In vivo CRISPR-based upregulation of a compensatory pathway for normalization of phenotypes of a disease-causing gene

Benjamin Diner, Meng Wu, Mansi Thakkar, Ameya Apte, Vikram Soman, Steve Bottega, Luis Agosto, Paul Wrighton, Stephen Sherman, James Bochicchio, Gulum Kosova, Linda Burkly, Jenny Xie

Editas Medicine, Inc., Cambridge, MA, United States

Poster #2324

Introduction: Editas' differentiated *in vivo* gene editing upregulation strategy aims to deliver best-in-class potentially curative medicines for genetic diseases

By editing non-coding gene regulatory regions, such as 5' and 3' untranslated regions (UTRs), we seek to induce upregulation of wild-type alleles, functional paralogs, or compensatory pathways to mitigate diseases caused by pathogenic mutations:

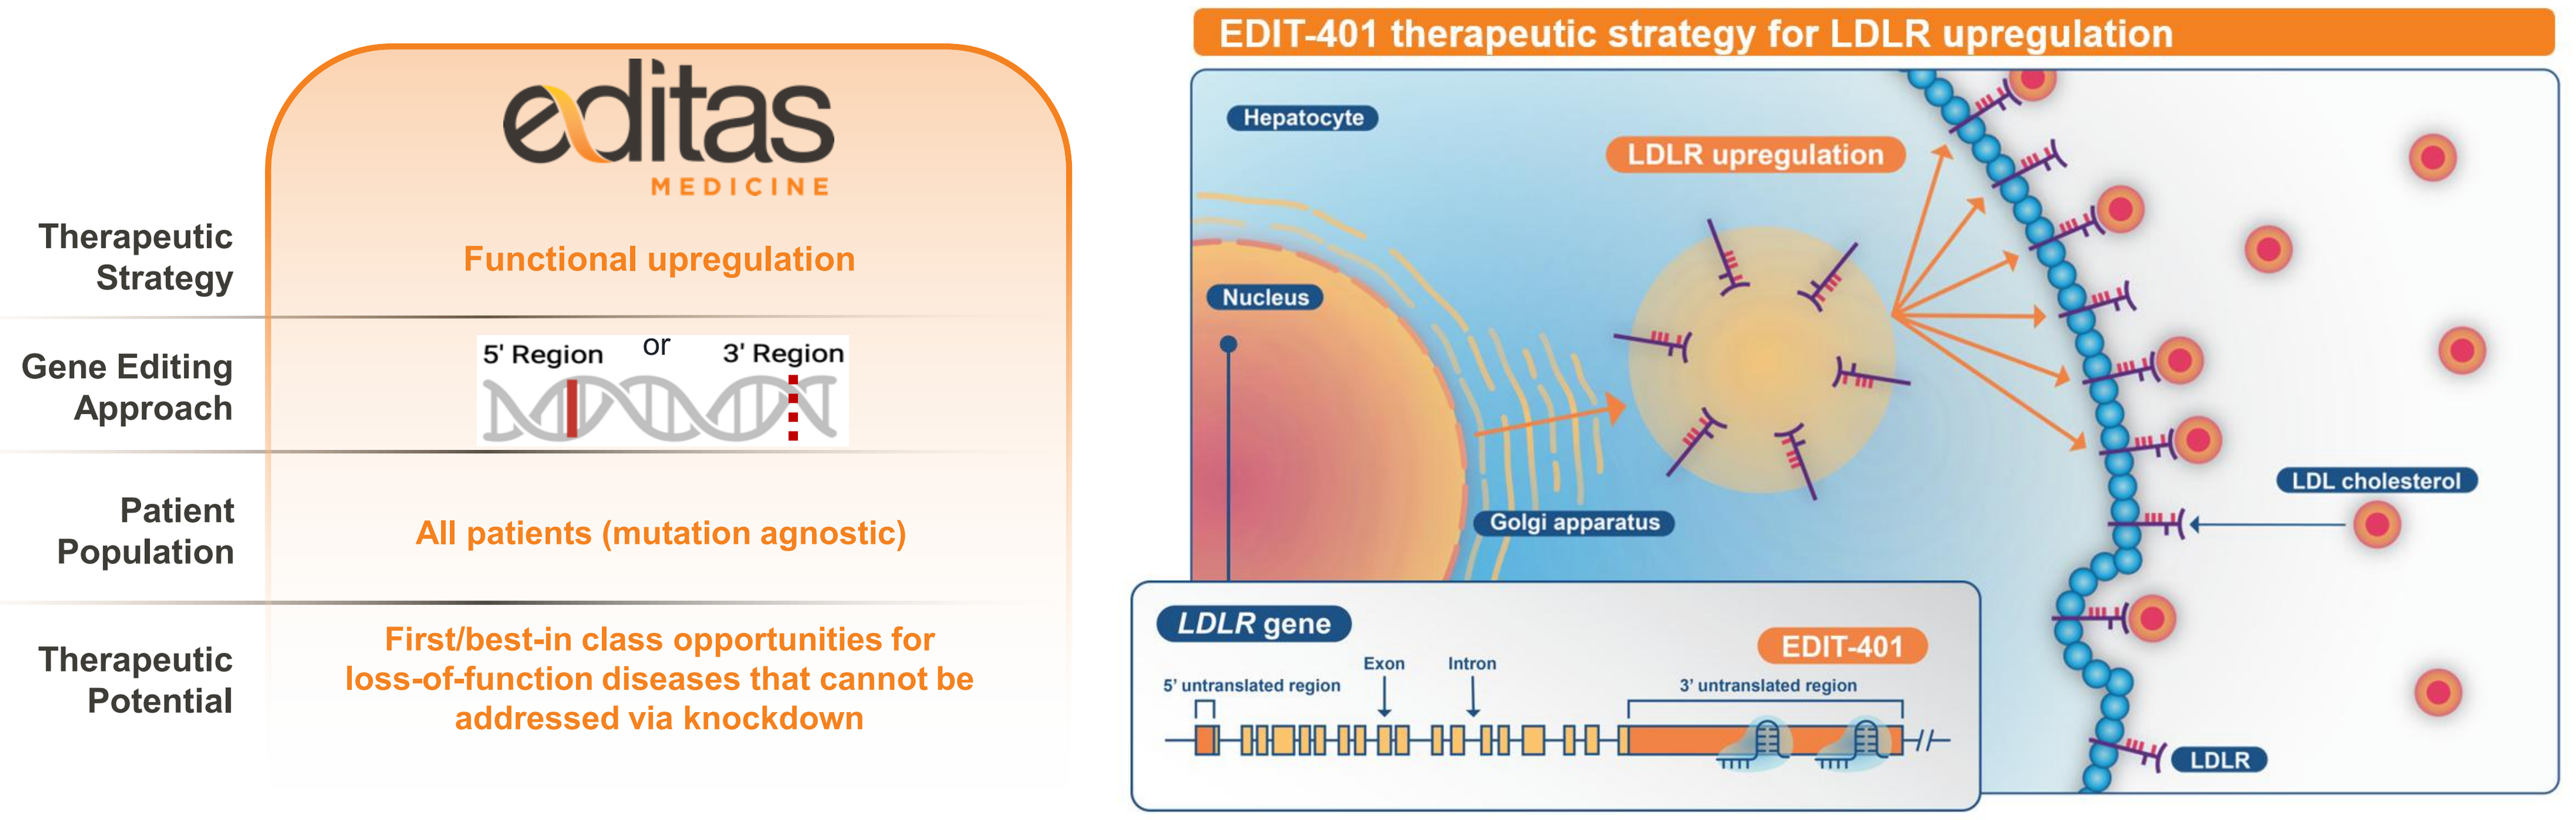

Therapeutic Strategy
Functional upregulation
Gene Editing Approach
5' Region or 3' Region
Patient Population
All patients (mutation agnostic)
Therapeutic Potential
First/best-in class opportunities for loss-of-function diseases that cannot be addressed via knockdown
EDIT-401 therapeutic strategy for LDLR upregulation
Hepatocyte
Nucleus
LDLR upregulation
Golgi apparatus
LDL cholesterol
LDLR gene
Exon
Intron
5' untranslated region
3' untranslated region
EDIT-401
LDLR

We harnessed the gene editing capabilities of Clustered Regularly Interspaced Short Palindromic Repeats (CRISPR) to develop EDIT-401 – an investigational therapeutic designed to upregulate the expression of liver LDLR and reduce LDL-cholesterol for individuals with heterozygous familial hypercholesterolemia at risk of experiencing a major cardiovascular event.

Guided by DNA large language models (DNA-LLMs), we build upon this validated CRISPR-based strategy to pursue the development of multiple novel *in vivo* therapeutics that upregulate genes-of-interest *Target A*, *Target B*, and *Target C* to compensate for loss-of-function mutations that cause monogenic diseases with severe unmet needs.

Methods: Workflow for designing and validating novel CRISPR-based strategies of gene upregulation

We use DNA-LLMs to delineate target gene repressor elements in non-coding regions, assess on- and off-target activity of a focused gRNA panel, and screen gene editing strategies combinatorially in relevant disease models to demonstrate target gene upregulation:

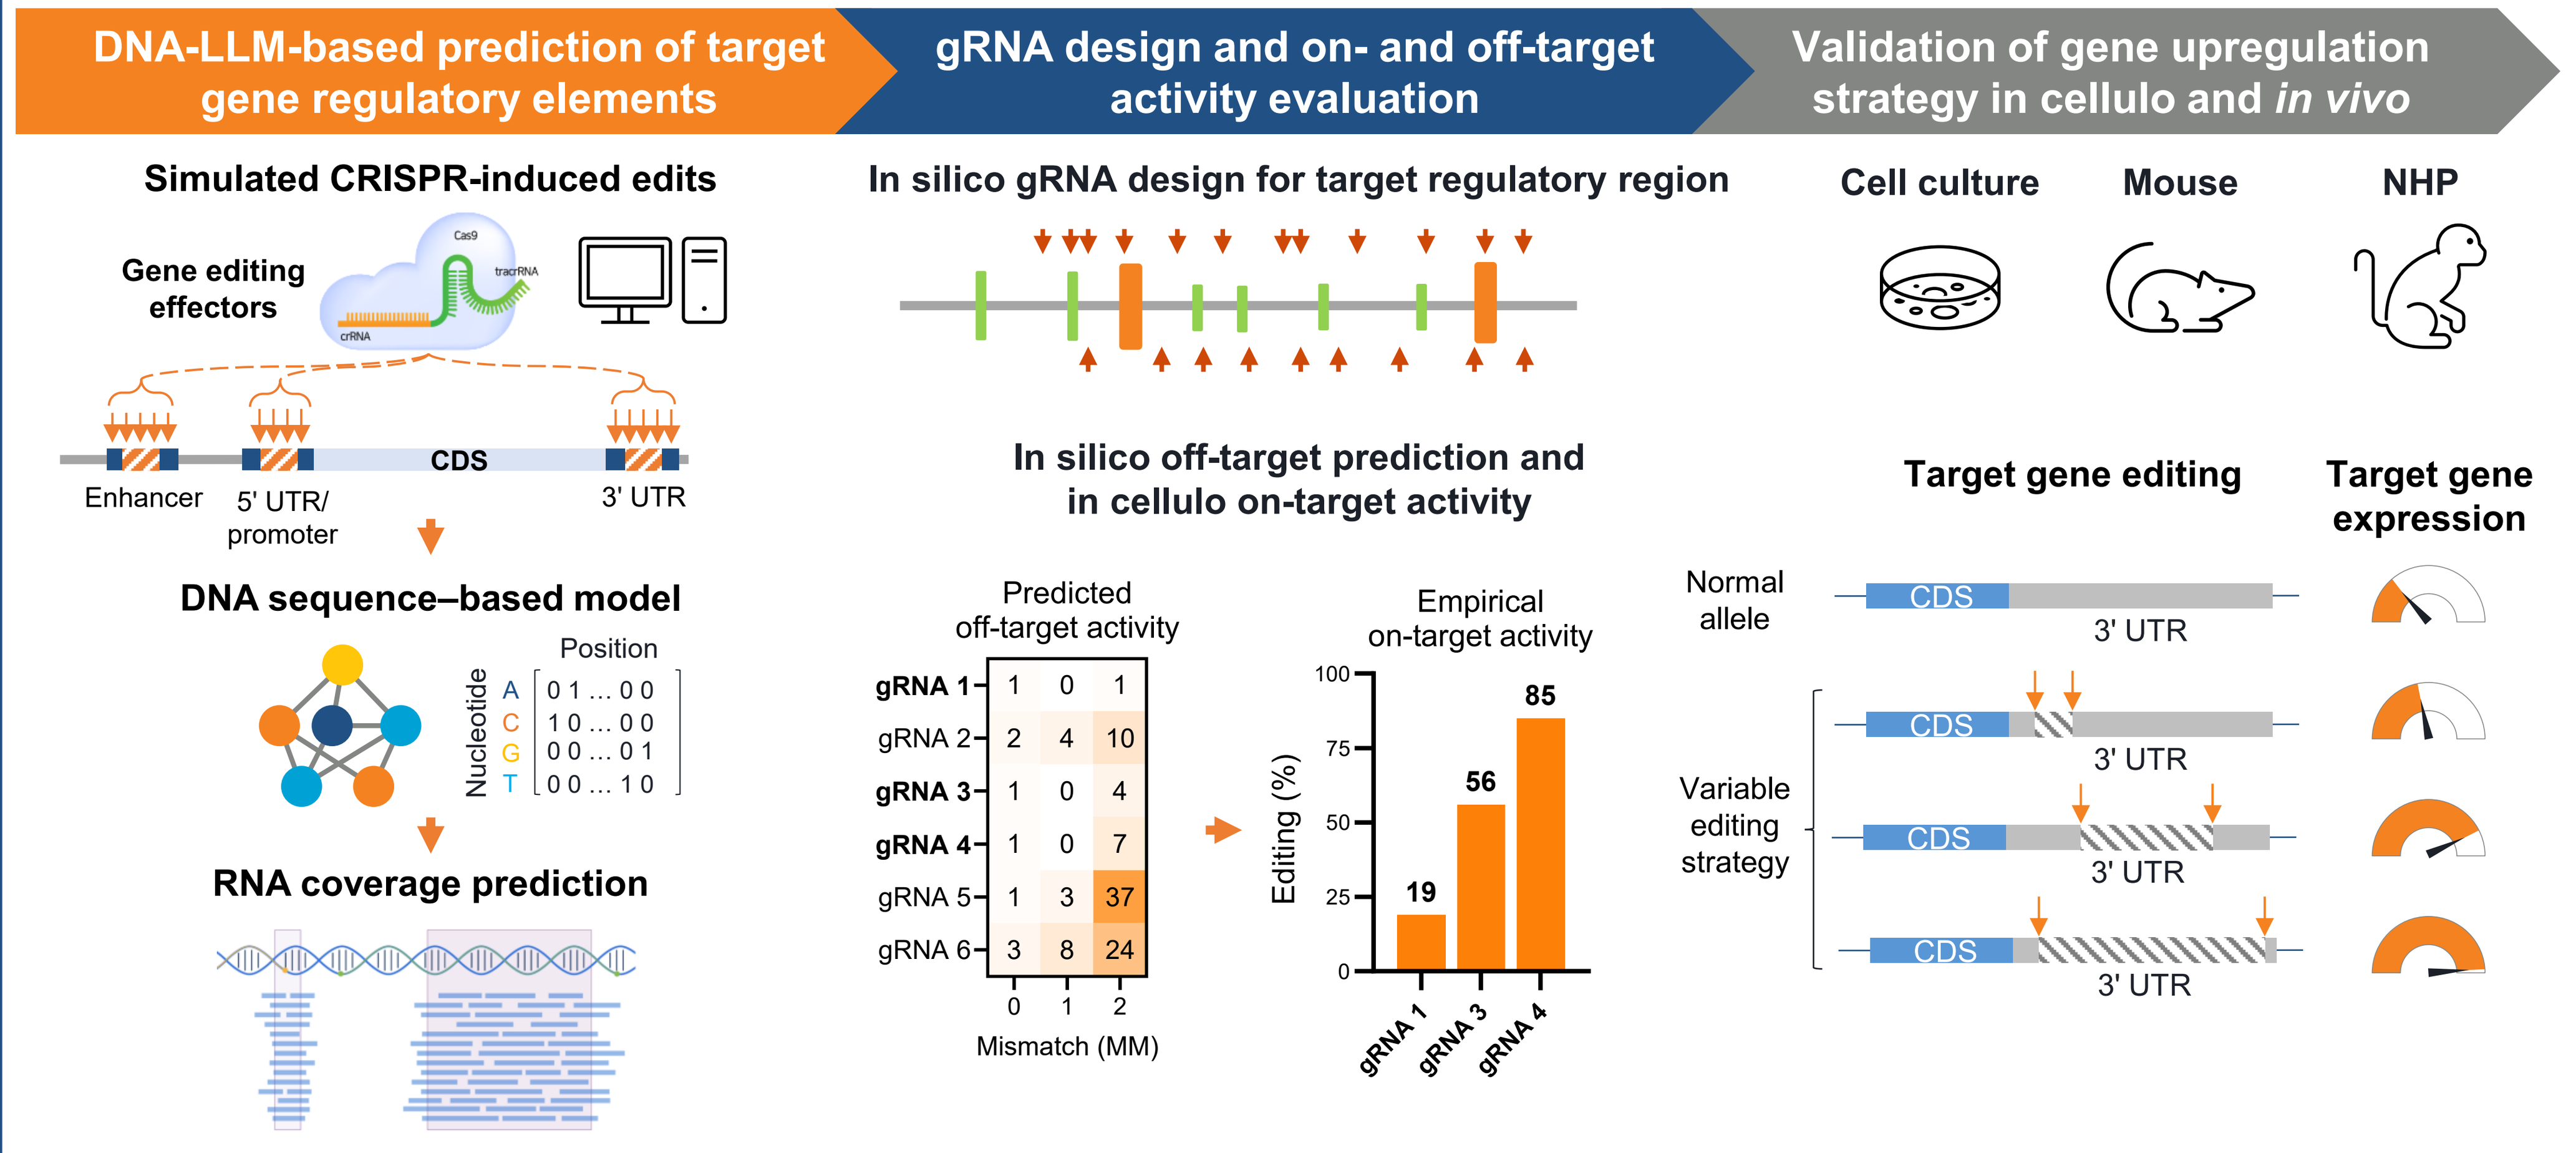

DNA-LLM-based prediction of target gene regulatory elements
gRNA design and on- and off-target activity evaluation
Validation of gene upregulation strategy in cellulo and in vivo
Simulated CRISPR-induced edits
Gene editing effectors
Enhancer
5' UTR/ promoter
CDS
3' UTR
DNA sequence–based model
Position
Nucleotide
RNA coverage prediction
In silico gRNA design for target regulatory region
In silico off-target prediction and in cellulo on-target activity
Predicted off-target activity
gRNA 1 1 0 1
gRNA 2 2 4 10
gRNA 3 1 0 4
gRNA 4 1 0 7
gRNA 5 1 3 37
gRNA 6 3 8 24
Mismatch (MM)
Empirical on-target activity
Editing (%)
19 56 85
gRNA 1 gRNA 3 gRNA 4
Cell culture
Mouse
NHP
Target gene editing
Target gene expression
Normal allele
Variable editing strategy
CDS
3' UTR

Predicted increases in target mRNA abundance suggests the presence of negative regulatory elements

gRNA-nuclease combos with high predicted specificity are assessed for their on-target activity in cellulo

Specific/active effector combos are tested in relevant biological systems to identify top upregulation strategies

Figure 1: A DNA-LLM identified purported cis repressor elements within the non-coding regions of multiple genes-of-interest

Using a DNA-LLM, we performed comprehensive *in silico* scanning mutagenesis of the 5' and 3' UTRs of target genes *A*, *B*, and *C*:

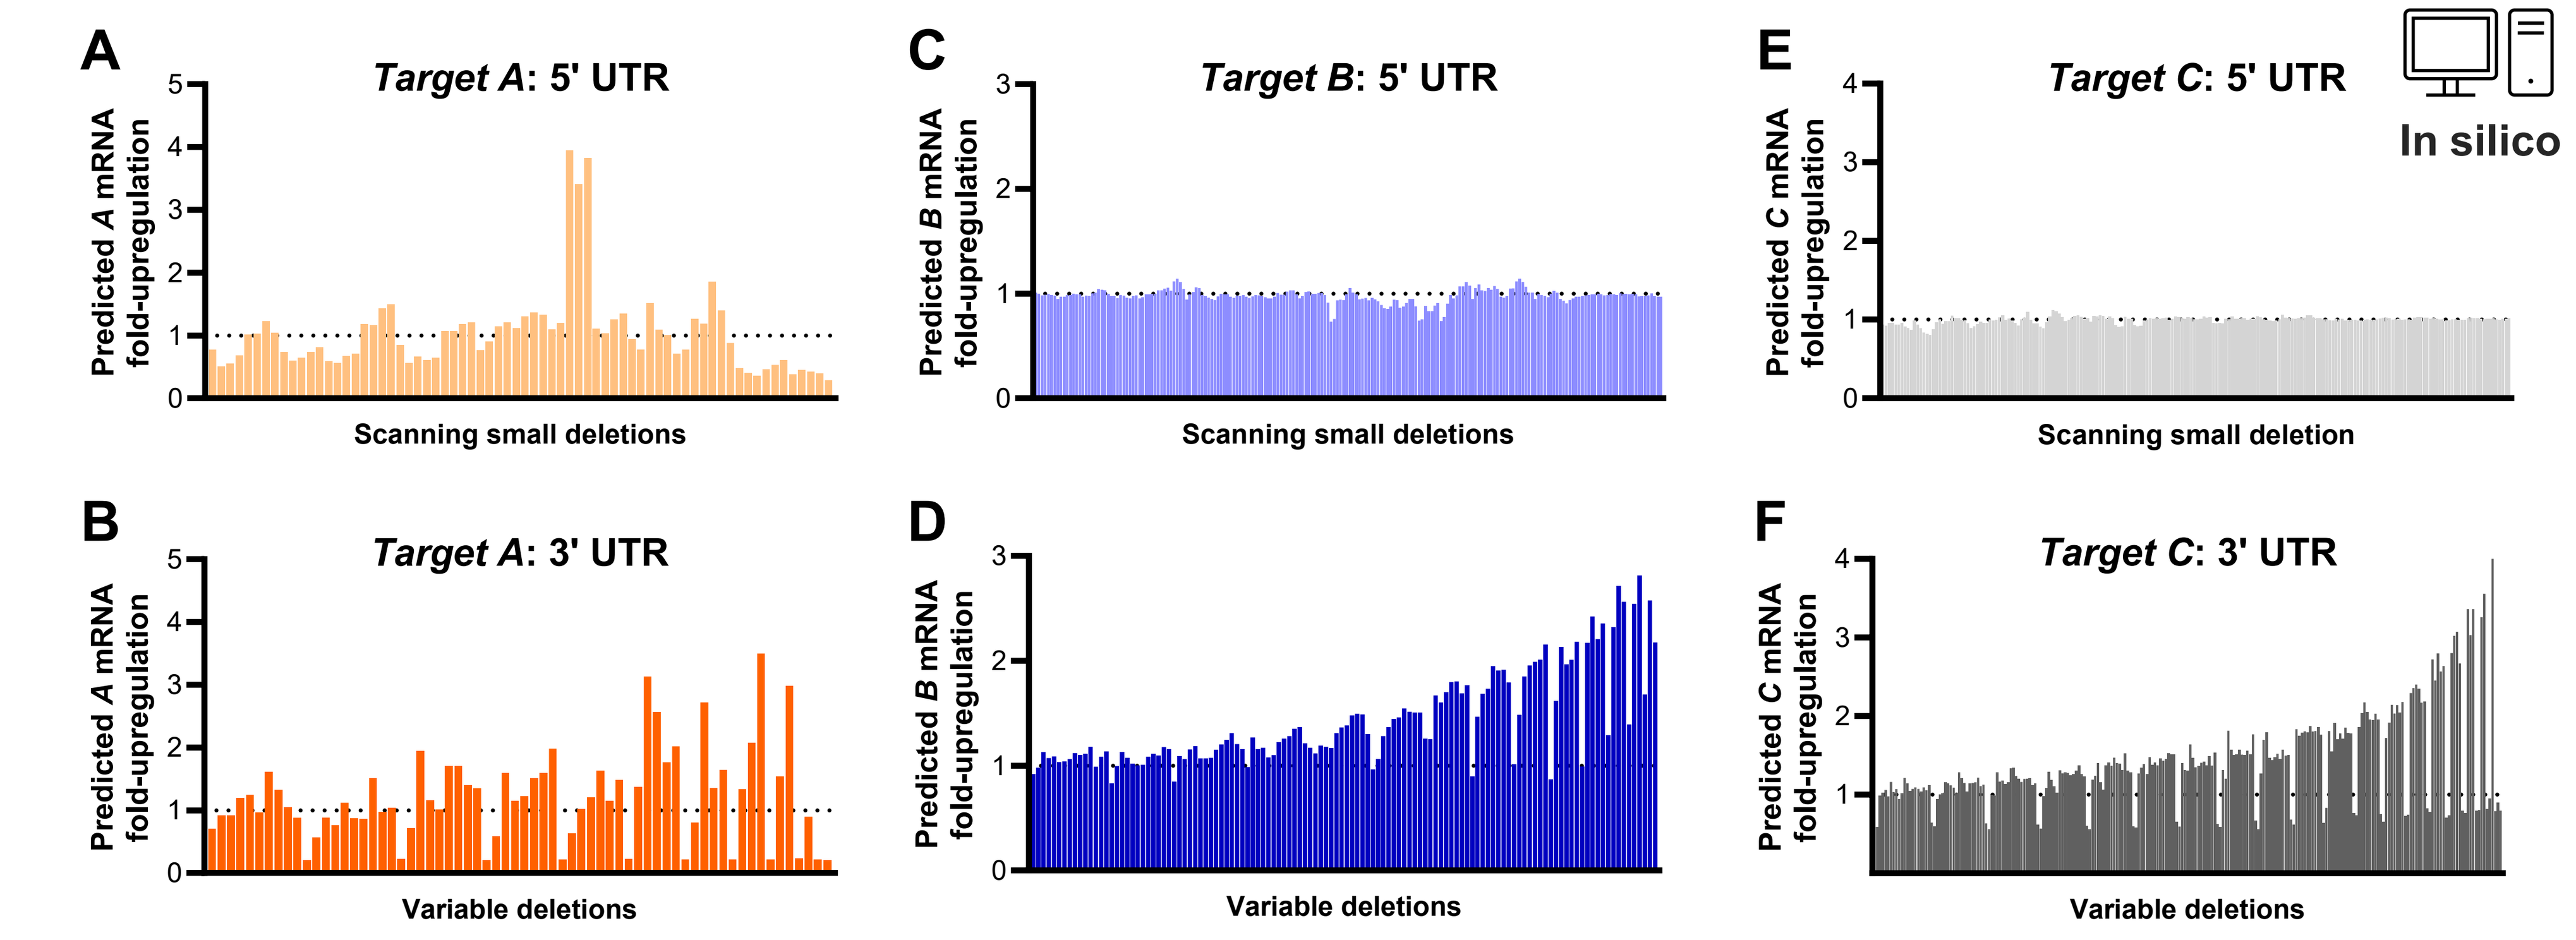

A Target A: 5' UTR
Predicted A mRNA fold-upregulation
Scanning small deletions
B Target A: 3' UTR
Variable deletions
C Target B: 5' UTR
Predicted B mRNA fold-upregulation
Scanning small deletions
D
Variable deletions
E Target C: 5' UTR
In silico
Predicted C mRNA fold-upregulation
Scanning small deletion
F Target C: 3' UTR
Variable deletions

For all three gene targets, disruption of purported 5' and 3' UTR repressor were predicted to de-repress gene expression ~3- to 4-fold

Figure 1: DNA-LLM-based prediction of *Target A*, *B*, and *C* regulatory region disruption and the impact on their mRNA expression. Variable genomic editing outcomes were simulated *in silico*, and the resulting edited DNA sequences were provided as input to a sequence-to-function DNA-LLM prediction model to infer regulatory effects of gene editing. For 5' UTRs, 10 bp deletions with a 5 bp sliding window from the beginning of the annotated promoter to the coding sequence start codon were simulated **(A, C, E)**, while for 3' UTRs, deletions of variable size and position between the coding sequence stop codon and consensus polyA signal were simulated **(B, D, F)**. Predicted gene expression changes were quantified by comparing model-derived expression profiles over annotated exon regions for each edited sequence relative to the unedited reference sequence and expressed as fold-upregulation.

Figure 2: Informed gRNA mapping of target gene regulatory regions validated several editing strategies that induce robust gene upregulation

To develop novel CRISPR-based upregulation strategies, we combinatorially tested a panel of potent and specific gRNAs to disrupt DNA-LLM-predicted repressor elements within the UTRs of target genes *A*, *B*, and *C* and measured the impact on their mRNA expression in relevant primary human cell cultures:

Disruption of a single repressor element within the 5' UTR of *Target A* achieved only ~1.5-fold upregulation **(A)**.

3' UTR–targeting strategies for *Targets A*, *B*, and *C* exhibited robust upregulation on gene transcripts (~4-, ~2-, and ~3.5-fold upregulation, respectively) **(B–D)**.

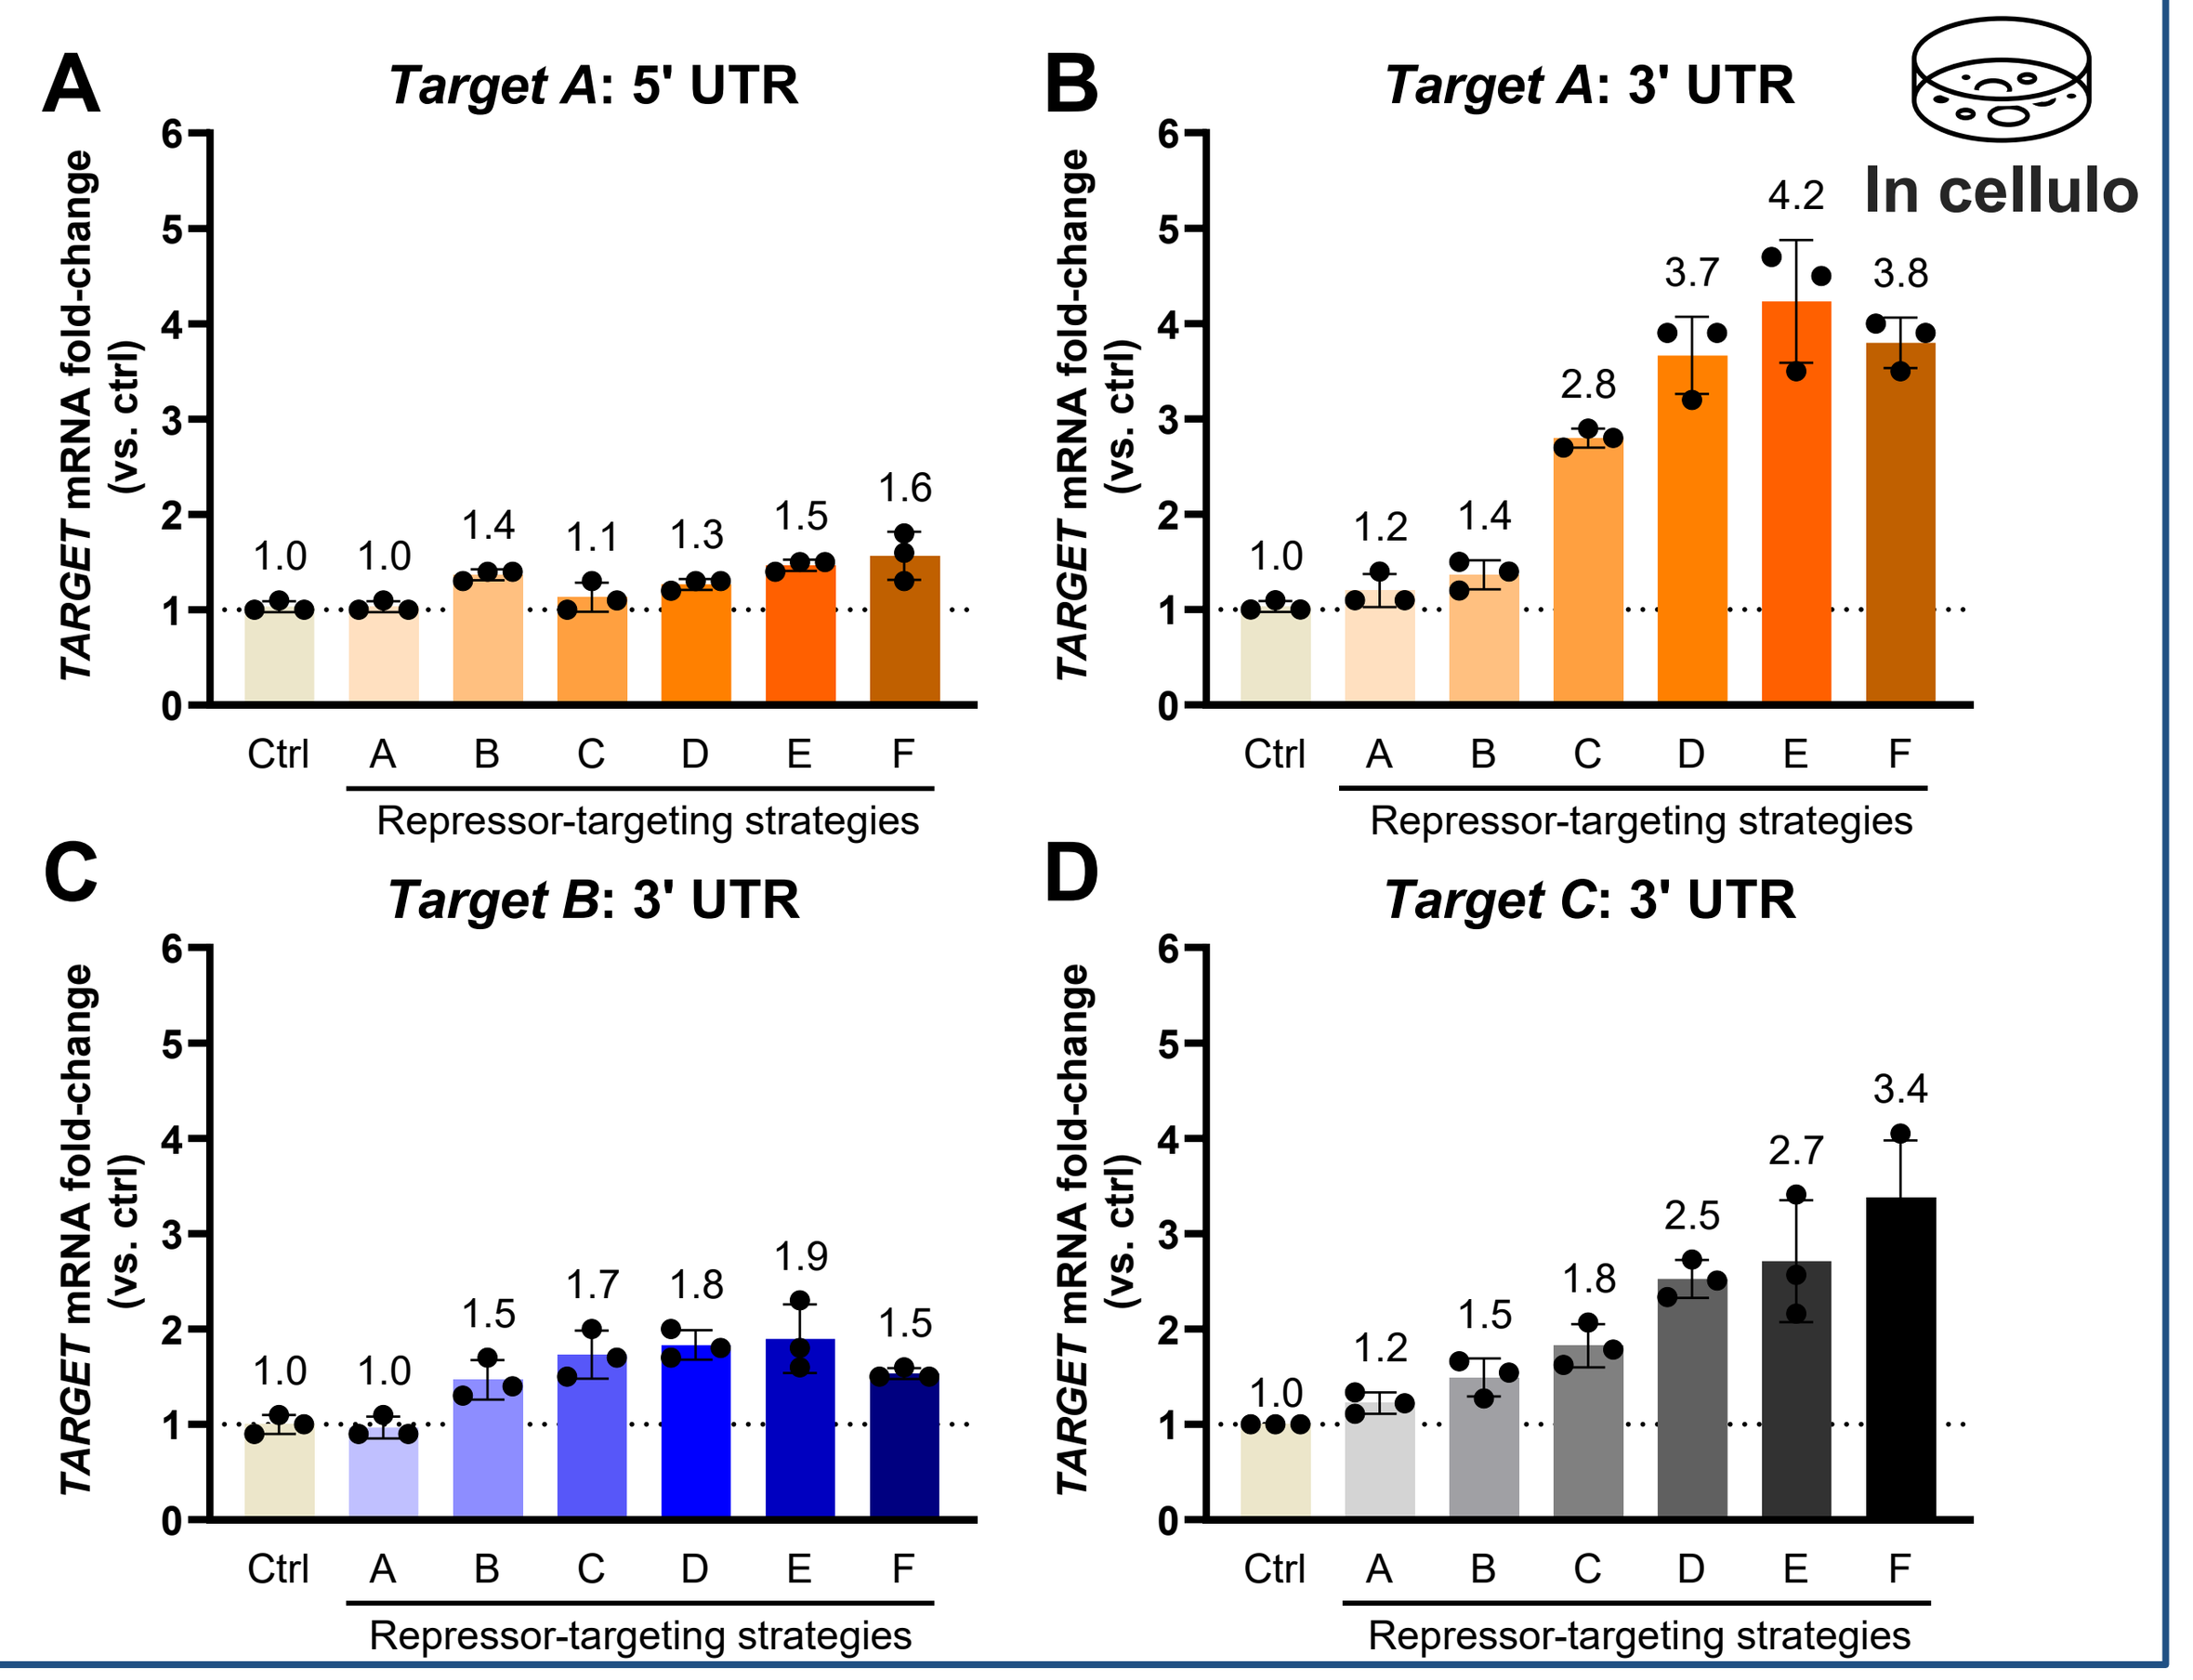

A Target A: 5' UTR
1.0 1.0 1.4 1.1 1.3 1.5 1.6
B Target A: 3' UTR
1.0 1.2 1.4 2.8 3.7 4.2 3.8
In cellulo
C Target B: 3' UTR
1.0 1.0 1.5 1.7 1.8 1.9 1.5
D Target C: 3' UTR
1.0 1.2 1.5 1.8 2.5 2.7 3.4
TARGET mRNA fold-change (vs. ctrl)
Ctrl A B C D E F
Repressor-targeting strategies

Figure 2: Screening of variable CRISPR cargos directed against different human *Target A*, *B*, and *C* gene regulatory regions-of-interest for their ability to induce gene expression in primary human cell cultures. Primary human cultures were RNA-transfected at a maximally effective dose and after 3 days post-transfection, RNA and gDNA were isolated from cell lysates to quantify target gene mRNA transcript levels by RT-ddPCR (n=3) **(A–D)**. mRNA abundances are reported relative to a non-edited "ctrl" condition with baseline levels represented by a dotted line.

Figure 3: DNA-LLM-simulated gene editing is highly predictive of resulting target gene expression in relevant biological systems

To assess the accuracy of our DNA-LLM prediction modeling, we determined the correlations between the empirically-determined in cellulo CRISPR-based upregulation and predicted upregulation for the 3' UTRs of *Targets A*, *B*, *C*, and EDIT-401 target gene *LDLR:*

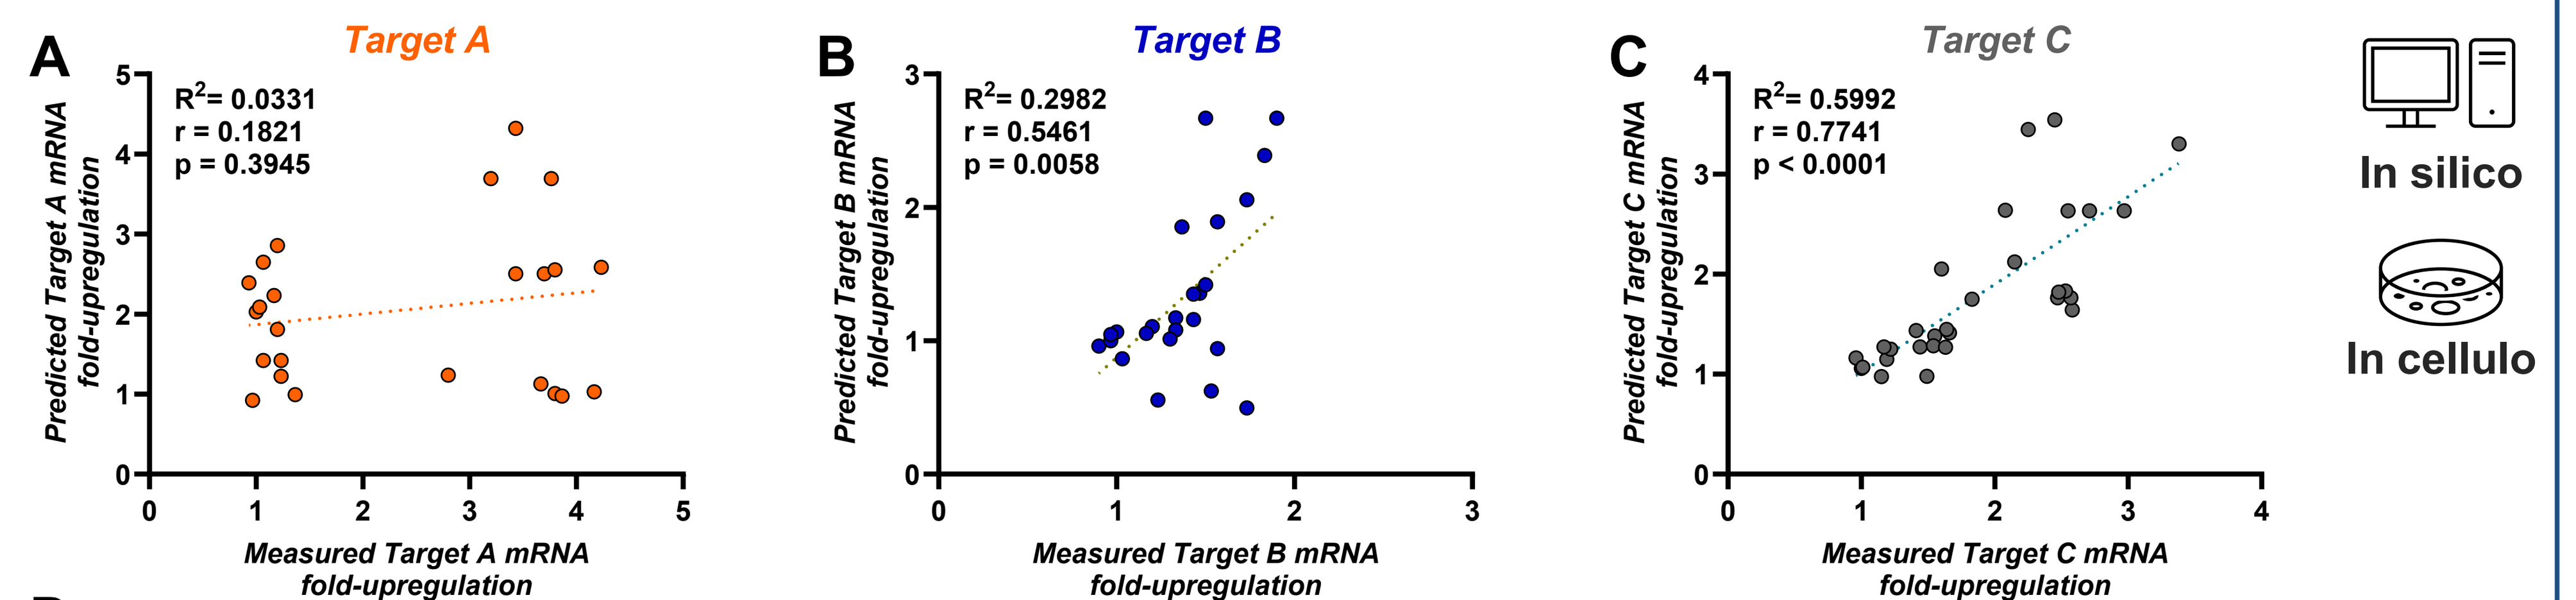

A Target A
R²= 0.0331
r = 0.1821
p = 0.3945
Predicted Target A mRNA fold-upregulation
Measured Target A mRNA fold-upregulation
B Target B
R²= 0.2982
r = 0.5461
p = 0.0058
Predicted Target B mRNA fold-upregulation
Measured Target B mRNA fold-upregulation
C Target C
R²= 0.5992
r = 0.7741
p < 0.0001
Predicted Target C mRNA fold-upregulation
Measured Target C mRNA fold-upregulation
In silico
In cellulo

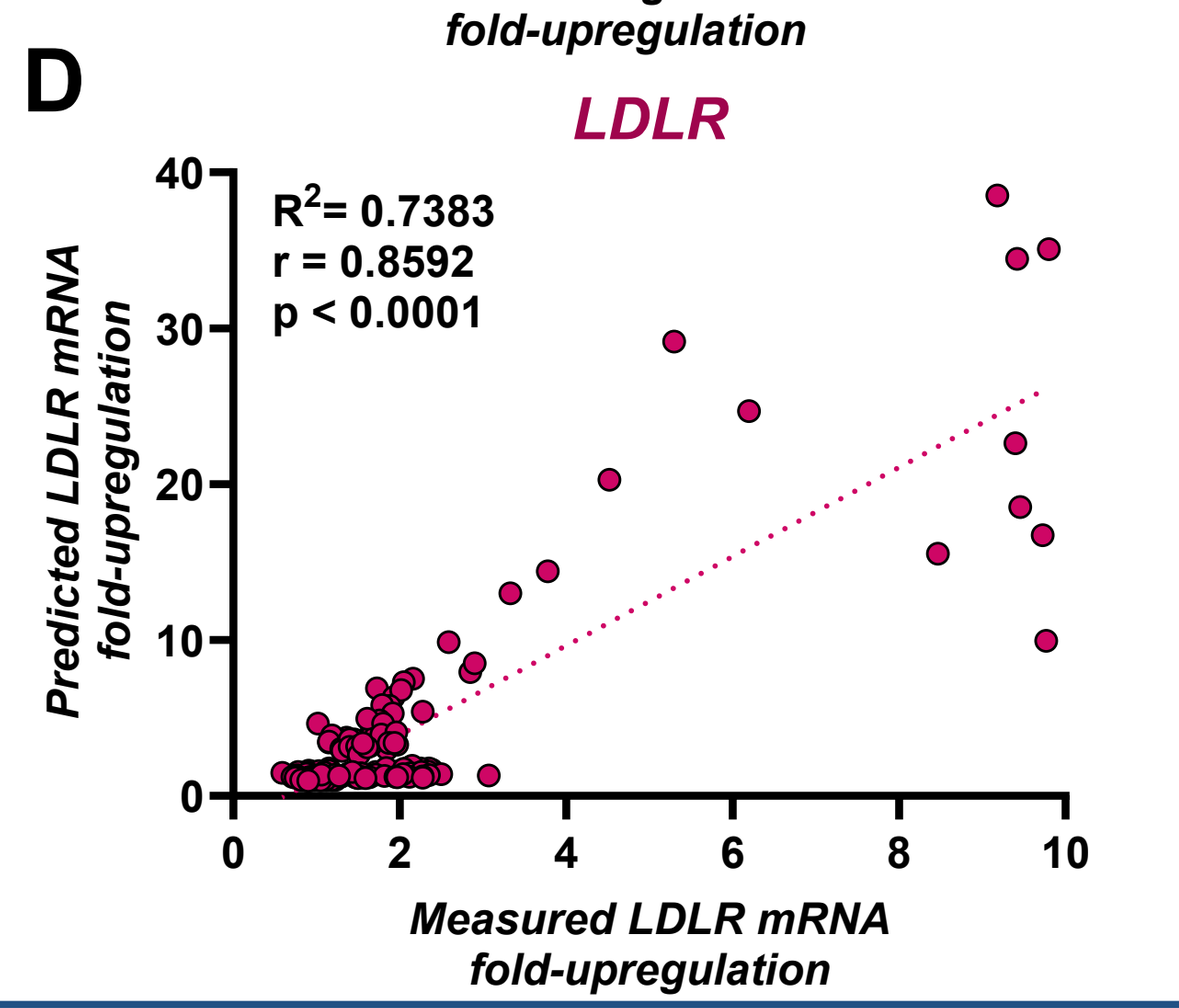

D LDLR
R²= 0.7383
r = 0.8592
p < 0.0001
Predicted LDLR mRNA fold-upregulation
Measured LDLR mRNA fold-upregulation

For *Targets B*, *C*, and *LDLR*, there was a strong correlation (r = 0.5461, 0.7741, and 0.8592, respectively) between predicted and measured gene upregulation.

For *Target A*, disruption of a second repressor element – originally predicted to not contribute to gene expression – drove significant de-repression, whereas disruption of the primary predicted repressor contributed little to de-repression.

Figure 3: Establishing a correlation between DNA-LLM-simulated and in cellulo gene editing effects on target gene upregulation. Primary human cultures were RNA-transfected at a maximally effective dose and at 3 days post-transfection, RNA was isolated from culture lysates to quantify *Target A*, *Target B*, *Target C*, and *LDLR* mRNA transcript levels by RT-ddPCR (n=3) **(A–D)**. "Measured" fold-upregulation was calculated as change in mRNA abundance relative to a non-edited control condition, whereas "predicted" fold-upregulation was calculated as change in model-derived mRNA abundance of each corresponding edited sequence relative to the unedited reference sequence. Both a Pearson's correlation (correlation coefficient "r", statistical significance "p") and linear regression analysis (dotted line, correlation coefficient "R^2") are displayed.

Figure 4: Target A repressor elements are conserved in mouse, and their disruption induces hepatic gene upregulation *in vivo*

We designed surrogate CRISPR strategies of the human *Target A* 3' UTR to disrupt the homologous murine locus, determine the conservation of purported repressor elements, and demonstrate our upregulation strategy using a lipid nanoparticle (LNP)/RNA modality *in vivo*:

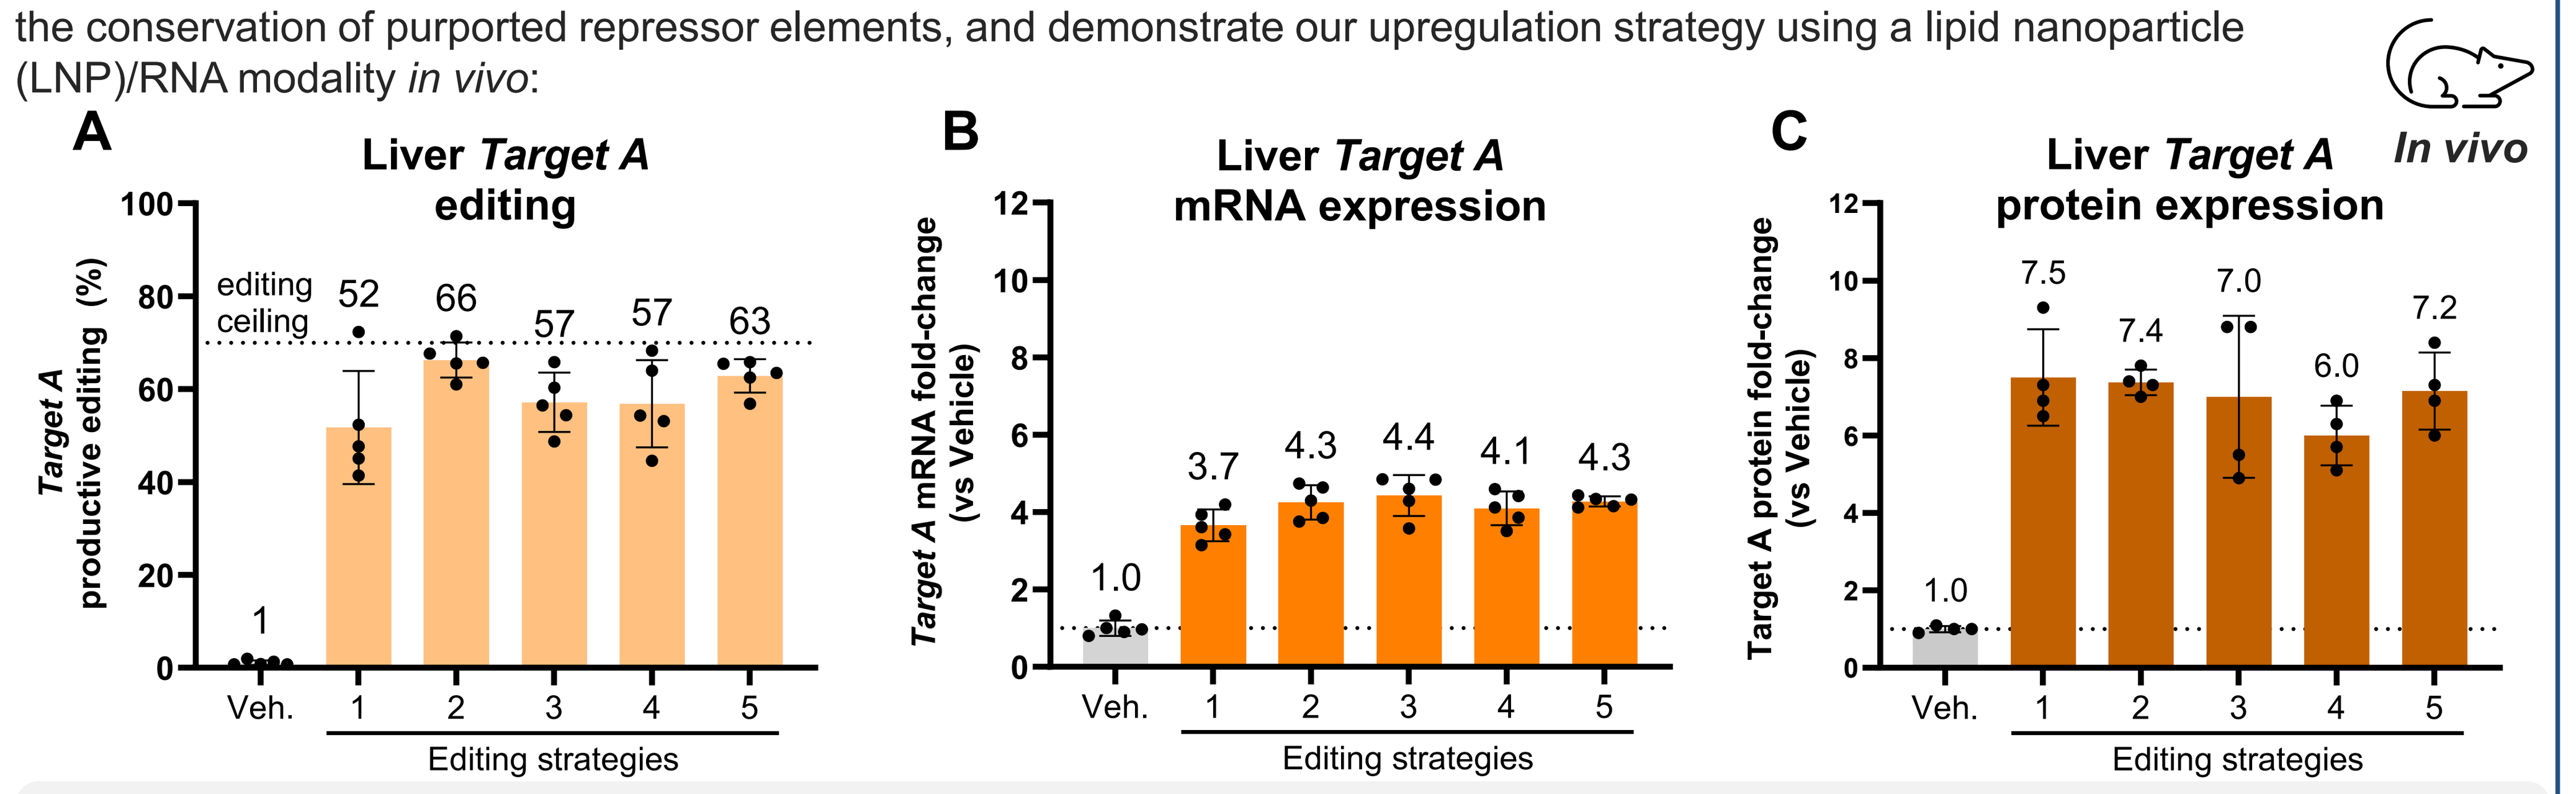

A Liver Target A editing
Target A productive editing (%)
editing ceiling
1 52 66 57 57 63
B Liver Target A mRNA expression
Target A mRNA fold-change (vs Vehicle)
1.0 3.7 4.3 4.4 4.1 4.3
C Liver Target A protein expression
Target A protein fold-change (vs Vehicle)
1.0 7.5 7.4 7.0 6.0 7.2
In vivo
Veh. 1 2 3 4 5
Editing strategies

Tested editing strategies achieved robust liver productive editing of murine *Target A* 3' UTR, resulting in significant upregulation of *Target A* mRNA transcript (up to ~4.5-fold) and Target A protein (up to ~7.5-fold) *in vivo*

Figure 4: Gene editing and subsequent upregulation of hepatic murine *Target A* expression *in vivo* using a liver-tropic LNP/RNA. Wild-type C57BL/6 mice were dosed with PBS ("Veh.") or standard liver-tropic LNP carrying Cas effector mRNA and mouse *Target A* gRNAs intravenously. At 7-days post-treatment, whole-liver homogenates were collected and gDNA was extracted to assess productive editing of the *Target A* 3' UTR by NGS (n = 5) **(A)**, RNA was extracted to assess *Target A* mRNA transcript by RT-ddPCR (n = 5) **(B)**, and protein was extracted to assess Target A protein by JESS-based Western Blot (n = 4) **(C)**. Dotted lines represent the theoretical hepatocyte upper editing limit **(A)** or baseline *Target A* gene expression **(B, C)**.

Conclusions

- Utilizing a DNA-LLM, we established a cost-effective, accelerated workflow for developing *in vivo* CRISPR-based upregulation strategies for multiple target genes in parallel.
- Simulated mutagenesis and gene editing events proved sequence-to-function DNA-LLMs can be highly predictive of resulting mRNA expression, but with some limitations.
- Informed gRNA screening in relevant primary human cells confirmed repressor elements governing *Target A*, *B*, *C* expression and identified optimal upregulation strategies.
- We validated our methodologies *in vivo* by demonstrating robust liver *Target A* productive editing and mRNA and protein upregulation using LNP/RNA in mice.

Acknowledgments and disclosures

We would like to thank our Editas Medicine colleagues and collaborators who provided support in sequencing, RNA and LNP manufacturing, animal studies, and scientific discourse. All authors are current or former employees and shareholders of Editas Medicine, Inc. Editorial assistance was provided by Porterhouse Medical and was funded by Editas Medicine, Inc. according to Good Publication Practice (GPP) guidelines.

Presented at the 29th ASGCT Annual Meeting • May 11–15, 2026 • Boston, MA, USA

© 2026 Editas Medicine