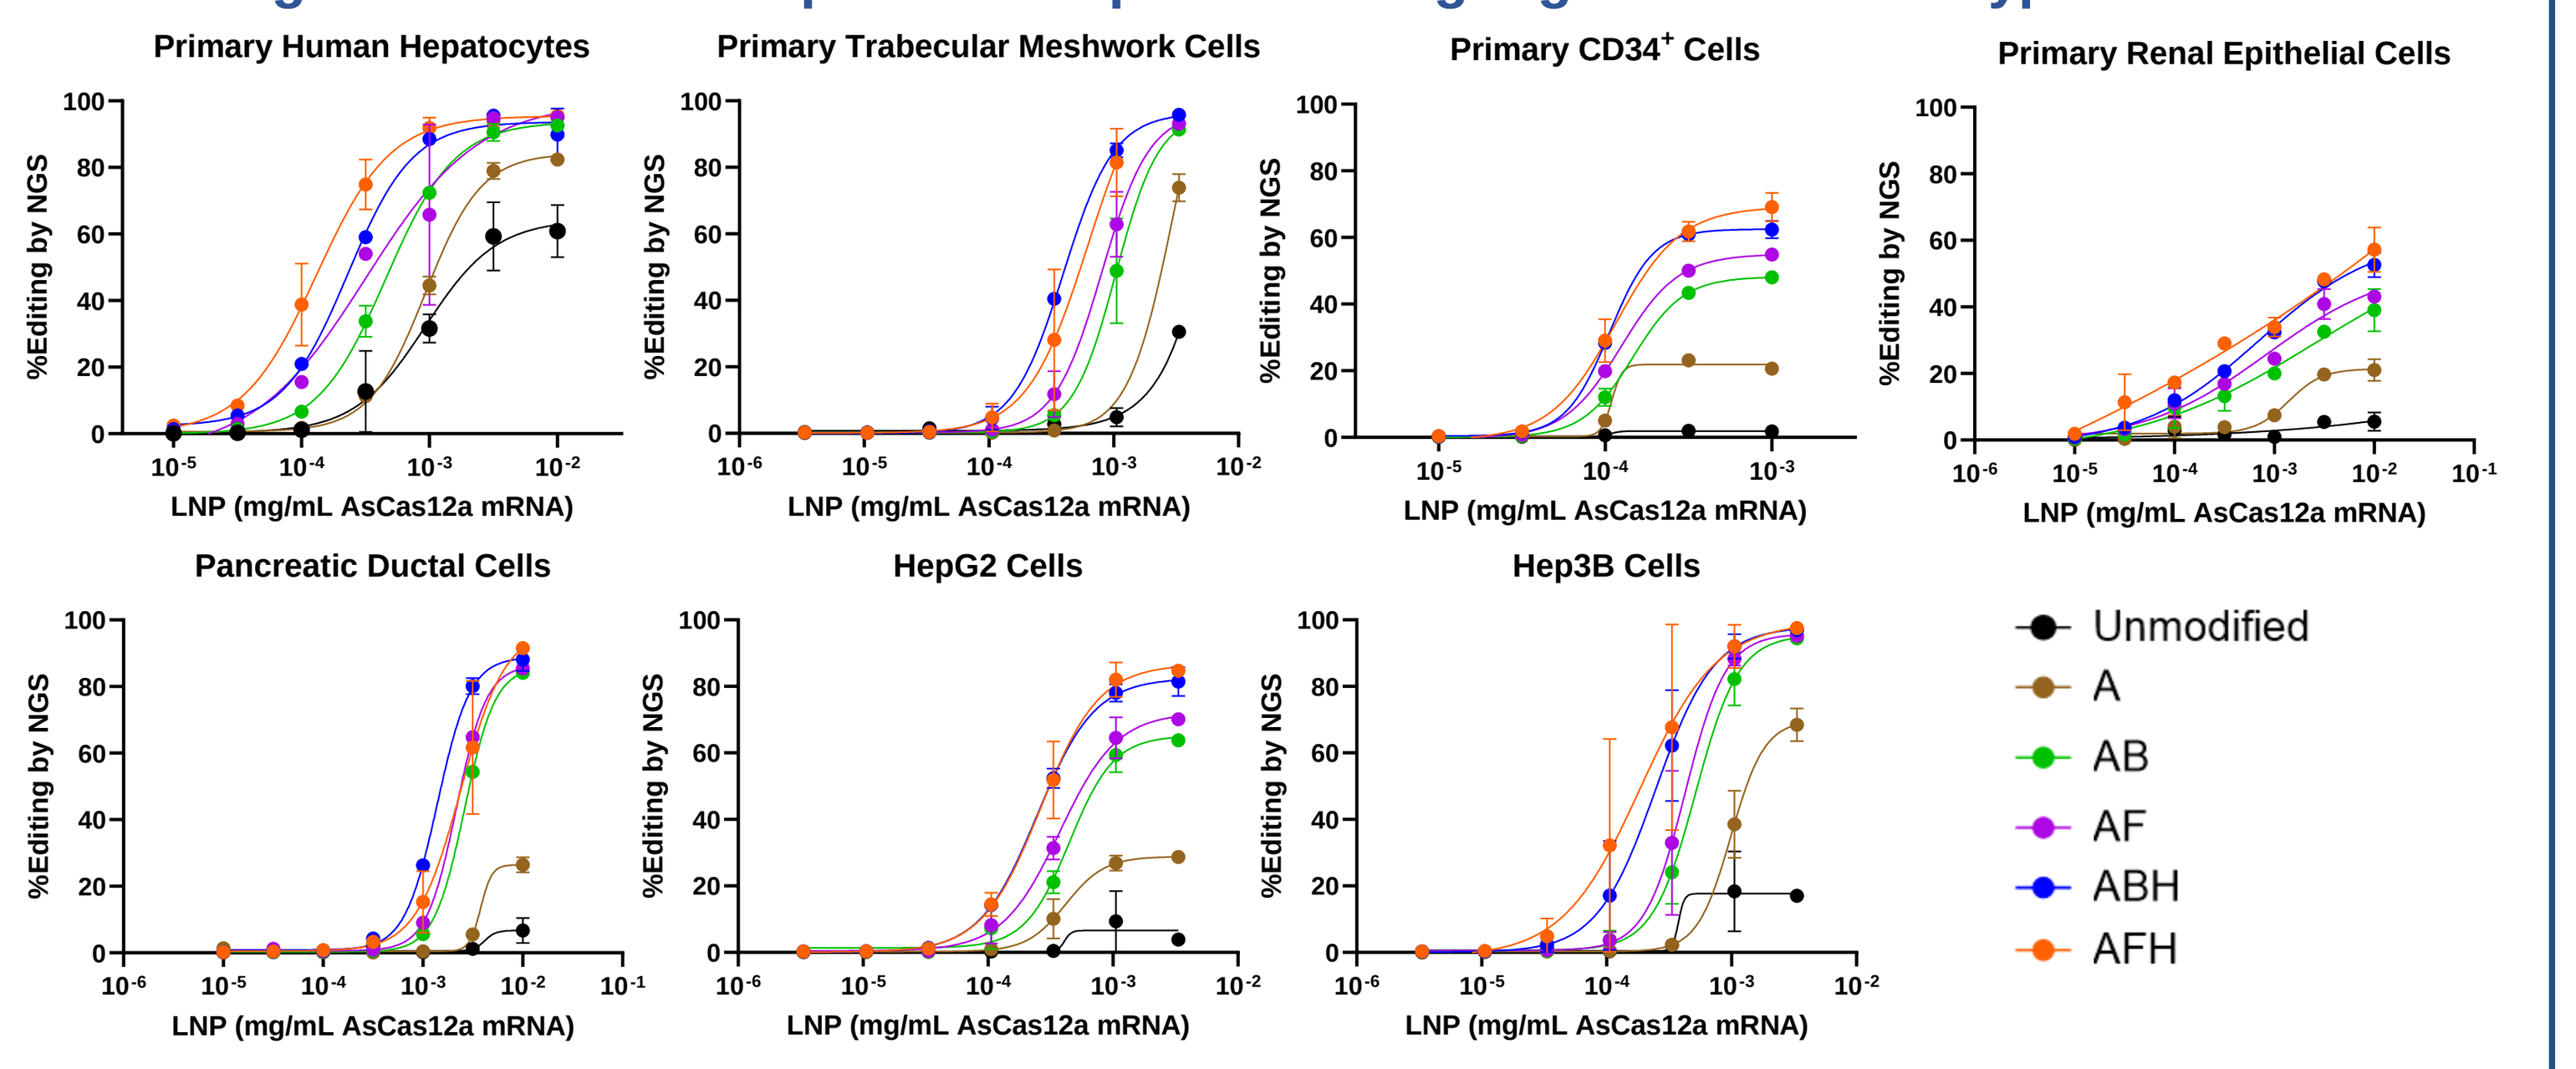
Primary Human Hepatocytes
Primary Trabecular Meshwork Cells
Primary CD34+ Cells
Primary Renal Epithelial Cells
Pancreatic Ductal Cells
HepG2 Cells
Hep3B Cells
%Editing by NGS
LNP (mg/mL AsCas12a mRNA)
Unmodified
A
AB
AF
ABH
AFH

- Rank ordering of guide potency shows consistency across diverse cell types.

To explore whether modification patterns increase editing potency regardless of the cell type, a variety of primary cell types and cell lines were treated *in vitro* with LNPs containing AsCas12a mRNA and a range of modified gRNAs targeting the same hMYOC site.

Results: Structure-function relations

Minimal model for understanding gRNA modification effects inside the cell

Editing ≈ Stability x Binding Affinity x DNA Cleavage Activity

- Strong correlation of modification pattern rank order suggests similar pressures on gRNA in diverse cell types.
- Modifications may alter gRNA behavior through entire intracellular pathway to editing.
- Chemical space of modifications is exponentially large:
 - Cas9 single guide RNA (sgRNA), RNA + 3 mods/position: ~10^{47}
 - Cas12a gRNA, RNA + 3 mods: ~10^{21}
- Assays for specific mechanistic steps can help guide gRNA engineering and narrow chemical space of guides to test.

Results: gRNA-protein binding affinity (1/2)

Relative binding affinity correlates with editing potency in proof-of-concept target and liver target

A

1) Add the test modified gRNA (modRNA) in increasing concentrations and the labeled gRNA, incubate with Cas12a.

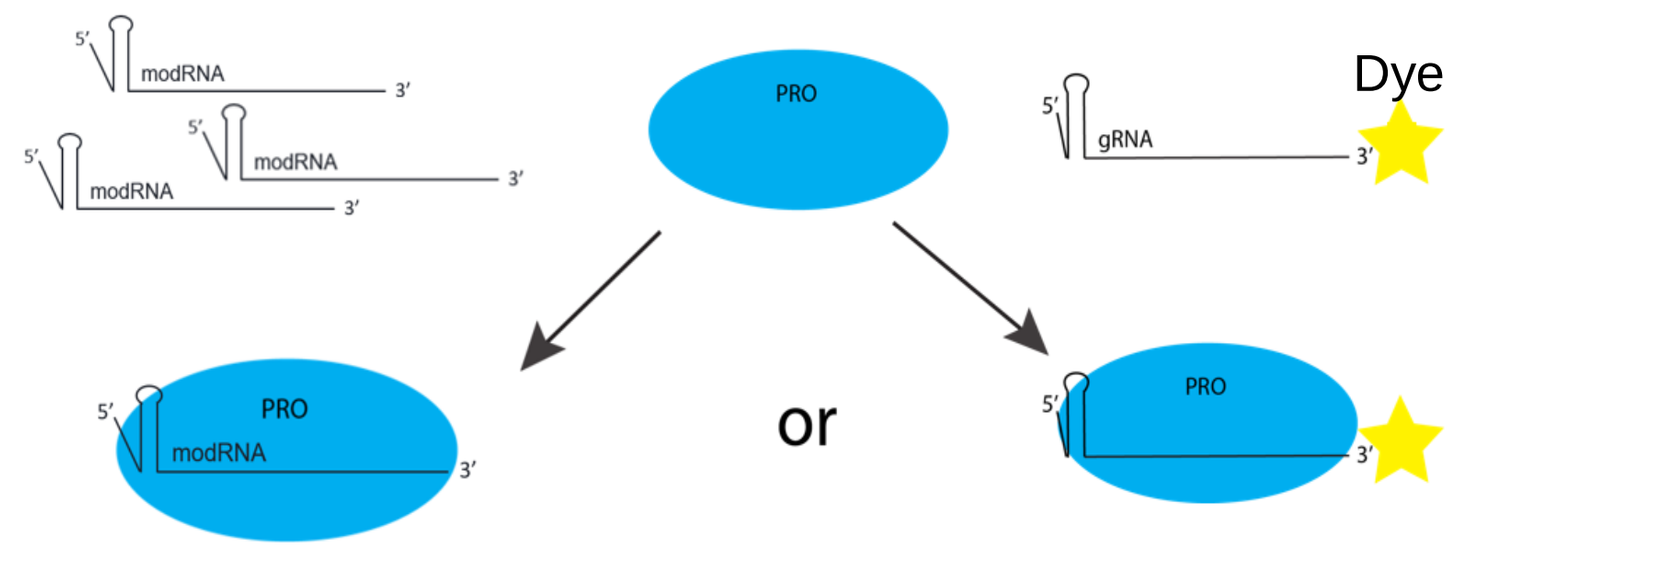
modRNA
PRO
gRNA
Dye
or

2) Separate bound RNP from free RNA.

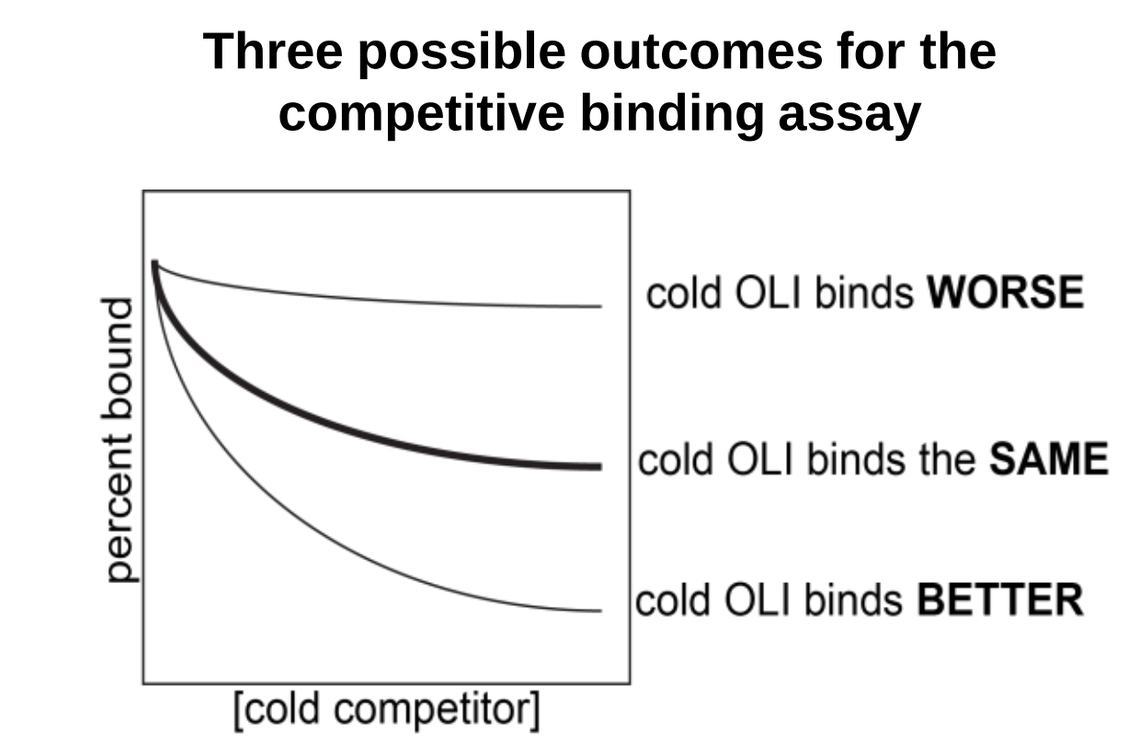
Three possible outcomes for the competitive binding assay
percent bound
[cold competitor]
cold OLI binds WORSE
cold OLI binds the SAME
cold OLI binds BETTER

Bold line indicates binding curve of unlabeled, unmodified control oligonucleotides (OLIs) (iso-affinity).

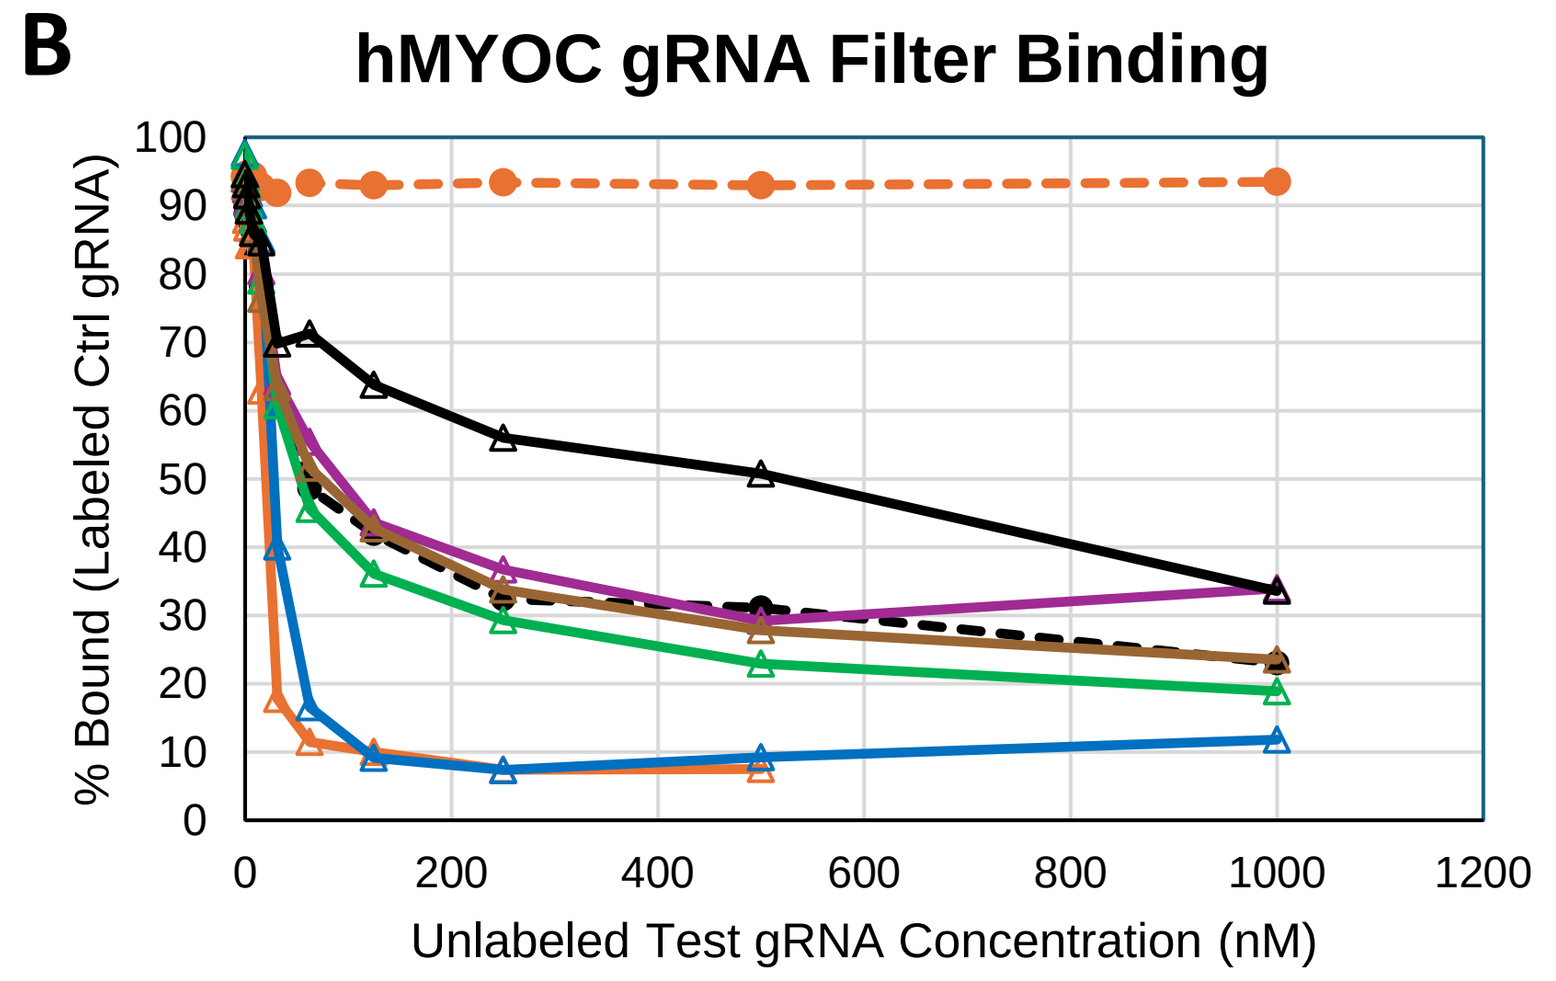
B
hMYOC gRNA Filter Binding
% Bound (Labeled Ctrl gRNA)
Unlabeled Test gRNA Concentration (nM)

Symbol	Mod Pattern	Binding	EC_{50} in PHH
△	Unmod	Poor	1.10E-03
△	A	Good	6.00E-04
△	AB	Good	3.20E-04
△	AF	Good	1.80E-04
△	ABH	Excellent	1.00E-04
△	AFH	Excellent	7.30E-05

Negative Ctrl Positive Ctrl

Results: gRNA-protein binding affinity (2/2)

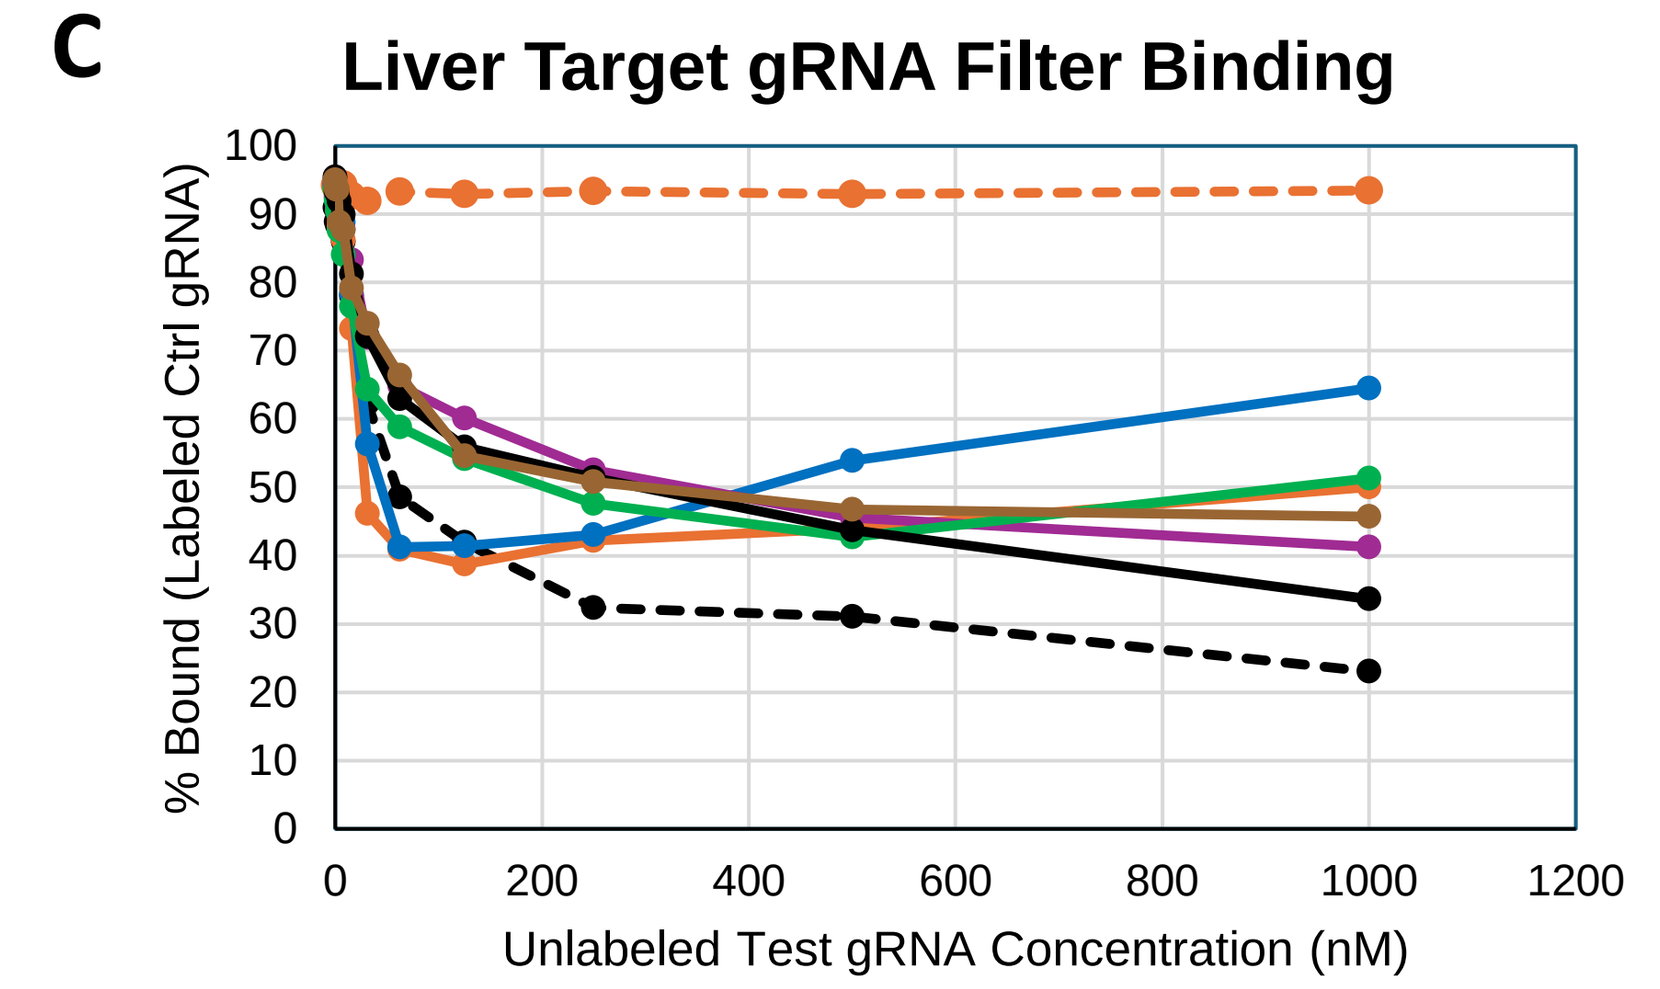
C
Liver Target gRNA Filter Binding
% Bound (Labeled Ctrl gRNA)
Unlabeled Test gRNA Concentration (nM)

Symbol	Mod Pattern	Binding	EC_{50} in PHH
●	Unmod	Poor	4.60E-04
●	A	Poor	6.80E-04
●	AB	Poor	4.80E-04
●	AF	Poor	3.20E-04
●	ABH	Good	3.10E-04
●	AFH	Good	2.40E-04

Negative Ctrl Positive Ctrl

- gRNA behavior is sequence-dependent even with the same modification patterns.
- Increased potency correlates with increased Cas12a binding affinity.

An *in vitro* binding affinity assay **(A)** was used to evaluate whether gRNA modifications impact binding to the AsCas12a nuclease and correlate with the effect on editing potency. The percentage of bound fluorescent control guide is read out through a filter binding assay to generate a binding isotherm. The binding affinity of the modified guides was categorized as “poor”, “good”, or “excellent” based on comparison of their binding isotherm to that of an unmodified gRNA positive control. Modified gRNAs targeting hMYOC **(B)** or a proof-of-concept liver target **(C)** were evaluated using this filter binding assay.

Results: AsCas12a mechanism

Accelerated R-loop formation is a primary mechanistic determinant of increased potency in engineered AsCas12a

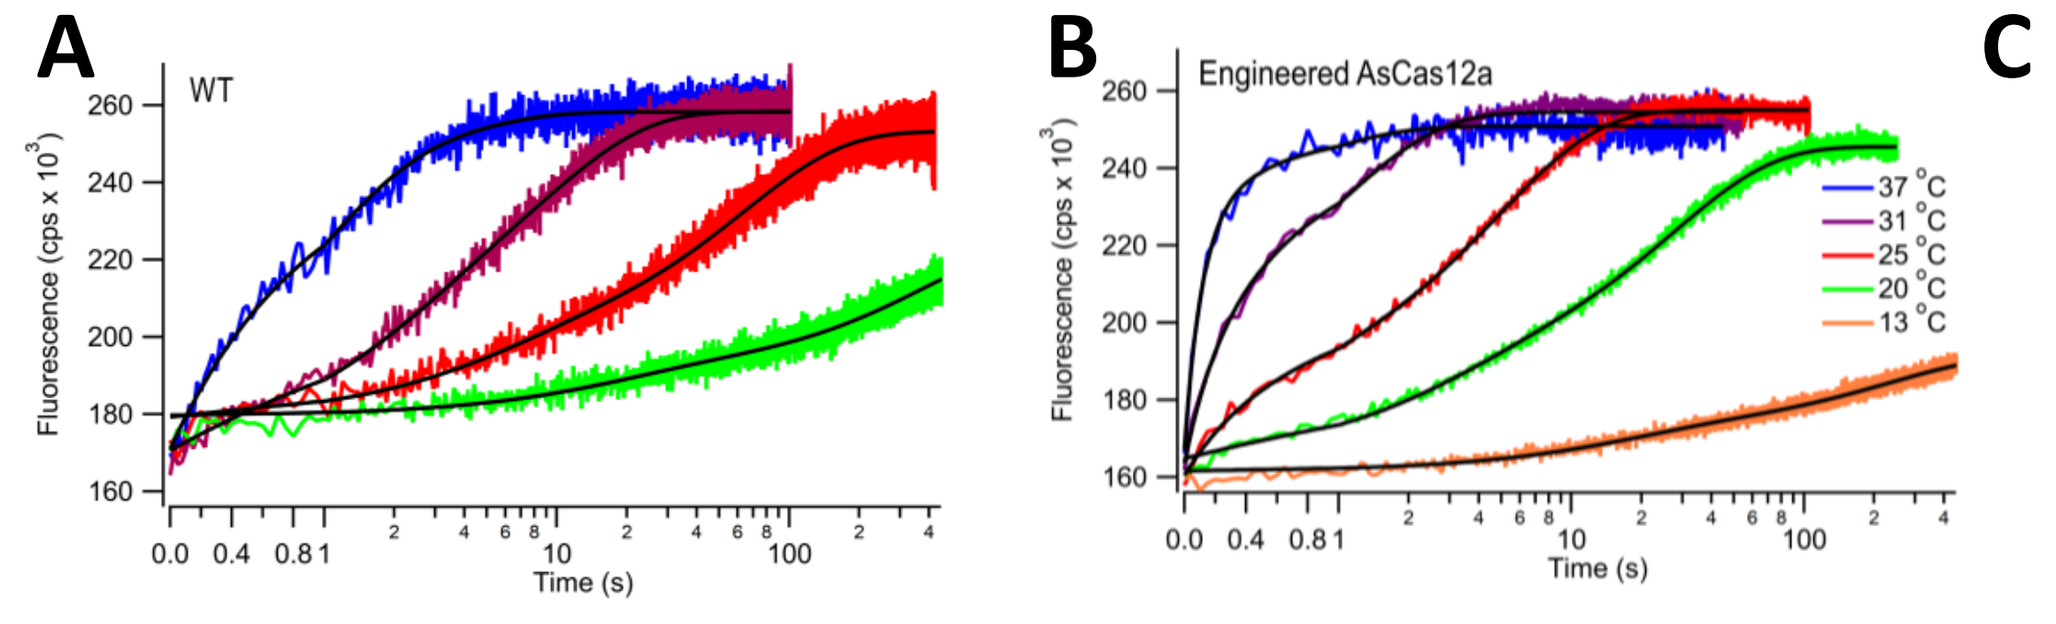
A
WT
B
Engineered AsCas12a
Fluorescence (cps x 10^3)
Time (s)
37 °C
31 °C
25 °C
20 °C
13 °C

C

T (°C)	$\tau_{R\text{-loop}}$ WT (s)	$\tau_{R\text{-loop}}$ Engineered (s)
37	1.35 ± 0.37	0.28 ± 0.13
31	6.81 ± 0.19	0.77 ± 0.06
25	49.79 ± 0.60	3.90 ± 0.06
20	309.22 ± 5.04	21.12 ± 0.19
13	nd	161.01 ± 3.54

- R-loop formation kinetics are accelerated ~4x at 37°C in engineered AsCas12a, highlighting the importance of ensuring modification patterns do not interfere with enzyme dynamics.

Time-resolved fluorescence kinetics of R-loop formation in wild-type (WT) **(A)** and engineered AsCas12a[1,2] **(B)** monitored by stopped-flow fluorescence of a tC° labelled DNA template (tC° at Position 9 of non-target strand). Excitation: 360 nm and Emission: 450 nm. Data were fit to the sum of two exponentials (black traces) and $\tau_{R\text{-loop}}$, calculated as the amplitude weighted lifetime: $\tau_{obs} = \sum_i \frac{a_i \tau_i}{a_i}$, is reported in **(C)**. nd, not determined.

Results: DNA cleavage rate and additional modifications

Modifications impact DNA cleavage rate

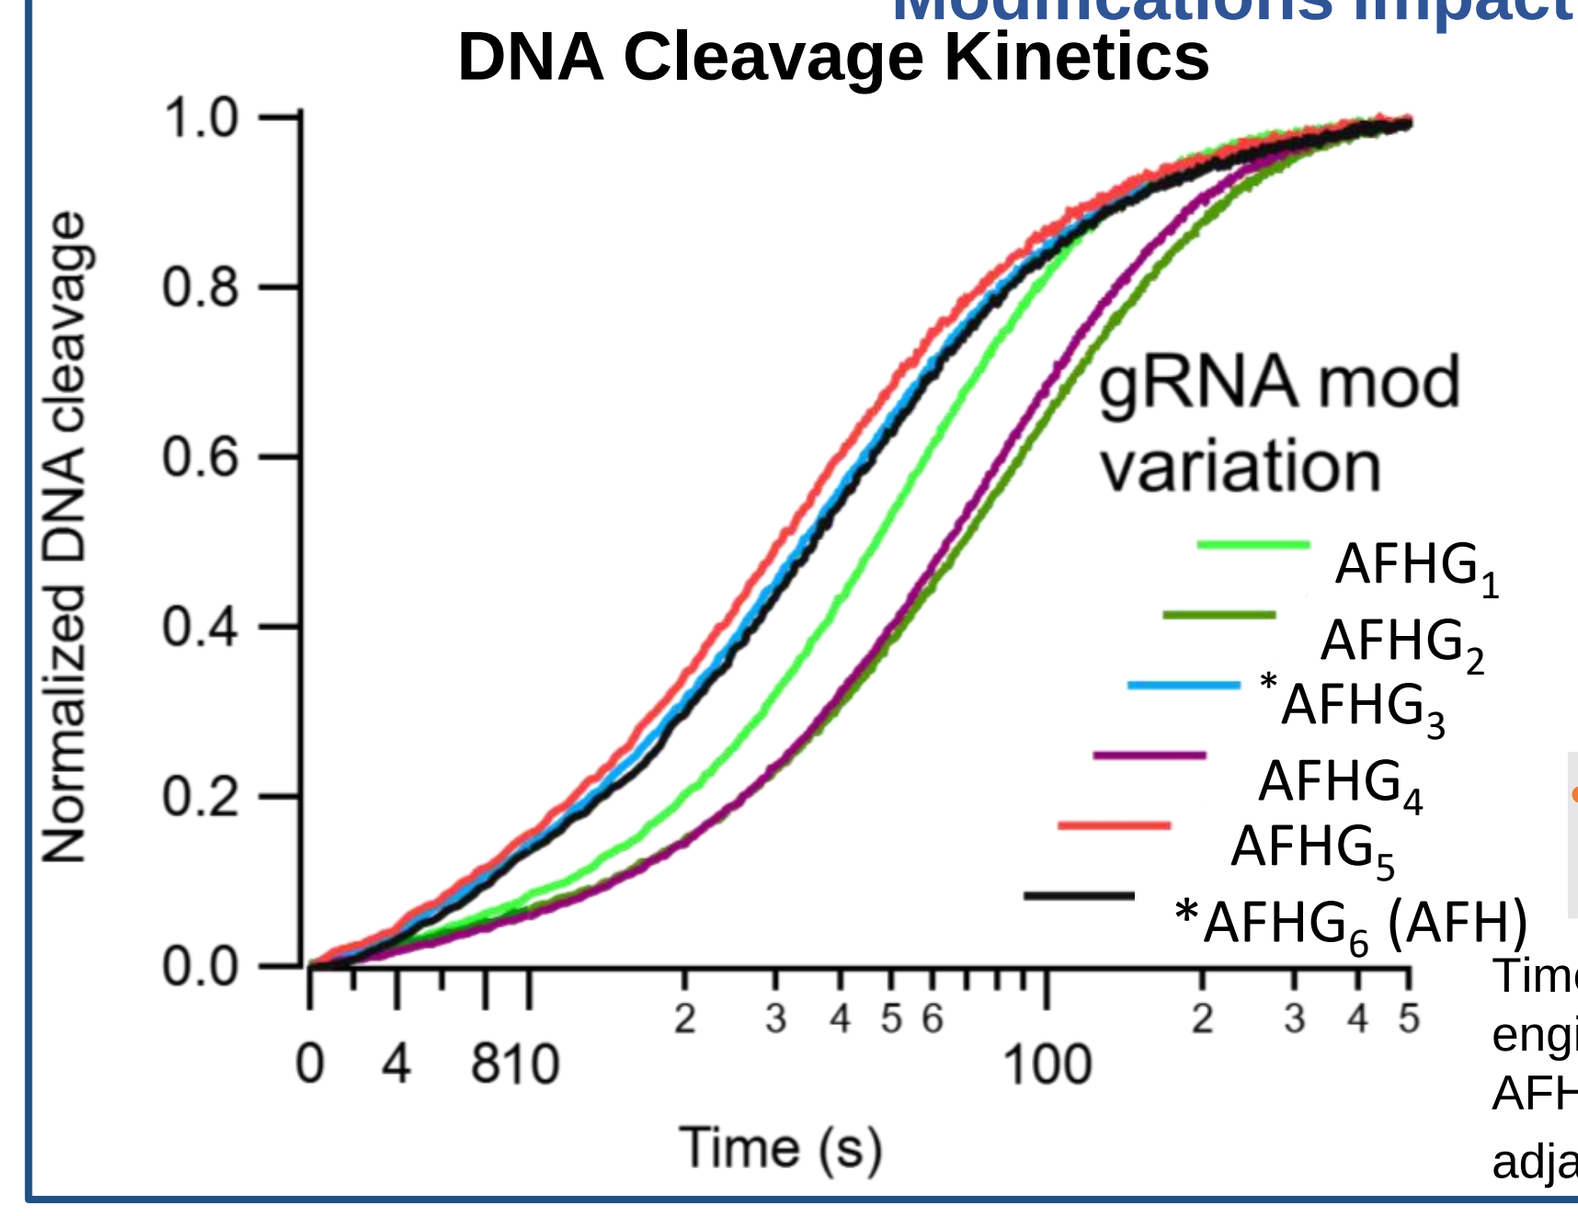
DNA Cleavage Kinetics
Normalized DNA cleavage
Time (s)
gRNA mod variation
AFHG1
AFHG2
*AFHG3
AFHG4
AFHG5
*AFHG6 (AFH)

- **G_n** represent 2 added modification strategies, 1) G relies on an mRNA variation, 2) subscript n denotes variations on a second strategy.

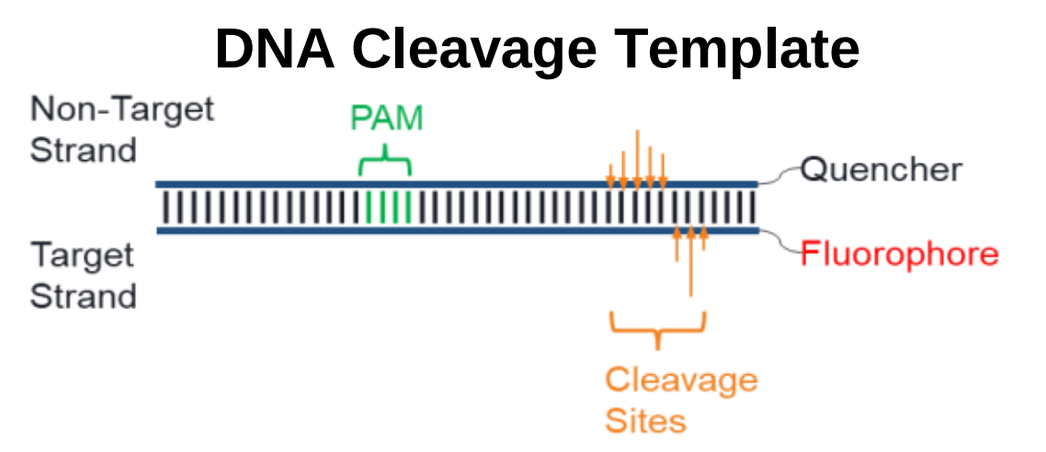
DNA Cleavage Template
Non-Target Strand
PAM
Quencher
Target Strand
Fluorophore
Cleavage Sites

- Modification patterns that slow DNA cleavage mechanism prevent enhanced editing potency.

Time-resolved fluorescence kinetics of DNA template cleavage in engineered AsCas12a. Excitation: 585 nm and Emission: 603 nm. AFH is equivalent to $AFHG_5$ in this experimental design. PAM, protospacer adjacent motif. *Enhanced editing potency over base AFH pattern.

Conclusions

- Editas has developed and refined modification patterns for Cas12a gRNA that significantly enhance *in vitro* and *in vivo* editing potency in LNP mediated gene editing.
 - gRNA chemical modifications enhance editing potency by increasing Cas12a binding affinity, preserving intrinsic DNA cleavage kinetics and slowing degradation kinetics in cytoplasm.
 - Rational design to improve gRNA potency can be successfully guided by biophysical characterization of the functional impact of chemical modifications.
 - Efforts targeting further improvements to gRNA potency are ongoing.

Results: gRNA stability and additional modifications

Chemical modification patterns impact gRNA stability in human cytoplasm model

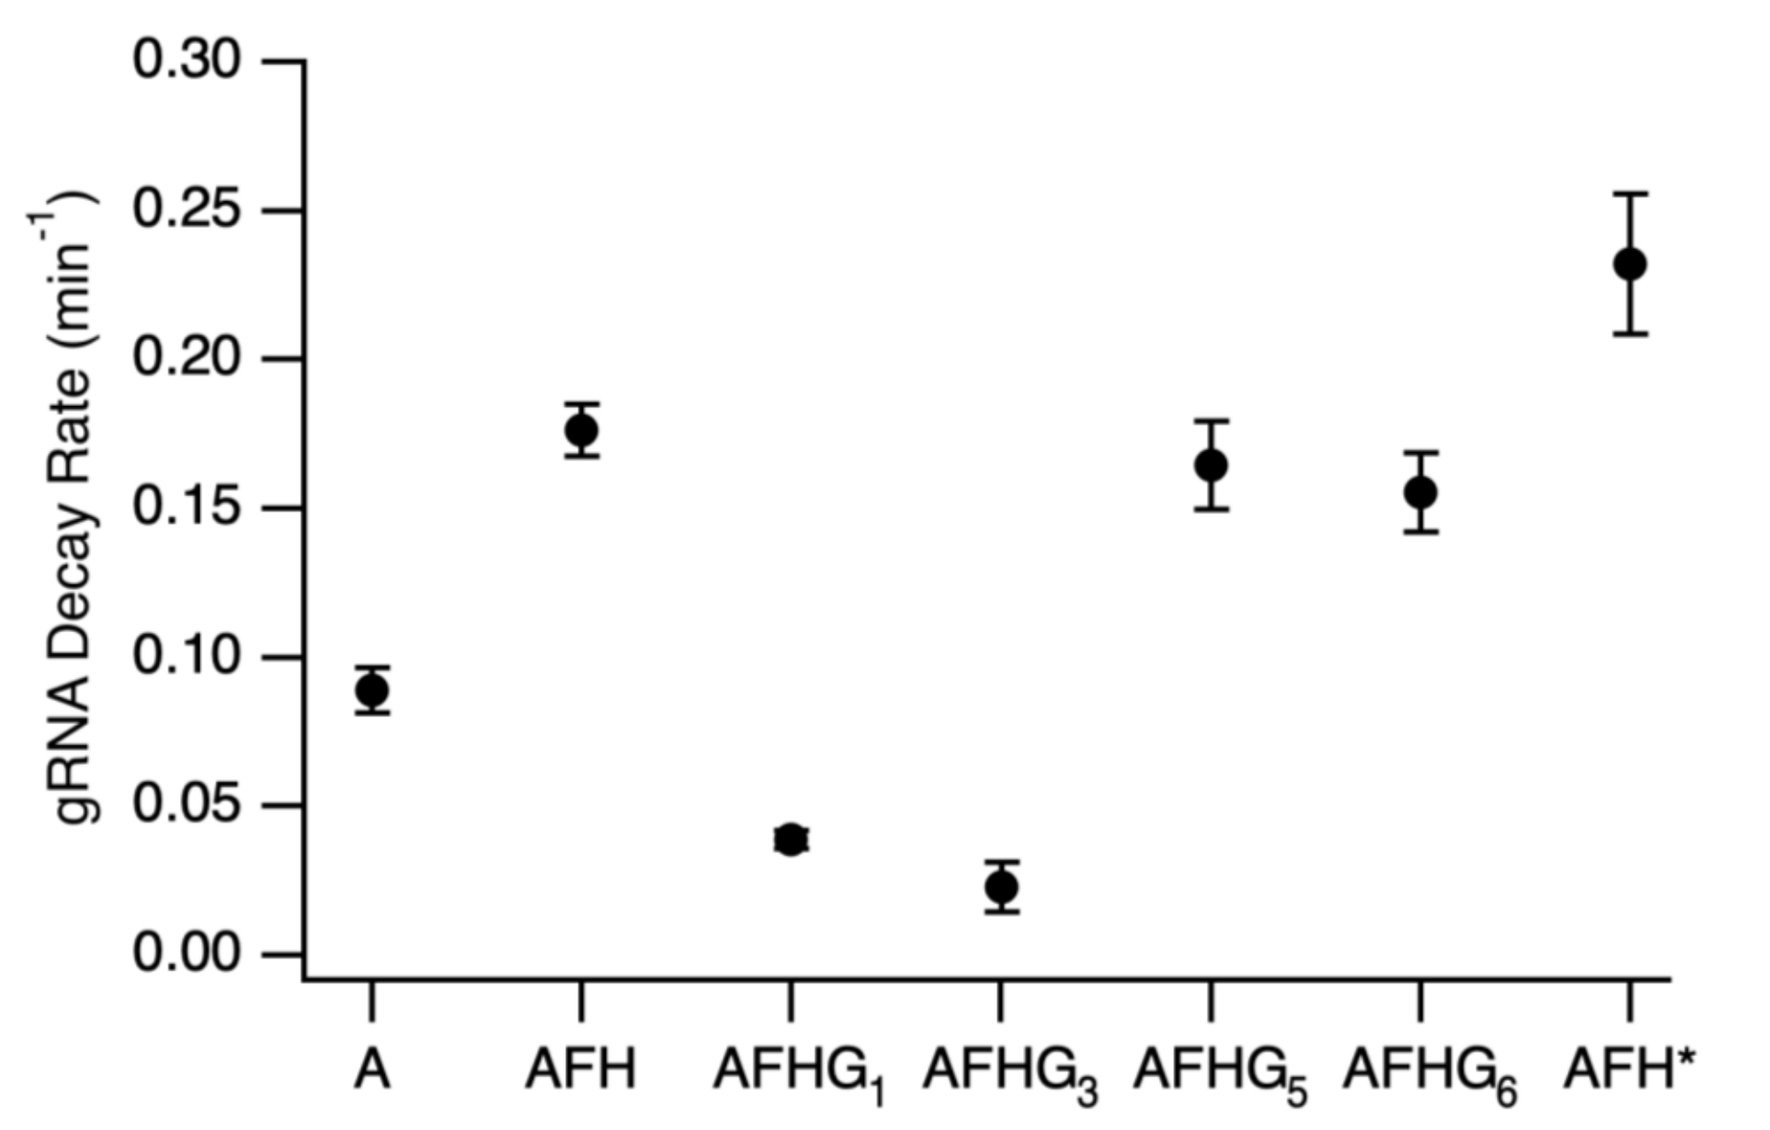
gRNA Decay Rate (min^-1)
A
AFH
AFHG1
AFHG3
AFHG5
AFHG6
AFH*

- Modification patterns have strong impact on gRNA stability but interplay with other factors determines potency outcomes.

gRNA incubated in PBS buffer with 2% human cytoplasm. Degradation is quenched at 0-, 3-, 5-, and 10-minute time points by the addition of excess engineered AsCas12a. Intact gRNA is read out by addition of excess DNA cleavage template and quantified by fluorescence. AFH* refers to a modified version of the H (H*) motif in the context of the AFH pattern.

Results: Translation of added modifications to editing potency

Translation of refined modifications to *in vitro* and *in vivo* editing

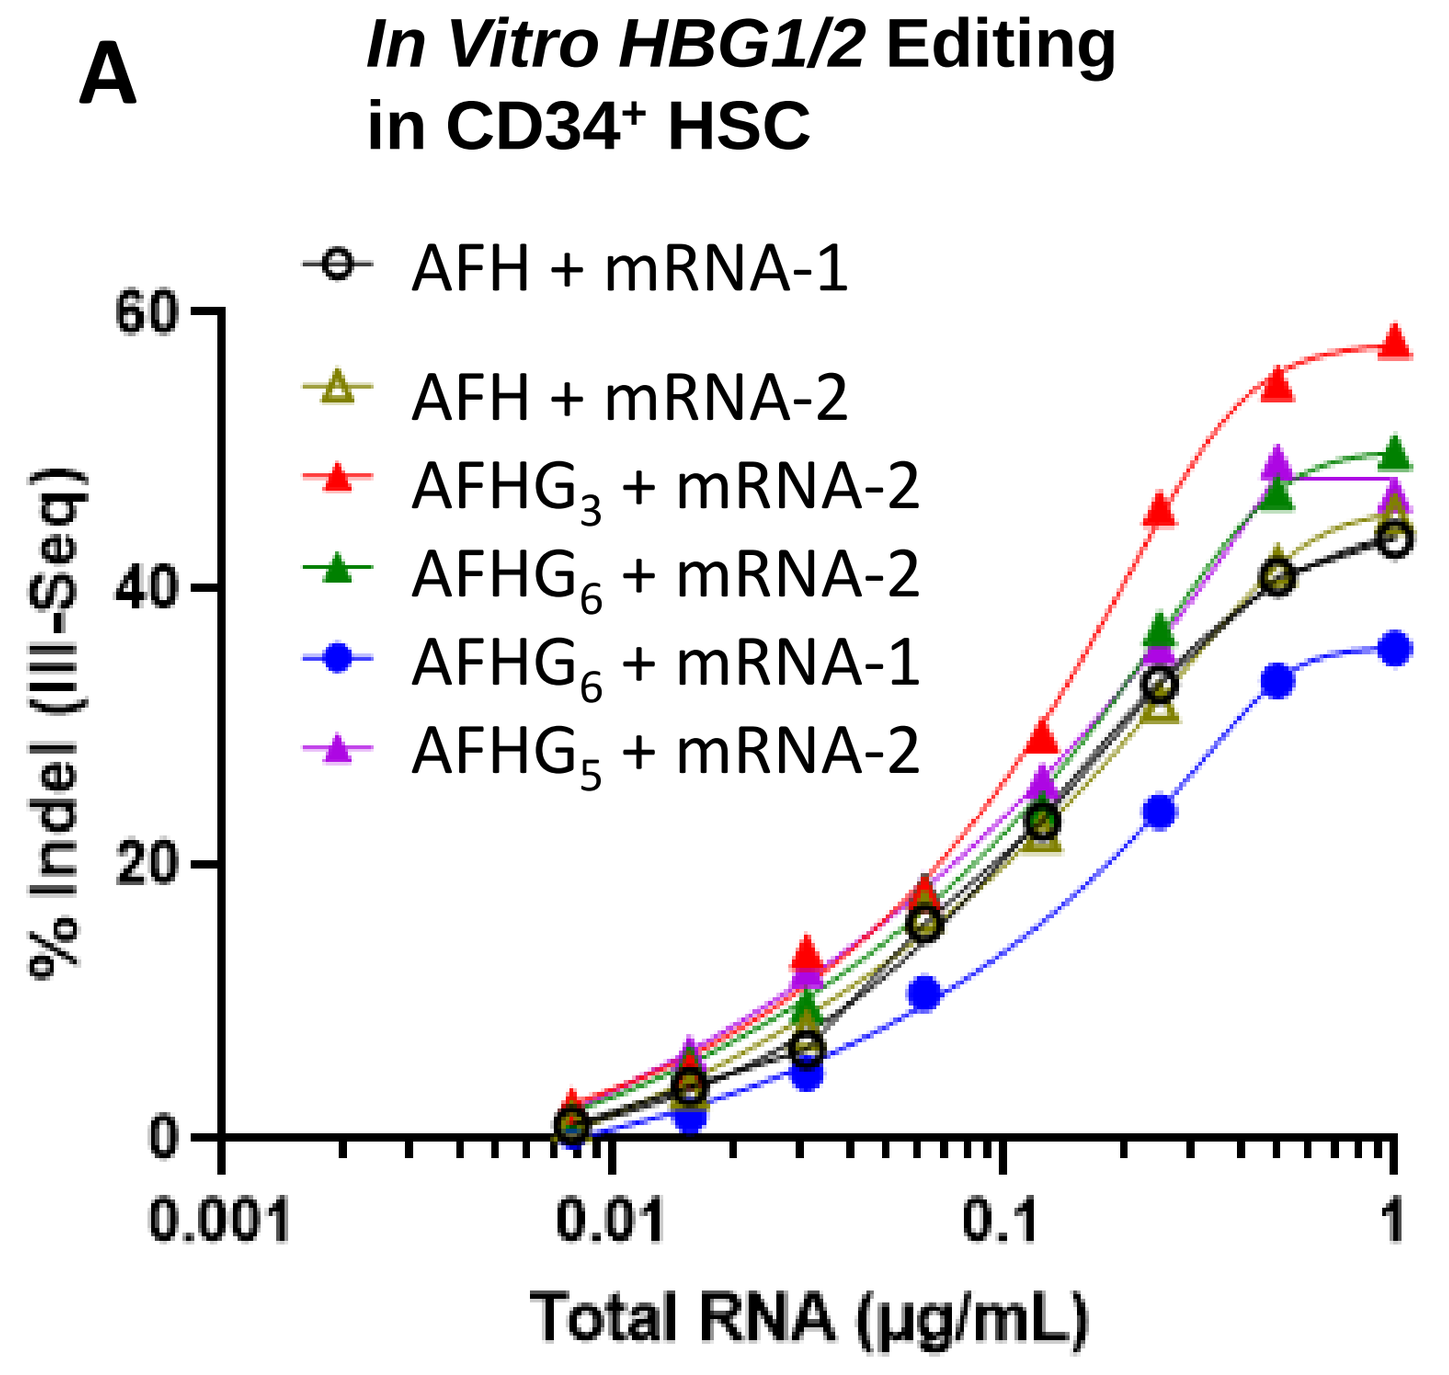
A
In Vitro HBG1/2 Editing in CD34+ HSC
AFH + mRNA-1
AFH + mRNA-2
AFHG3 + mRNA-2
AFHG6 + mRNA-2
AFHG6 + mRNA-1
AFHG5 + mRNA-2
% Indel (III-Seq)
Total RNA (μg/mL)

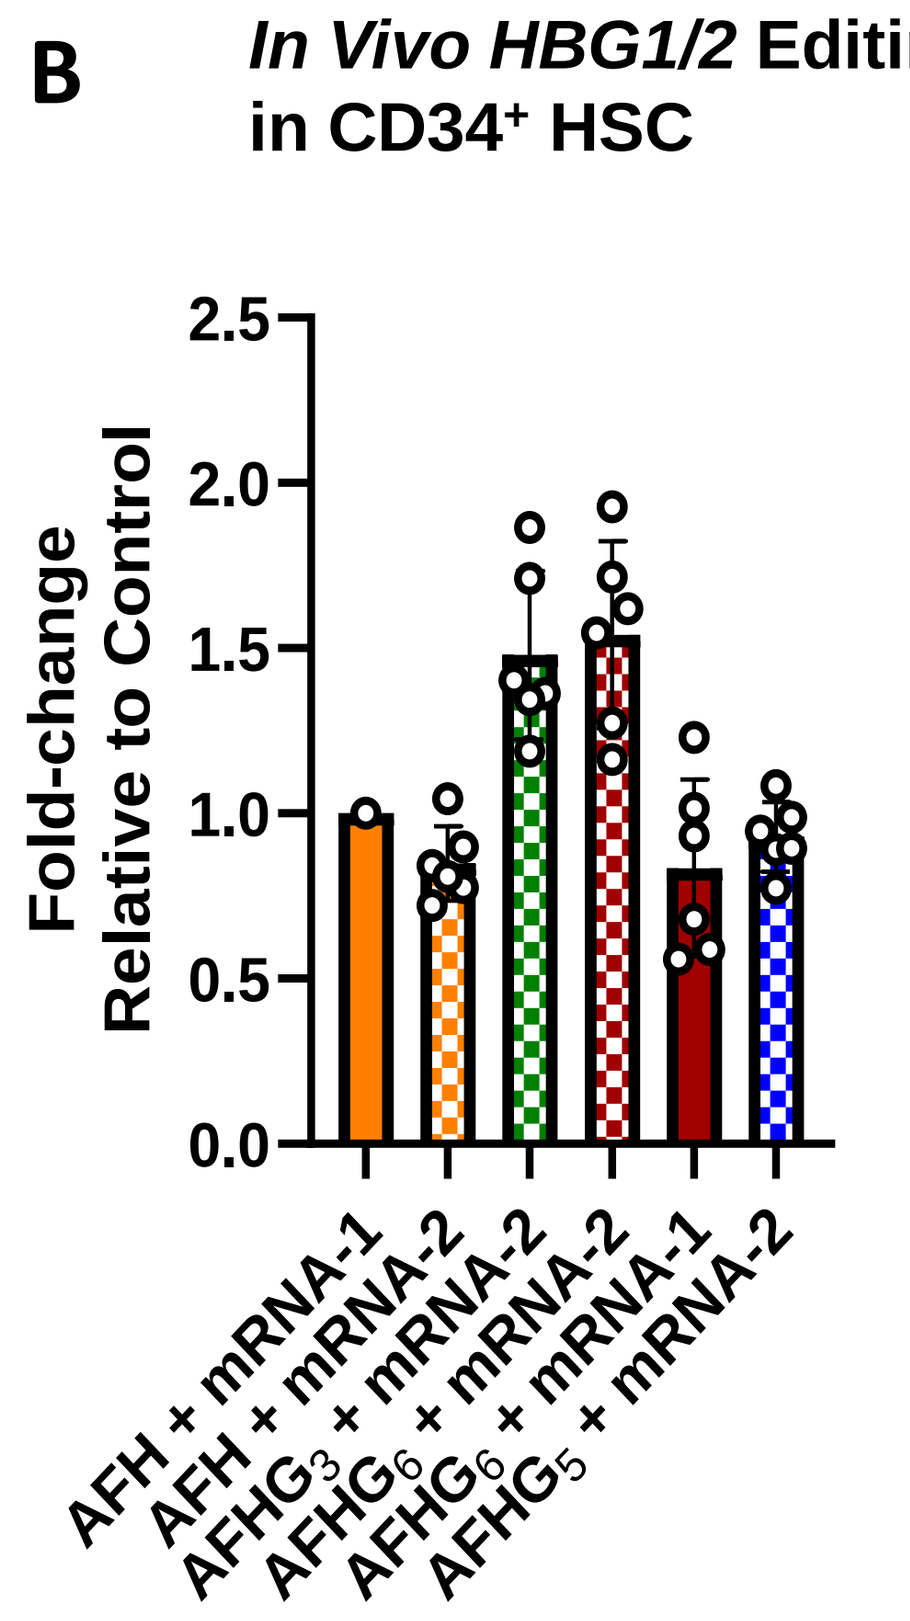
B
In Vivo HBG1/2 Editing in CD34+ HSC
Fold-change Relative to Control
AFH + mRNA-1
AFH + mRNA-2
AFHG3 + mRNA-2
AFHG6 + mRNA-2
AFHG6 + mRNA-1
AFHG5 + mRNA-2

- Further refinement of chemical modification pattern leads to enhanced *in vitro* and *in vivo* editing potency.

(A) *In vitro* testing of selected *HBG1/2* gRNA variants compared with initial AFH design. Cells were treated with targeted LNPs (tLNPs) at specified total RNA doses. LNPs were cultured for 5 days and editing at *HBG1/2* was assessed via Ill-Seq. Two variant mRNAs were examined where mRNA-2 is tailored to specifically enable an undisclosed mechanism with the AFH-x variants. **(B)** *In vivo* testing of selected gRNA variants in NSBGW HSPC mice comparing editing level relative to AFH-mRNA-1 control. HSC, hematopoietic stem cell.

References

1. Zhang L *et al*. *Nat Comms* 2021; 12: 3908. 2. Jansson *et al*. *Commun Biol* 2024; in revision.

Acknowledgements and disclosures

We would like to thank our Editas Medicine colleagues and collaborators who provided support in sequencing, RNA and LNP manufacturing, animal studies, and scientific discourse. All authors are current or former employees and shareholders of Editas Medicine, Inc. The authors have filed a patent application on the data presented here. Medical writing and editorial assistance were provided by Porterhouse Medical US and were funded by Editas Medicine, Inc. according to Good Publication Practice (GPP) guidelines.

Presented at the 28th ASGCT Annual Meeting ● May 13–17, 2025 ● New Orleans, LA, USA

© 2025 Editas Medicine